Long-Term Clinical Outcomes in HCV Genotype 1-Infected Patients Receiving Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir ± Ribavirin: First Interim Safety and Efficacy Results From TOPAZ-I

865

K Agarwal[1], GB Gaeta[2], SS Lee[3], E Dumas[4], A Streinu-Cercel[5], E Schott[6], RJ Andrade[7], AM Perez-Rios[8], J George[9], I Bakulin[10], R Marinho[11], R Ozaras[12], M Charafeddine[4], L Liu[4], T Pilot-Matias[4], K Howieson[4], A Joy[4], D Cohen[4], F Zoulim[13]

[1]Institute of Liver Studies, King's College Hospital NHS Foundation Trust, London, United Kingdom; [2]Second University of Naples, Naples, Italy; [3]University of Calgary, Alberta, Canada; [4]AbbVie Inc., North Chicago, Illinois, United States; [5]Carol Davila University of Medicine and Pharmacy, National Institute for Infectious Diseases "Prof. Dr. Matei Balș", Bucharest, Romania; [6]Charité Universtätsmedizin Berlin, Berlin, Germany; [7]University Hospital of Málaga, IBIMA, CIBERehd, Andalusia, Spain; [8]Centro de Investigaciòn Farmaceutica Especializada SC Guadalajara, Mexico; [9]Westmead Hospital, Westmead Institute for Medical Research and the University of Sydney, Westmead, Sydney, Australia; [10]Central Scientific Research Institute for Gastroenterology, Moscow, Russia; [11]Hospital Santa Maria, Medical School of Lisbon, Lisbon, Portugal; [12]Department of Infectious Diseases, Cerrahpasa Medical School, Istanbul University, Istanbul, Turkey; [13]Institut National de la Santé et de la Recherche Médicale, Paris, France

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

BACKGROUND

- HCV-infected individuals are at risk for adverse clinical outcomes, such as liver cirrhosis and hepatocellular carcinoma (HCC), resulting in increased liver-related mortality[1]
- The all-oral three direct-acting antiviral (3-DAA) regimen of ombitasvir, paritaprevir with the pharmacokinetic enhancer ritonavir, and dasabuvir (OBV/PTV/r + DSV) ± ribavirin (RBV) is available for the treatment of patients with HCV genotype (GT) 1 infection in more than 60 countries[2,3]
- The TOPAZ-I (outside of the United States) and TOPAZ-II (United States) studies were undertaken to assess the impact of interferon-free DAA treatment on long-term clinical outcomes among HCV-infected patients who achieve a sustained virologic response (SVR)[4]
- Here we present interim results from TOPAZ-I

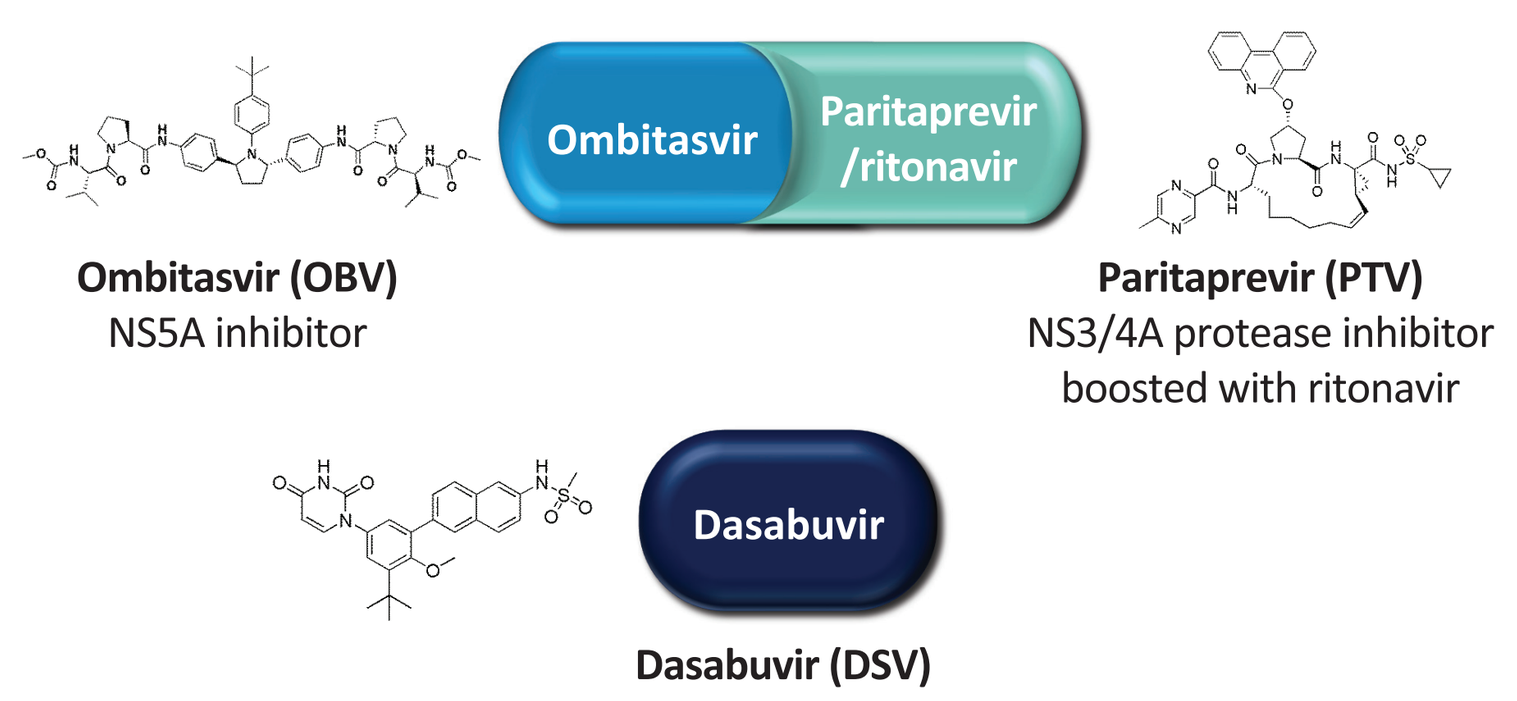
Ombitasvir
Paritaprevir /ritonavir
Dasabuvir

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir

Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

PTV was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVES

- To assess the efficacy (measured by SVR12 rate) and safety of the 3-DAA regimen with or without RBV in treatment-naive and treatment-experienced HCV GT1-infected patients, with or without cirrhosis
- To evaluate the impact of SVR on the long-term progression of liver disease as measured by all-cause death, liver-related death, liver decompensation, liver transplantation, and HCC
- To evaluate the long-term progression of fibrosis among patients treated with the 3-DAA regimen, as measured by a change from baseline in liver stiffness determined by transient elastography (FibroScan®)

METHODS

STUDY DESIGN

- TOPAZ-I is an ongoing Phase 3b, open-label, multi-center study with 1596 patients from 187 sites in 27 countries **(Figure 1)**

Figure 1. Distribution of the Study Population by Country

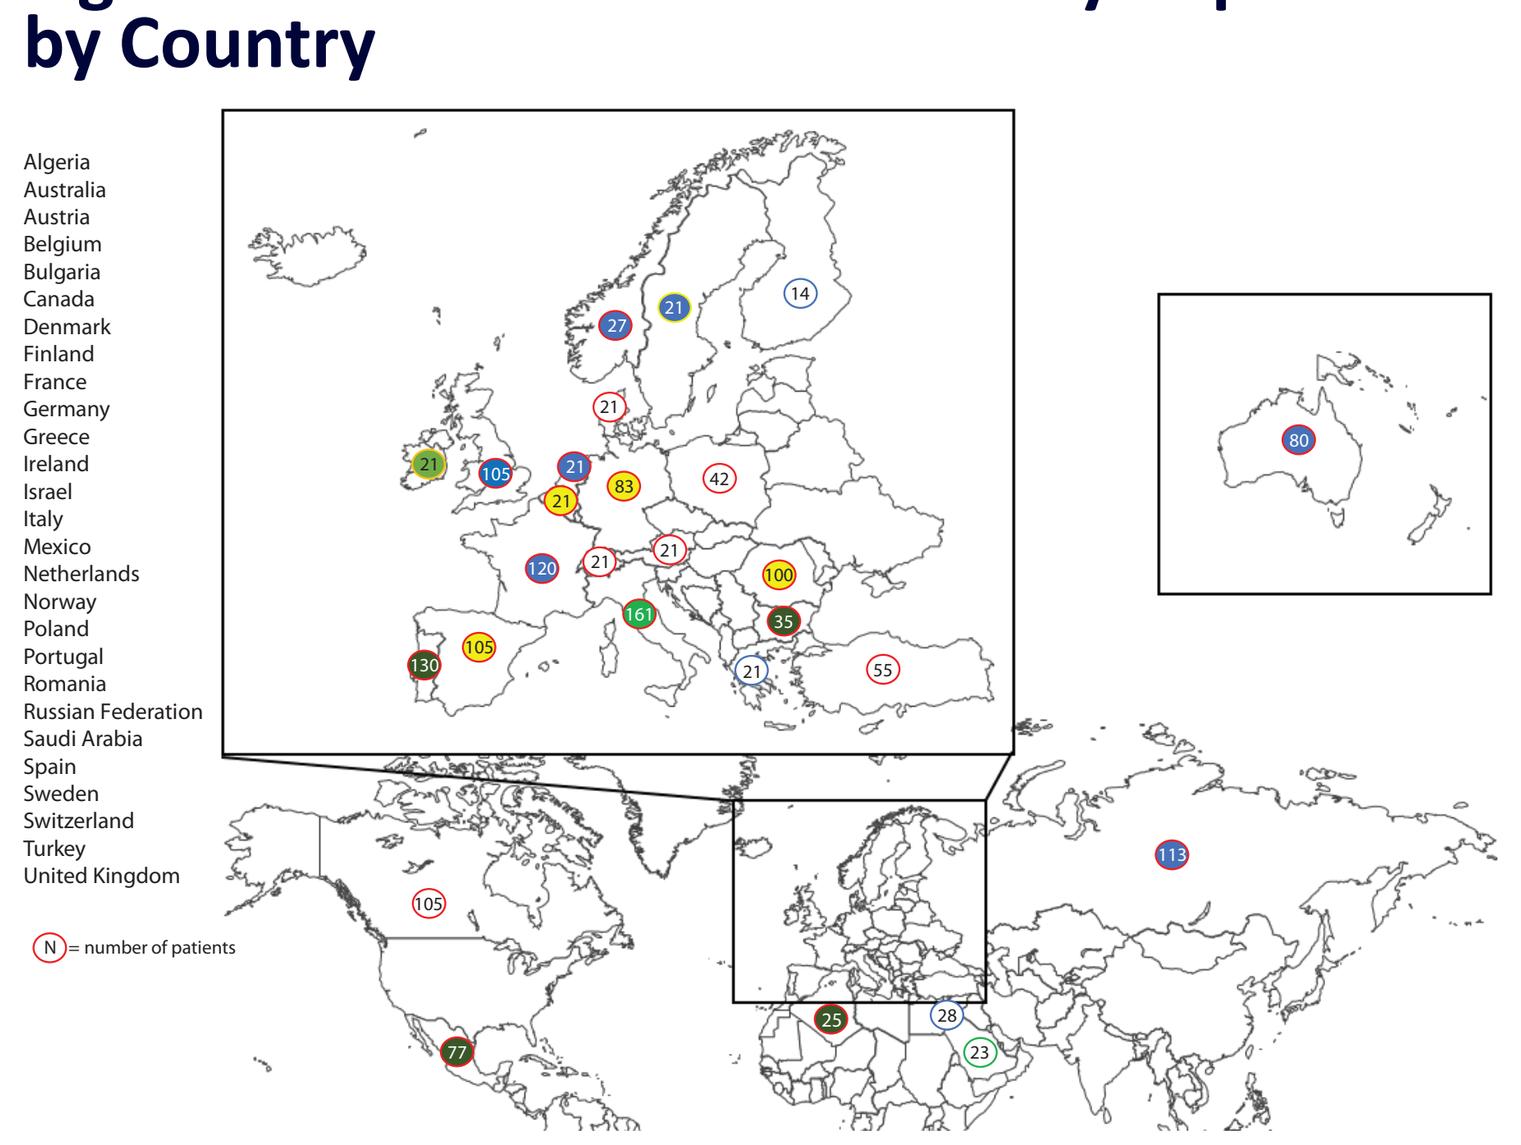
Algeria
Australia
Austria
Belgium
Bulgaria
Canada
Denmark
Finland
France
Germany
Greece
Ireland
Israel
Italy
Mexico
Netherlands
Norway
Poland
Portugal
Romania
Russian Federation
Saudi Arabia
Spain
Sweden
Switzerland
Turkey
United Kingdom
N = number of patients

METHODS (CONTINUED)

- HCV GT1-infected patients who were either treatment-naive or interferon-based treatment-experienced received the 3-DAA regimen ± RBV for 12 or 24 weeks, based on HCV subtype and the presence or absence of cirrhosis **(Figure 2)**
 - A post-treatment follow-up duration of 5 years is planned for all patients who received at least 1 dose of study drug **(Figure 2)**

Figure 2. Study Design

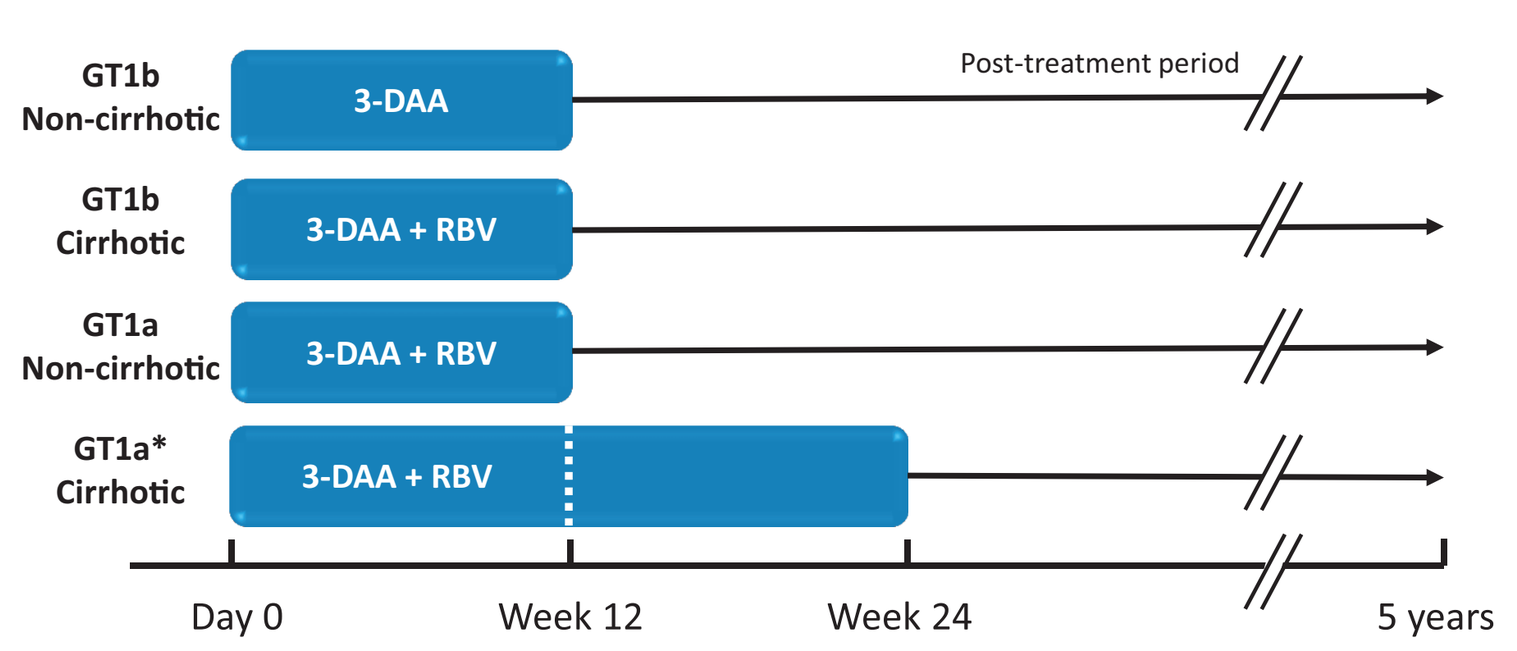
GT1b Non-cirrhotic
3-DAA
Post-treatment period
GT1b Cirrhotic
3-DAA + RBV
GT1a Non-cirrhotic
3-DAA + RBV
GT1a* Cirrhotic
3-DAA + RBV
Day 0
Week 12
Week 24
5 years

*Treatment for 12 weeks was permitted for some patients where consistent with the approved local label. Follow-up visits at post-treatment (PT) weeks 4, 12, 24, 52, 104, 156, 208, and 260, or at premature discontinuation visit.

KEY ELIGIBILITY CRITERIA

- Adult patients with documented chronic HCV GT1 infection
- Patients were excluded if they had
 - Abnormal laboratory values (calculated creatinine clearance <30 mL/min; albumin <2.8 g/dL; hemoglobin < lower limit of normal; platelets <25 000 cells per µL; total bilirubin >3.0 mg/dL)
 - HIV or HBV coinfection
 - A history of solid organ transplant or a confirmed presence of HCC within 3 months prior to screening
 - Any current or past clinical evidence of Child-Pugh B or C classification or clinical history of liver decompensation

STUDY ANALYSES

- The primary endpoint is assessed in the intent-to-treat (ITT) population, which consisted of all enrolled patients in this study who received at least one dose of study drug
- The primary endpoint is the incidence of all-cause death, liver-related death, liver decompensation, liver transplantation, HCC, and the composite of any of the above outcomes observed during the 5 years post-treatment period
- Secondary endpoints include
 - The percentage of subjects achieving SVR12 and corresponding 2-sided 95% confidence interval
 - The mean change from baseline in FibroScan score, when available, during treatment period and post-treatment
- For patients failing to achieve SVR, population sequencing (approximately 15% level of detection) was performed on samples from baseline and after virologic failure to detect resistance-associated substitutions in NS3, NS5A, and NS5B
- Treatment-emergent adverse events (TEAEs) and laboratory abnormalities were assessed in the safety population, defined as all patients receiving at least 1 dose of study drug
- Interim results of primary and secondary endpoints are presented here

RESULTS

STUDY DISPOSITION

- 1596 patients were enrolled and received study drug; 1573 (99%) have completed study treatment to date
- 21 (1.3%) patients discontinued study drug: primary reasons were adverse event (n = 6), withdrew consent (n = 4), lost to follow-up (n = 3), non-compliance (n = 1), and other reasons (n = 7)
- 1564 (98%) patients have reached the PT week 12 time point, 1374 (86%) have reached PT week 24, and 644 (40%) have reached PT week 52
- To date, 34 (2.1%) patients have discontinued study participation: primary reasons were withdrew consent (n = 9), lost to follow-up (n = 9), adverse event (n = 6), and other reasons (n = 10)

Table 1. Baseline Demographics and Clinical Characteristics

Baseline characteristics	GT1a cirrhotic (n = 97)	GT1a non-cirrhotic (n = 597)	GT1b cirrhotic (n = 139)	GT1b non-cirrhotic (n = 760)	Non-GT1 or missing (n = 3)	Total (N = 1596)
Male, n (%)	71 (73)	332 (56)	78 (56)	314 (41)	2 (67)	797 (50)
White, n (%)	95 (98)	576 (97)	134 (96)	736 (97)	3 (100)	1544 (97)
Age (yrs) <65, n (%)	86 (89)	578 (97)	98 (71)	648 (85)	3 (100)	1413 (89)
BMI (kg/m²) ≥30, n (%)	25 (26)	74 (12)	38 (27)	118 (16)	0	255 (16)
HOMA-IR (mU × mmol/L²) ≥ 3, n (%)	45 (46)	128 (21)	64 (46)	214 (28)	0	451 (28)
IL28B non-CC, n (%)	77 (80)	430 (73)	113 (82)	616 (82)	0	1236 (78)
HCV RNA ≥800 000 IU/mL, n (%)	79 (81)	441 (74)	95 (68)	461 (61)	2 (100)	1078 (68)
Treatment-experienced, n (%)	57 (59)	261 (44)	97 (70)	398 (52)	1 (33)	814 (51)
Albumin <35 g/L, n (%)	8 (8)	2 (<1)	16 (12)	4 (1)	0	30 (2)
Platelets <90 × 10⁹/L, n (%)	20 (21)	4 (1)	27 (20)	7 (1)	0	58 (4)
History of depression or bipolar disorder, n (%)	12 (12)	92 (15)	7 (5)	55 (7)	0	166 (10)
Current or former alcohol use, n (%)	69 (71)	493 (83)	63 (65)	331 (55)	1 (33)	888 (56)

BMI, body mass index; HOMA-IR, homeostatic model assessment of insulin resistance.

EFFICACY

- As the study is still ongoing, 32 subjects who have not reached PT Week 12 were excluded from the calculation of all SVR12 rates in a modified ITT (mITT) population (ITT population counting patients with missing data as failures but excluding patients who have not reached PT Week 12)
- The overall SVR12 rate in the mITT population was 97% (1517/1564) **(Figure 3)**
 - Of the 47 patients without SVR12, 4 were due to viral breakthrough, 21 were due to relapse, 17 due to study drug discontinuation, and 5 had missing SVR12 data
- SVR12 rates of 93–99% were observed across all subtypes with and without cirrhosis **(Figure 3)**

Figure 3. SVR12 Rates in Patients With and Without Cirrhosis by HCV Subtype (mITT Population)

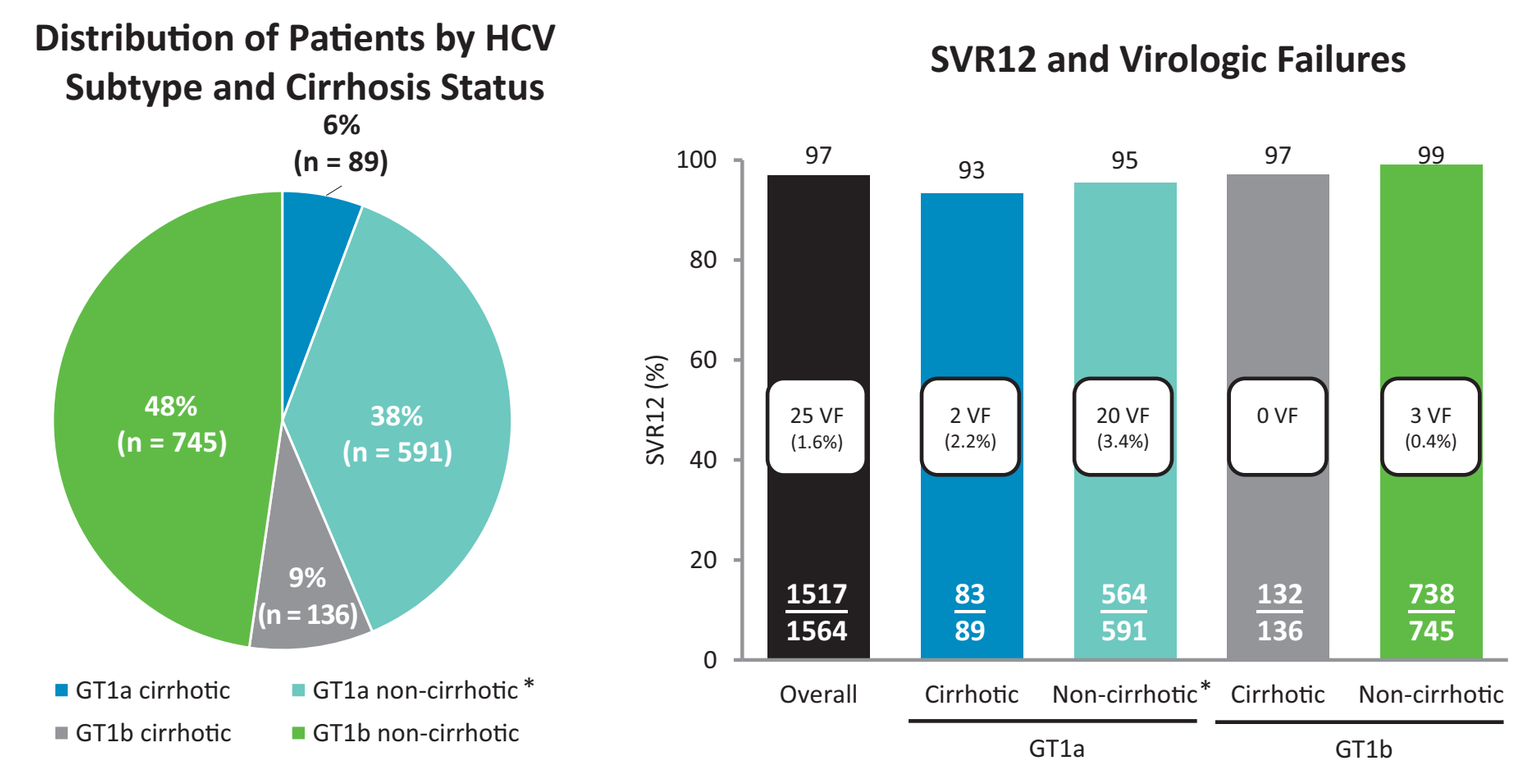
Distribution of Patients by HCV Subtype and Cirrhosis Status
6% (n = 89)
48% (n = 745)
38% (n = 591)
9% (n = 136)
GT1a cirrhotic
GT1a non-cirrhotic*
GT1b cirrhotic
GT1b non-cirrhotic
SVR12 and Virologic Failures
SVR12 (%)
97
93
95
97
99
25 VF (1.6%)
2 VF (2.2%)
20 VF (3.4%)
0 VF
3 VF (0.4%)
1517/1564
83/89
564/591
132/136
738/745
Overall
Cirrhotic
Non-cirrhotic*
Cirrhotic
Non-cirrhotic
GT1a
GT1b

VF, virologic failure.
*The GT1a non-cirrhotic arm contained patients infected with HCV GT1-other (n = 3).

- Overall, 1.6% (25/1564) of the subjects met the criteria for virologic failure: 0.3% (3/881) of patients with HCV GT1b and 3.2% (22/680) of patients with HV GT1a
- Among HCV GT1a patients with cirrhosis, there were 2 virologic failures in the 24 week treatment group and none in the 12-week treatment group

- SVR12 rates were ≥96% when analyzed by multiple baseline characteristics **(Figure 4)**

Figure 4. SVR12 Rates by Baseline Characteristic (mITT Population)

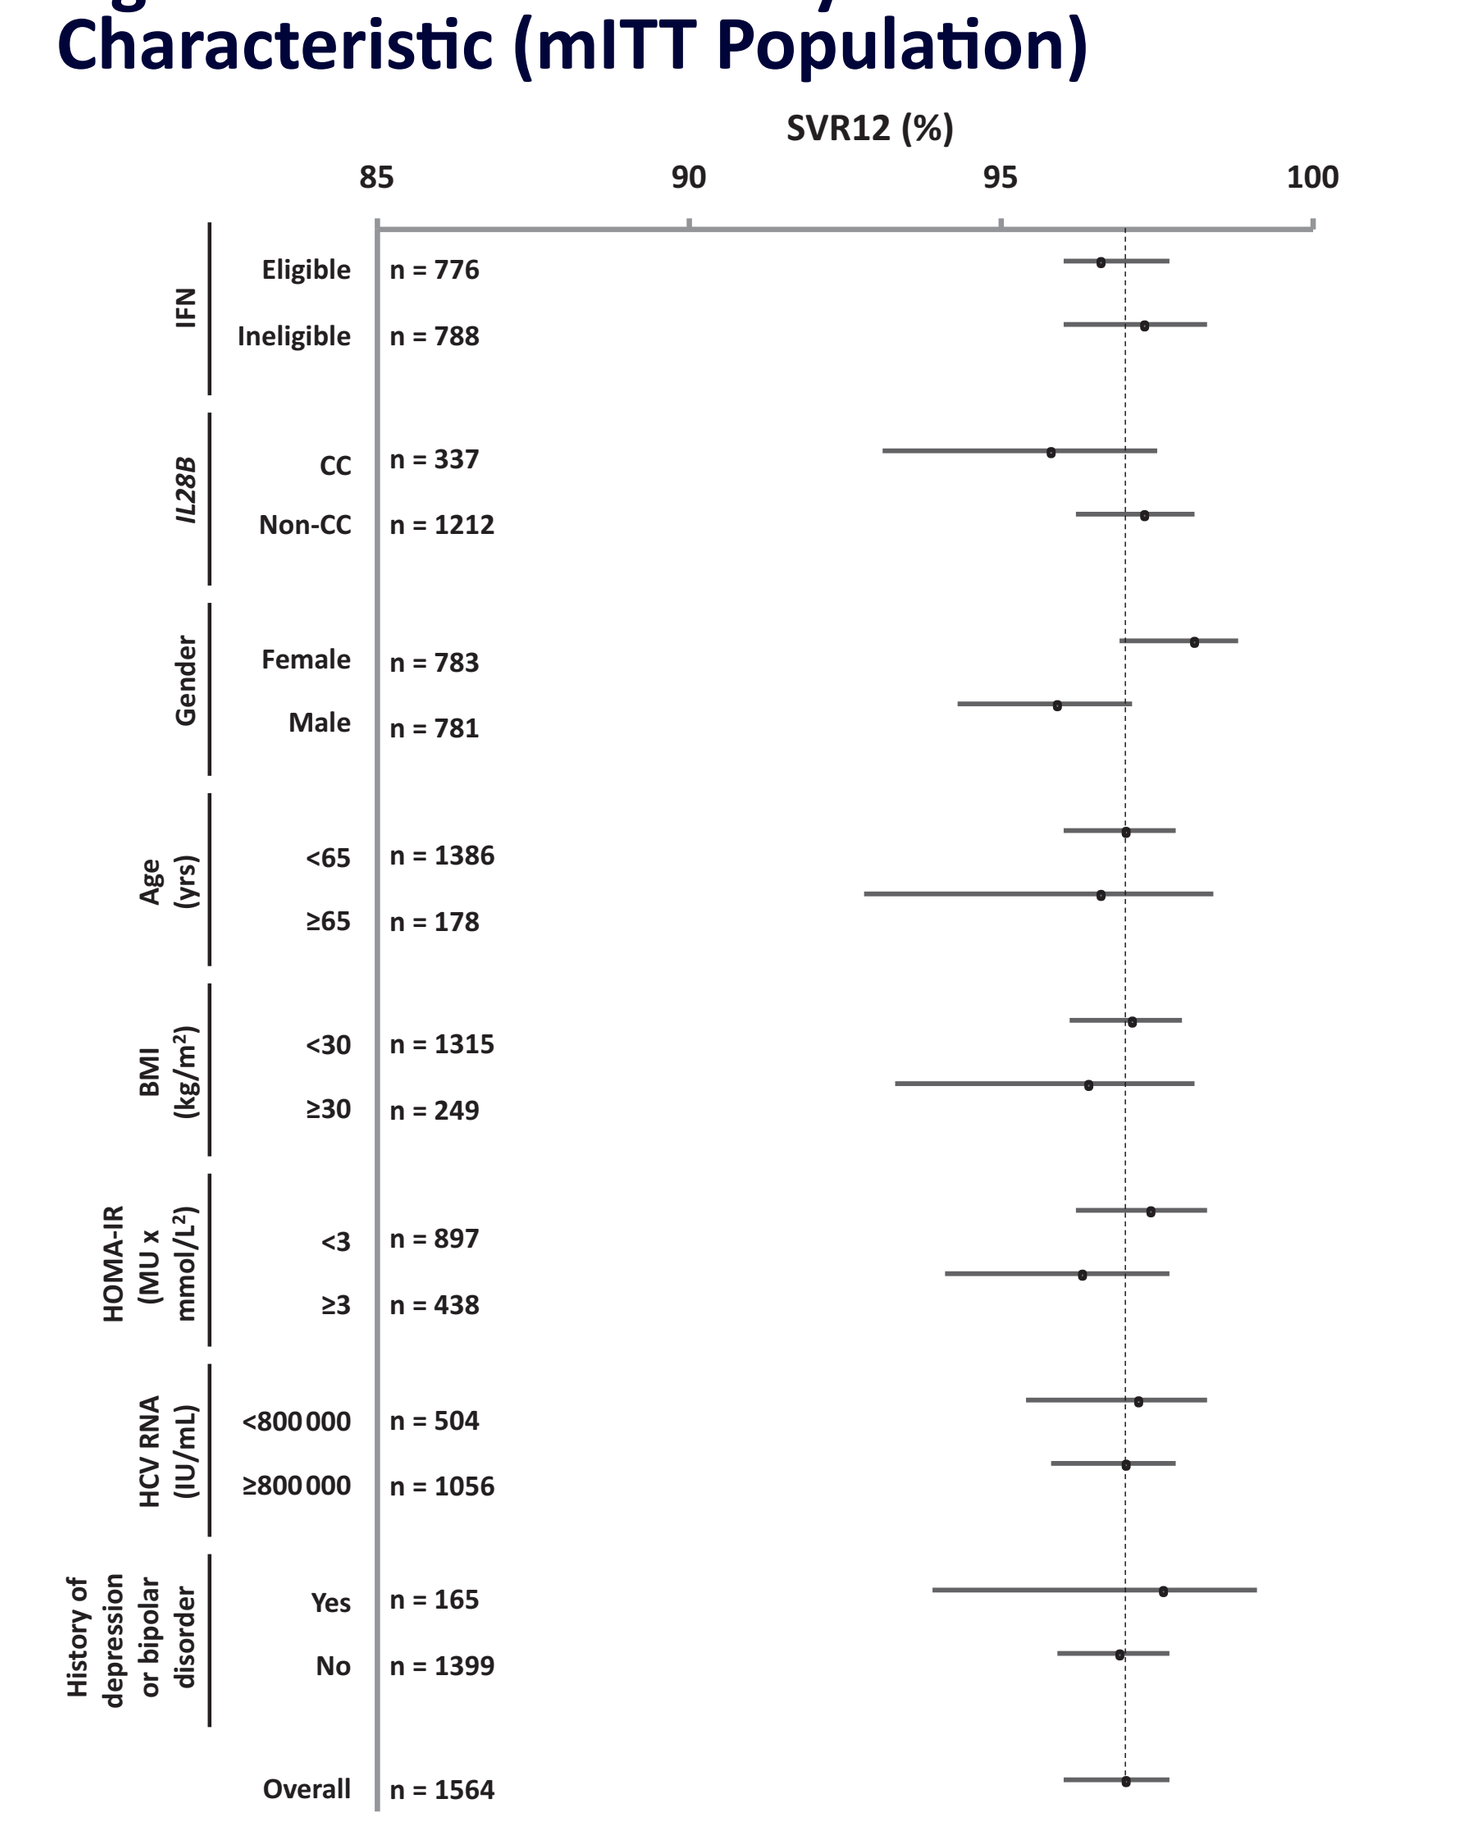
SVR12 (%)
85
90
95
100
IFN
Eligible n = 776
Ineligible n = 788
IL28B
CC n = 337
Non-CC n = 1212
Gender
Female n = 783
Male n = 781
Age (yrs)
<65 n = 1386
≥65 n = 178
BMI (kg/m²)
<30 n = 1315
≥30 n = 249
HOMA-IR (MU x mmol/L²)
<3 n = 897
≥3 n = 438
HCV RNA (IU/mL)
<800000 n = 504
≥800000 n = 1056
History of depression or bipolar disorder
Yes n = 165
No n = 1399
Overall n = 1564

BMI, body mass index; HOMA-IR, homeostatic model assessment of insulin resistance.
Black dots indicate the mITT SVR12 and lines represent the range of the 95% confidence interval. The dotted black line represents the mITT SVR12 of the overall population.

