Treatment Uptake and Real World Effectiveness of Sofosbuvir/Ledipasvir and Paritaprevir/Ritonavir/Ombitasvir/Dasabuvir Among HCV-Infected Persons With Chronic Kidney Disease

ERCHIVES
Electronically Retrieved Cohort of HCV Infected Veterans

M. Alkadi[1,2]; Y. Ren[3]; A. Puenpatom[4]; R. Kumar[4]; J.M. Arduino[4]; A.-B. Abou-Samra[1,2]; A.A. Butt[1,2,3]

[1]Weill Cornell Medical College, New York, NY, USA; [2]Hamad Medical Corporation, Doha, Qatar; [3]VA Pittsburgh Healthcare System, Pittsburgh, PA, USA; [4]Merck & Co., Inc., Kenilworth, NJ, USA

Contact Information:
Adeel A. Butt
Building 30, Mailstop 151; University Drive C Pittsburgh, PA 15240
Email: aabutt@pitt.edu

Introduction

- New direct-acting antiviral agents (DAA) have revolutionized treatment of HCV infection
- With newer DAA regimens, SVR rates of >90%-95% are the norm
- Patients with chronic kidney disease (CKD) were often not initiated on HCV treatment due to safety, tolerability, and efficacy concerns
- Newer DAA regimens are more safe, tolerable, and efficacious than the older interferon/ribavirin-based regimens, even in the CKD population
- While treatment initiation rates in the CKD population in the interferon/ribavirin era were low, such data are lacking in the DAA era

Aims

- To determine treatment initiation rates and effectiveness with the sofosbuvir/ledipasvir (SOF/LDV) and paritaprevir/ritonavir/ ombitasvir/dasabuvir (PrOD) regimens among persons with HCV/CKD
- To assess factors associated with SOF/ODV or PrOD treatment initiation in HCV/CKD patients

Methods

- We used the Electronically Retrieved Cohort of HCV Infected Veterans (ERCHIVES), a well-established national cohort of HCV-infected persons and age-, sex-, and race-matched controls who received care within the Veterans Health Administration (VHA)
- Demographic, clinical, laboratory, pharmacy, utilization, and vital status data were retrieved from VHA's Corporate Data Warehouse
- Included all HCV RNA+ persons with ≥2 available eGFR 3 months apart and prior to baseline and at least 1 eGFR value ≥12 weeks after baseline. eGFR was estimated using the CKD-EPI equation
- Excluded HIV- and hepatitis B surface antigen-positive persons
- CKD stages were categorized using the National Kidney Foundation criteria
- Treatment initiation was defined as a prescription of SOF/ODV or PrOD for >14 days
- Severity of liver disease was estimated using the FIB-4 score, with cirrhosis defined as FIB-4 score >3.5
- Baseline characteristics were compared by CKD stages
- Factors associated with treatment initiation were determined using logistic regression analysis
- SVR was defined as the proportion of persons with undetectable HCV RNA >12 weeks after treatment completion