RESISTANCE ANALYSIS IN PATIENTS WITH VIROLOGIC FAILURE

- Of the 25 subjects who experienced virologic failure, 72% (18/25) had at least 1 treatment-emergent substitution
- Among the 22 GT1a-infected patients with virologic failure, the most frequent treatment-emergent substitutions were Y56H and D168V (NS3), M28T and Q30R (NS5A), and S556G (NS5B)
- 3 GT1b-infected patients experienced virologic failure; all had NS5A resistance-associated polymorphisms at baseline

EFFICACY BY BASELINE RENAL FUNCTION

- Baseline eGFR did not impact SVR12 rates **(Figure 5)**

Figure 5. Baseline eGFR Distribution and Corresponding SVR12 Rates by Kidney Function Stages (mITT Population)*

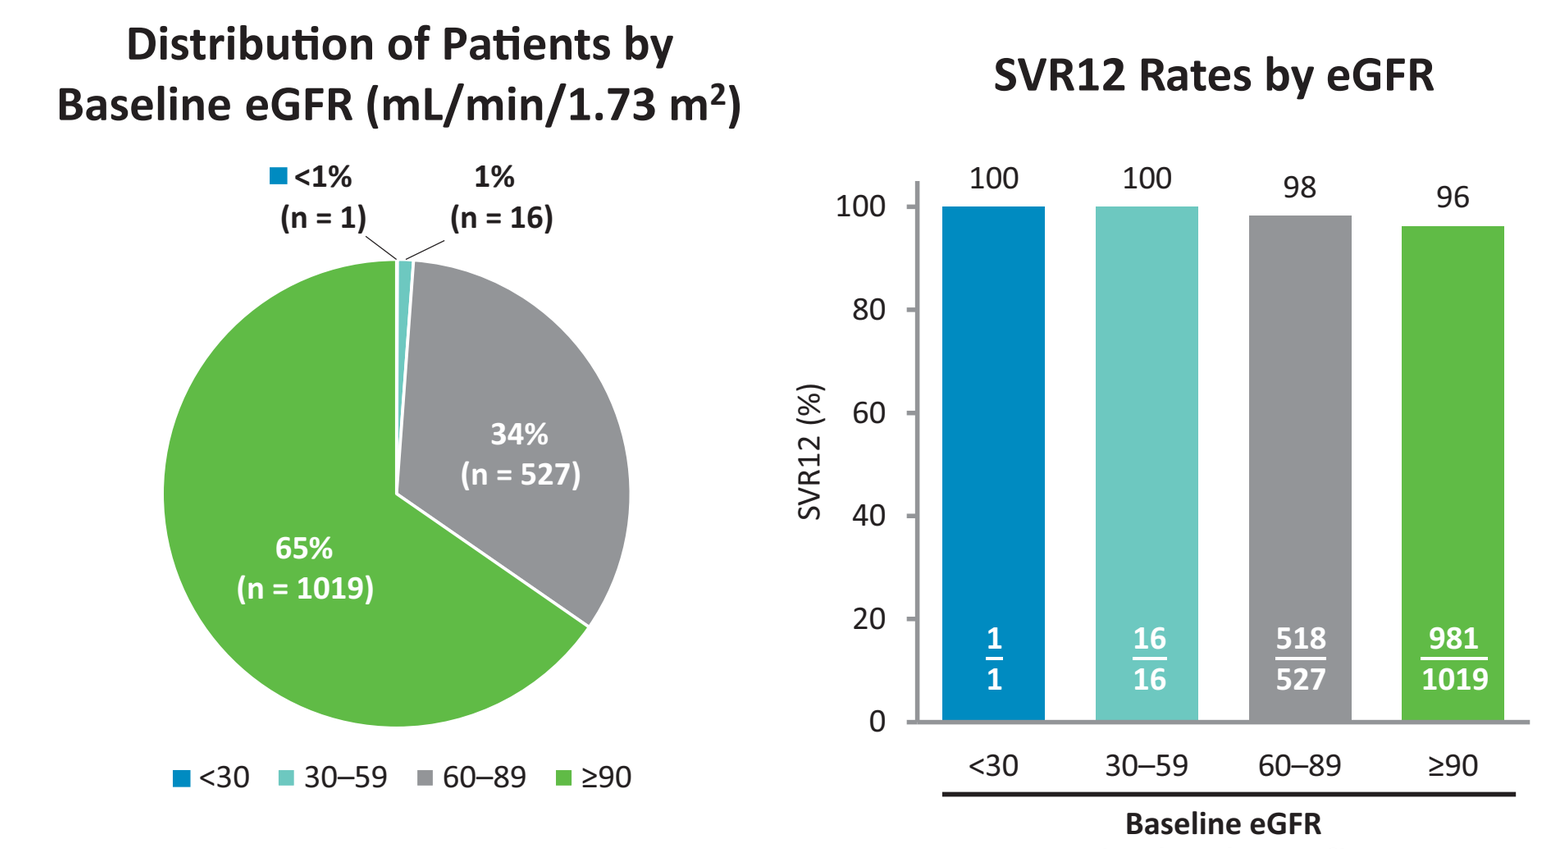
Distribution of Patients by Baseline eGFR (mL/min/1.73 m²)
<1% (n = 1)
1% (n = 16)
34% (n = 527)
65% (n = 1019)
<30
30–59
60–89
≥90
SVR12 Rates by eGFR
SVR12 (%)
100
100
98
96
1/1
16/16
518/527
981/1019
Baseline eGFR (mL/min/1.73 m²)

*One patient had missing value for eGFR at baseline and achieved SVR12.

EFFICACY BY BASELINE FIBROSIS SCORE

- SVR12 rates ranged from 96–97% when stratified by baseline fibrosis score
- The rate of virologic failure was comparable across all baseline fibrosis scores **(Figure 6)**

Figure 6. Baseline Distribution of Liver Fibrosis (Metavir Score) and Corresponding SVR12 Rates (mITT Population)

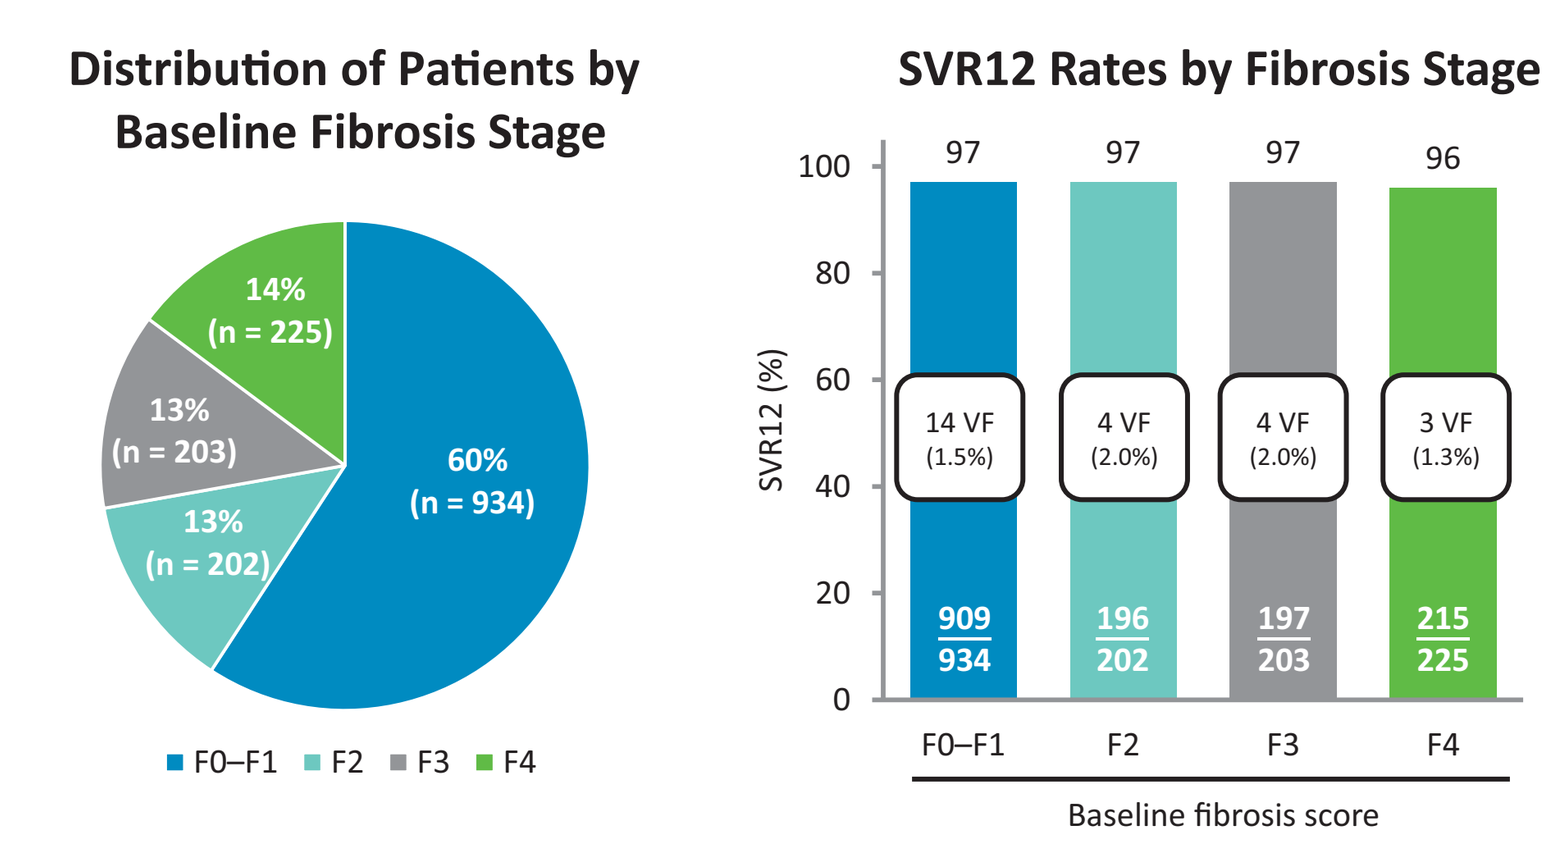
Distribution of Patients by Baseline Fibrosis Stage
14% (n = 225)
13% (n = 203)
13% (n = 202)
60% (n = 934)
F0–F1
F2
F3
F4
SVR12 Rates by Fibrosis Stage
SVR12 (%)
97
97
97
96
14 VF (1.5%)
4 VF (2.0%)
4 VF (2.0%)
3 VF (1.3%)
909/934
196/202
197/203
215/225
Baseline fibrosis score

LONG-TERM IMPACT OF TREATMENT ON LIVER STIFFNESS

- In non-cirrhotic subjects, liver stiffness scores remained stable during and after treatment
- Cirrhotic patients experienced decreases in liver stiffness during treatment, and further decreases were observed in the post-treatment period **(Figure 7)**

Figure 7. Median Change From Baseline in FibroScan® Scores (Safety Population)

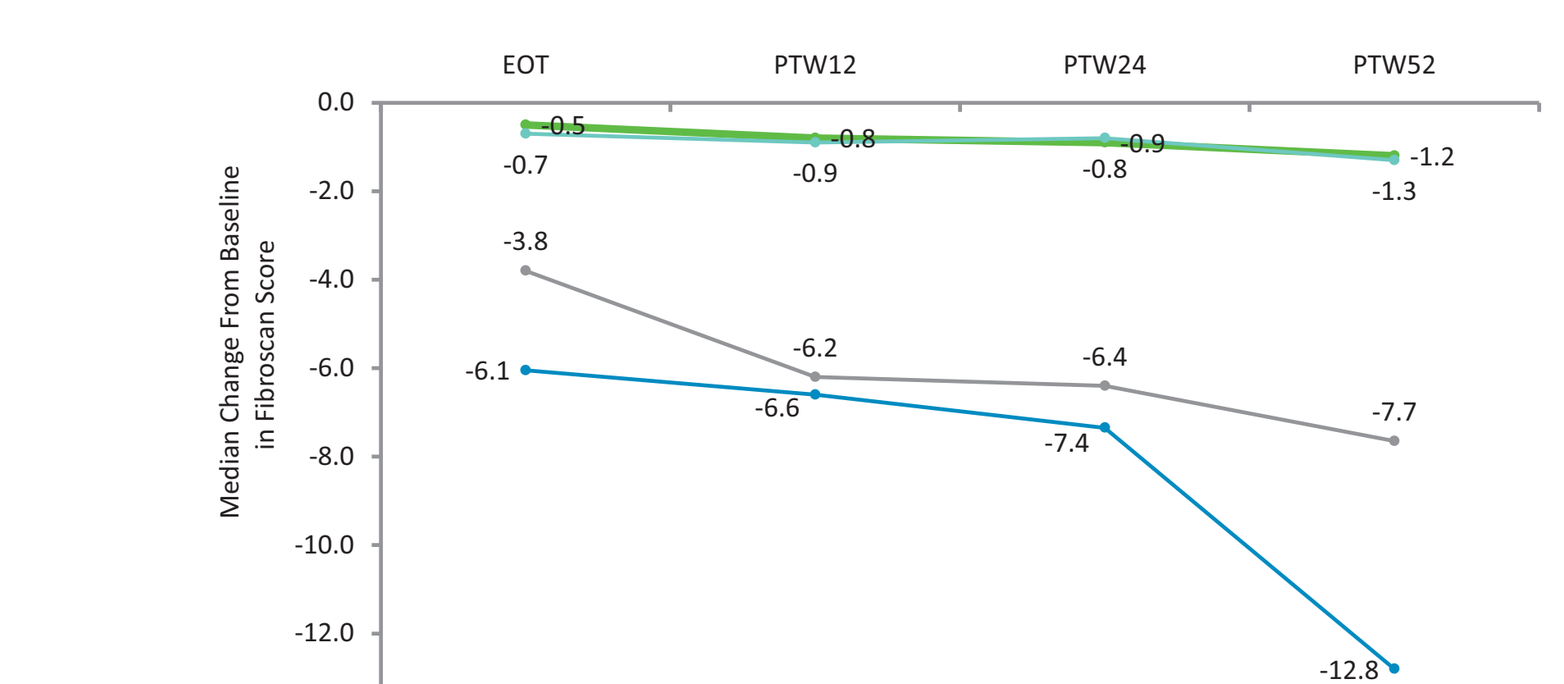
EOT
PTW12
PTW24
PTW52
Median Change From Baseline in Fibroscan Score
-0.5
-0.7
-0.8
-0.9
-0.9
-0.8
-1.2
-1.3
-3.8
-6.2
-6.4
-7.7
-6.1
-6.6
-7.4
-12.8

	EOT N	EOT Range	PTW12 N	PTW12 Range	PTW24 N	PTW24 Range	PTW52 N	PTW52 Range
GT1b non-cirrhotic	483	-7.9 to 11.8	520	-12.3 to 6.1	492	-12.3 to 57.1	229	-6.8 to 7.3
GT1b cirrhotic	79	-52.8 to 16.9	89	-32.7 to 26	77	-33.1 to 10.9	34	-26.3 to 12.1
GT1a non-cirrhotic*	393	-9.9 to 4.8	469	-10.3 to 14.1	423	-10.4 to 7.0	257	-8.0 to 7.8
GT1a cirrhotic	68	-29.4 to 9.6	66	-42.2 to 30.6	62	-41.1 to 5.2	29	-46.1 to 11.1

EOT, end of treatment; PTW, post-treatment week.
*The GT1a non-cirrhotic arm contained patients infected with HCV GT1-other (n = 3).

CLINICAL OUTCOMES

- A total of 11 (0.7%) patients experienced 1 or more clinical outcome event
- 4 patients had non-liver-related events
 - 3 deaths: cerebrovascular embolism, ischemic heart disease, and car accident
 - 1 case of acute renal failure with ascites following an event of pyelonephritis
- 7 patients had liver-related events
 - Aspiration pneumonia leading to multi-organ failure and death (n = 1)
 - 2 cases of HCC: both in patients without a history of HCC, and with negative ultrasound at screening and baseline METAVIR scores of F3 or F4
 - 4 patients with hepatic decompensation events: all were cirrhotic patients assigned to receive 3-DAA + RBV for 24 weeks; consistent with other studies, these patients either had baseline Child-Pugh A-6 score or high (>17 KPa)/FibroTest results (>0.75) **(Table 2)**
- The overall rate of decompensation among patients with cirrhosis was 1.8% (4/225), and the rate of HCC was 0.5% (2/428) among patients with advanced liver disease and cirrhosis

Table 2. Baseline Characteristics of Hepatic Decompensation Patients and Outcome Description

	Baseline characteristics						Description of outcome events			
	Age	Alcohol intake	Non-invasive methods results	Child-Pugh Score	Platelets (× 10⁹/L)	Albumin (g/dL)	Outcome events	Event occurrence	Relation to study drug*	Comment
Pt 1	69	Former	FibroScan: 62.7 FibroTest: N/A	CPA-5	74	3.6	Hepatic encephalopathy and ascites	Weeks 8–12	Reasonable possibility	Study drugs discontinued at Week 8 Both events resolved
Pt 2	59	Never	FibroScan: 17.8 FibroTest: N/A	CPA-6	64	3.5	Hepatic encephalopathy Ascites and hepatorenal syndrome	Week 8	Reasonable possibility	Study drugs discontinued at Week 8 and subject lost to follow-up
Pt 3	59	Former	FibroScan: 27.4 FibroTest: 0.9	CPA-6	94	3.3	Ascites and spontaneous bacterial peritonitis	Weeks 6–10	Reasonable possibility	Study drugs discontinued at Week 10 All events resolved
							Variceal bleeding	PT Week 1		
							Hepatic encephalopathy	PT Week 2		
Pt 4	42	Former	FibroScan: NA FibroTest: 0.88	CPA-5	80	3.8	Variceal bleeding	Week 14	No reasonable possibility	Did not discontinue study drugs Resolved after ligation of esophageal varices

Pt = patient. *As assessed by primary investigator.

SAFETY

- AEs, SAEs, and laboratory abnormalities are presented in **Table 3**
- The majority of AEs were mild or moderate in severity
- Four subjects experienced SAEs considered possibly related to study drugs: palpitation and peripheral edema due to drug–drug interaction with lacidipine (n = 1), hepatic failure and hepatorenal syndrome (n = 1), encephalopathy (n = 1), esophageal varices, ascites and bacterial peritonitis (n = 1). The last 3 patients are described in **Table 2** (Patients 1, 2, and 3)
- Grade 3 laboratory abnormalities were uncommon
- No Grade 4 post-baseline laboratory abnormalities in hemoglobin, ALT, or AST were reported **(Table 3)**

Table 3. Summary of Adverse Events and Post-baseline Laboratory Abnormalities

Safety summary (n, %)	Total (N = 1596)
Any AE	1075 (67.4)
Any serious AE	38 (2.4)
Death	4 (0.3)
AE leading to discontinuation of study drug	8 (0.5)
AEs in ≥10% of patients (n, %)	
Fatigue	300 (18.8)
Headache	283 (17.7)
Nausea	184 (11.5)
Pruritus	182 (11.4)
Insomnia	175 (11.0)
Asthenia	164 (10.3)
Post-baseline laboratory abnormalities, n/N (%)	
Hemoglobin	
Grade 3 (<8.0–6.5 g/dL)	6/1590 (0.4)
Grade 4 (<6.5 g/dL)	0/1590
Total bilirubin	
Grade 3 (>3.0–10 × ULN)	49/1590 (3.1)
Grade 4 (>10 × ULN)	2/1590 (0.1)
ALT	
Grade 3 (>5.0–20 × ULN)	7/1590 (0.4)
Grade 4 (>20 × ULN)	0/1590
AST	
Grade 3 (>5.0–20 × ULN)	4/1589 (0.3)
Grade 4 (>20 × ULN)	0/1589

ULN, upper limit of normal.

RBV DOSE MODIFICATION

- RBV dose was modified due to AE in 11% (92/836) of patients, mostly due to hemoglobin decrease

CONCLUSIONS

- Interim results from the TOPAZ-I study confirm that SVR12 rates, and safety and tolerability profiles are consistent with those seen in the registration trials of OBV/PTV/r + DSV ± RBV in HCV GT1-infected patients with and without cirrhosis
- Efficacy in this trial was high regardless of fibrosis stage or renal function at baseline
- In the post-treatment period, liver stiffness remained stable in patients without cirrhosis, while decreases were observed in patients with cirrhosis
- Clinical outcome events, including all-cause death, liver-related death, liver decompensation, and HCC, occurred in <1% of the patients to date

REFERENCES

1. Dore GJ, et al. *J Viral Hepat.* 2014;21(Suppl 1):1–4.
2. Viekirax EU Summary of Product Characteristics. Available at https://www.medicines.org.uk/emc/medicine/29784 (accessed Oct 2016).
3. Exviera EU Summary of Product Characteristics. Available at https://www.medicines.org.uk/emc/medicine/29785 (accessed Oct 2016).
4. Dumas EO, et al. *J Hepatol.* 2015;62(Suppl):S855.

DISCLOSURES

The design, study conduct, analysis, and financial support of the study (NCT02219490) were provided by AbbVie. AbbVie participated in the interpretation of data, review, and approval of the content. All authors had access to all relevant data and participated in writing, review, and approval of this presentation.

K Agarwal: Speaker Bureau/Consultancy: Achilion, Arbutus, BMS, Gilead, GSK, Intercept, Janssen, Merck, Novartis, Roche. Research Support: AbbVie, BMS, Gilead, Roche.

GB Gaeta: Speaker or Advisor for: AbbVie, BMS, Merck, Gilead, Janssen.

SS Lee: Consultant/Research Grants: AbbVie, Achillion, Bayer, BMS, Gilead, Janssen, Merck, Novartis, Roche, Pendopharm, Roche. Speaker for: AbbVie, BMS, Gilead, Janssen, Merck, Roche.

E Dumas, M Charafeddine, D Cohen, T Pilot-Matias, L Liu, K Howieson, and A Joy: Employees of AbbVie and may hold stock or options.

A Streinu-Cercel: Speaker/Principal Investigator in HCV trials by: AbbVie, Boehringer Ingelheim, Janssen, Merck.

E Schott: Speaker Bureau/Consultancy/Travel Support: AbbVie, BMS, Gilead, Intercept, Janssen, MSD, Roche.

RJ Andrade: Advisor/Speaker Bureau: AbbVie, Bayer, BMS, Gilead, GSK, Intercept, Janssen, Madaus, MSD, Novartis, Roche. Research Grant: Boehringer Ingelheim, Schwabe Pharmaceuticals.

AM Perez-Rios: Advisory Boards: Roche, MSD, Gilead, AbbVie, BMS. Speaker for: Roche, MSD, Janssen, AbbVie, BMS, Stendhal, Gilead.

J George: Advisory Boards: Merck, Gilead, AbbVie, BMS, Pharmaxis.

I Bakulin: Grants/Clinical Trials: BMS, GSK, Gilead, Falk, Johnson & Johnson, Roche, Schering-Plough/Merck, Gilead, Novartis. Speaking/Teaching: BMS, AbbVie, GSK, Johnson & Johnson, Roche, Schering-Plough/Merck, Gilead, Novartis. Advisory Boards: BMS, Johnson & Johnson, AbbVie, GSK, Roche, Schering-Plough/Merck.

R Marinho: Talks and Advisory Boards: AbbVie, MSD, Gilead, Janssen, BMS.

R Ozaras: Nothing to disclose.

F Zoulim: Consultant for AbbVie and Gilead.

ACKNOWLEDGMENTS

Medical writing support was provided by Scott Battle of Medical Expressions, funded by AbbVie.

44587-161467-865 — AASLD 2016 — AbbVie — proof 7 (rjh) — november 9, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051
CREATED @ 100%

Predictors of Improvement in Glomerular Filtration Rate Among Patients Treated With Ombitasvir/Paritaprevir/r and Dasabuvir With or Without Ribavirin

D Bernstein[1], A Tran[2], P Martin[3], K Kowdley[4], M Bourlière[5], MS Sulkowski[6], PJ Pockros[7], L Larsen[8], D Shuster[8], DE Cohen[8], B Renjifo[8], IM Jacobson[9]

[1]Department of Medicine, Hofstra Northwell School of Medicine, Manhasset, New York, United States; [2]University Hospital of Nice, Digestive Centre, Nice, F-06202, Cedex 3, France; [3]Division of Hepatology, School of Medicine, University of Miami, Miami, Florida, United States; [4]Swedish Medical Center, Seattle, Washington, United States; [5]Hôpital Saint Joseph, Marseilles, France; [6]Viral Hepatitis Center, Johns Hopkins University, Baltimore, Maryland, United States; [7]Scripps Clinic and Scripps Translational Science Institute, La Jolla, California, United States; [8]AbbVie Inc., North Chicago, Illinois, United States; [9]Mount Sinai Beth Israel, New York, New York, United States

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

INTRODUCTION

- Patients with hepatitis C virus (HCV) infection have a high rate of chronic kidney disease (CKD)[1]
- Management of HCV in patients with advanced CKD remains a challenge because the safety and efficacy profile of direct antiviral agents in these patients has not been fully established
- We previously reported that treatment of HCV genotype (GT) 1-infected patients with the all-oral regimen of ombitasvir, paritaprevir boosted with ritonavir, and dasabuvir (OBV/PTV/r + DSV) resulted in high sustained virologic response rates and was not associated with overall changes in renal function[2,3]
- However, we observed a mean change in estimated glomerular filtration rate (eGFR) at the end of treatment (EOT) of +6, +1.3, and -5.6 mL/min/1.73 m² in patients with baseline (BL) eGFR, ≤60, 60–90 or >90 mL/min/1.73 m², repectively[2,3]
- A better understanding of predictors associated with changes in renal function when treated with OBV/PTV/r + DSV may help to guide clinicians treating HCV-infected patients

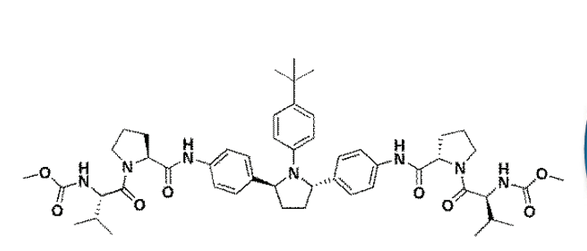
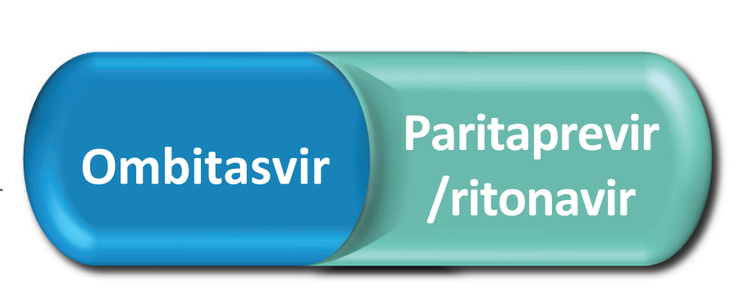
Ombitasvir
Paritaprevir /ritonavir
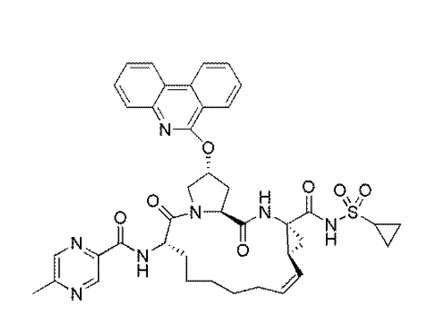

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)

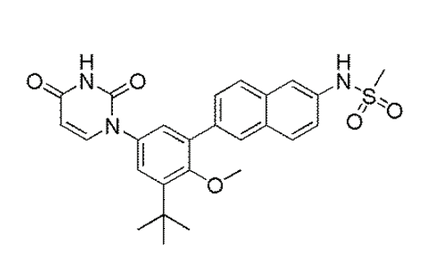
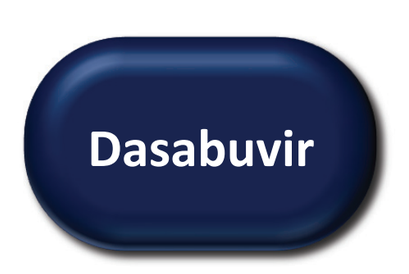
Dasabuvir

Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

Paritaprevir was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

ACKNOWLEDGEMENTS

Medical writing support was provided by Gillian Patman of Medical Expressions, funded by AbbVie.

DISCLOSURES

The design, study conduct, analysis, and financial support of all the clinical studies were provided by AbbVie. AbbVie participated in the interpretation of data, review, and approval of the content. All authors had access to all relevant data and participated in writing, review, and approval of this presentation.

D Bernstein: Research support: AbbVie, BMS, Gilead, Merck; Consultant/Speaker: AbbVie, BMS, Gilead, Merck.

A Tran: Board Member and Speaker: AbbVie, BMS, Gilead, Janssen, MSD.

P Martin: Investigator: AbbVie, Gilead, Merck; Consultant: AbbVie, Gilead, Merck.

K Kowdley: Grant/Research Funding: AbbVie, Gilead, Merck; Consultant/Advisor: AbbVie, Gilead, Merck, Trio Health Advisory Group.

M Bourlière: Board Member and Speaker: AbbVie, BMS, Gilead, GSK, Janssen, MSD.

MS Sulkowski: Research grants with funds paid to Johns Hopkins University: AbbVie, BMS, Gilead, Janssen, Merck; Scientific Advisor: AbbVie, BMS, Cocrystal Pharma, Gilead, Janssen, Merck, Trek.

PJ Pockros: Speaker/Consultant/Advisor: AbbVie, BMS, Gilead, Merck; Research Support: AbbVie, BMS, Conatus, Gilead, Janssen, Merck, Roche Molecular.

B Renjifo, L Larsen, D Shuster, DE Cohen: Employees of AbbVie and may hold AbbVie stock or options.

IM Jacobson: Research Support: AbbVie, BMS, Gilead, Janssen, Merck; Consultant/Advisor: AbbVie, BMS, Gilead, Intercept, Janssen, Merck, Trek; Speaker: AbbVie, BMS, Gilead, Intercept, Janssen.

OBJECTIVES

- To describe changes in eGFR among patients treated with OBV/PTV/r + DSV ± ribavirin (RBV)
- To compare the changes in eGFR among patients treated with OBV/PTV/r + DSV ± RBV vs placebo
- To investigate BL predictors of eGFR changes for patients treated with placebo or OBV/PTV/r + DSV ± RBV
- To further characterize the safety profile of OBV/PTV/r + DSV ± RBV in patients categorized by BL eGFR

METHODS

- Renal function was categorized by eGFR using the Modification of Diet in Renal Disease (MDRD) Study equation and patients were grouped in categories according to BL eGFR: >90, 60–90, or <60 mL/min/1.73 m²
- Analysis included patients treated with OBV/PTV/r + DSV ± RBV for 12 or 24 weeks from 9 clinical trials (SAPPHIRE-I, SAPPHIRE-II, TURQUOISE-II, TURQUOISE-III, TOPAZ-II, PEARL-II, PEARL-III, PEARL-IV, RUBY-I [excluding patients on dialysis]) and patients in the double-blind placebo arm of the SAPPHIRE-I and SAPPHIRE-II clinical trials
- The analysis of safety outcomes included:
 - Adverse events (AEs) occurring in ≥10% of patients
 - Grade 3 and 4 laboratory abnormalities during treatment
 - Renal-associated AEs defined by the Standardized MedDRA Queries (SMQ) search terms “acute renal failure” (SMQ20000003) and “CKD” (SMQ20000213) plus the Company MedDRA Query search term “DME acute renal failure” (CMQ80000038)
- BL factors associated with a ≥10 mL/min/1.73 m² increase in eGFR at EOT were examined by stepwise logistic regression in 7 trials that collected BL urinalysis per protocol

RESULTS

STUDY POPULATION

- The pooled analysis of the 9 clinical trials included 2733 patients with a BL eGFR <60, 60–90, or >90 mL/min/1.73 m² that received OBV/PTV/r + DSV ± RBV
 - 26% (705/2733) of patients received OBV/PTV/r + DSV without RBV and 74% (2028/2733) of patients were treated with OBV/PTV/r + DSV + RBV
 - The majority of patients were white and two-thirds were treatment-naïve. GT1a and GT1b were equally represented in this cohort
 - 3%, 54%, and 43% had BL eGFR in the categories of <60, 60–90, and >90 mL/min/1.73 m², respectively
 - BL characteristics of these 2733 patients are listed in **Table 1**
- Analysis also included 255 patients in the placebo arms of the SAPPHIRE-I and SAPPHIRE-II clinical trials[4,5]
 - The demographics of the 255 patients in the placebo arms were similar to those of the 769 patients treated with OBV/PTV/r + DSV + RBV[4,5]
 - 2%, 60%, and 38% had BL eGFR in the categories of <60, 60–90, and >90 mL/min/1.73 m², respectively

RESULTS (CONTINUED)

Table 1. Baseline Characteristics of 2733 Patients Treated With OBV/PTV/r + DSV ± RBV in the Pooled Analysis

	eGFR (mL/min/1.73 m²)		
Characteristic	<60 N = 82	60–90 N = 1479	>90 N = 1172
Age, years, mean ± SD	59.9 ± 6.6	54.8 ± 9.7	49.1 ± 11.6
Female, n (%)	47 (57)	683 (46)	397 (34)
Race, n (%)*			
White	72 (88)	1348 (91)	1026 (88)
Black	9 (11)	99 (7)	110 (9)
Asian	1 (1)	19 (1)	18 (2)
Other	0	13 (1)	18 (2)
HCV genotype subtype, n (%)			
1a	45 (55)	808 (55)	662 (57)
1b	37 (45)	670 (45)	509 (43)
Other	0	1 (<0.1)	1 (<0.1)
BMI, <30 kg/m², n (%)	58 (71)	1177 (80)	896 (77)
IL28B non-CC genotype, n (%)	56 (68)	1129 (76)	941 (80)
Baseline HCV RNA level, $\log_{10}$ IU/mL, mean ± SD	6.52 ± 0.57	6.46 ± 0.63	6.36 ± 0.63
Prior treatment history, n (%)			
Treatment-naïve	52 (63)	996 (67)	779 (67)
Treatment-experienced	30 (37)	483 (33)	393 (34)
Fibrosis stage, n (%)†			
F0–F1	37 (45)	793 (54)	650 (55)
F2	11 (13)	229 (15)	145 (12)
F3	7 (9)	190 (13)	112 (10)
F4	27 (33)	265 (18)	264 (23)
History of diabetes, n (%)	10 (12)	119 (8)	97 (8)
History of hypertension, n (%)	41 (50)	467 (32)	291 (25)
BL proteinuria, n/N (%)‡	21/67 (31)	325/1449 (22)	277/1151 (24)
BL hematuria, n/N (%)‡	3/67 (5)	64/1449 (4)	63/1151 (6)

BL, baseline; BMI, body mass index; eGFR, estimated glomerular filtration rate; HCV, hepatitis C virus; *IL28B*, interleukin 28B; SD, standard deviation.
*Missing data for 1 patient in the Other category; †Data missing for 3 patients; ‡ Excludes RUBY-I and TURQUOISE-III.

DECREASE IN eGFR FROM BASELINE

- In the SAPPHIRE trials, there was no difference between placebo (55/255, 20%) vs OBV/PTV/r + DSV + RBV (170/769, 22%) in the proportion of patients with a post-BL eGFR decrease ≥10 mL/min/1.73m² at EOT **(Figure 1)**

Figure 1. Patients With a Post-baseline eGFR Decrease ≥10 mL/min/1.73 m² at End of Treatment

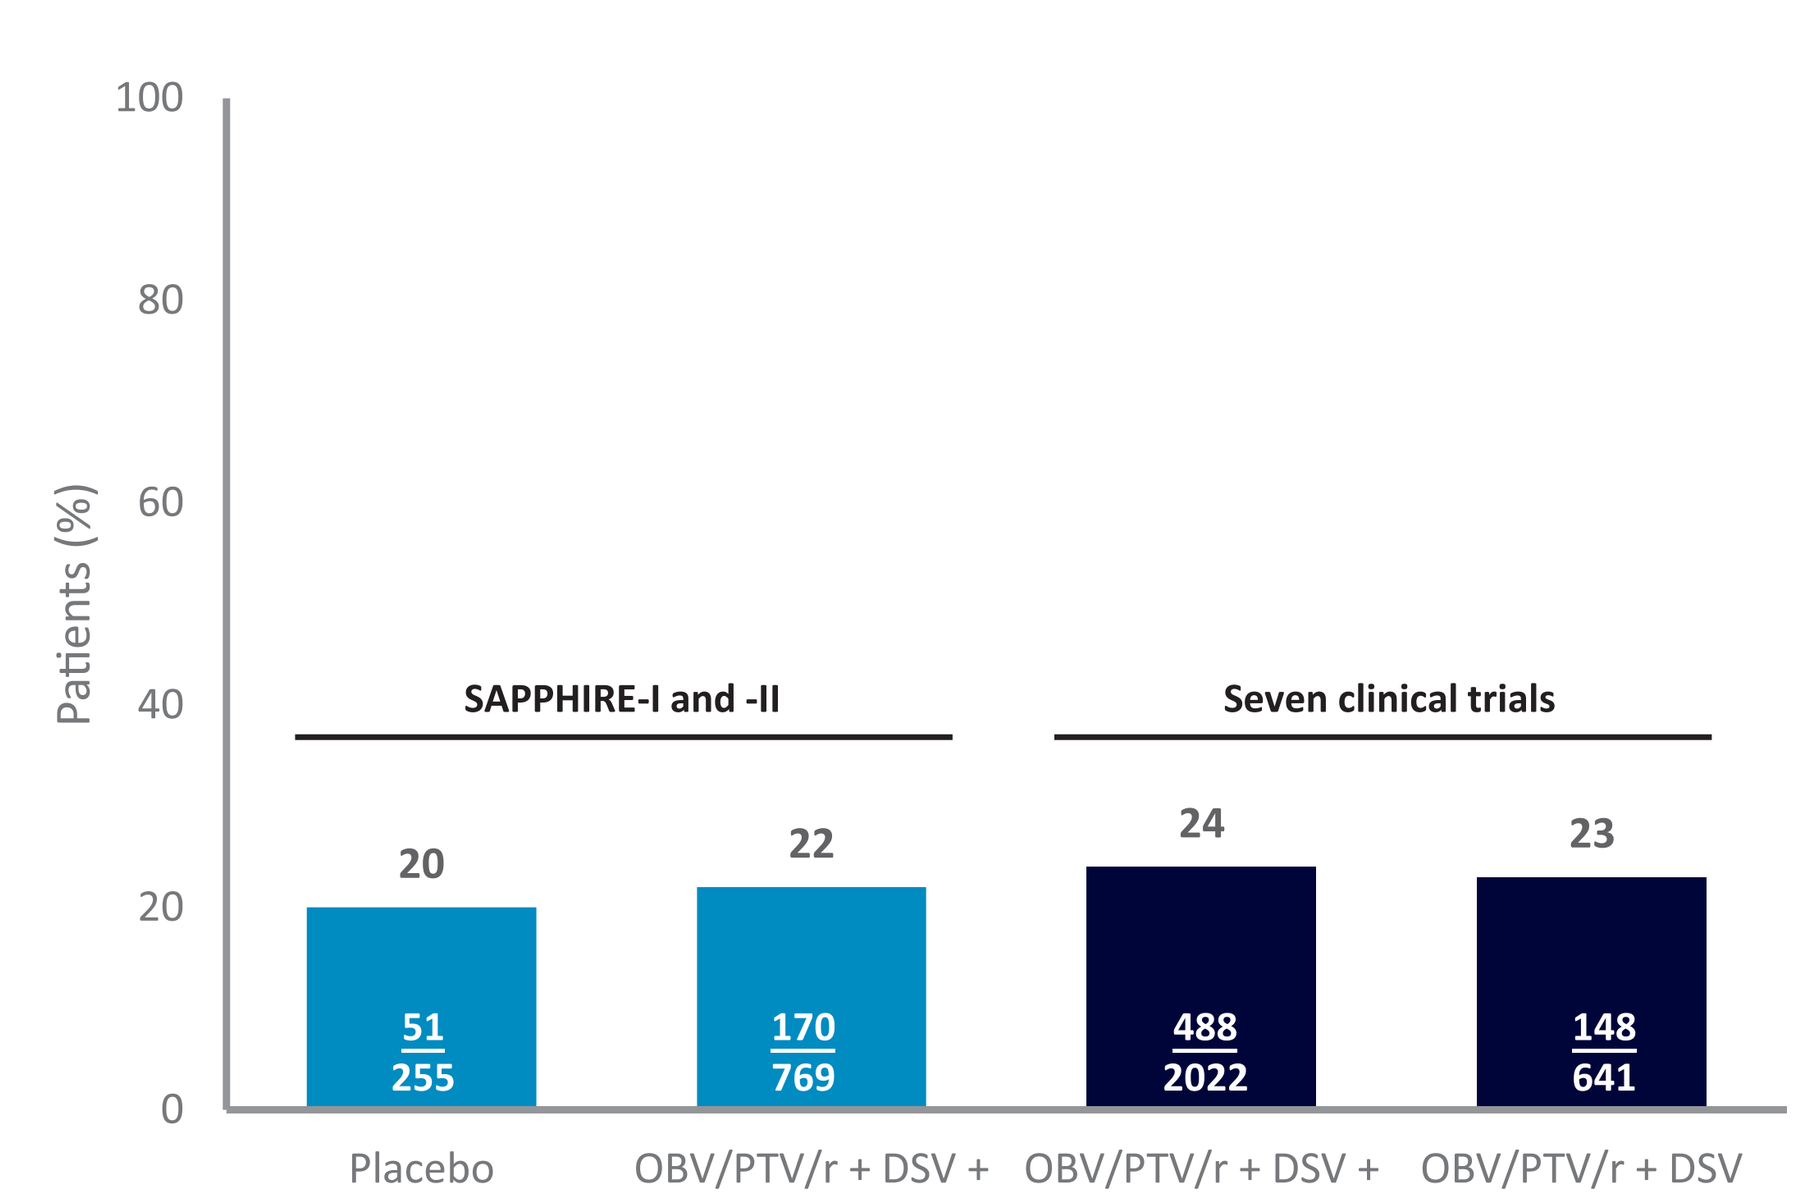
100
80
60
40
20
0
Patients (%)
SAPPHIRE-I and -II
Seven clinical trials
20
22
24
23
51
255
170
769
488
2022
148
641
Placebo
OBV/PTV/r + DSV + RBV
OBV/PTV/r + DSV + RBV
OBV/PTV/r + DSV

- When eGFR changes were assessed according to BL eGFR, a similar pattern was also observed between the placebo and OBV/PTV/r + DSV + RBV arms of the SAPPHIRE trials or OBV/PTV/r + DSV ± RBV in 7 clinical trials that had available BL urinalysis **(Table 2)**
- None of the patients in the placebo (N = 5) or OBV/PTV/r + DSV + RBV (N = 14) with a BL <60 mL/min/1.73 m² experienced eGFR decrease ≥10 mL/min/1.73 m² at EOT

Table 2. Patients With a Post-baseline eGFR Decrease ≥10 mL/min/1.73 m² at End of Treatment

	SAPPHIRE-I and -II		Seven Clinical Trials	
BL eGFR category (mL/min/1.73 m²) n/N (%)	Placebo N = 255	OBV/PTV/r + DSV + RBV N = 769	OBV/PTV/r + DSV + RBV N = 2022	OBV/PTV/r + DSV N = 641
<60	0/5 (0)	0/14 (0)	1/48 (2)	1/19 (5)
60–90	11/152 (7)	50/455 (11)	120/1065 (11)	55/381 (9)
>90	40/98 (41)	120/310 (39)	367/909 (40)	92/241 (38)
TOTAL	51/255 (20)	170/769 (22)	488/2022 (24)	148/641 (23)

INCREASE IN eGFR FROM BASELINE

- In the SAPPHIRE trials, there was no difference between placebo (56/255, 22%) vs OBV/PTV/r + DSV + RBV (170/769, 22%) in the proportion of patients with post-baseline eGFR increase ≥10 mL/min/1.73m² at EOT **(Figure 2 and Table 3)**

Figure 2. Patients With a Post-baseline eGFR Increase ≥10 mL/min/1.73 m² at End of Treatment

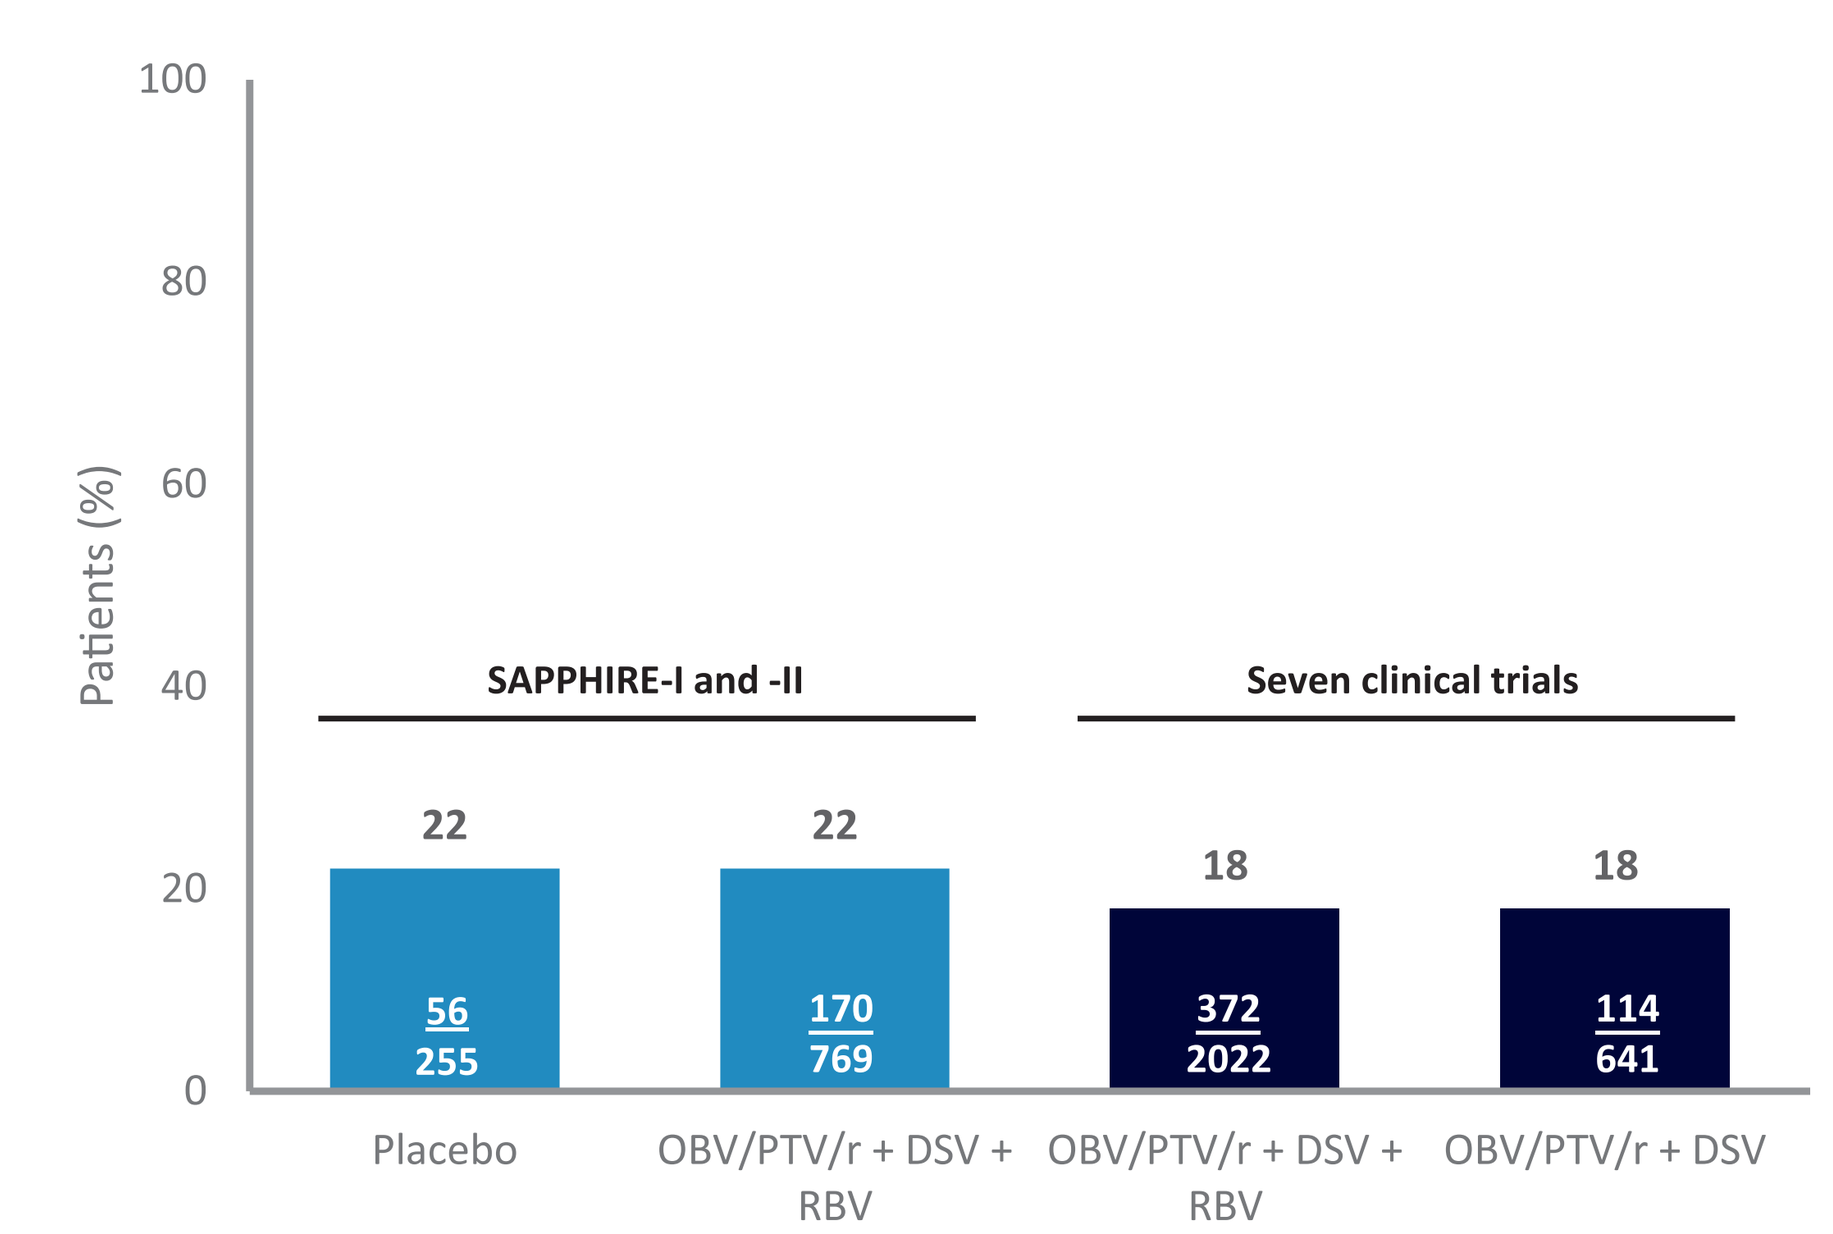
100
80
60
40
20
0
Patients (%)
SAPPHIRE-I and -II
Seven clinical trials
22
22
18
18
56
255
170
769
372
2022
114
641
Placebo
OBV/PTV/r + DSV + RBV
OBV/PTV/r + DSV + RBV
OBV/PTV/r + DSV

Table 3. Patients With a Post-baseline eGFR ≥10 mL/min/1.73 m² Increase at End of Treatment

	SAPPHIRE-I and -II		Seven Clinical Trials	
BL eGFR category (mL/min/1.73 m²) n/N (%)	Placebo N = 255	OBV/PTV/r + DSV + RBV N = 769	OBV/PTV/r + DSV + RBV N = 2022	OBV/PTV/r + DSV N = 641
<60	3/5 (60)	1/14 (7)	12/48 (25)	5/19 (26)
60–90	37/152 (24)	119/445 (27)	253/1065 (24)	86/381 (23)
>90	16/98 (16)	50/310 (16)	107/909 (12)	23/241 (10)
TOTAL	56/255 (22)	170/769 (22)	372/2022 (18)	114/641 (18)

PREDICTORS OF eGFR IMPROVEMENT

- In the 7 clinical trials that had available BL urinalysis, 18% (486/2663) of patients had an increase of ≥10 mL/min/1.73 m² in eGFR at the EOT. BL factors associated with an increase in eGFR of ≥10 mL/min/1.73 m² included body mass index (BMI), non-black race, BL proteinuria, and a positive history of diabetes **(Table 4)**

Table 4. Logistic Regression Analysis in 7 Clinical Trials – Excluding RUBY-I and TURQUOISE-III*

	≥10 mL/min/1.73 m² eGFR improvement		
Variable	OR	95% CI	*P*-value
Baseline proteinuria (positive vs negative)	1.65	1.32–2.05	<.001
Baseline BMI	0.95	0.93–0.97	<.001
Race (black vs non-black)	0.60	0.38–0.92	.021
History of diabetes (yes vs no)	1.51	1.06–2.16	.023

BMI, body mass index; CI, confidence interval; eGFR, estimated glomerular filtration rate; HCV, hepatitis C virus; OR, odds ratio.
*Baseline characteristics that were evaluated: urine blood (positive or negative), urine protein (positive or negative), fibrosis score (F0–1, F2, F3, F4), BMI (continuous, kg/m²), age (continuous, years), sex (male or female), race (black or non-black), history of hypertension (yes or no), history of diabetes (yes or no), baseline HCV RNA (continuous, $\log_{10}$ IU/mL).

- BL predictors were also investigated among patients in the placebo group from the SAPPHIRE trials. The stepwise logistic regression analysis did not identify any BL factor associated with eGFR changes

SAFETY

- The safety during the double-blind period of patients receiving OBV/PTV/r + DSV + RBV compared with patients receiving placebo in the SAPPHIRE trials has been already published[3,4]
- In 9 clinical trials, AEs were reported in 74% (522/705) and 87% (1768/2028) of patients treated with OBV/PTV/r + DSV without and with RBV, respectively
- Of the AEs occurring in ≥10% of patients, the most frequent were fatigue, headache, and nausea regardless of eGFR category **(Table 5)**. More AEs of fatigue and anemia were reported in subjects administered RBV with eGFR <60 mL/min/1.73 m²

Table 5. Adverse Events in ≥10% of Patients in Any Subgroup

	OBV/PTV/r + DSV			OBV/PTV/r + DSV + RBV		
	eGFR, mL/min/1.73 m²			eGFR, mL/min/1.73 m²		
n (%)	<60 N = 29	60–90 N = 414	>90 N = 262	<60 N = 53	60–90 N = 1065	>90 N = 910
Fatigue	5 (17)	106 (26)	68 (26)	25 (47)	395 (37)	277 (30)
Headache	8 (28)	91 (22)	61 (23)	15 (28)	311 (29)	224 (25)
Nausea	1 (3)	35 (9)	19 (7)	12 (23)	227 (21)	162 (18)
Pruritus	4 (14)	33 (8)	18 (7)	10 (19)	162 (15)	128 (14)
Insomnia	3 (10)	19 (5)	18 (7)	11 (21)	168 (16)	112 (12)
Diarrhea	3 (10)	43 (10)	28 (11)	14 (26)	130 (12)	97 (11)
Asthenia	0	14 (3)	7 (3)	4 (8)	125 (12)	76 (8)
Rash	0	18 (4)	8 (3)	6 (11)	110 (10)	78 (9)
Cough	1 (3)	24 (6)	9 (3)	6 (11)	86 (8)	73 (8)
Dizziness	0	22 (5)	11 (4)	7 (13)	93 (9)	52 (6)
Decreased appetite	1 (3)	10 (2)	11 (4)	7 (13)	60 (6)	52 (6)
Anemia	0	0	1 (< 1)	16 (30)	94 (9)	29 (3)
Vomiting	1 (3)	8 (2)	5 (2)	6 (11)	62 (6)	34 (4)

DSV, dasabuvir; eGFR, estimated glomerular filtration rate; OBV, ombitasvir; PTV, paritaprevir; r, ritonavir; RBV, ribavirin.

- Patients with laboratory abnormalities of at least grade 3 were infrequent; however, grade 2+ hemoglobin occurred more frequently in those treated with RBV and in the lowest eGFR category **(Table 6)**

Table 6. Laboratory Abnormalities at Baseline

	OBV/PTV/r + DSV			OBV/PTV/r + DSV + RBV		
	eGFR, mL/min/1.73 m²			eGFR, mL/min/1.73 m²		
n/N (%)	<60 N = 29	60–90 N = 414	>90 N = 262	<60 N = 53	60–90 N = 1065	>90 N = 910
Hemoglobin (g/dL)						
≥Grade 2 (<10)	2 (7)	0	0	16/52 (31)	86/1061 (8)	37/909 (4)
≥Grade 3 (< 8)	0	0	0	3/52 (6)	6/1061 (1)	0
ALT (U/L)						
≥Grade 2 (>3 × ULN)	0	6 (1)	2 (1)	1/52 (2)	20/1061 (2)	13/909 (1)
≥Grade 3 (>5 × ULN)	0	2 (<1)	0	1/52 (2)	12/1061 (1)	7/909 (1)
AST (U/L)						
≥Grade 2 (>3 × ULN)	0	7 (2)	0	1/52 (2)	11/1060 (1)	6/909 (1)
≥Grade 3 (>5 × ULN)	0	2 (<1)	0	0	6/1060 (1)	3/909 (<1)
Total bilirubin (µmol/L)						
≥Grade 2 (>1.5 × ULN)	2 (7)	25 (6)	18 (7)	20/52 (38)	283/1061 (27)	208/909 (23)
≥Grade 3 (>3 × ULN)	0	0	2 (1)	5/52 (10)	52/1061 (5)	40/909 (4)

ALT, alanine aminotransferase; AST, aspartate aminotransferase; DSV, dasabuvir; eGFR, estimated glomerular filtration rate; OBV, ombitasvir; PTV, paritaprevir; r, ritonavir; RBV, ribavirin; ULN, upper limit of normal.
Note: post-baseline grade must be at least 1 grade increased from baseline grade.

- Renal-associated AEs were experienced by only 1.9% (52/2733) of patients **(Table 7)**

Table 7. Renal-associated Adverse Events

	OBV/PTV/r + DSV			OBV/PTV/r + DSV + RBV		
	eGFR, mL/min/1.73 m²			eGFR, mL/min/1.73 m²		
	<60 N = 29	60–90 N = 414	>90 N = 262	<60 N = 53	60–90 N = 1065	>90 N = 910
Patients with renal-associated AEs, n (%)	1 (3)	9 (2)	4 (2)	5 (9)	22 (2)	11 (1)

AE, adverse event; DSV, dasabuvir; eGFR, estimated glomerular filtration rate; OBV, ombitasvir; PTV, paritaprevir; r, ritonavir; RBV, ribavirin.
Standardized MedDRA Queries (SMQ) search terms: acute renal failure (SMQ20000003) and CKD (SMQ20000213).
Company MedDRA Query search term: DME acute renal failure (CMQ80000038).

CONCLUSIONS

- OBV/PTV/r + DSV ± RBV was not associated with overall changes in renal function[2]
- OBV/PTV/r + DSV ± RBV effect on eGFR was similar to that observed with placebo, regardless of BL eGFR
- BL proteinuria, BL BMI, non-black race, and a history of diabetes were BL predictors associated with an increase of ≥10 mL/min/1.73 m² eGFR at EOT among patients treated with OBV/PTV/r + DSV ± RBV
- OBV/PTV/r + DSV ± RBV was well tolerated across all patients, regardless of BL eGFR stage, with infrequent lab abnormalities and renal-associated AEs

REFERENCES

1. Kidney Disease: Improving Global Outcomes (KDIGO). *Kidney Int Suppl.* 2008;109:S1–99.
2. Jacobson IM, et al. Poster 56. HepDART, 2015.
3. Sulkowski MS, et al. Poster 58. HepDART, 2015.
4. Feld JJ, et al. *N Engl J Med.* 2014;370:1594–603.
5. Zeuzem S, et al. *N Engl J Med.* 2014;370:1604–14.

44587-161467-eGFR — AASLD 2016 — AbbVie — proof 2 (rjh) — november 8, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051
CREATED @ 100%

An Open-Label, Multicenter Study of Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir With or Without Ribavirin in US Veterans With Genotype 1 Chronic Hepatitis C Infection: Efficacy and Safety Results of TOPAZ-VA

Michael Fuchs[1], Timothy Morgan[2], Mariem Charafeddine[3], Norbert Bräu[4], Warren Schmidt[5], Michael Kozal[6], Susanna Naggie[7], Ramsey Cheung[8], Tami Pilot-Matias[3], Yao Yu[3], Kristine Richards[3], Victoria Mullally[3], Daniel Cohen[3], Alexander Monto[9], Doris Toro[10]

[1]Hunter Holmes McGuire VA Medical Center, Richmond, Virginia, United States; [2]VA Long Beach Healthcare System, Long Beach, California, United States; [3]AbbVie Inc., North Chicago, Illinois, United States; [4]James J. Peters VA Medical Center, Bronx, New York, United States; [5]Iowa City VA Healthcare System, Iowa City, Iowa, United States; [6]VA Connecticut Healthcare System and Yale University School of Medicine, New Haven, Connecticut, United States; [7]Durham VA Medical Center, Durham, North Carolina, United States; [8]VA Palo Alto Healthcare System, Palo Alto, California, United States; [9]San Francisco VA Medical Center, San Francisco, California, United States; [10]VA Caribbean Healthcare System, San Juan, Puerto Rico

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

INTRODUCTION

- Hepatitis C virus (HCV) infection is highly prevalent among US veterans receiving care through Veterans Affairs (VA) Medical Centers, and 80% are infected with HCV genotype 1 (GT1)[1,2]
- The use of IFN-based regimens in this population was limited because of unfavorable drug safety profiles, major treatment contraindications, and low success rates[3]
- Psychiatric disorders and substance use, highly prevalent among US veterans, may be a major reason for the low success rates of IFN-based regimens in this population[4]
- The introduction of highly potent new direct-acting antivirals (DAAs) with favorable safety profiles offers a great opportunity for US veterans infected with HCV
- Large studies of real-world effectiveness of DAAs in US VA populations have shown sustained virologic response (SVR) rates of greater than 90%[5,6]; however, there is a dearth of interventional prospective studies to confirm the safety and efficacy of new DAA regimens in those patients
- The 3-direct-acting antiviral regimen of ombitasvir/paritaprevir/ritonavir (paritaprevir identified by AbbVie and Enanta, co-dosed with ritonavir) and dasabuvir (3D regimen) ± ribavirin (RBV) has been associated with high SVR rates and a favorable safety profile in patients infected with HCV genotype 1 (GT1) from the general population[7–9]; however, these results have yet to be confirmed in a population with a high prevalence of psychiatric disorders and substance use, such as US veterans

Multi-Targeted 3D HCV Regimen

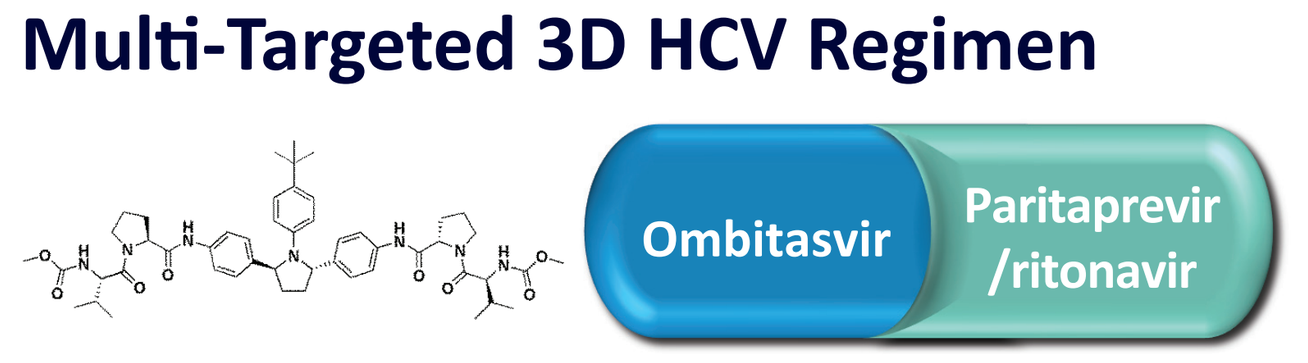
Ombitasvir
Paritaprevir /ritonavir

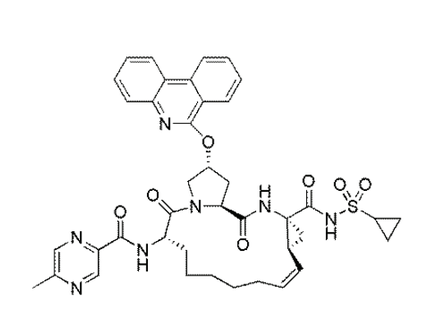

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)

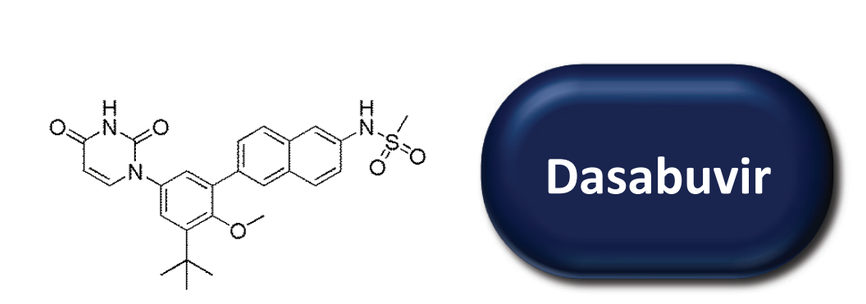
Dasabuvir

Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

PTV was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVE

- The objective of the TOPAZ-VA study was to evaluate the efficacy and safety of the 3D regimen among US veterans infected with HCV GT1, and assess the impact of ongoing psychiatric disorders and substance use on treatment parameters

DISCLOSURES

M Fuchs: Research Support from: AbbVie, Gilead, Intercept, Conatus, Galectin, Genfit.