Results

Figure 1. Study flow sheet

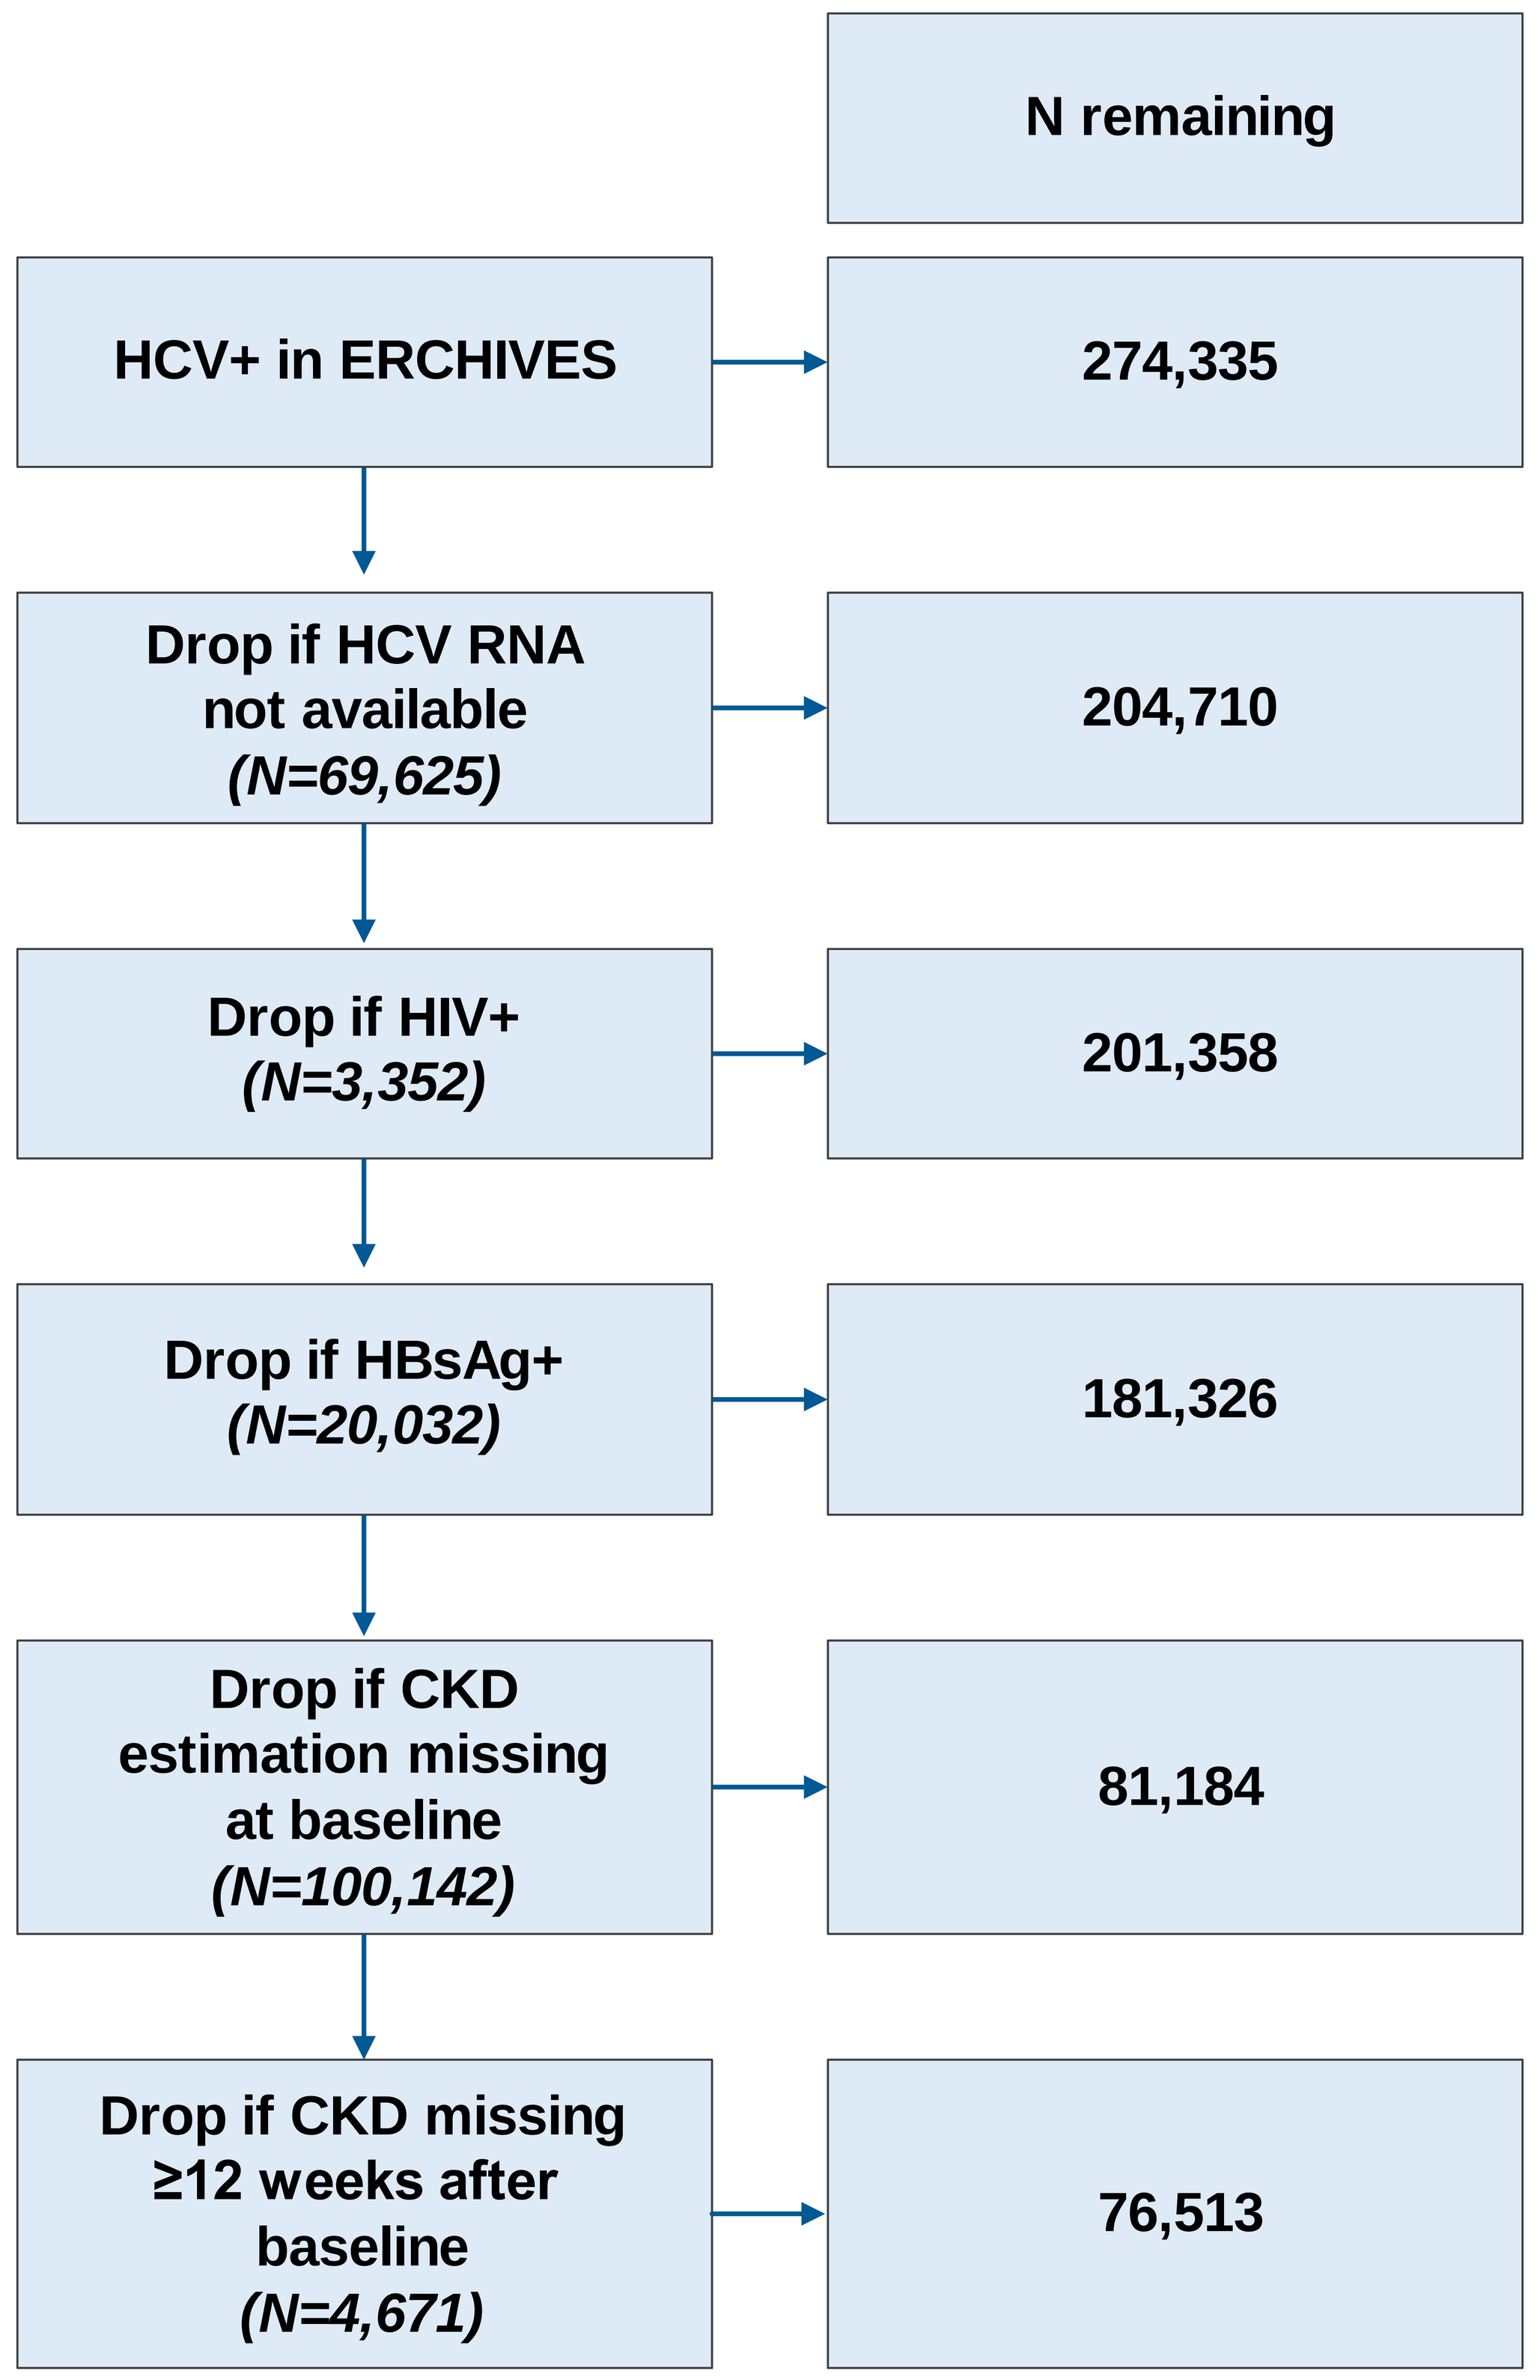
N remaining
HCV+ in ERCHIVES
274,335
Drop if HCV RNA not available (N=69,625)
204,710
Drop if HIV+ (N=3,352)
201,358
Drop if HBsAg+ (N=20,032)
181,326
Drop if CKD estimation missing at baseline (N=100,142)
81,184
Drop if CKD missing ≥12 weeks after baseline (N=4,671)
76,513