T Morgan: Participated in AbbVie-sponsored clinical studies.

M Charafeddine, T Pilot-Matias, Y Yu, K Richards, V Mullally, D Cohen: AbbVie employees and may hold AbbVie stock.

N Bräu: Research Grants: AbbVie, Gilead, BMS; Speaker Bureau: AbbVie, Gilead, Merck; Advisory Boards: AbbVie, Gilead, BMS.

W Schmidt: Participated in AbbVie-sponsored clinical studies.

M Kozal: Yale/VACT receives Grant Support from: Merck, Pfizer, Gilead, Hologic, AbbVie, ViiV, CytoDyn, TiaMed, and BMS for studies that Dr. Kozal serves as the PI. Dr. Kozal is an employee of the federal government and does not receive any salary support from these grants.

S Naggie: Durham VA Medical Center Receives Research support from: AbbVie, Bristol-Myers Squibb, Gilead Sciences, Inc, Janssen Therapeutics, Tacere, Merck & Co, and Vertex Pharmaceuticals, Inc; Scientific Advisor: Merck & Co, Inc.

R Cheung: Participated in AbbVie-sponsored clinical studies.

A Monto: Participated in AbbVie-sponsored clinical studies.

D Toro: Participated in AbbVie-sponsored clinical studies.

METHODS

STUDY DESIGN

- TOPAZ-VA (NCT02442284) was an open-label, multicenter, Phase 3b study conducted in 11 VA centers in the US
- Eligible participants were assigned to receive the 3D regimen ± RBV for 12 or 24 weeks, based on GT1 subtype and cirrhosis status **(Figure 1)**

Figure 1. Study Design

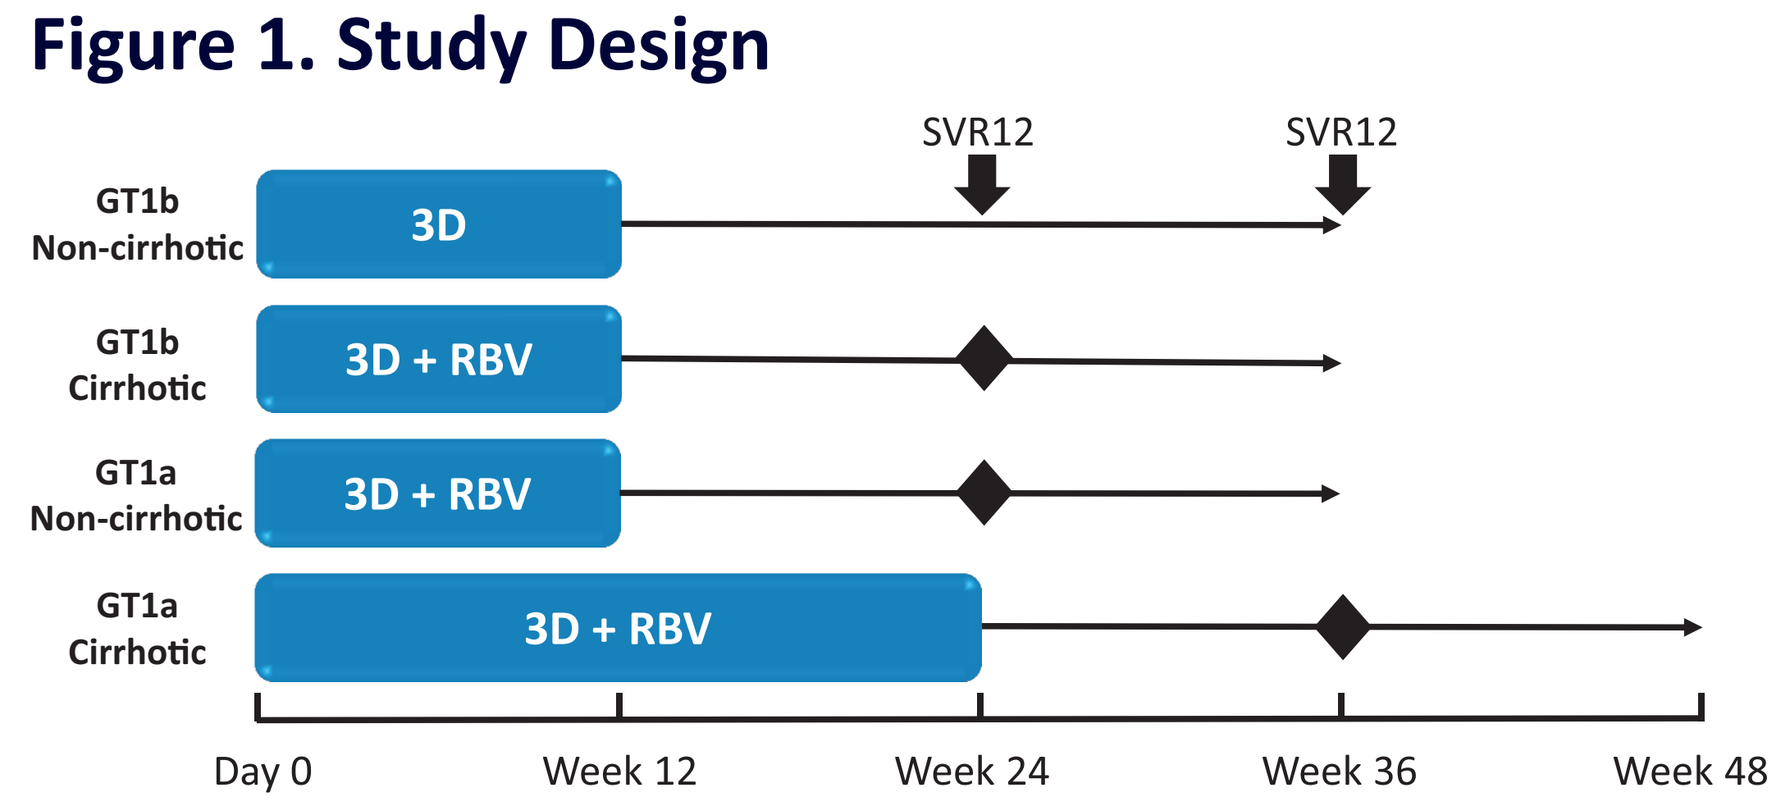
SVR12
SVR12
GT1b Non-cirrhotic
3D
GT1b Cirrhotic
3D + RBV
GT1a Non-cirrhotic
3D + RBV
GT1a Cirrhotic
3D + RBV
Day 0
Week 12
Week 24
Week 36
Week 48

3D = paritaprevir/ritonavir/ombitasvir (150 mg/100 mg/25 mg per day) + dasabuvir (250 mg twice daily) ± ribavirin (RBV) (weight-based dosing of 1000 or 1200 mg, divided twice-daily).

ENROLLMENT CRITERIA

- US military veterans, male or female, currently receiving healthcare through the Veterans Health Administration, and aged 18 years and older
- Chronic HCV GT1 infection (plasma HCV RNA > 1000 IU/mL)
- Treatment-naive or IFN, pegIFN, RBV, or sofosbuvir treatment-experienced
- No current or past clinical evidence of Child-Pugh B or C cirrhosis
- No current heavy alcohol use (more than 5 drinks on the same occasion on each of 5 or more days in the past 30 days)
- Ongoing psychiatric disorders were allowed (defined as a clinical diagnosis of bipolar disorder, depression, schizophrenia, anxiety, post-traumatic stress disorder, or other psychiatric disorder and currently requiring pharmacotherapy)

EFFICACY, ADHERENCE, AND SAFETY PARAMETERS

- The primary efficacy endpoint was sustained virologic response 12 weeks after the last administered study drug dose (SVR12)
- A secondary efficacy endpoint was the percentage of participants achieving SVR12 among those with ongoing comorbid psychiatric disorders
- Adherence was calculated as the percentage of tablets taken relative to total tablets expected to be taken; a patient was considered adherent if the percentage was between 80% and 120%
- Safety evaluations included adverse events (AEs) and clinical laboratory parameters

RESISTANCE ANALYSES

- Population sequencing (approximately 15% detection) was performed on serum samples from patients who experienced virologic failure to detect resistance-associated substitutions (RASs) in NS3, NS5A, and NS5B at baseline and after virologic failure

STATISTICAL ANALYSES

- Efficacy data were calculated with a 2-sided 95% confidence interval, using the Wilson score method
- Safety data were analyzed using descriptive statistics
- Efficacy, safety, and demographic analyses were performed on the intent-to-treat (ITT) population, defined as all enrolled participants who received at least 1 dose of the study drug
- Additional efficacy analyses were performed on a modified ITT (mITT) population, which excluded the subjects who discontinued the trial for reasons other than virologic failure

RESULTS

PARTICIPANTS

- A total of 115 patients were screened and 99 were enrolled and received the study drugs
- Baseline demographics are shown in **Table 1**
- There were 6 (6%) study discontinuations **(Figure 2)**
- There were 9 (9%) patients who discontinued study drugs: 6 (6%) due to AEs, 2 (2%) due to withdrawal of consent, and 1 (1%) due to virologic breakthrough

Figure 2. Study Flow

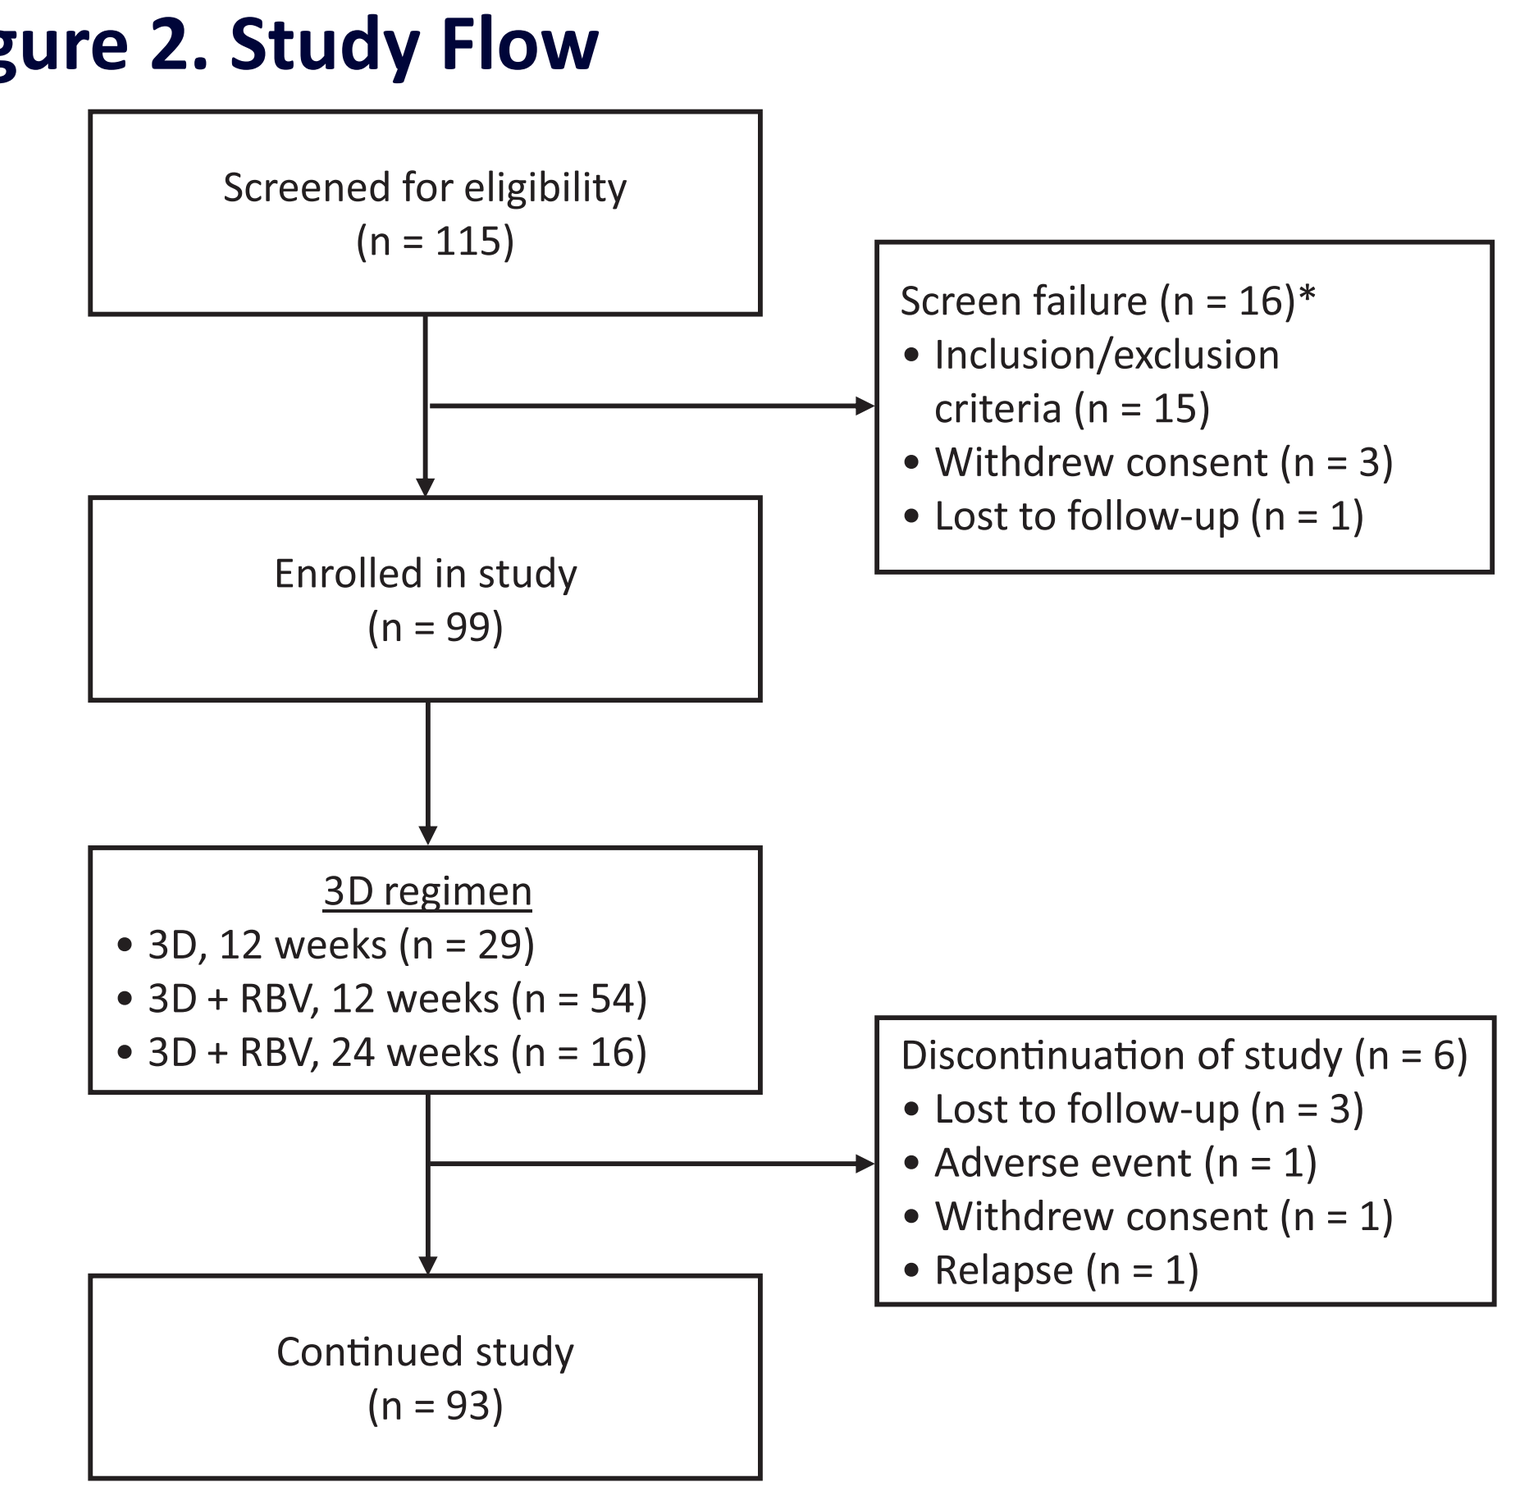
Screened for eligibility
(n = 115)
Screen failure (n = 16)*
• Inclusion/exclusion criteria (n = 15)
• Withdrew consent (n = 3)
• Lost to follow-up (n = 1)
Enrolled in study
(n = 99)
3D regimen
• 3D, 12 weeks (n = 29)
• 3D + RBV, 12 weeks (n = 54)
• 3D + RBV, 24 weeks (n = 16)
Discontinuation of study (n = 6)
• Lost to follow-up (n = 3)
• Adverse event (n = 1)
• Withdrew consent (n = 1)
• Relapse (n = 1)
Continued study
(n = 93)

*Reasons for screen failure do not add up to total due to patients failing for multiple reasons.

Table 1. Baseline Characteristics

Characteristic	3D ± RBV, N = 99
Male, n (%)	95 (96)
Race, n (%)	
White	59 (60)
Black or African American	37 (37)
Other	3 (3)
Age (years), mean ± SD	61.5 ± 5.9
BMI ≥30 kg/m², n (%)	30 (30)
Genotype, n (%)	
GT1a	67 (68)
GT1b	32 (32)
Treatment-naive, n (%)	71 (72)
Treatment-experienced, n (%)	28 (28)
HCV RNA ≥800 000 IU/mL, n (%)	81 (82)
Fibrosis stage, n (%)	
F0–F1	43 (43)
F2	14 (14)
F3	22 (22)
F4	20 (20)
History of IDU, n (%)	59 (60)
Alcohol use, n (%)	
Current	21 (21)
Former	74 (75)
Never	4 (4)
Ongoing psychiatric disorder, n (%)	48 (49)
Type of psychiatric disorder, n (%)*	
Depression	36 (75)
Bipolar disorder	2 (4)
Schizophrenia	5 (10)
Anxiety and/or post-traumatic stress disorder	31 (65)

BMI, body mass index; IDU, injection drug use.
*Percent of each type of psychiatric disorder calculated as percent of patients with ongoing psychiatric disorder; patients can have more than 1 psychiatric disorder.

TREATMENT ADHERENCE

- Treatment adherence was high, with 98–99% patients classified as adherent to each component of the 3D ± RBV regimen

VIROLOGIC RESPONSE

- The SVR12 rate was 94% (93/99) in the ITT population, and 97% (93/96) in the mITT population, which excluded all ITT patients who discontinued the trial for reasons other than virologic failure **(Figure 3)**

Figure 3. Treatment Efficacy in All Patients and Stratified by HCV Subtype and Cirrhosis Status (ITT and mITT)

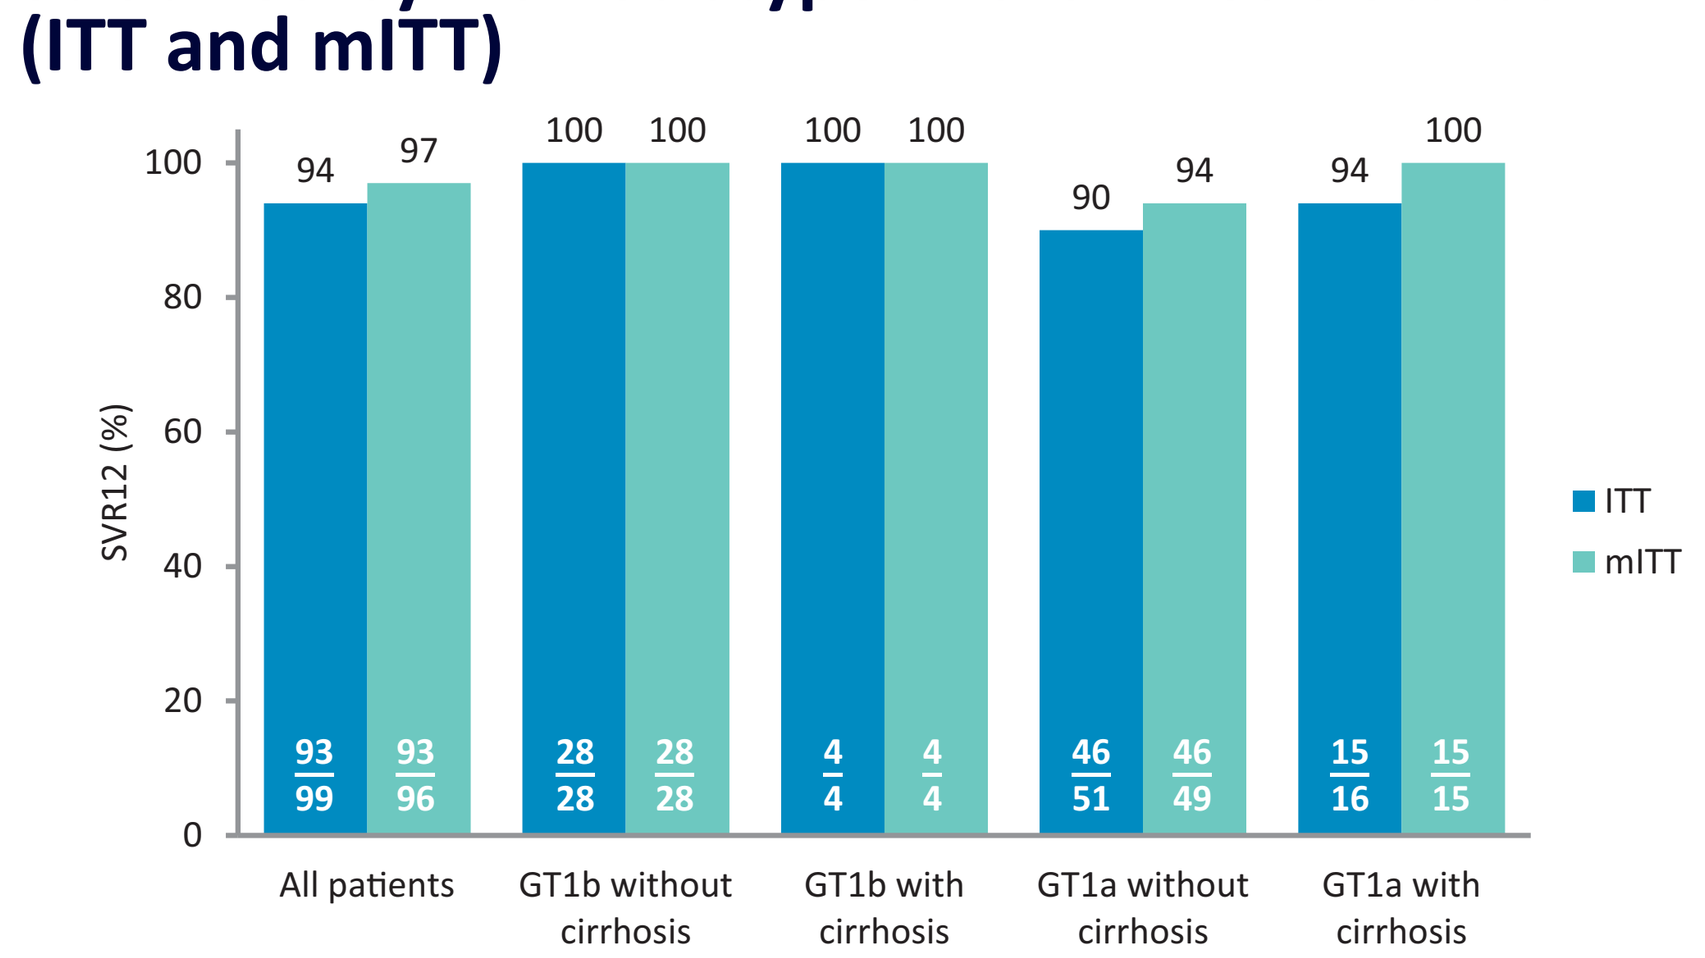
SVR12 (%)
100
80
60
40
20
0
94
97
100
100
100
100
90
94
94
100
93/99
93/96
28/28
28/28
4/4
4/4
46/51
46/49
15/16
15/15
All patients
GT1b without cirrhosis
GT1b with cirrhosis
GT1a without cirrhosis
GT1a with cirrhosis
ITT
mITT

- The 6 patients who did not achieve SVR12 included 1 on-treatment virologic failure at treatment Week 4, 2 relapses, and 3 patients who prematurely discontinued study drug
- The 3 patients who experienced virologic failure were non-cirrhotic and had GT1a infection; these 3 patients are described in more detail **(Table 2)**
 - None of the virologic failure patients required an RBV dose modification

Table 2. Characteristics of Patients Who Experienced Virologic Failure

HCV subtype	Fibrosis stage	Gender	Age	Prior treatment experience	*IL28B*	Psychiatric history	Alcohol use	IDU	Time of virologic failure
GT1a	F3	Male	60	Treatment-naive	CC	Depression/Bipolar disorder – asymptomatic (no treatment)	Former	Former	PTW12
GT1a	F3	Male	64	Treatment-experienced	TT	Schizophrenia – asymptomatic (no treatment)	Former	Former	PTW4
GT1a	F0-F1	Male	67	Treatment-naive	CT	Post-traumatic stress disorder – asymptomatic (no treatment)	Former	No	On-treatment Week 4

IDU, injection drug use; PTW, post-treatment week.

- The presence of an ongoing comorbid psychiatric disorder or history of injection drug use (IDU) did not impact SVR12 rates **(Figure 4)**

Figure 4. SVR12 in Patients With and Without Ongoing Psychiatric Disorders or History of IDU (ITT Population)

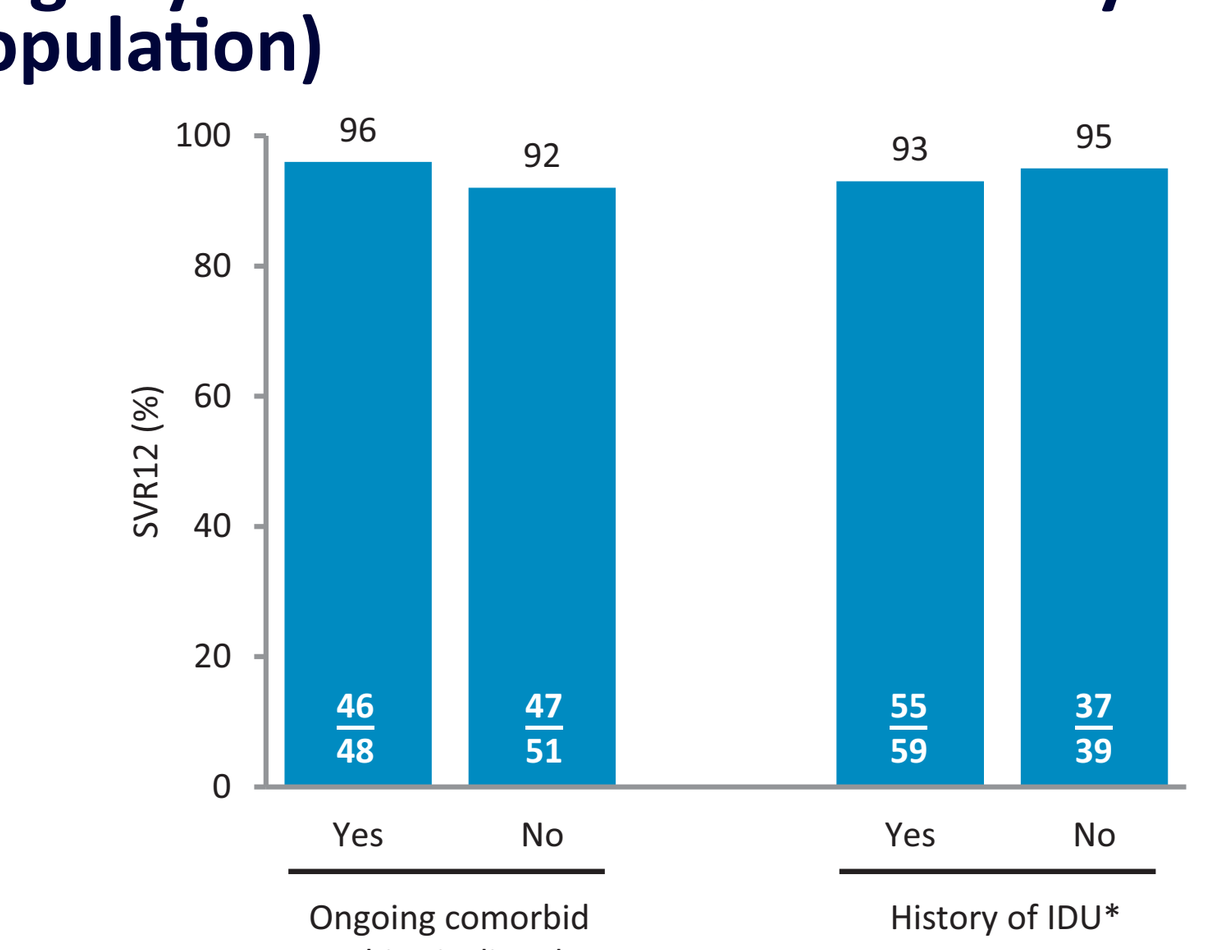
SVR12 (%)
100
80
60
40
20
0
96
92
93
95
46/48
47/51
55/59
37/39
Yes
No
Yes
No
Ongoing comorbid psychiatric disorders
History of IDU*

IDU, injection drug use.
*History of IDU was missing for 1 patient; this patient achieved SVR12.

- SVR12 rates ranged from 93–100% in subgroups stratified by baseline eGFR **(Figure 5)**

Figure 5. SVR12 in Patients by Baseline eGFR (ITT Population)*

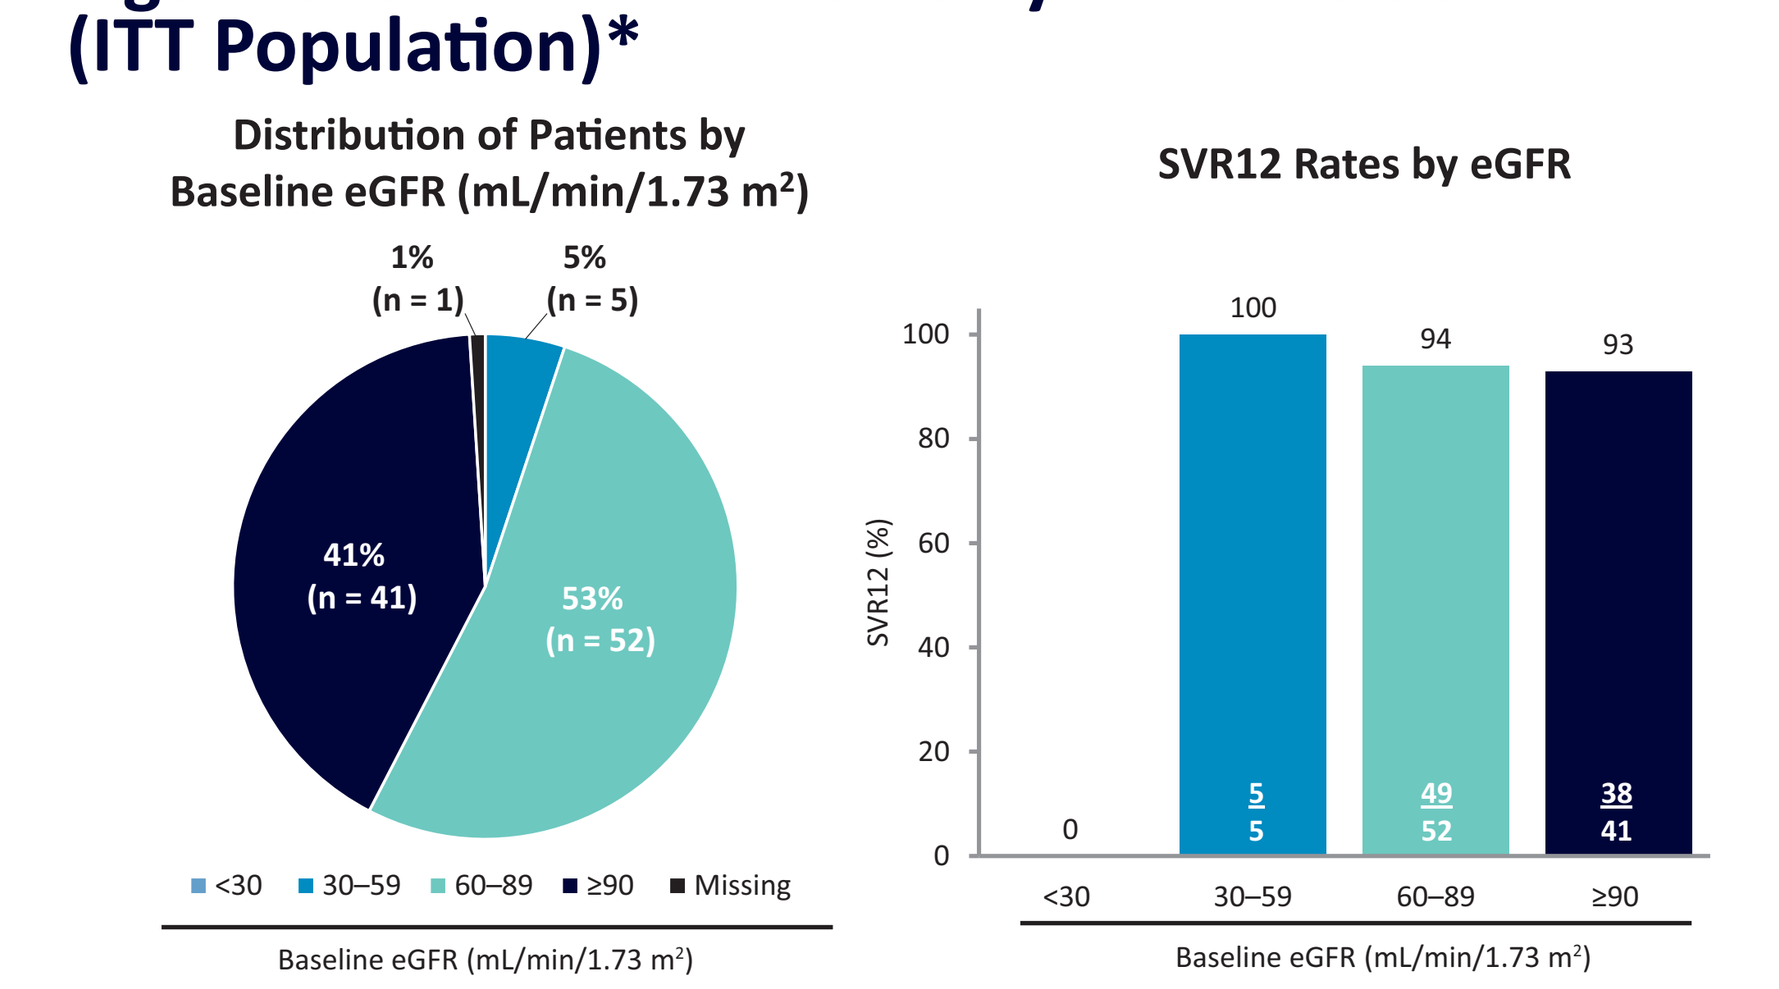
Distribution of Patients by Baseline eGFR (mL/min/1.73 m²)
1% (n = 1)
5% (n = 5)
41% (n = 41)
53% (n = 52)
<30
30–59
60–89
≥90
Missing
Baseline eGFR (mL/min/1.73 m²)
SVR12 Rates by eGFR
SVR12 (%)
100
80
60
40
20
0
0
100
94
93
5/5
49/52
38/41
<30
30–59
60–89
≥90
Baseline eGFR (mL/min/1.73 m²)

*1 patient had a missing baseline eGFR value; this patient achieved SVR12.

RESISTANCE ANALYSIS

- One of the 3 patients who experienced virologic failure had baseline NS5A polymorphisms **(Table 3)**
- At the time of failure, all 3 patients had a treatment-emergent substitution in at least 1 of the targets **(Table 3)**

Table 3. Resistance Profile of Patients Who Experienced Virologic Failure

HCV subtype	Fibrosis stage	Prior treatment experience	Virologic failure	NS3 substitutions		NS5A substitutions		NS5B substitutions	
				At baseline	At VF	At baseline	At VF	At baseline	At VF
GT1a	F3	Treatment-naive	Relapse PTW12	None	D168H	Q30H, Y93H	Q30H, Y93H	None	None
GT1a	F3	Treatment-experienced	Relapse PTW4	None	D168Y	None	M28T	None	None
GT1a	F0–F1	Treatment-naive	Breakthrough at Week 4	None	Y56H + D168V	None	M28T	None	S556G

PTW, post-treatment week; VF, virologic failure.

SAFETY

- A total of 79 patients (80%) experienced an AE **(Table 4)**; 43% were assessed as being possibly related to DAAs, and 46% as possibly related to RBV
- The majority of AEs were mild or moderate in severity
- The most common AEs were similar to those previously reported with the 3D regimen and RBV **(Table 4)**
- Ten serious AEs (SAEs) were reported in 7 patients, only 1 of which was considered possibly related to DAA therapy (mental state change due to drug interaction between DAAs and the antipsychotic quetiapine)
- Of patients receiving RBV, 19% (13/70) experienced AEs leading to RBV dose modification, of which 38% (5/13) were due to hemoglobin decrease **(Table 4)**

Table 4. Summary of Adverse Events

AEs	3D ± RBV, N = 99
Any AE, n (%)	78 (79)
SAE*, n (%)	7 (7)
AE leading to discontinuation of study drug, n (%)	6 (6)
AE leading to RBV dose modification, n/N (%)	13/70 (19)
AEs in ≥10% of patients, n (%)	
Fatigue	28 (28)
Headache	20 (20)
Nausea	15 (15)
Insomnia	10 (10)
Pruritus	10 (10)

*SAEs were myocardial infarction, pancreatitis, cholangitis and cholecystitis due to gallstones, viral gastroenteritis, appendicitis, full mouth teeth extraction due to periodontal disease, chronic obstructive pulmonary disease exacerbation, pneumonia, exacerbation of depressive symptoms, drug interaction causing altered mental state, and schizophrenia.

- Grade 3 laboratory abnormalities were rare **(Table 5)** and there were no Grade 4 laboratory abnormalities observed
- The most common laboratory abnormality was reduced hemoglobin level (n = 38), all cases of which were Grade 2 or less **(Table 5)**
- There were no clinically significant transaminase elevations or decreases in creatinine clearance (data not shown)

Table 5. Summary of Clinical Laboratory Parameters

Laboratory parameter	n/N* (%)
Hemoglobin (g/L)	
Grade 2 (<10.0–8.0 g/dL)	5/98 (5)
Grade 3 (<8.0 g/dL)	0
ALT†	
Grade 2 (>3.0–5.0 × ULN)	0
Grade 3 (>5.0–20.0 × ULN)	0
AST†	
Grade 2 (>3.0–5.0 × ULN)	1/98 (1)
Grade 3 (>5.0–20.0 × ULN)	0
Total bilirubin (μmol/L)	
Grade 2 (>1.5–3.0 × ULN)	10/98 (10)
Grade 3 (>3.0–10.0 × ULN)	1/98 (1)

ALT, alanine aminotransferase; AST, aspartate aminotransferase; ULN, upper limit of normal.
*The number of patients with post-baseline values for the respective parameter;
†Number of post-nadir Grade 2 or 3 elevations.

CONCLUSIONS

- In HCV GT1-infected US veterans, 12- or 24-week treatment with 3D ± RBV resulted in an overall ITT SVR12 rate of 94%, which is consistent with rates observed in real-world observational studies[5,6]
- There were no treatment failures among patients with GT1b infection
- The presence of ongoing psychiatric disorders or history of IDU had no impact on treatment efficacy
- In this population with a high rate of comorbidities, no new safety signals were observed; 1 patient experienced an SAE of mental status change considered to have a reasonable possibility of being related to DAAs due to drug-drug interaction with quetiapine

REFERENCES

1. Dominitz JA, et al. *Hepatology.* 2005;41(1):88–96.
2. Beste LA, et al. *Epidemiol Rev.* 2015;37:131–43.
3. Cawthorne CH, et al. *Am J Gastroenterol.* 2002;97(1):149–55.
4. Rosen CS, et al. *Psychiatr Serv.* 2008;59(3):290–3.
5. Ioannou GN, et al. *Gastroenterology.* 2016;151(3):457–71.
6. McCombs J, et al. *J Hepatol.* 2016;64(Suppl 2):S217.
7. Feld JJ, et al. *N Engl J Med.* 2014;370(17):1594–603.
8. Poordad F, et al. *N Engl J Med.* 2014;370(21):1973–82.
9. Ferenci P, et al. *N Engl J Med.* 2014;370(21):1983–92.

ACKNOWLEDGEMENTS

AbbVie sponsored the study, contributed to the design, participated in the collection, analysis, and interpretation of the data, and in the writing, reviewing, and approval of the publication. Medical writing support was provided by Scott Battle of Medical Expressions, funded by AbbVie.

abbvie

44587-161467-TOPAZ-VA — AASLD 2016 — AbbVie — proof 5 (rjh) — november 8, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051

CREATED @ 100%

935

RUBY-II: Efficacy and Safety of a Ribavirin-Free Ombitasvir/Paritaprevir/Ritonavir ± Dasabuvir Regimen in Patients With Severe Renal Impairment or End-Stage Renal Disease and HCV Genotype 1a or 4 Infection

Edward Gane[1], Ricard Sola[2], Eric Cohen[3], Stuart K Roberts[4], Jacob George[5], Richard Skoien[6], Stephen Riordan[7], Niloufar Mobashery[3], Manal Abunimeh[3], Daniel E Cohen[3], Kosh Agarwal[8]

[1]Auckland City Hospital, Auckland, New Zealand; [2]Hospital del Mar. IMIM, Universitat Autónoma de Barcelona, Barcelona, Spain; [3]AbbVie Inc., North Chicago, Illinois, United States; [4]Alfred Hospital, Melbourne, Australia; [5]Westmead Hospital, Westmead Institute for Medical Research and University of Sydney, Westmead, Australia; [6]Royal Brisbane and Women's Hospital, Brisbane, Australia; [7]University of New South Wales, Randwick, Australia; [8]Institute of Liver Studies, Kings College Hospital NHS Foundation Trust, London, United Kingdom

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

BACKGROUND

- Many direct-acting antiviral (DAA) therapies are renally metabolized or excreted (eg, sofosbuvir) or have increased exposures in patients with severe renal impairment. As a result, these therapies are not recommended for patients with an estimated glomerular filtration rate of <30 mL/min/1.73 m^2 [1–4]
- By contrast, ombitasvir (OBV), paritaprevir (PTV), ritonavir (r), and dasabuvir (DSV) are all hepatically metabolized and, therefore, require no dose adjustment in patients with any degree of renal impairment[5]
- OBV/PTV/r ± DSV–containing regimens have been shown to retain efficacy regardless of baseline resistance-associated polymorphisms,[6] unlike the combination of grazoprevir and elbasvir, which often requires resistance profiling before treatment[7]
- In the phase 3b RUBY-I study, patients with stage 4 or 5 chronic kidney disease (CKD) who received this 3-DAA regimen had OBV, PTV, r, and DSV exposures comparable to those observed in patients with healthy renal function, and 18/19 (95%) patients that completed treatment and follow-up achieved sustained virologic response at post-treatment Week 12 (SVR12)[6]
- Although dosing recommendations for RBV in renal insufficiency have been published,[8] RBV-free hepatitis C virus (HCV) treatment regimens are strongly desired for patients with end-stage renal disease (ESRD) to obviate the risk for hematologic toxicity associated with RBV coadministration
- Prior phase 3 trial of OBV/PTV/r + DSV suggested that GT1a-infected patients outside the US may have a higher SVR rate when treated without RBV,[9] possibly related to the lower prevalence of the Q80K polymorphism outside the US.[10] In addition, hemodialysis has been associated with reduction of plasma HCV RNA,[11] suggesting these patients might be treatable without RBV coadministration

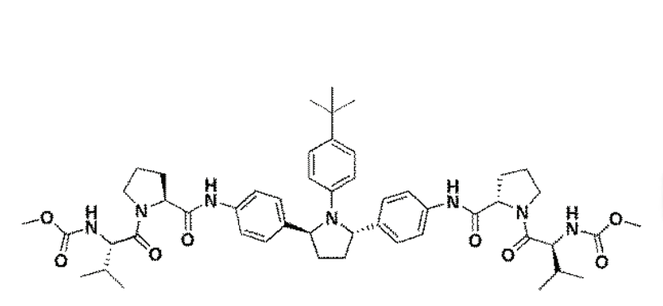

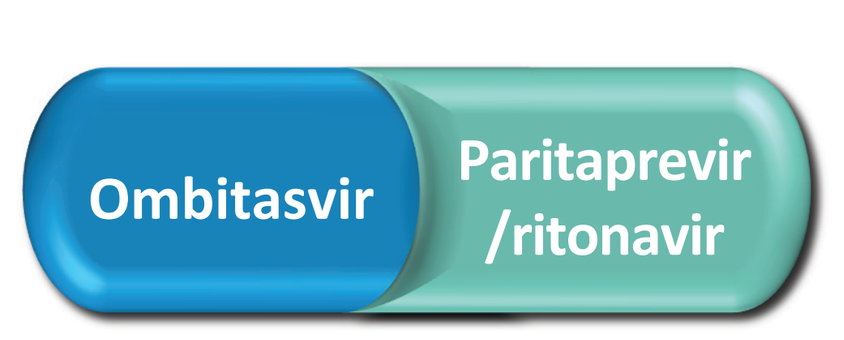
Ombitasvir
Paritaprevir /ritonavir

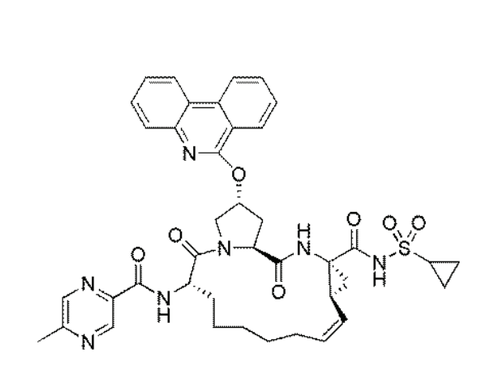

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)

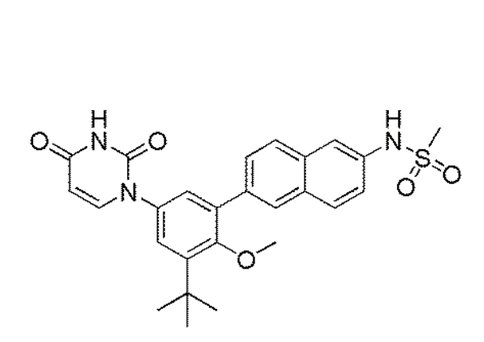

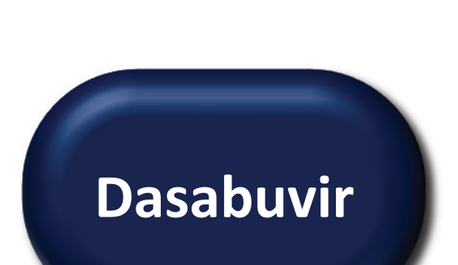
Dasabuvir

Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

Paritaprevir identified by AbbVie and Enanta.
Ritonavir has no activity against HCV.

OBJECTIVE

- To examine the efficacy and safety of the RBV-free regimen of OBV/PTV/r ± DSV in patients with stage 4 or 5 CKD (receiving dialysis) with chronic HCV GT1a or 4 infection

METHODS

ELIGIBILITY CRITERIA

Inclusion

- Age ≥18 years
- Chronic infection with HCV GT1a or GT4
- Stage 4 or 5 CKD, including hemodialysis or peritoneal dialysis
- Absence of cirrhosis (METAVIR score ≤3 or equivalent)
- No prior HCV treatment experience

Exclusion

- Co-infection with hepatitis B or human immunodeficiency virus
- Current or past clinical evidence of cirrhosis
- Albumin <3.5 g/dL, hemoglobin <8 g/dL, platelets <120 000 cells per mm^3, total bilirubin ≥3 mg/dL, or international normalized ratio >2.3

Treatment

- Patients with GT1a infection received the RBV-free regimen of OBV/PTV/r (25/150/100 mg once daily) + DSV (250 mg twice daily) for 12 weeks **(Figure 1)**
- Patients with GT4 infection received the RBV-free regimen of OBV/PTV/r (25/150/100 mg once daily) for 12 weeks
- Efficacy was assessed as SVR12, defined as plasma HCV RNA level <25 IU/mL at 12 weeks post-treatment

Figure 1. RUBY-II: Phase 3b, Open-label, Multicenter Study Design

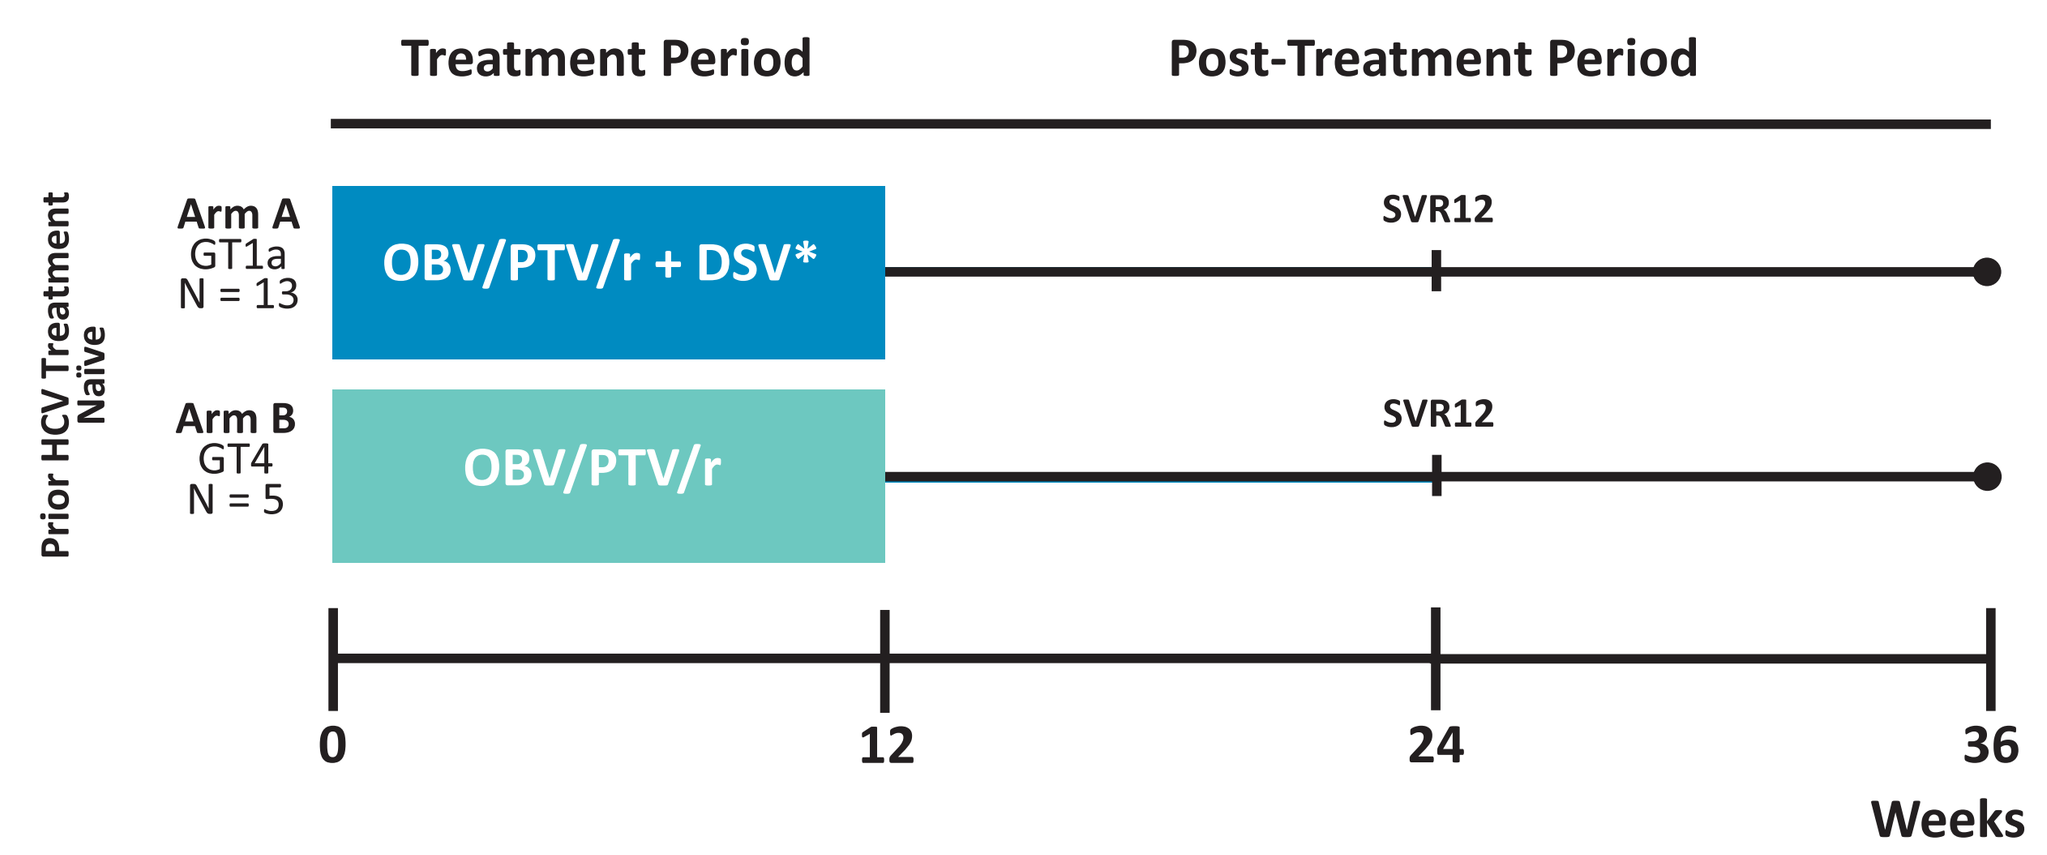
Treatment Period
Post-Treatment Period
Prior HCV Treatment Naïve
Arm A GT1a N = 13
OBV/PTV/r + DSV*
SVR12
Arm B GT4 N = 5
OBV/PTV/r
SVR12
0
12
24
36
Weeks

DSV, dasabuvir; GT, genotype; HCV, hepatitis C virus; OBV, ombitsavir; p, paritaprevir; r, ritonavir; SVR, sustained virologic response.
*No RBV coadministration in this study, despite USPI/SmPC label recommendation for RBV coadministration in patients with HCV GT1a.

- SVR12 was analyzed in all patients who received at least 1 dose of the study drugs (intent-to-treat [ITT] population); a modified ITT (mITT) population in which non-virologic failures were excluded was also evaluated
- Adverse events (AEs) and laboratory abnormalities were assessed in all patients who received at least 1 dose of study drugs
- A total of 18 patients (13 with GT1a and 5 with GT4 infection) were enrolled at 8 study sites in Australia, New Zealand, Spain, and the United Kingdom
- Baseline demographics are shown in **Table 1**

RESULTS

Table 1. Baseline Patient Demographics

Characteristic	HCV GT1a OBV/PTV/r + DSV 12 Weeks N = 13	HCV GT4 OBV/PTV/r 12 Weeks N = 5
Male, n (%)	9 (69)	3 (60)
White race, n (%)	8 (62)	3 (60)
Age, median (range) years	57 (34–76)	58 (31–67)
BMI, median (range) kg/m^2	26.4 (20.1–31.8)	28.4 (18.7–40.9)
IL28B non-CC genotype, n (%)	7 (54)	5 (100)
HCV RNA, median (range) $\log_{10}$ IU/mL	5.8 (4.6–7.3)	5.7 (4.6–6.2)
CKD stage		
4	0	1 (20)
5	13 (100)	4 (80)
Baseline dialysis		
Peritoneal dialysis	5 (39)	0
Hemodialysis	8 (62)	4 (80)*
eGFR, mean ± SD mL/min/1.73 m^2	6.7 ± 3.0	10.1 ± 4.6
Fibrosis stage, n (%)		
F0–F1	8 (62)	3 (60)
F2	1 (8)	1 (20)
F3	4 (31)	1 (20)

BMI, body-mass index; CKD, chronic kidney disease; DSV, dasabuvir; eGFR, estimated glomerular filtration rate; GT, genotype; HCV, hepatitis C virus; *IL28B*, interleukin 28B; OBV, ombitsavir; PTV, paritaprevir; r, ritonavir.
*1 patient not on baseline dialysis.

- At baseline, 71% (12/17) of patients were receiving hemodialysis and 29% (5/17) were receiving peritoneal dialysis (data were missing for 1 subject)
- Baseline HCV RNA levels were ≥800 000 IU/mL (≥5.9 $\log_{10}$ IU/mL) in 8 (44%) patients

Figure 2. Sustained Virologic Response at Post-treatment Week 12

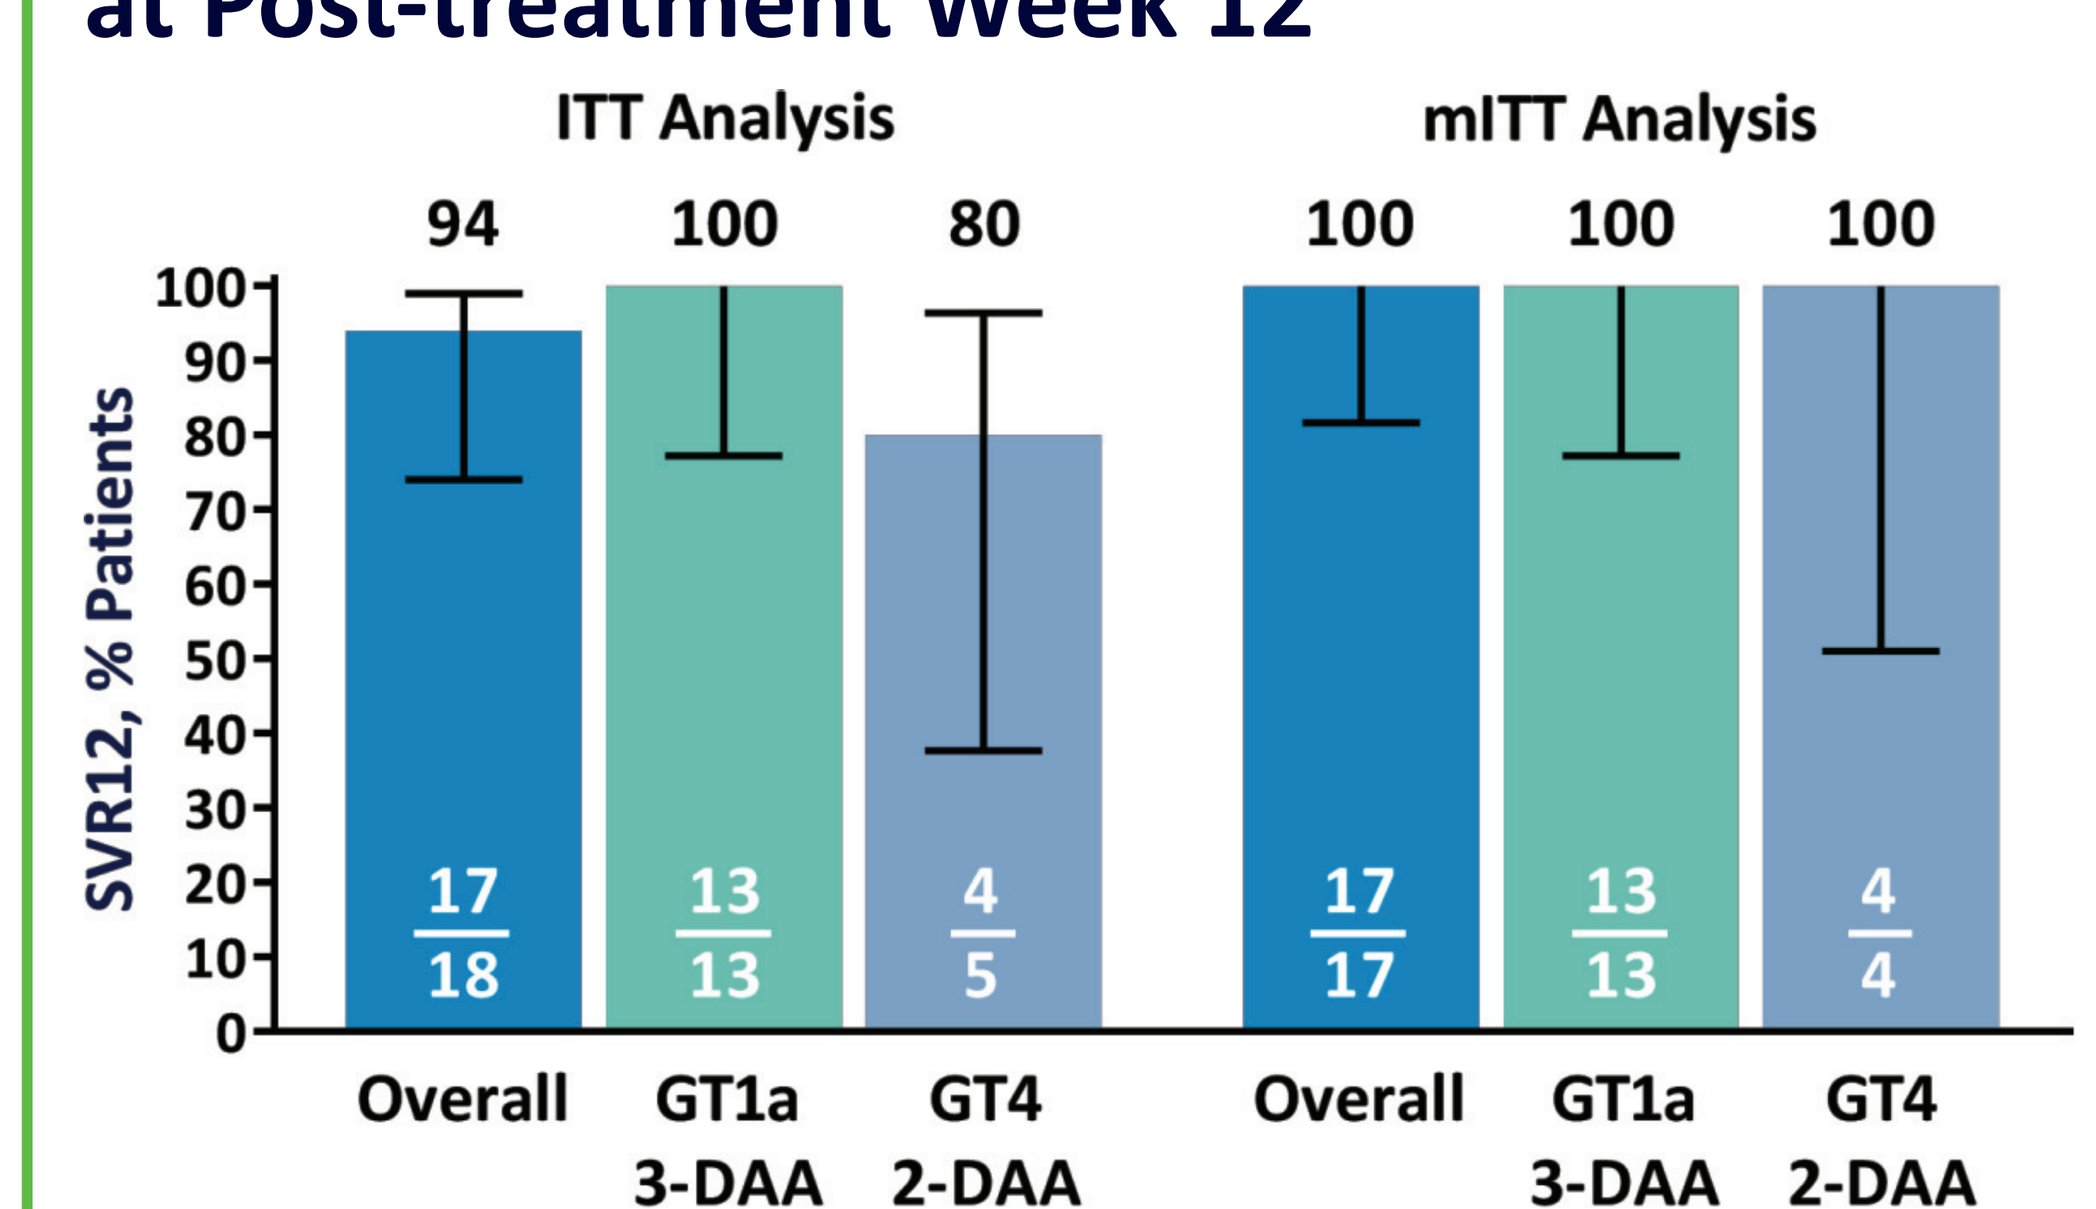
ITT Analysis
mITT Analysis
94
100
80
100
100
100
SVR12, % Patients
100
90
80
70
60
50
40
30
20
10
0
17/18
13/13
4/5
17/17
13/13
4/4
Overall
GT1a 3-DAA
GT4 2-DAA
Overall
GT1a 3-DAA
GT4 2-DAA

DAA, direct-acting antiviral; GT, genotype; ITT, intent-to-treat population; mITT, modified intent-to-treat population; SVR12, sustained virologic response at post-treatment Week 12.
One HCV GT4-infected patient underwent elective renal transplantation and withdrew consent at treatment Week 2 and did not achieve SVR12.

EFFICACY

- The overall ITT SVR12 was 94% (17/18; 95% CI 74–99) for patients receiving the RBV-free 2-DAA or 3-DAA regimens, including 100% (13/13; 95% CI 77–100) SVR12 in patients with HCV GT1a infection
- The overall mITT SVR12 was 100% (17/17)
 - No patient had virologic failure
 - One patient with HCV GT4 infection discontinued study drug at treatment Week 2 to undergo an elective renal transplantation and did not achieve SVR12

SAFETY

- The majority of AEs were mild to moderate in severity **(Table 2)**

Table 2. Adverse Events and Laboratory Abnormalities

Event, n (%)	HCV GT1a OBV/PTV/r + DSV 12 Weeks N = 13	HCV GT4 OBV/PTV/r 12 Weeks N = 5
Any AE	13 (100)	5 (100)
Serious AE	3 (23)	1 (20)
AE leading to study drug discontinuation	1 (8)*	1 (20)†
AEs occurring in ≥15% of all patients		
Abdominal pain	4 (31)	0
Fatigue	3 (23)	1 (20)
Diarrhea	4 (31)	0
Headache	3 (23)	0
Hypertension	3 (23)	1 (20)
Nausea	4 (31)	0
Pruritus	2 (15)	1 (20)
Hemoglobin		
Grade 2 (<10.0–8.0 g/dL)	4 (31)	2 (40)
Grade 3 (<8.0–6.5 g/dL)	0	0
Alanine aminotransferase		
Grade 2 (>3–5 × ULN)	0	0
Grade 3 (>5–20 × ULN)	1 (8)	1 (20)
Total bilirubin, Grade ≥2 (>1.5 × ULN)	0	0

AE, adverse event; DSV, dasabuvir; eGFR, estimated glomerular filtration rate; GT, genotype; HCV, hepatitis C virus; OBV, ombitsavir; PTV, paritaprevir; r, ritonavir; SVR, sustained virologic response; ULN, upper limit of normal.
*Discontinued study drug, but still achieved SVR12.
†Discontinued study drug due to renal failure and transplantation.