Table 1. Baseline characteristics

Characteristic	CKD Stage 1-2 N=68,469	CKD Stage 3 N=6,086	CKD Stage 4-5 N=1,958
Age, median [IQR]	53 (49, 58)	59 (54, 67)	57 (53, 63)
Race/ethnicity, %			
White	55.15	51.69	32.74
Black	32.67	34.80	56.59
Hispanic	4.30	4.12	4.24
Other	7.88	9.38	6.44
Sex (M), %	96.67	96.88	98.01
HCV RNA, $\log_{10}$, median [IQR]	6.04 (4.32, 6.92)	5.93 (3.01, 6.85)	5.89 (3.69, 6.77)
HCV genotype, %			
1a	26.85	19.96	19.87
1b	9.58	9.79	8.07
2	4.50	4.01	2.45
3	3.13	1.63	0.66
4/5/6	0.32	0.18	0.15
Mixed	0.12	0.12	0.05
Missing	55.49	64.31	68.74
Body mass index, median [IQR]	26.88 (23.8, 30.5)	27.61 (24.3, 31.2)	26.57 (23.2, 30.8)
Diabetes, %	17.67	32.71	53.42
CVD, %	8.94	27.26	42.13
Alcohol abuse/dependence, %	43.58	29.23	26.10
FIB-4 score, median [IQR]	1.54 (1.0, 2.5)	1.91 (1.3, 3.1)	1.68 (1.1, 2.7)
Cirrhosis, %	14.78	20.69	14.30
Hepatic decompensation, %	0.77	1.27	1.63
eGFR, median [IQR]	90.64 (79.0, 101.7)	50.9 (43.5, 56.2)	12.99 (7.1, 22.2)
ACE-I/ARB use overall, %	6.52	15.54	25.38