- Serious AEs occurred in 4 patients; these events (elective renal transplant, worsening hypertension, gastroenteritis, and pulmonary edema) were assessed as not related to study drugs by the investigator
- Two patients prematurely discontinued study drugs
 - One HCV GT1a-infected patient discontinued study drug on treatment Day 77 due to elevated alanine aminotransferase (grade 3: >5 × upper limit of normal and ≥2 × baseline); patient still achieved SVR12
 - One HCV GT4-infected patient underwent elective renal transplantation, withdrew consent at treatment Week 2, and did not achieve SVR12

CONCLUSIONS

- In this study of non-cirrhotic, treatment-naïve patients with stage 4 or 5 CKD, including those receiving dialysis, the RBV-free regimen of OBV/PTV/r ± DSV resulted in an overall ITT SVR12 rate of 94% for GT1a and 4 (100% in GT1a) and an mITT SVR12 rate of 100%, with no on-treatment virologic failure or relapse
- The RBV-free 2-DAA and 3-DAA regimens were generally well tolerated in this patient population. Most AEs were mild to moderate in severity, and there were no serious AEs deemed related to study drugs
- These data suggest that RBV may not be necessary in some GT1a- or GT4-infected patients with severe renal impairment treated with OBV/PTV/r ± DSV; however, larger trials are needed to confirm the results of this exploratory study

REFERENCES

1. Perz JF, Armstrong GL, Farrington LA, et al. The contributions of hepatitis B virus and hepatitis C virus infections to cirrhosis and primary liver cancer worldwide. *J Hepatol.* 2006;45:529–38.
2. Webster DP, Klenerman P, Dusheiko GM. Hepatitis C. *Lancet.* 2015;385:1124–35.
3. Bansal S, Singal AK, McGuire BM, et al. Impact of all oral anti-hepatitis C virus therapy: A meta-analysis. *World J Hepatol.* 2015;7:806–13.
4. Zeuzem S, Ghalib R, Reddy KR, et al. Grazoprevir-Elbasvir Combination Therapy for Treatment-Naive Cirrhotic and Noncirrhotic Patients With Chronic Hepatitis C Virus Genotype 1, 4, or 6 Infection: A Randomized Trial. *Ann Intern Med.* 2015;163:1–13.
5. Khatri A, Dutta S, Marbury T, et al. The pharmacokinetics and safety of the direct acting antiviral regimen of ABT-450/r, ombitasvir with/without dasabuvir in subjects with mild, moderate and severe renal impairment compared to subjects with normal renal function. *Hepatology.* 2014;60:320A.
6. Pockros PJ, Reddy KR, Mantry PS, et al. Efficacy of Direct-Acting Antiviral Combination for Patients With Hepatitis C Virus Genotype 1 Infection and Severe Renal Impairment or End-Stage Renal Disease. *Gastroenterology.* 2016;150:1590–8.
7. Zepatier (elbasvir and grazoprevir) tablets, for oral use [package insert]. Whitehouse Station, NJ: Merck & Co; 2016.
8. Rebetol (ribavirin USP) capsules, for oral use and oral solution [highlights of prescribing information]. Whitehouse Station, NJ: Merck & Co; 2016.
9. Ferenci P, Bernstein D, Lalezari J, et al. ABT-450/r–Ombitasvir and Dasabuvir with or without Ribavirin for HCV. *N Engl J Med.* 2014;370:1983–92.
10. Sarrazin C, Lathouwers E, Peeters M, et al. Prevalence of the hepatitis C virus NS3 polymorphism Q80K in genotype 1 patients in European region. *Antiviral Res*. 2015;116:10–6.
11. Fabrizi F, Bunnapradist S, Lunghi G, et al. Kinetics of hepatitis C virus load during hemodialysis: novel perspectives. *J Nephrol*. 2003;16(4):467–75.

ACKNOWLEDGEMENTS

Medical writing was provided by Ryan J Bourgo, PhD, and Deborah M Eng, MS, ELS, of AbbVie.

DISCLOSURES

AbbVie sponsored the study (NCT02487199), contributed to its design, the collection, analysis, and interpretation of the data, and participated in the writing, review, and approval of the poster. All authors had access to relevant data. **E Cohen, M Abunimeh, N Mobashery, and DE Cohen:** Employees of AbbVie and may hold stock or stock options. **E Gane:** Advisor: AbbVie, Gilead, Achillion, Novartis, Roche, Merck, Janssen. **R Sola Lamoglia:** Consultant: Roche, Bristol-Meyers Squibb, Roche/Genentech, Janssen Cilag, AbbVie; Lecturer: Bristol-Meyers Squibb, Novartis, Roche/Genentech, Janssen, AbbVie. **SK Roberts:** Advisor: AbbVie, Gilead, MSD, Janssen, Bristol-Meyers Squibb, Roche. **J George:** Advisory boards: AbbVie, Merck, Roche. **R Skoien:** Consultant: AbbVie, Bayer Australia Ltd, MSD, Roche, Janssen-Cilag Pty Ltd. **S Riordan:** Nothing to disclose. **K Agarwal:** Speaker bureau/consultancy: Achilion, Arbutus, BMS, Gilead, GSK, Intercept, Janssen, Merck, Novartis, Roche; Research support: AbbVie, BMS, Gilead, Roche.

44577-161416-935 — AASLD 2016 — AbbVie — proof 4 (rjh) — november 1, 2016
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051
CREATED @ 100%

Preliminary Results From TOPAZ-III: A Phase 3b Study Evaluating the Efficacy and Safety of Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir ± Ribavirin in Naive or Experienced Adults in Brazil With HCV Genotype 1 Infection and Advanced Fibrosis/Cirrhosis

MG Pessoa[1], JV Madruga[2], K Alves[3], E Nunes[4], H Cheinquer[5], CE Brandão-Mello[6], MC Mendes-Correa[7], ML Ferraz[8], SA Lari[3], L Liu[3], R Tripathi[3], T Pilot-Matias[3], DE Cohen[3], NS Shulman[3], AL Martinelli[9]

[1]Department of Gastroenterology, University of São Paulo School of Medicine, São Paulo, Brazil; [2]Centro de Referência e Treinamento DST/AIDS, São Paulo, Brazil, [3]AbbVie Inc., North Chicago, Illinois, United States; [4]National Institute of Infectious Diseases/FIOCRUZ, Brazil; [5]Universidade Federal de Rio Grande de Sul, Gastroenterology and Hepatology Unit, Hospital de Clinicas de Porto Alegre, Brazil; [6]Internal Medicine Department, College of Medicine & Surgery, Universidade Federal do Estado do Rio de Janeiro – UNIRIO, Rio de Janeiro, Brazil; [7]Department of Infectious Diseases, São Paulo University Medical School, University of São Paulo, Brazil; [8]Department of Gastroenterology, Escola Paulista de Medicina, UNIFESP, São Paulo, Brazil; [9]Department of Medicine, Gastroenterology Division, Ribeirao Preto School of Medicine, University of São Paulo, Brazil

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

INTRODUCTION

- In the Brazilian population, the prevalence of chronic hepatitis C virus (HCV) infection is approximately 1.5%, with genotype (GT) 1 being the most common across all regions[1,2]
- Brazil's universal healthcare system offers free treatment for citizens with chronic hepatitis C including individuals with HIV coinfection, renal insufficiency, idiopathic thrombocytopenic purpura, cryoglobulinemia, hematologic cancers, and fibrosis (METAVIR stages F3 or above or greater than 3 years METAVIR stage F2)[3,4]
- Phase 3 trials of AbbVie's all-oral, direct-acting antiviral (3-DAA) regimen of ombitasvir, ritonavir-enhanced paritaprevir (paritaprevir identified by AbbVie and Enanta), and dasabuvir (OBV/PTV/r + DSV), with or without weight-based ribavirin (RBV), have demonstrated high rates of sustained virologic response at post-treatment Week 12 (SVR12) in GT1-infected patients with or without compensated cirrhosis[5–10]

Multi-Targeted 3-DAA HCV Regimen

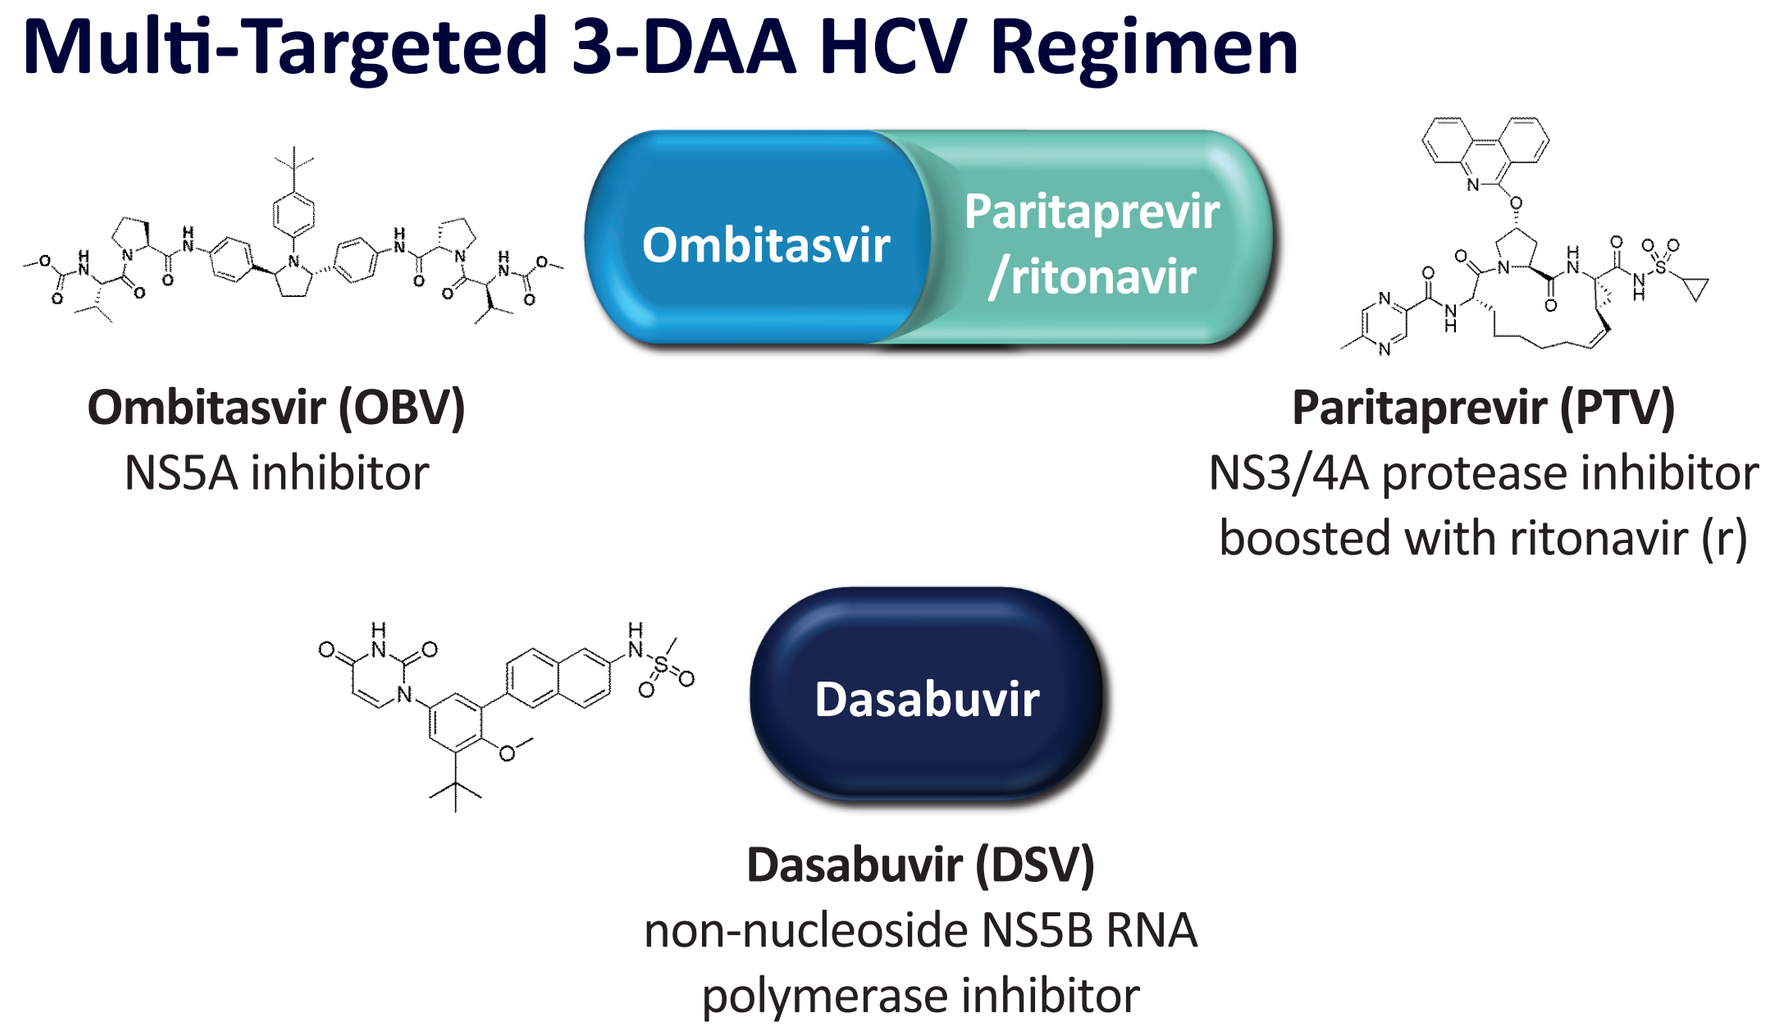
Ombitasvir
Paritaprevir /ritonavir
Dasabuvir
Ombitasvir (OBV)
NS5A inhibitor
Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)
Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

Paritaprevir (PTV) was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVES

- To assess the efficacy and safety of 3-DAA ± RBV in GT1-infected, treatment-naive, or interferon (IFN)-experienced Brazilian patients with advanced bridging fibrosis or compensated cirrhosis (METAVIR F3–F4) **(Figure 1)**

Figure 1. TOPAZ-III Study Schematic

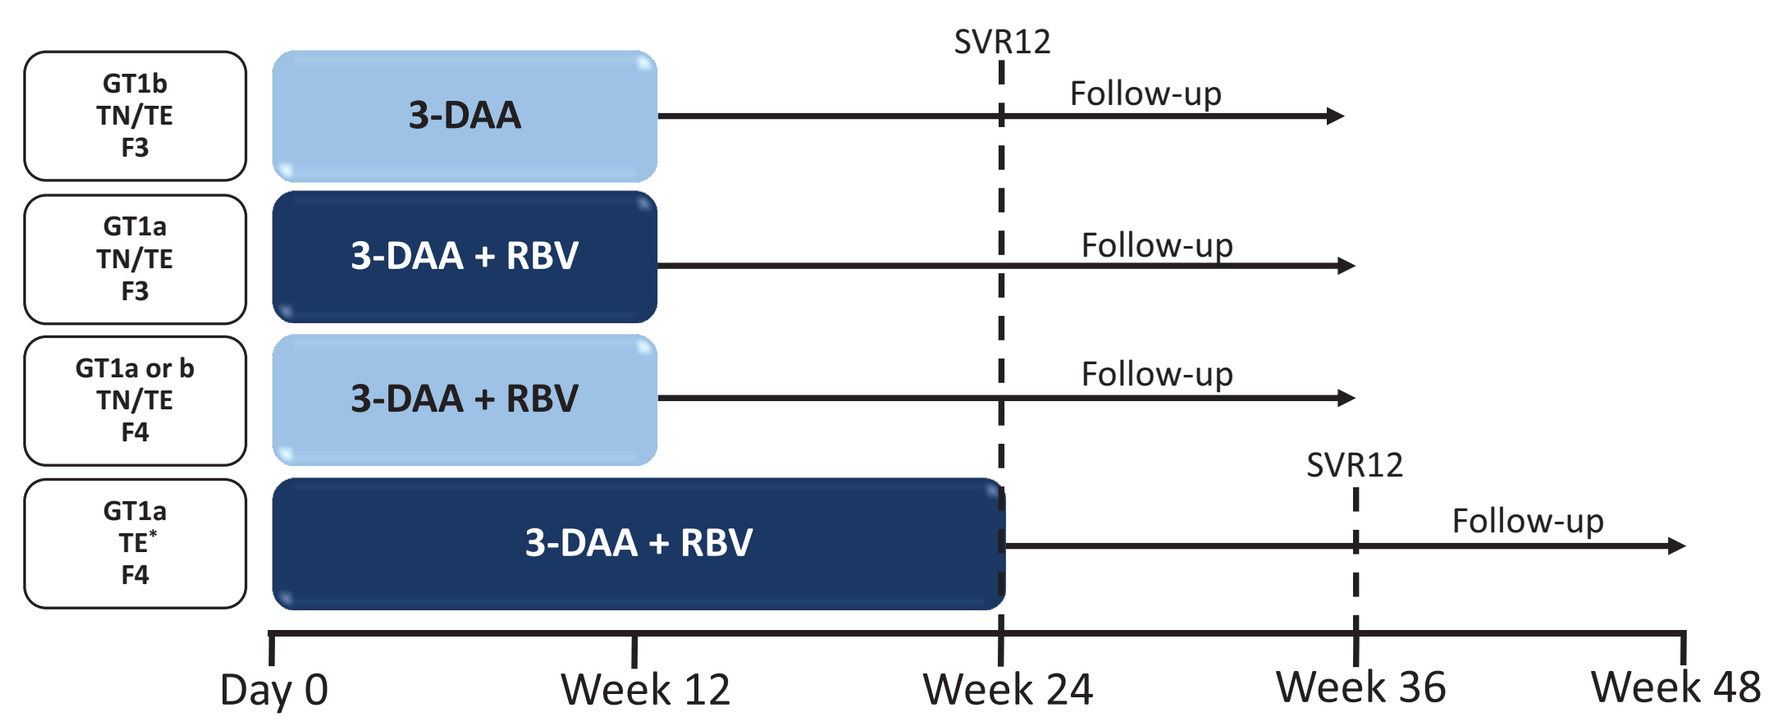
GT1b TN/TE F3
3-DAA
GT1a TN/TE F3
3-DAA + RBV
GT1a or b TN/TE F4
3-DAA + RBV
GT1a TE* F4
3-DAA + RBV
SVR12
Follow-up
Follow-up
Follow-up
SVR12
Follow-up
Day 0
Week 12
Week 24
Week 36
Week 48

*F4, GT1a-infected participants and prior IFN/RBV or pegIFN/RBV null responders or nonresponders received 24 weeks of treatment, all other F4, GT1-infected participants with cirrhosis received 12 weeks of treatment.
3-DAA: OBV/PTV/r (25/150/100 mg daily) + DSV (250 mg twice daily) ± RBV (weight-based); patients with an unknown GT1 subtype or with mixed GT1 infection were treated as GT1a-infected; TN, treatment-naive; TE, treatment-experienced (IFN/RBV or pegIFN/RBV experienced patients).

ACKNOWLEDGMENTS

Medical writing support was provided by Alexis Cogswell of Medical Expressions, funded by AbbVie.

METHODS

STUDY DESIGN

- Phase 3, non-randomized, open-label, multicenter trial
- All participants received co-formulated, once-daily OBV/PTV/r (25/150/100 mg daily) + DSV (250 mg twice daily) ± RBV (1000 mg daily for body weight <75 kg or 1200 mg daily for body weight ≥75 kg) for 12 or 24 weeks according to genotype and METAVIR fibrosis staging **(Figure 1)**
- Post-treatment follow-up was 24 weeks

STUDY POPULATION

- HCV genotype 1-infected adult patients with METAVIR F3 or F4 fibrosis staging, who are treatment naive or previously treated with an IFN-based regimen

KEY ELIGIBILITY CRITERIA

- Fibrosis stage F3 or greater, documented by 1 of the following
 - Liver biopsy showing bridging fibrosis (METAVIR F3, Ishak 4, or equivalent) within 24 months prior to first dose of study drug or any previous liver biopsy showing cirrhosis (METAVIR F3/4 or F4, Ishak 5 or 6, or equivalent)
 - FibroScan result of ≥9.6 kPa within 6 months prior to first dose of study drug
 - FibroTest result of 0.59 or greater at the time of screening
- HCV RNA ≥1000 IU/mL at the time of Screening
- Compensated liver disease with no prior history of hepatocellular carcinoma or hepatic decompensation
- Patients with mild and moderate renal insufficiency (creatinine clearance of ≥30 mL/min) and advanced liver disease (platelets ≥25 000 cells per mm^3 and total bilirubin <3.0 mg/dL) were included

EFFICACY AND SAFETY ASSESSMENTS

- The primary efficacy endpoint was sustained virologic response (the proportion of patients with HCV RNA below lower limit of quantification [LLOQ]) at 12 weeks (SVR12) after the final dose of study drug
- Secondary efficacy endpoints were SVR12 rates by fibrosis stage, prior treatment experience (naive, or previous IFN-based regimen), and IFN eligibility (ineligible, intolerant, eligible)
- Clinical laboratory data were monitored throughout the study
- FibroScan was measured at screening, baseline, end of treatment, and post-treatment Weeks 12 and 24 to assess progression of liver stiffness
- Treatment-emergent adverse events (TEAEs) were monitored from initiation of treatment until 30-days after discontinuation of study drugs

DATA ANALYSES

- The percentage of patients achieving SVR12 was calculated and a 2-sided 95% confidence interval (CI) was computed based on the Wilson's score method
- Safety data were analyzed using descriptive statistics

RESULTS

BASELINE CHARACTERISTICS

- The study enrolled 222 individuals across 16 study centers in Brazil; 99.5% (221/222) completed treatment
- The majority of patients were white (82.4%), male (55.4%), had a BMI <30 kg/m^2 (76.1%) and <65 years of age (77.0%) **(Table 1)**
- Approximately half (54.1%) of patients received prior IFN-based therapy **(Table 1)**

Table 1. TOPAZ-III Demographic and Baseline Characteristics

Patient Characteristics, n (%)	GT1a 3-DAA + RBV 12 or 24† weeks F4 n = 64	GT1b 3-DAA + RBV 12 weeks F4 n = 69	GT1a 3-DAA + RBV 12 weeks F3 n = 46	GT1b 3-DAA 12 weeks F3 n = 43	Total n = 222
Male	35 (54.7)	35 (50.7)	28 (60.9)	25 (58.1)	123 (55.4)
Age, median (range)	56 (29–73)	61 (35–78)	54 (26–71)	60 (34–77)	57 (26–78)
White	53 (82.8)	56 (81.2)	40 (87.0)	34 (79.1)	183 (82.4)
BMI (kg/m^2)					
<30	47 (73.4)	53 (76.8)	37 (80.4)	32 (74.4)	169 (76.1)
≥30	17 (26.6)	16 (23.2)	9 (19.6)	11 (25.6)	53 (23.9)
HCV Genotype					
1a	64 (100)	0	46 (100)	0	110 (49.5)
1b	0	69 (100)	0	43 (100)	112 (50.5)
Prior HCV Treatment History					
Treatment-naive	26 (40.6)	26 (37.7)	27 (58.7)	23 (53.5)	102 (45.9)
Treatment-experienced*	38 (59.4)	43 (62.3)	19 (41.3)	20 (46.5)	120 (54.1)
Baseline Fibrosis Stage					
F3	0	0	46 (100)	43 (100)	89 (40.1)
F4	64 (100)	69 (100)	0	0	133 (59.9)
Baseline HCV RNA level, IU/mL					
<800,000	13 (20.3)	16 (23.2)	9 (19.6)	9 (20.9)	47 (21.2)
≥800,000	51 (79.7)	53 (76.8)	37 (80.4)	34 (79.1)	175 (78.8)
IL28B genotype					
CC	7 (10.9)	19 (27.5)	7 (15.2)	17 (39.5)	50 (22.5)
CT	40 (62.5)	35 (50.7)	29 (63.0)	20 (46.5)	124 (55.9)
TT	17 (26.6)	15 (21.7)	10 (21.7)	6 (14.0)	48 (21.6)
Baseline Platelet Counts 10^9/L					
<90	23 (35.9)	18 (26.1)	4 (8.7)	1 (2.3)	46 (20.7)
≥90	41 (64.1)	51 (73.9)	42 (91.3)	42 (97.7)	176 (79.3)
Baseline Albumin (g/L)					
<35	11 (17.2)	11 (15.9)	2 (4.3)	0	24 (10.8)
≥35	53 (82.8)	58 (84.1)	44 (95.7)	43 (100)	198 (89.2)
Baseline EGFR by MDRD (mL/min/1.73 m^2)					
30–59	1 (1.6)	2 (2.9)	2 (4.3)	0	5 (2.3)
60–89	22 (34.4)	30 (43.5)	20 (43.5)	18 (41.9)	90 (40.5)
≥90	41 (64.1)	37 (53.6)	24 (52.2)	25 (58.1)	127 (57.2)
History of Diabetes					
Yes	10 (15.6)	20 (29.0)	9 (19.6)	8 (18.6)	47 (21.2)
No	54 (84.4)	49 (71.0)	37 (80.4)	35 (81.4)	175 (78.8)

*Treatment experienced includes prior IFN-based regimens only.
†GT1a prior IFN/RBV or pegIFN/RBV null-responders or nonresponders were treated for 24 weeks.

EFFICACY

- The SVR12 rate was 96.4% (214/222) with a 2-sided 95% confidence interval of (93.1%, 98.2%) in the overall ITT population **(Figure 2)**

Figure 2. Efficacy of 12- or 24-week Treatment With 3-DAA ± RBV by Genotype and Fibrosis Stage

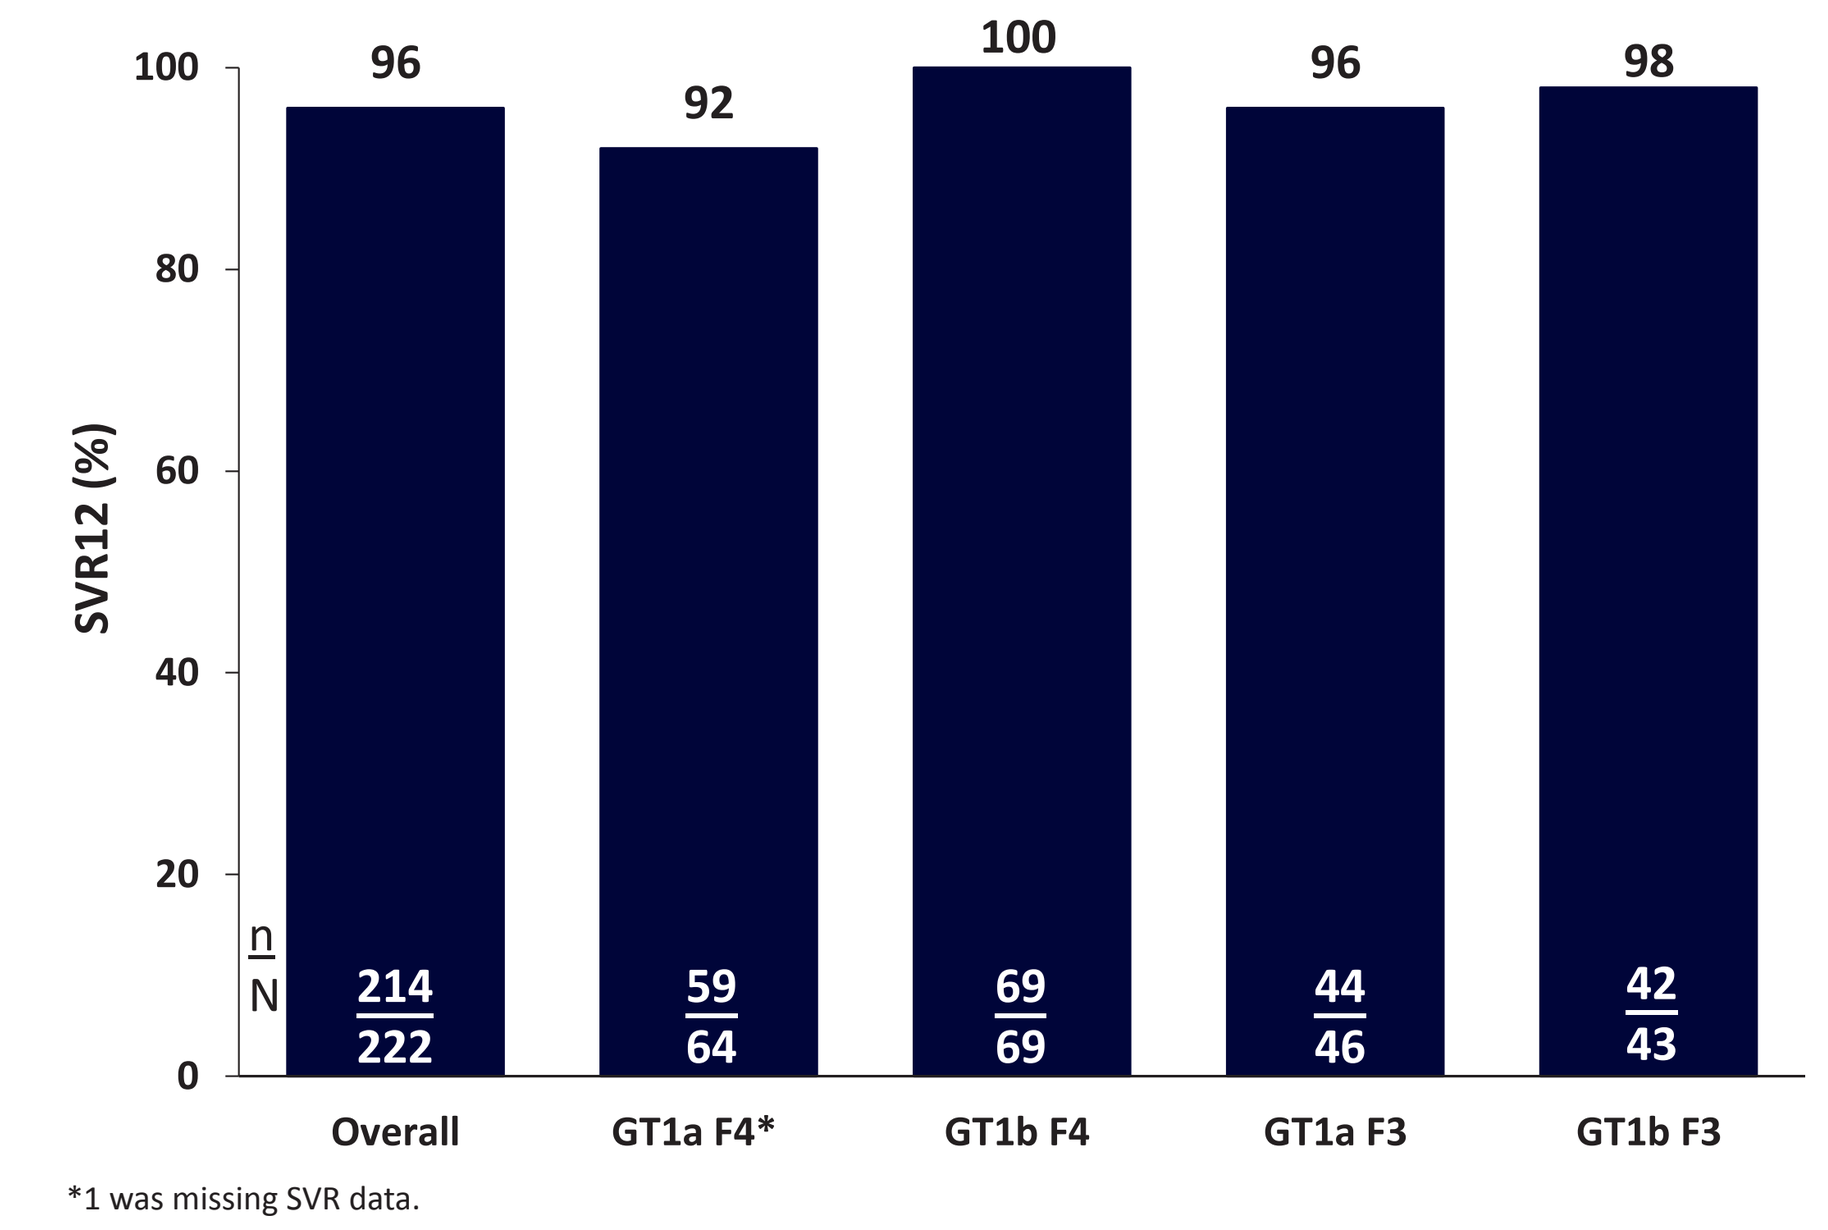
SVR12 (%)
100
80
60
40
20
0
96
92
100
96
98
n
N
214
222
59
64
69
69
44
46
42
43
Overall
GT1a F4*
GT1b F4
GT1a F3
GT1b F3

*1 was missing SVR data.

- Eight patients did not achieve SVR12 **(Table 2)**
 - One experienced on-treatment virologic failure (breakthrough at Week 12)
 - Six patients experienced relapse by post-treatment Week 12
 - One patient was missing SVR12 data due to premature study discontinuation at post-treatment Day 35; HCV RNA was undetectable at end of treatment

Table 2. Resistance Data for Virologic Failures

	Reason	Treatment status/duration	NS3 BL	NS3 VF	NS5A BL	NS5A VF	NS5B BL	NS5B VF
GT1a F4	Relapse	Naive, 12-weeks	None	V36M, R155K, D168V	M28V, Q30R	M28V, Q30R	None	M414I, S556G
	Relapse	Experienced 12-weeks	None	D168V	None	M28T, Q30R	None	None
	Relapse	Experienced 24-weeks	None	R155K	None	Q30R	Y448H	Y448H
	Breakthrough	Experienced 24-weeks	None	R155K	Q30H, Y93H	Q30R	None	None
GT1a F3	Relapse	Experienced 12-weeks	None	None	None	Q30R	None	C316Y, S556G
	Relapse	Naive 12-weeks	None	R155K, D168V	None	Q30R	None	None
GT1b F3	Relapse	Experienced 12-weeks	None	None	Y93H	Y93H	C316N	C316N, M414I

BL, baseline; VF, virologic failure.

- Mean changes in liver stiffness (measured by FibroScan) between baseline and post-treatment Week 12 among patients who achieved SVR12 who had baseline METAVIR F3 fibrosis staging was -2.9 (SD 3.5) and in patients with METAVIR F4 this change was -6.8 (SD 8.8). Overall, the median percent change from baseline was -29.2 (-68.1 to -114.5) **(Table 3)**

Table 3. FibroScan Percent Change From Baseline

Patients	n	FibroScan (kPa) Baseline mean	SVR12 mean	Mean change	SD	Percent change from baseline, median (range)
SVR12	141	19.9	14.5	-5.4	7.6	-29.2 (-68.1–114.5)
F3	51	11.4	8.6	-2.9	3.5	-30.0 (-64.6–114.5)
F4	90	24.7	17.9	-6.8	8.8	-28.9 (-68.1–37.9)

- The SVR12 rates were similar between the subsets with METAVIR F3 and F4 fibrosis (97% vs 96%, respectively) **(Figure 3)**

Figure 3. Efficacy of 12- or 24-week Treatment With 3-DAA ± RBV in GT1a- or 1b-infected Patients With METAVIR F3 or F4 Fibrosis

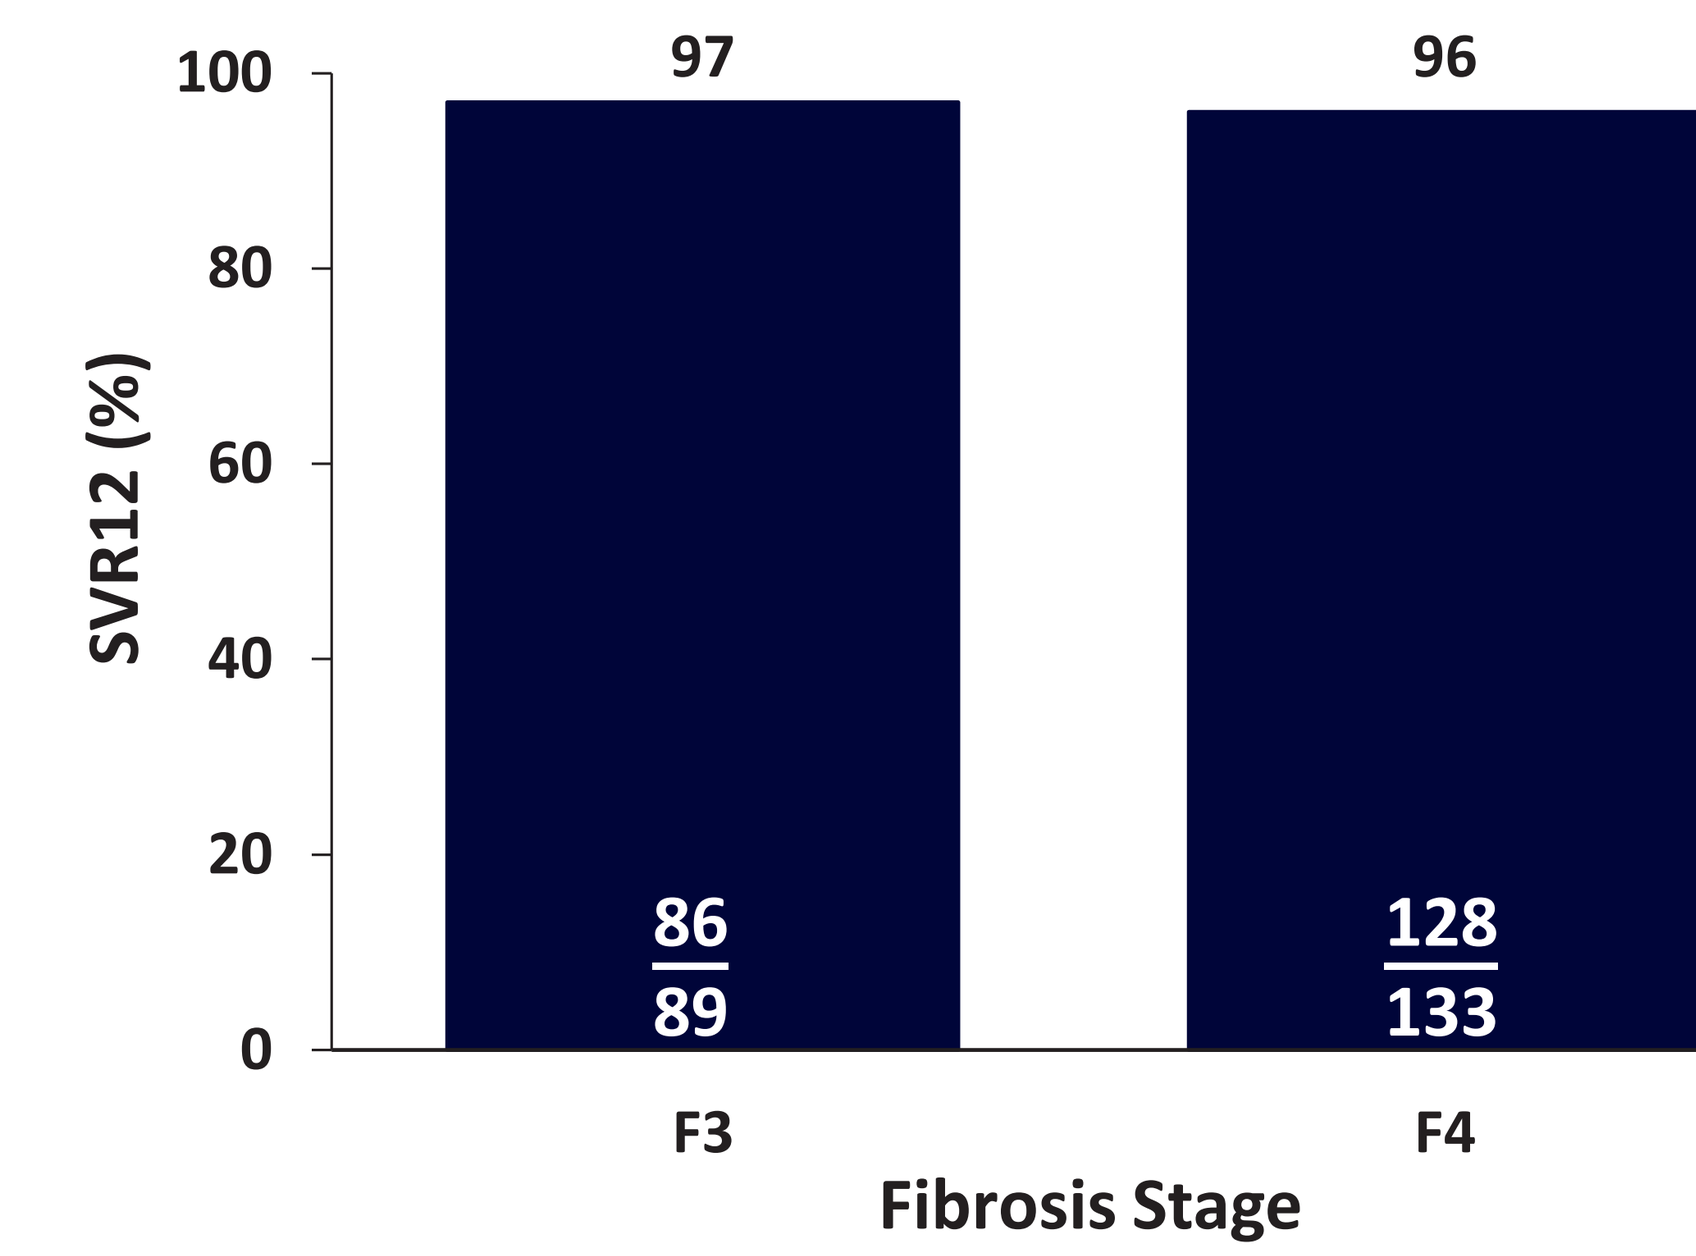
SVR12 (%)
100
80
60
40
20
0
97
96
86
89
128
133
F3
F4
Fibrosis Stage

- SVR12 rates were similar between treatment-naive and -experienced patients (96% vs 97%, respectively) **(Figure 4)**

Figure 4. Efficacy of 12- or 24-week Treatment With 3-DAA ± RBV by Treatment Status

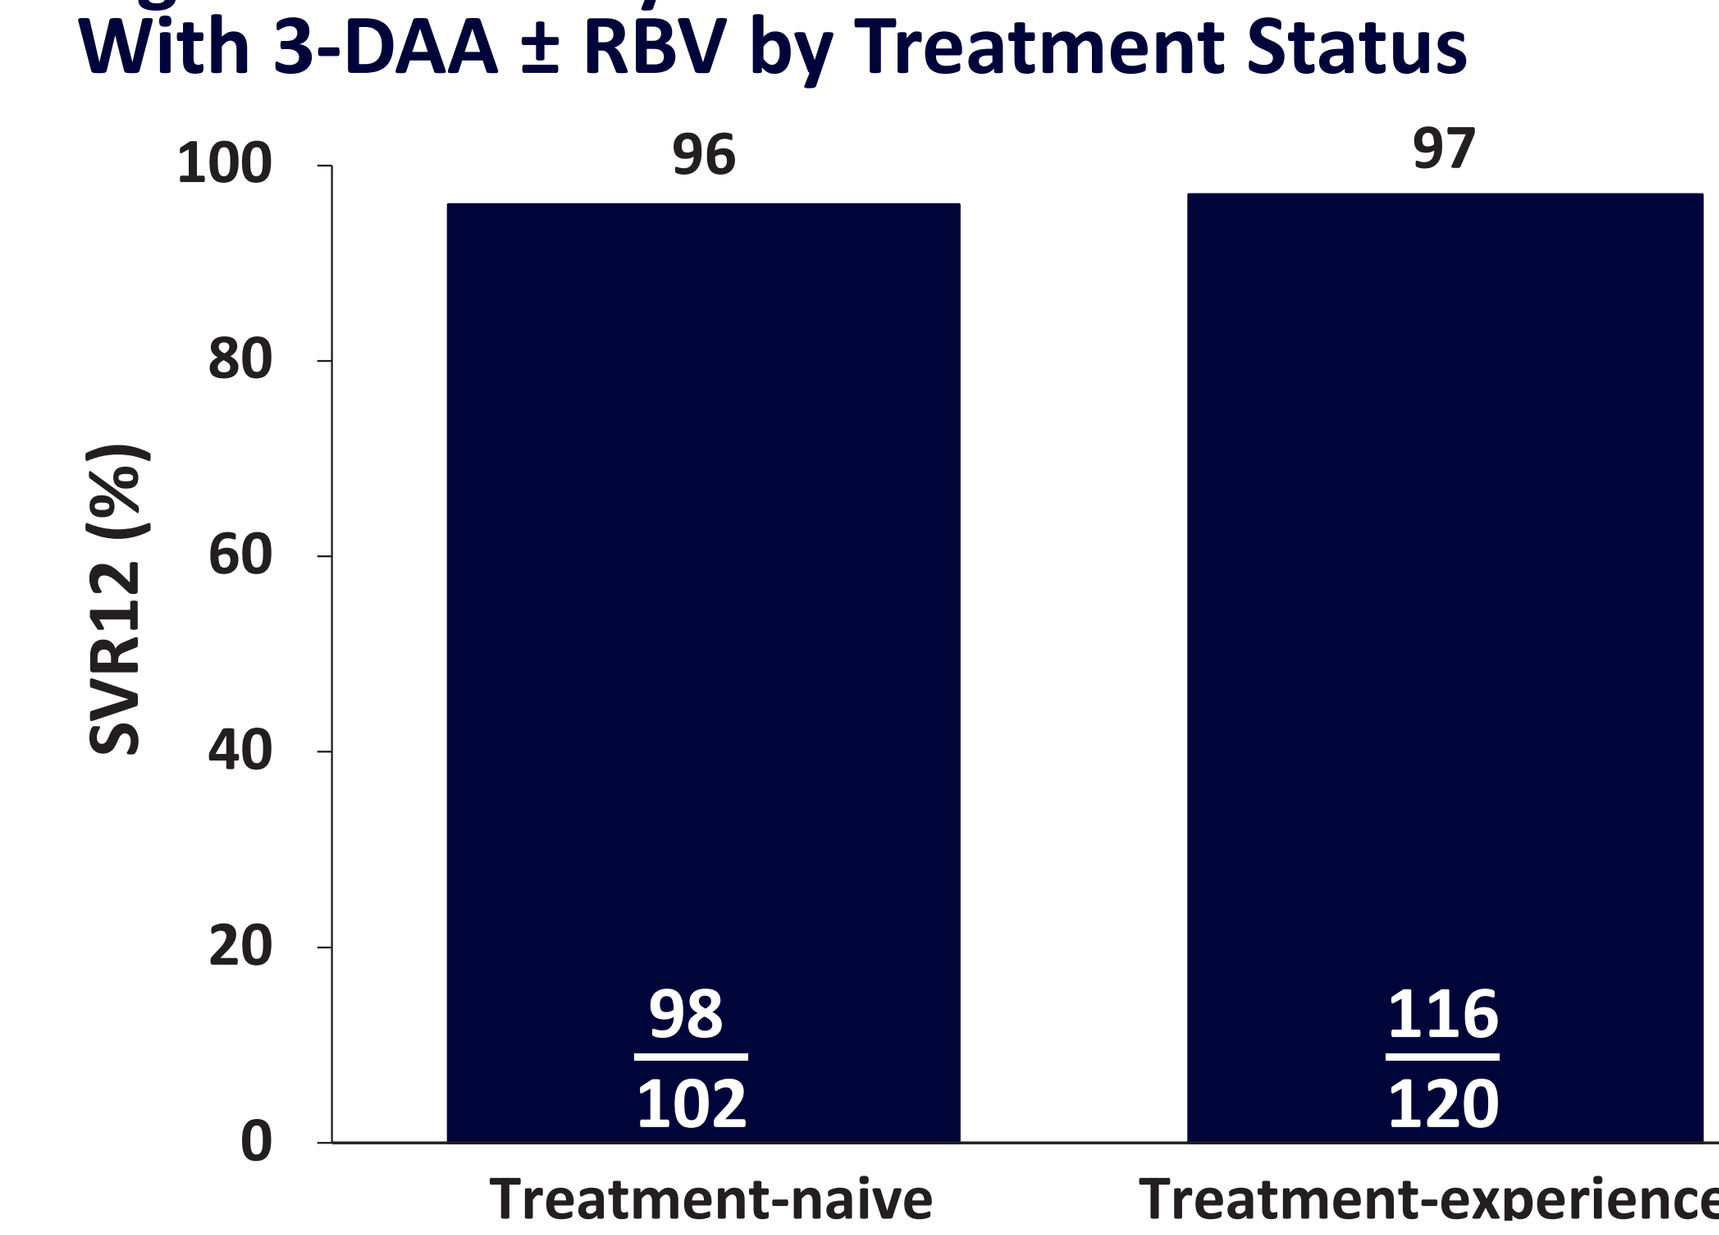
SVR12 (%)
100
80
60
40
20
0
96
97
98
102
116
120
Treatment-naive
Treatment-experienced

SAFETY

- A total of 174 (78.4%) patients reported experiencing at least 1 TEAE
- There were few serious adverse events (SAEs) and 1 AE which led to the discontinuation of study drugs **(Table 4)**
- Of the 6 SAEs (metastatic lung adenocarcinoma, esophageal varices hemorrhage, renal failure, transient ischemic attack, diarrhea, and hepatic decompensation), only 1 (hepatic decompensation) was considered by the investigator to be possibly related to treatment
 - SAE of hepatic decompensation occurred after the end of treatment (post-treatment Day 141) and was thought possibly related to study drugs. A secondary diagnosis of viral infection was hypothesized but not confirmed
- Two deaths occurred during the post-treatment period: 1 due to hepatocellular carcinoma (post-treatment Day 155) and 1 due to septicemia and hepatic decompensation (post-treatment Day 141) after complications from total hip arthroplasty

Table 4. Summary of Adverse Events

Characteristic, n (%)	3-DAA ± RBV N = 222
Any TEAE	174 (78.4)
Serious AEs	6 (2.7)
Any AE leading to discontinuation of study drug*	1 (0.5)
Any AE leading to interruption of study drug	3 (1.4)
Any AE leading to RBV dose modification	20 (9.0)
TEAEs in ≥10% of patients	
Headache	47 (21.2)
Fatigue	41 (18.5)
Nausea	33 (14.9)
Pruritus	32 (14.4)

*One patient discontinued treatment on Day 76 due to a moderate non-treatment related event of ascites.
TEAE, treatment-emergent adverse event. SAE, serious adverse event.

- Grade 3 laboratory abnormalities were rare **(Table 5)**
- One F4 patient receiving 3-DAA + RBV for 12 weeks experienced a grade 4 decrease in hemoglobin
- There were no ALT elevations of ≥ 3 x ULN during treatment

Table 5. Summary of Clinical Laboratory Parameters†

Laboratory parameter	n/N (%)
Hemoglobin (g/L)	
Grade 2 (<10–8 g/dL)	12/222 (5.4)
Grade 3 (<8–6.5 g/dL)	1/222 (0.5)
Grade 4 (<6.5 g/dL)	1/222 (0.5)
ALT	
Grade 3 (>5.0–20.0 × ULN)	0
AST	
Grade 3 (>5.0–20.0 × ULN)	0
Total bilirubin (μmol/L)	
Grade 3 (>3.0–10.0 × ULN)	12/222 (5.4)

†Each individual was only counted once in the highest grade category.
ALT, alanine aminotransferase; AST, aspartate aminotransferase; ULN, upper limit of normal.

CONCLUSIONS

- Treatment with 3-DAA ± RBV in treatment-naive and -experienced individuals resulted in 96.4% SVR12 rates in HCV GT1-infected Brazilian patients with bridging fibrosis (METAVIR F3) or compensated cirrhosis (METAVIR F4)
- Achieving SVR12 reduced liver stiffness in individuals with advanced fibrosis or cirrhosis
- 3-DAA ± RBV was well tolerated and had a similar safety profile in Brazilian patients when compared to other global studies

REFERENCES

1. Kershenobich D, et al. *Liver Int*. 2011;31(Suppl 2):18–29.
2. Boletim Epidemiológico—Hepatites Virais. Brasília, Brazil: Ministério da Salúde; 2015.
3. Castro R, et al. *Int J Hepatol*. 2015;doi:10.1155/2015/852968.
4. Brazil: Ministério da Salúde. Clinical Protocol and Therapeutic Guidelines for Hepatitis C and Coinfections; 2015.
5. Viekira Pak Prescribing Information, available online at http://www.rxabbvie.com/pdf/viekirapak_pi.pdf; Revised 06/2016 (accessed August 2016).
6. Andreone P, et al. *Gastroenterology.* 2014;147:359–65.
7. Feld JJ, et al. *N Engl J Med.* 2014;370:1594–603.
8. Ferenci P, et al. *N Engl J Med.* 2014;370:1983–92.
9. Poordad F, et al. *N Engl J Med.* 2014;370:1973–82.
10. Zeuzem S, et al. *N Engl J Med.* 2014;370:1604–14.

abbvie

DISCLOSURES

The design, study conduct, analysis, and financial support of the study (NCT02442271) were provided by AbbVie. AbbVie participated in the interpretation of data, review, and approval of the content. All authors had access to all relevant data and participated in writing, review, and approval of this presentation.

K Alves, DE Cohen, SA Lari, L Liu, R Tripathi, T Pilot-Matias, NS Shulman: AbbVie employees may hold stock or options

MG Pessoa: Recipient of research grants and speaker for AbbVie, Alexion, Gilead, Janssen, and MSD

JV Madruga: Received honoraria and served on Advisory boards for AbbVie, BMS, Gilead, GSK/ViiV, Pfizer, and Roche; Lecturer for AbbVie, Boehringer Ingelheim, BMS, Gilead, GSK/ViiV, MSD, Pfizer, Roche, and Janssen; conducted clinical trials for AbbVie, Boehringer Ingelheim, BMS, Gilead, GSK/ViiV, MSD, Pfizer, Roche, and Janssen

E Nunes: Recipient of research grants and speaker for Gilead and AbbVie

H Cheinquer: Recipient of research grants for AbbVie, MSD, BMS, Janssen, and Gilead

CE Brandão-Mello: has received honoraria and served on Advisory Boards for Roche, MSD, Gilead, BMS, Janssen, AbbVie; Member of the Advisory Board of Viral Hepatitis Department of Ministry of Health and of the Advisory Board of Viral Hepatitis of the Health State Secretariat of Rio de Janeiro

MC Mendes-Correa: Recipient of research grants and speaker for Gilead, MSD, Janssen, and AbbVie. Member of the Committee for Viral Hepatitis, Ministry of Health, Brazil

ML Ferraz: Recipient of research grants and speaker for Gilead, MSD, Janssen, AbbVie and Janssen. Member of the Committee for Viral Hepatitis, Ministry of Health, Brazil

AL Martinelli: Recipient of research grants and speaker for AbbVie, BMS, Gilead, and Janssen

44587-161467-TOPAZ-III — AASLD 2016 — AbbVie — proof 4 (rjh) — november 8, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051

CREATED @ 100%

Abstract/Poster # 934

Exposure-Response Analyses to Demonstrate Similar Efficacy and Better Tolerability for Low Dose Ribavirin Compared to Weight-Based Ribavirin with the 3D Regimen (Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir) in HCV GT1 Infection

Akshanth R. Polepally[1], Balakrishna S. Hosmane[2], Chih-Wei Lin[1], Mukul Minocha[1], Haoyu Wang[2], Amit Khatri[1], Rajeev M. Menon[1]

[1]AbbVie Inc., Clinical Pharmacokinetics and Pharmacodynamics, North Chicago, IL, 60064, USA [2]AbbVie Inc., Biometrics, North Chicago, IL, 60064, USA

Contact Information: akshanth.r.polepally@abbvie.com

THE LIVER MEETING® AASLD 2016 BOSTON • NOVEMBER 11-15

BACKGROUND

The 3 direct acting antiviral (DAA) combination (3D) of paritaprevir/r (NS3/4A protease inhibitor identified by AbbVie and Enanta and dosed with ritonavir), ombitasvir (NS5A inhibitor) and dasabuvir (NS5B polymerase inhibitor) ± ribavirin (RBV) was evaluated in >2300 hepatitis C virus (HCV) genotype 1-infected subjects as part of 6 phase 3 studies and was generally well tolerated with a 12-week sustained virologic response (SVR_{12}) of 92% to 100% in patients with or without cirrhosis.[1-5]

The 3D regimen (ombitasvir/paritaprevir/ritonavir and dasabuvir) ± weight based RBV is approved for the treatment of HCV genotype 1 (GT1) infection.

RATIONALE

With the potent 3D regimen, RBV dose reduction is expected to improve tolerability with minimal impact on efficacy.

OBJECTIVES

- To predict the efficacy (percent 12-week sustained virologic response [%SVR_{12}]), and
- To predict the safety event incidence rates (total bilirubin [TBIL] elevation and hemoglobin [Hgb] reduction) . . .

for the 3D regimen + low dose (600 mg) RBV (3D + LDR) compared to the 3D regimen + weight based (1000 or 1200 mg daily) RBV (3D + WBR).

METHODS

Datasets:

In general, the studies (in **Table 1**) enrolled:

- GT1 HCV-infected subjects who were 18 years or older and are either HCV treatment naïve or treatment experienced to previous interferon (IFN) or pegylated IFN ± RBV therapy.
- Studies excluded subjects who were with a positive test result for hepatitis B or human immunodeficiency virus or recent (within 6 months prior to study drug administration) history of drug or alcohol abuse that could preclude adherence to the study protocol.

Efficacy data

- Data from GT1a subjects from four Phase 3 and two Phase 2 studies (2D* or 3D ± WBR; N = 1253) were included in the analysis (**Table 1**).
 - All the GT1a subjects included in the dataset were treated with the regimens for 12 weeks except for cirrhotic null responders (to interferon based therapies) who were treated for 24 weeks.
 - Subjects who discontinued prematurely due to non-virologic reasons (adverse events or withdrawal of informed consent) irrespective of whether they achieved SVR_{12} or not, or those with missing SVR_{12} data (loss to follow-up) were excluded for the analysis.

Safety data

- Data from GT1a and GT1b subjects from six Phase 3 studies and one Phase 2 study (3D ± WBR; N = 2346) were included in the analyses (**Table 1**).

Table 1. List of Studies with Efficacy and Safety Data Included in the Analyses

Phase	Study	Patients	Treatment Regimen	N for Efficacy Analysis (GT1a only)	N for Safety Analyses
3	PEARL-II	GT1b treatment-experienced	3D regimen ± RBV	3	186
	PEARL-III	GT1b treatment-naïve	3D regimen ± RBV	0	419
	PEARL-IV	GT1a treatment-naïve	3D regimen ± RBV	298	305
	TURQUOISE-II	GT1 treatment-naive and treatment-experienced with compensated cirrhosis	3D regimen + RBV for 12 or 24 weeks	204	380
	SAPPHIRE-I	GT1 treatment-naïve	3D regimen + RBV	313	626
	SAPPHIRE-II	GT1 treatment-experienced	3D regimen + RBV	172	392
2	AVIATOR	GT1 treatment-naive and treatment-experienced	2D* or 3D regimen ± RBV	232	0
	M14-103	GT1 treatment-naïve and treatment-experienced taking methadone or buprenorphine	3D regimen + RBV	31	38

*2D regimen represents a regimen without ombitasvir or without dasabuvir

METHODS (Continued)

Exposures from Pharmacokinetic Analyses

- Population pharmacokinetic models were built for paritaprevir, ombitasvir, dasabuvir, ritonavir and RBV using a nonlinear mixed effects approach using the NONMEM (v7.3) software with First Order Conditional Estimation with interaction (FOCE-I) estimation method.[6,7]
- The empirical Bayes (*post hoc*) estimates of the pharmacokinetic parameters of the DAAs of each subject from the respective population-pharmacokinetic models were used to obtain the steady state DAA and ribavirin exposure variables (trough plasma concentration [C_{trough}] and area under the plasma concentration time curve [AUC]).

Efficacy Analysis

- Multiple linear logistic regression (MLR) model was developed (SAS 9.2) to establish the relationship between SVR_{12} and C_{trough} values of DAAs and RBV for the GT1a infected subjects only.
- Age, weight, body mass index (BMI), sex, ethnicity, baseline HCV viral load, IL28B genotype (CC, Non-CC [CT or TT]), prior treatment experience (Naïve, treatment experienced) and compensated liver cirrhosis (Child-Pugh A) were tested as covariates.
- Backward elimination procedure was used to select the covariates. Relevant covariates were included in the final model based on statistical and clinical significance.
- The Hosmer-Lemeshow test was used to assess the goodness of fit of the logistic regression model. The Hosmer-Lemeshow test p-value > 0.05 indicates that the logistic regression model fits the data well.
- The final model was used to predict the %SVR_{12} for the 3D + LDR compared to the 3D + WBR for the following 4 subpopulations; predictions for the 3D + LDR were conducted at 50% of the geometric mean value of RBV C_{trough} from the 3D + WBR.

 P1: Non-cirrhotic females with IL28B CC genotype ("easy-to-cure")

 P2: Non-cirrhotic males with IL28B CC genotype

 P3: Cirrhotic females with IL28B non-CC genotype

 P4: Cirrhotic males with IL28B non-CC genotype ("hard-to-cure")

- In addition to the above, efficacy analysis was done excluding AVIATOR study which included 2D and 3D regimens administered with different formulations compared to the marketed formulations used in other studies.

Safety Analysis

- Separate MLR models for the relationship between safety incidence rates (by severity for total bilirubin elevation and hemoglobin reduction) and area under the concentration-time curve (AUC) values were developed (R 3.0.1).
- In the analyses, the predictor variables were logarithmic values of the steady-state AUCs for paritaprevir, ombitasvir, dasabuvir, ritonavir, and RBV derived from population pharmacokinetic analyses, treatment effects (no RBV versus RBV, or placebo versus active treatment), and covariates including age, weight, sex, study effects, use of estrogen or progesterone containing medication, and baseline status of the safety variable.
- Forward selection procedure was used to select the pharmacokinetic variables and covariates based on the Bayesian Information Criterion (BIC).
- The final model was used to predict the grade 3 total bilirubin elevation and grade 2 hemoglobin reduction for the 3D + LDR compared to the 3D + WBR; predictions for the 3D + LDR were conducted at 40% to 50% of RBV AUC values from the 3D + WBR.

DISCLOSURES

AbbVie contributed to the design, research, and interpretation of data, writing, reviewing, and approving the publication. All authors are AbbVie employees/contractors and may hold AbbVie stocks and/or options.

REFERENCES

1. Feld J, et al. *NEJM*. 2014; 370:1594–1603.
2. Zeuzem S, et al. *NEJM*. 2014; 370:1604–1614.
3. Andreone P, et al. *Gastroenterology*. 2014; 147:359–365.
4. Poordad F, et al. *NEJM*. 2014; 370:1973–1982.
5. Ferenci P, et al. *NEJM*. 2014; 370:1983–1992.
6. Mensing S, et al. *Journal of Hepatology*. 2015; 62(S2):S644.
7. Mensing S, et al. *AAPS J*. 2016; 18(1):270-80.

RESULTS

***Efficacy Analysis:* Logistic Regression Model**

- Based on exploratory analysis, change in RBV C_{trough} values over 2-fold showed only a 2.4% change in SVR_{12} rates (**Figure 1**).

Figure 1. Percent SVR_{12} vs RBV C_{trough} (GT1a)

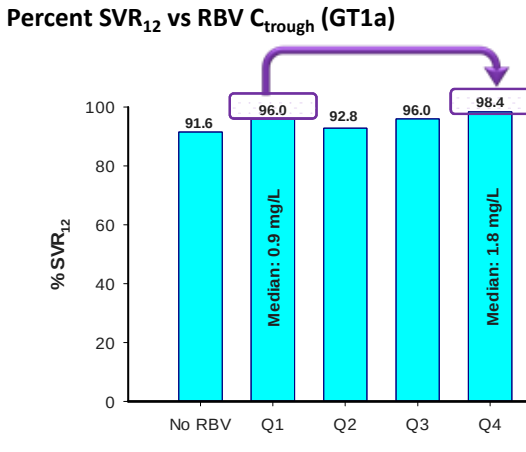

100
80
60
40
20
0
% SVR12
91.6
96.0
92.8
96.0
98.4
Median: 0.9 mg/L
Median: 1.8 mg/L
No RBV
Q1
Q2
Q3
Q4
Ribavirin Ctrough Quartile

Median (range) RBV C_{trough} (mg/L): Q1, 0.9 (0. 001 to 1.0); Q2, 1.1 (1.0 to 1.2); Q3, 1.4 (1.2 to 1.5); Q4, 1.8 (1.5 to 4.3)

- In the logistic regression analyses, C_{trough} for DAAs, RBV, age, baseline viral load and interleukin 28 (IL28B) genotype (CC *vs* Non-CC) were significant covariates ($p < 0.05$); sex ($p = 0.07$) and cirrhosis (presence *vs* absence; $p = 0.2$) were retained in the model for prediction purposes (**Table 2**).

Table 2. Model Parameter Estimates and Odds Ratios for Predictor Variables Based on C_{trough} (2D or 3D ± RBV Regimens)

Predictor variable	β	SE	P value	Odds ratio estimates: Point estimate	Odds ratio estimates: 95% Wald confidence limits
Baseline viral load, log_{10} IU/mL	-0.8424	0.3088	0.0064	0.431	0.235–0.789
Age, Years	-0.0547	0.0181	0.0026	0.947	0.914–0.981
Ln ombitasvir C_{trough}, mg/L[a]	0.6615	0.1909	0.0005	1.938	1.333–2.817
Ln dasabuvir C_{trough}, mg/L[a]	0.3390	0.0843	< 0.0001	1.404	1.190–1.656
Ln paritaprevir C_{trough}, mg/L[a]	0.3697	0.1367	0.0068	1.447	1.107–1.892
Ln ribavirin C_{trough}, mg/L[a]	0.2132	0.0490	< 0.0001	1.238	1.124–1.362
Male sex	-0.7413	0.4046	0.0669	0.476	0.216–1.053
Presence of Cirrhosis	-0.5589	0.4361	0.2000	0.572	0.243–1.344
IL28B non-CC genotype	-1.0844	0.4048	0.0074	0.338	0.153–0.747

β, regression coefficient; SE, standard error

[a]1 natural log (Ln) unit increase in C_{trough} represents 2.7-fold increase

- Observed and predicted RBV C_{trough} values vs. SVR_{12} relationship are shown in **Figure 2**.
- The Hosmer-Lemeshow test p-value was 0.44 which indicates that the logistic regression model described the data well.

Figure 2. Observed and Predicted RBV C_{trough} *vs.* Response Relationship in HCV GT1a-Infected Subjects by IL28B genotype

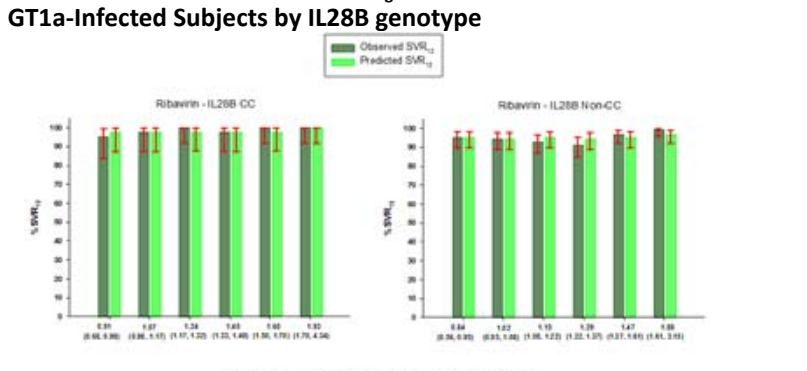

Observed SVR12
Predicted SVR12
Ribavirin - IL28B CC
Ribavirin - IL28B Non-CC
Ribavirin Ctrough, mg/L (median and range of each sextile)

SVR_{12} predictions represent the individual predictions at their respective RBV C_{trough} values and other categorical and continuous covariate values. Dark green and light green bars represent the observed proportion of subjects that achieved SVR_{12} and mean of the individual predicted SVR_{12}, respectively. Error bars represent the 95% exact binomial confidence intervals.