Table 2. Treatment uptake and SVR12 of SOF/LDV and PrOD regimens, by CKD stage

	All stages	CKD Stage 1-2	CKD Stage 3	CKD Stage 4-5
Number of patients with HCV/CKD	76,513	68,469	6,086	1,958
Patients who initiated SOF/LDV or PrOD	6,715 (8.8%)	6,317 (9.2%)	352 (5.8%)	46 (2.3%)
Number and rate of SOF/LDV + or – RBV initiation	5,181 (6.8%)	4,878 (7.1%)	267 (4.4%)	36 (1.8%)
Number of patients who initiated PrOD + or - RBV	1,534 (2.0%)	1,439 (2.1%)	85 (1.4%)	10 (0.5%)
SVR12 (SOF/LDV or PrOD regimens)	93.9% (2,709/2,885)	93.8% (2,545/2,712)	94.8% (164/173)	

Results (continued)

Table 3. Factors associated with treatment initiation

Characteristic	OR	95% CI
Age, per 10-year increase	1.08	(1.05, 1.12)
Race/ethnicity: White (comparator)	1	-
Black	1.03	(0.97, 1.09)
Hispanic	0.90	(0.78, 1.04)
Male sex (compared to female)	0.84	(0.74, 0.97)
HCV RNA, per $\log_{10}$, increase	1.09	(1.08, 1.11)
HCV genotype 1a (comparator)	1	
1b	1.05	(0.98, 1.13)
2	0.02	(0.01, 0.04)
3	0.35	(0.29, 0.40)
4/5/6	0.76	(0.44, 0.96)
Body mass index, per unit increase	1.03	(1.02, 1.03)
Diabetes	0.83	(0.78, 0.90)
Cardiovascular disease diagnosis	0.75	(0.67, 0.82)
Alcohol abuse or dependence	0.76	(0.72, 0.80)
Moderate to severe anemia (Hb<10)	0.68	(0.54, 0.86)
Cirrhosis at baseline (by FIB-4 >3.5)	1.15	(1.07, 1.23)
CKD stage 1-2 (comparator)	1	-
CKD stage 3	0.66	(0.59, 0.73)
CKD stage 4-5	0.28	(0.21, 0.38)

Conclusions

- **Treatment uptake for HCV with SOF/LDV or PrOD in CKD population is low (8.8%). There is a significant unmet need for HCV treatment in the CKD population.**
- **Male sex, non-1 genotype, and comorbidities are associated with a lower likelihood of treatment initiation with SOF/LDV or PrOD**
- **More advanced CKD is significantly associated with lower likelihood of treatment initiation with SOF/LDV or PrOD**
- **Overall SVR12 was 93.9%. Among those with CKD stage 3-5, SVR was 94.8% (N = 164/173)**

References

- Flisiak R, Janczewska E, Wawrzynowicz-Syczewska M, et al. Real-world effectiveness and safety of ombitasvir/paritaprevir/ritonavir +/- dasabuvir +/- ribavirin in hepatitis C: AMBER study. *Aliment Pharmacol Ther.* 2016;44(9):946-956.
- Li T, Qu Y, Guo Y, Wang Y, Wang L. Efficacy and safety of direct-acting antivirals-based antiviral therapies for hepatitis C virus patients with stage 4-5 chronic kidney disease: a meta-analysis. *Liver Int.* 2016.
- Dumortier J, Bailly F, Pageaux GP, et al. Sofosbuvir-based antiviral therapy in hepatitis C virus patients with severe renal failure. *Nephrol Dial Transplant.* 2016.
- Pockros PJ, Reddy KR, Mantry PS, et al. Efficacy of direct-acting antiviral combination for patients with hepatitis C virus genotype 1 infection and severe renal impairment or end-stage renal disease. *Gastroenterology.* 2016;150(7):1590-1598.
- Butt AA, Ren Y, Marks K, Shaikh OS, Sherman KE; ERCHIVES study. Do directly acting antiviral agents for HCV increase the risk of hepatic decompensation and decline in renal function? Results from ERCHIVES. *Aliment Pharmacol Ther.* 2017;45(1):150-159.
- Saxena V, Koraishy FM, Sise ME, et al. Safety and efficacy of sofosbuvir-containing regimens in hepatitis C-infected patients with impaired renal function. *Liver Int.* 2016;36(6):807-816.

This study was funded by Merck & Co., Inc., Kenilworth, NJ, USA

Copyright © 2017 Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Kenilworth, NJ, USA. All rights reserved.