***Efficacy Analysis:* Impact of Low-Dose RBV with 3D Regimen on SVR_{12} Rates**

- SVR_{12} predictions are shown in **Table 3** for 4 subpopulations.
- The expected change in SVR_{12} for 3D + LDR compared to 3D + WBR was < 1.3% (upper confidence bound) across easy-to-treat (non-cirrhotic, female, IL28B CC) to hard-to-treat (cirrhotic, male, IL28B non-CC) GT1a subpopulations (**Table 3**).

Table 3. Prediction Results: The Effect of 50% Lower RBV C_{trough} on SVR_{12}

Population	Observed SVR_{12} %	Predicted SVR_{12} % (95% CI): 3D + WBR (Reference)	Predicted SVR_{12} % (95% CI): 3D + LDR (Test)	Delta SVR_{12}% (95% CI)
P1: Noncirrhotic, female, IL28B CC	100	99.80 (99.56 to 100.03)	99.77 (99.50 to 100.03)	0.03 (0.00 to 0.07)
P2: Noncirrhotic, male, IL28B CC	96.60	98.97 (98.07 to 99.88)	98.81 (97.80 to 99.82)	0.16 (0.04 to 0.29)
P3: Cirrhotic, female, IL28B non-CC	100	99.08 (98.13 to 100.02)	98.93 (97.84 to 100.02)	0.15 (-0.01 to 0.30)
P4: Cirrhotic, male, IL28B non-CC	92.37	95.39 (92.18 to 98.61)	94.70 (91.02 to 98.38)	0.69 (0.13 to 1.26)

CI = confidence interval; IL28B = interleukin-28B genotype (CC or non-CC)

Note: SVR_{12} predictions were performed at mean baseline viral load, age and geometric mean for DAAs and RBV steady-state C_{trough} of each subpopulation; Predictions for 3D + LDR were at 50% of the geometric mean value of RBV C_{trough}.

- Based on the additional analysis excluding AVIATOR study data (model results not shown), the expected change in SVR_{12} for 3D + LDR compared to 3D + WBR was up to 3.2% (upper confidence bound) in GT1a non-cirrhotics (**Table 4**).

Table 4. Prediction Results (excluding AVIATOR data from modeling): The Effect of 50% Lower RBV C_{trough} on SVR_{12}

Population	Observed SVR_{12} %	Predicted SVR_{12} % (95% CI): 3D + WBR (Reference)	Predicted SVR_{12} % (95% CI): 3D + LDR (Test)	Delta SVR_{12}% (95% CI)
Non-cirrhotics	96.33	99.56 (99.00 to 100.12)	98.50 (96.11 to 100.90)	1.06 (-1.05 to 3.16)
Cirrhotics	95.60	99.09 (97.87 to 100.32)	96.96 (91.72 to 102.19)	2.14 (-2.37 to 6.65)

CI = confidence interval

Note: SVR_{12} predictions were performed at mean baseline viral load, age and geometric mean for DAAs and RBV steady-state C_{trough} of each subpopulation; Predictions for 3D + LDR were at 50% of the geometric mean value of RBV C_{trough}.

***Safety Analyses:* Improvement in Safety Profile with 3D + LDR**

- Paritaprevir and RBV AUC values and baseline total bilirubin were associated with total bilirubin elevation; RBV AUC, baseline hemoglobin, sex, and cirrhosis were associated with hemoglobin reduction.
- The predicted probability of Grade 3 total bilirubin elevation and Grade 2 hemoglobin reduction with 3D + LDR were 2.3% and 0.7% which were significantly lower than observed 5.1% and 6.9% with 3D + WBR, respectively (**Table 5**).

Table 5. Prediction Results: Effect of Reduced RBV AUC on Safety events

Adverse Event/Lab Toxicity	Observed Incidence Rate (% and 95% CI): 3D without RBV	Observed Incidence Rate (% and 95% CI): 3D + WBR	Predicted Incidence Rate (% and 95% PI): 3D + LDR
≥ Grade 3 Total Bilirubin Elevation	0.393 (0.00 to 1.42)	5.11 (4.11 to 6.30)	2.32 (1.43 to 3.75)
≥ Grade 2 Hemoglobin Reduction	0.000 (0.00 to 0.00)	6.88 (5.72 to 8.22)	0.651 (0.39 to 1.08)

CI = confidence interval; PI = Prediction Interval

Note: Predictions for 3D + LDR were at 40% to 50% of RBV AUC

CONCLUSIONS

- Exposure-response analysis using data from Phase 2 and 3 studies demonstrated that lowering the RBV dose (to 600 mg) from weight based dosing with 3D regimen would have minimal or no impact on SVR_{12} (< 1.3% change) in HCV GT1 infection.
 - Exposure-analyses using the data only from Phase 3 (and one Phase 2)studies with currently marketed 3D regimen resulted with up to 3.2% change in SVR_{12} in GT1a non-cirrhotic subjects.
- Lowering RBV total daily dose to 600 mg is expected to improve tolerability with meaningful decreases in Grade 3 bilirubin elevations as well as it is expected to provide a clinically meaningful reduction in adverse events related to decreased hemoglobin compared to weight based RBV dosing.

AASLD AMERICAN ASSOCIATION FOR THE STUDY OF LIVER DISEASES

GEODE-II: Efficacy and Safety of Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir With Low-Dose Ribavirin QD in Patients With Genotype 1a Chronic Hepatitis C Virus Infection Without Cirrhosis

Fred Poordad[1], Shahriar Sedghi[2], Paul J Pockros[3], Natarajan Ravendhran[4], Robert Reindollar[5], Michael R Lucey[6], Michael Epstein[7], Leslie Bank[8], David Bernstein[9], Roger Trinh[10], Preethi Krishnan[10], Tami Pilot-Matias[10], Akshanth R Polepally[10], Rajvineeth Kumar Pothacamury[10], Kristina Unnebrink[11], Marisol Martinez[10], David R Nelson[12]

[1]Texas Liver Institute, University of Texas Health Science Center, San Antonio, Texas, United States; [2]Associate Professor of Medicine and Surgery, Mercer University School of Medicine, Macon, Georgia, United States; [3]Division of Gastroenterology/Hepatology, Scripps Clinic, La Jolla, California, United States; [4]Digestive Disease Associates, Baltimore, Maryland, United States; [5]Piedmont Healthcare/Carolinas Center for Liver Disease, Statesville, North Carolina, United States; [6]Division of Gastroenterology/Hepatology, Department of Medicine, University of Wisconsin School of Medicine and Public Health, Madison, Wisconsin, United States; [7]Anne Arundel Medical Center, Digestive Disorders Associates, Annapolis, Maryland, United States; [8]Principal Investigator, Regional Clinical Research Inc., Endwell, New York, United States; [9]Hofstra Northwell School of Medicine, Manhasset, New York, United States; [10]AbbVie Inc., North Chicago, Illinois, United States; [11]AbbVie Deutschland GmbH & Co. KG, Ludwigshafen, Germany; [12]Department of Medicine, University of Florida, Gainesville, Florida, United States

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

BACKGROUND

- Hepatitis C virus (HCV) currently infects more than 170 million individuals worldwide; with more than 80% of those acutely infected developing chronic infection. When left untreated, chronic HCV infection can lead to liver cirrhosis, hepatocellular carcinoma (HCC), and liver-related morbidity and mortality[1]
- Genotype 1 (GT1) is the most prevalent HCV genotype in the United States, with the GT1a sub-genotype being most common followed by GT1b[2]
- The direct acting antiviral agents (DAAs) ombitasvir/paritaprevir/ritonavir + dasabuvir (3-DAA) administered for 12 weeks have been very effective in treating GT1b HCV infection, achieving SVR rates of 100% with a good safety profile.[3] In GT1a HCV-infected patients without cirrhosis, high SVR rates of 97% and 90% have also been achieved for those treated with and without RBV, respectively (PEARL-IV)[4]
- RBV is known to be associated with increased rates of adverse events, including decreases in hemoglobin and elevations of indirect bilirubin, and in some cases requiring RBV dose reduction or discontinuation
- Treatment guidelines recommend that RBV is administered according to weight, where patients weighing <75 kg receive 1000 mg RBV twice daily and those weighing ≥75 kg receive 1200 mg RBV twice daily

Multi-targeted 3 Direct-acting Antiviral Regimen

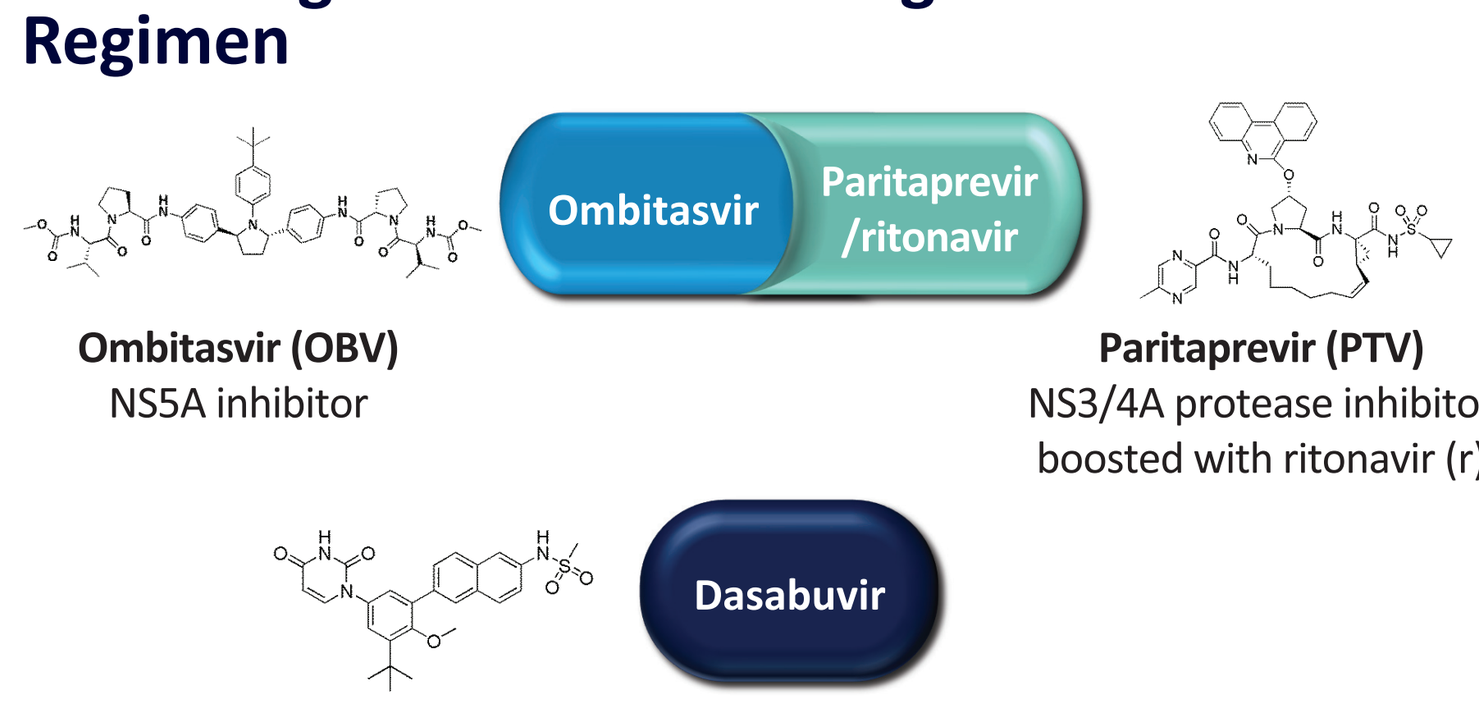
Ombitasvir
Paritaprevir
/ritonavir
Dasabuvir

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)

Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

PTV was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVE

- Evaluate the safety and efficacy of OBV/PTV/r + DSV with low-dose RBV (600 mg once daily) for 12 weeks in GT1a HCV-infected patients, naïve or experienced (with pegIFN/RBV), without cirrhosis and compare with the historical SVR12 rate for 3-DAA + full-dose, weight-based RBV as determined by the phase 3 PEARL-IV study[4]

METHODS

- Patients with GT1a infection without cirrhosis received OBV/PTV/r (25/150/100 mg once daily) + DSV (250 mg twice daily) + RBV (600 mg once daily) for 12 weeks **(Figure 1)**
- SVR12 rates in the presence or absence of baseline polymorphisms in NS3 or NS5A were compared by Fisher's exact test. The following polymorphisms (alone or as components of double variants) that confer ≥5-fold increase in EC_{50} were considered 3-DAA regimen-specific polymorphisms for GT1a
 - Paritaprevir-specific amino acid positions/variants: F43L, Y56H, R155G/K/S/T/W, A156S/T/V, or D168A/E/F/H/N/V/Y. Q80K (3-fold increase in EC50) was evaluated separately based on the significance of this polymorphism for the NS3 inhibitor simeprevir
 - Ombitasvir-specific amino acid positions/variants: M28T/V, Q30E/K/R/Y, L31V, P32L, H58D, or Y93C/F/H/L/N/S
 - Dasabuvir-specific amino acid positions/variants: L314H, C316Y, M414I/T/V, E446K/Q, Y448C/H, C451R, A553T, G554S, Y555H, S556G/R, G557R, G558R, D559G/N, or Y561H/N

METHODS (CONTINUED)

- Blood samples for the pharmacokinetic assessment of the study drugs were collected at treatment weeks 2, 4, 8, and 12. Drug concentrations were summarized as trough plasma concentrations (C_{trough}) which were binned based on sample collection time within the 22–26 hour or 10–14 hour bins for once or twice daily drugs, respectively

Figure 1. GEODE-II: Phase 3, Open-label, Multicenter Study Design

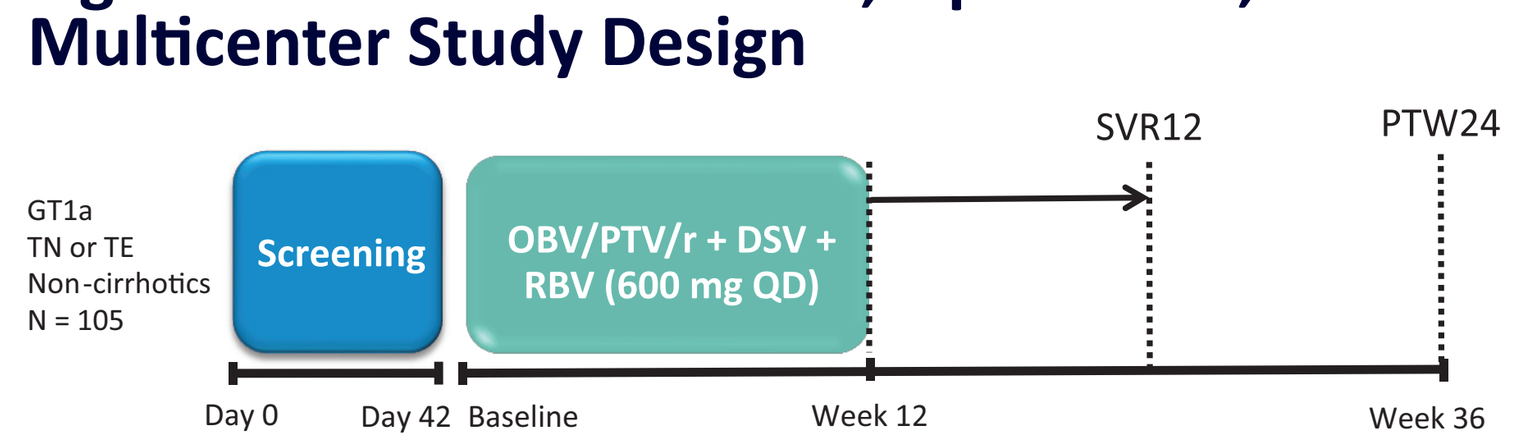
GT1a
TN or TE
Non-cirrhotics
N = 105
Screening
OBV/PTV/r + DSV + RBV (600 mg QD)
SVR12
PTW24
Day 0
Day 42
Baseline
Week 12
Week 36

KEY ELIGIBILITY CRITERIA

Key inclusion criteria:

- Male or female at least 18 years of age
- Chronic GT1a HCV infection (HCV RNA >1000 IU)
- Treatment-naïve or treatment-experienced (IFN/pegIFN and RBV)
- Absence of cirrhosis (verified by liver biopsy, FibroTest, or FibroScan)

Key exclusion criteria:

- Presence of HCV genotypes other than GT1a
- Co-infection with the hepatitis B virus (HBV) or human immunodeficiency virus (HIV)
- Any primary cause of liver disease other than chronic HCV infection
- Abnormal laboratory result that meets at least 1 of the following criteria (see **Table 1**)

Table 1. Laboratory Parameters Exclusion Criteria

Assessment	Value
CrCl*	<50 mL/min
Serum Albumin	<2.8 g/dL
INR†	>2.3
Hemoglobin	<LLN‡
Platelet Count	<25 000 cells/mm³
Total Bilirubin	>3.0 mg/dL

*Creatinine clearance estimated using the CockCroft-Gault equation.
†INR, International normalized ratio.
‡LLN, lower limit of normal.

STUDY ASSESSMENTS

Primary Efficacy Endpoint	Determine the percentage of patients with SVR12 and compare with the historical SVR12 rates of weight-based RBV*
Secondary Efficacy Objectives	Determine the percentage of patients with on-treatment virologic failure and post-treatment relapse
Primary Safety Endpoint	Determine the proportion of patients with hemoglobin <10 g/dL during treatment and hemoglobin decrease from baseline

STUDY COMPARISONS

- SVR12 rates are compared to the PEARL-IV study which assessed the safety and efficacy of the 3-DAA + RBV (full dose) in GT1a patients without cirrhosis for 12 weeks.[4] Non-inferiority of the 3-DAA + low dose RBV to the 3-DAA + full-dose RBV is achieved if the lower bound of the CI exceeds 92%
- Safety data are compared to pooled data from PEARL-II, III & IV studies which assessed the safety and efficacy of the 3-DAA + RBV (full dose) in GT1a and GT1b patients without cirrhosis for 12 weeks[4,5]

RESULTS

PARTICIPANTS

- 105 patients were enrolled in the study from 10 study centers in the United States **(Table 2)**
- **Figure 2** shows patient disposition

Table 2. Baseline Demographics and Disease Characteristics of Patients Enrolled in the Study

Characteristic	N = 105
Female, n (%)	55 (52)
Age, median (range), years	54 (24–69)
Race,* n (%)	
White	90 (86)
Black	15 (14)
Hispanic or Latino ethnicity,* n (%)	5 (5)
History of drug abuse, n (%)	69 (66)
Prior use of behavioral mood modifying drugs, n (%)	43 (41)
History of drug abuse or prior use of behavioral mood modifying drugs, n (%)	80 (76)
History of alcoholism, n (%)	85 (81)
Medical history of cognitive or psychiatric disorder condition/diagnosis, n (%)	57 (54)
BMI, mean ± SD, kg/m²	28 ± 6
HCV GT1a, n (%)	105 (100)
IL28B non-CC genotype, n (%)	77 (73)
HCV RNA, mean ± SD, log_{10} IU/mL	6.01 ± 0.85
HCV RNA ≥800 000 IU/mL, n (%)	62 (59)
Prior IFN/RBV treatment experience, n (%)	
Treatment-naïve	93 (89)
Null responder	6 (6)
Relapser	3 (3)
Unknown	2 (2)
IFN intolerant	1 (1)
Fibrosis stage	
F0–F1	85 (81)
F2	11 (11)
F3	9 (9)

BMI, body mass index; *IL28B*, interleukin 28.
*Race and ethnicity are self-reported.
†N = 104.

Figure 2. Patient Disposition Chart

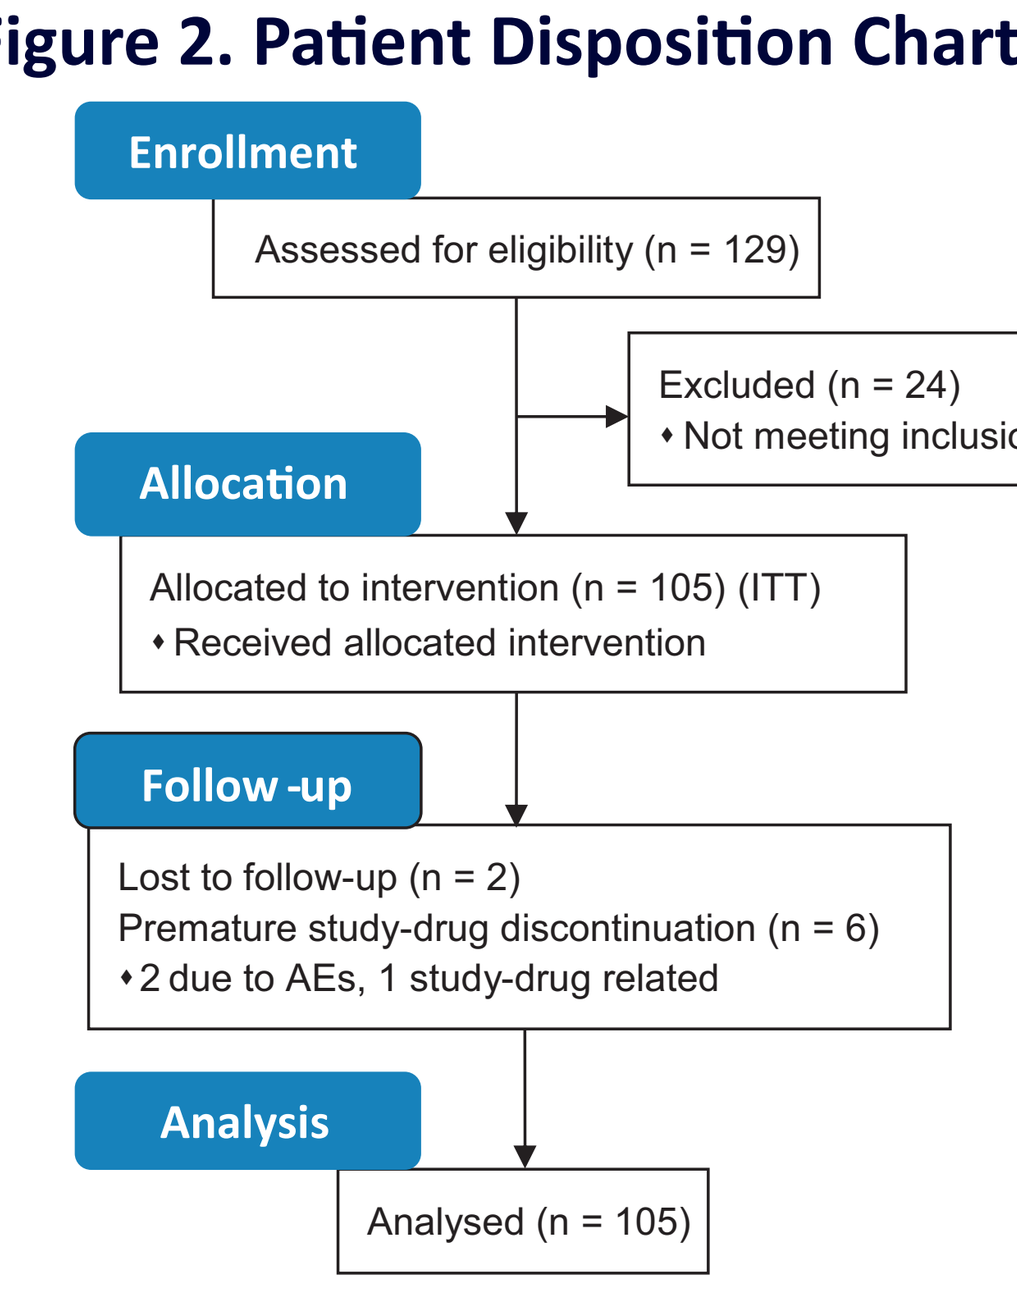
Enrollment
Assessed for eligibility (n = 129)
Excluded (n = 24)
• Not meeting inclusion criteria (n = 24)
Allocation
Allocated to intervention (n = 105) (ITT)
• Received allocated intervention
Follow-up
Lost to follow-up (n = 2)
Premature study-drug discontinuation (n = 6)
• 2 due to AEs, 1 study-drug related
Analysis
Analysed (n = 105)

EFFICACY

- 94/105 (90%) patients in the intent-to-treat (ITT) population achieved SVR12
- Of the 11 patients who did not achieve SVR12, 4 patients experienced a relapse by post-treatment week 12, 1 patient had a breakthrough, 4 patients prematurely discontinued study drug, and 2 patients were missing SVR12 data **(Table 3)**
- 94/99 (95%) patients in the modified ITT (mITT) population (which excludes SVR12 non-responders who were not categorized as virologic failures) achieved SVR12 **(Figure 3)**
- Non-inferiority to 3-DAA + full dose RBV (PEARL-IV) was not achieved

Figure 3. Sustained Virologic Response at Post-treatment Week 12 (SVR12)

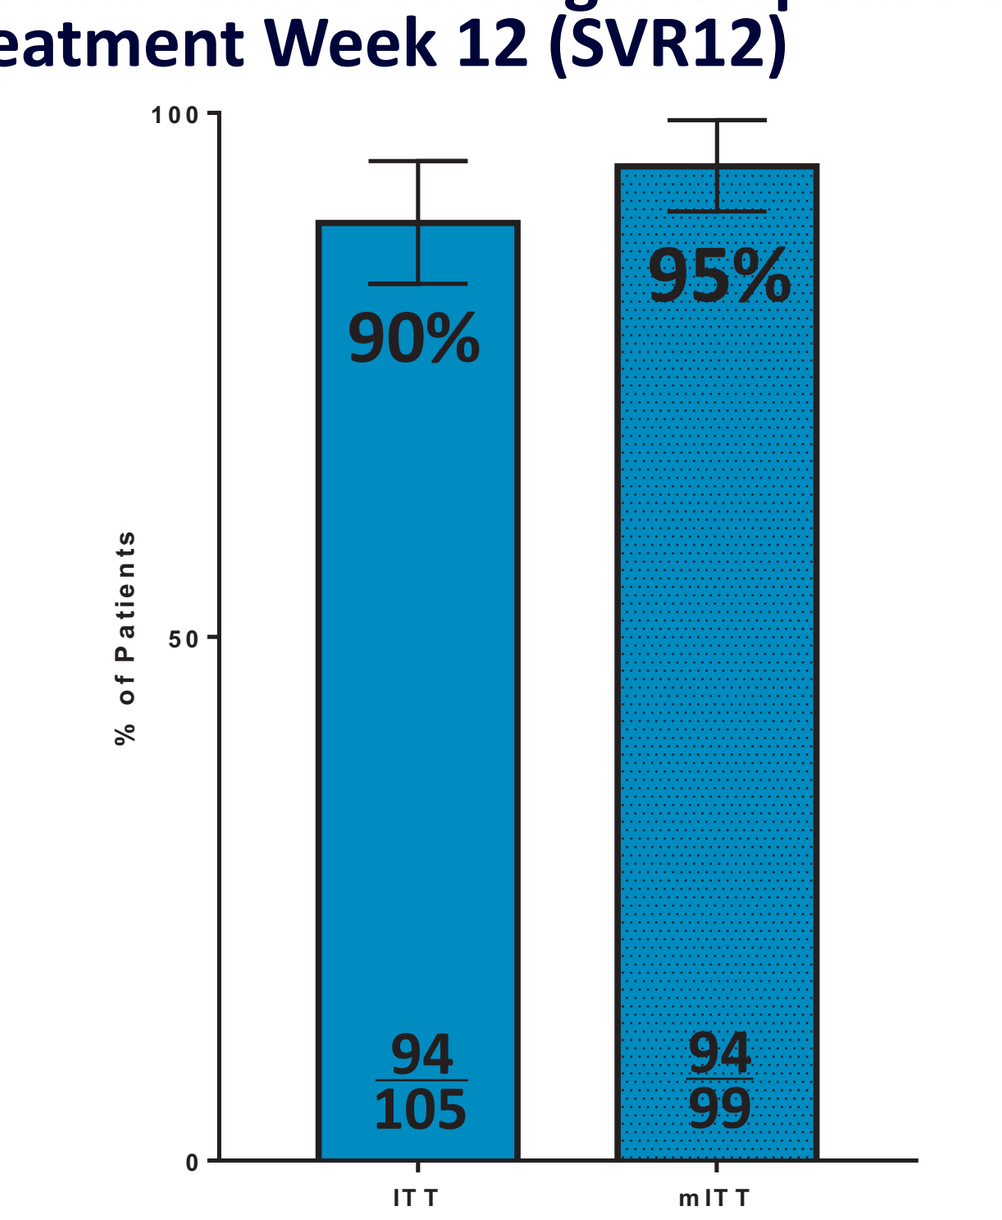
100
50
0
% of Patients
90%
95%
94
105
94
99
ITT
mITT

ITT, intent-to-treat population; mITT, modified ITT.

Table 3. Reasons for Not Achieving SVR12

Reason For Not Achieving SVR12	N = 105 n (%)
Virologic failure	5 (5)
Breakthrough	1 (1)
Relapse	4 (4)
Non-virologic failure	6 (6)
Drug discontinuation	4 (4)
Missing data*	2 (2)

*One patient had an undetectable HCV RNA load at end of treatment and the second patient achieved SVR4.

SAFETY

- Most adverse events (AEs) were mild or moderate in severity **(Table 4)**
- No cases of grade ≥3 hemoglobin decrease were observed **(Table 5)**
- No cases of grade ≥3 elevated total bilirubin were observed
- The most common AEs reported in the study were fatigue (28%), headache (13%), insomnia (11%), and nausea (11%)
- 3 patients had serious AEs. One serious AE was assessed as having a reasonable possibility of being study-drug related (bipolar-I disorder). The patient achieved SVR12
- AEs leading to study drug discontinuation occurred in 2 patients; 1 was assessed as having a reasonable possibility of being study-drug related as per the investigator's assessment (chest pain, heart rate increased, and dyspnea). The other patient had a psychotic disorder
- 2 AEs (decrease in hemoglobin) lead to RBV dose modification to 400 mg daily
- **Table 6** shows AEs and laboratory abnormalities in GT1a HCV-infected patients in GEODE-II and GT1 HCV-infected patients in PEARL II, III, & IV no-RBV and full-dose RBV arms

Table 4. Treatment-emergent Adverse Events

Adverse Event (AE)	N = 105 n (%)
Any AE	77 (73.3)
Serious AE*	3 (2.9)
AE leading to study drug D/C†	2 (1.9)
AE leading to RBV dose modification‡	2 (1.9)
Death	0
Common AEs (>10% frequency)	
Fatigue	29 (27.6)
Headache	14 (13.3)
Insomnia	12 (11.4)
Nausea	11 (10.5)

*One serious AE had a reasonable possibility of being study-drug related (Bipolar-I disorder). The patient reached SVR12.
†One AE had a reasonable possibility of being study-drug related (chest pain, heart rate increased, and dyspnea).
‡Decrease in hemoglobin.

Table 5. Post-baseline Laboratory Abnormalities

Lab parameters	N = 104 n (%)
Hemoglobin	
Grade 2 (<10–8 g/dL)	1 (1)
Grade 3+ (<8 g/dL)	0
Alanine aminotransferase	
Grade 2 (>3-5 × ULN)	1 (1)
Grade 3+ (>5 × ULN)	1 (1)
Aspartate aminotransferase	
Grade 2 (>3-5 × ULN)	0
Grade 3+ (>5 × ULN)	1 (1)
Total bilirubin	
Grade 2 (>1.5-3 × ULN)	5 (5)
Grade 3+ (>3 × ULN)	0

ULN = upper limit of the normal range.

Table 6. Comparison of Treatment-emergent Adverse Events and Laboratory Abnormalities in GT1a HCV-infected Patients in GEODE-II and GT1 HCV-infected Patients in PEARL II, III, and IV No-RBV and Full-dose RBV Arms

	GEODE-II	PEARL II,III & IV[4,5]	
Adverse Event (AE)	Low-Dose RBV N = 105 n (%)	No RBV N = 509 n (%)	Full-Dose RBV N = 401 n (%)
Any AE	77 (73.3)	383 (75.2)	332 (82.8)
Serious AE	3 (2.9)*	7 (1.4)†	9 (2.2)†
AE leading to study drug D/C	2 (1.9)	2 (0.4)	2 (0.5)
AE leading to RBV dose modification	2 (1.9)	0	34 (8.5)
Common AEs‡			
Fatigue	29 (27.6)	135 (26.5)	120 (29.9)
Headache	14 (13.3)	129 (25.3)	98 (24.4)
Insomnia	12 (11.4)	26 (5.1)	49 (12.2)
Nausea	11 (10.5)	43 (8.4)	63 (15.7)
Hemoglobin			
Grade 2 (<10–8 g/dL)	1 (1)§	0	23 (5.7)
Grade 3+ (<8 g/dL)	0	0	2 (0.5)
Total bilirubin			
Grade 3+ (>3 × ULN)	0	2 (0.4)	23 (5.7)

ULN = upper limit of the normal range.
*One AE had a reasonable possibility of being study-drug related.
†Two AEs had a reasonable possibility of being study-drug related.
‡AEs with >10% in GEODE-II.
§N = 104.

RESISTANCE-ASSOCIATED SUBSTITUTIONS

- The impact of NS3 Q80K and OBV-specific resistance-associated baseline polymorphisms was similar to that seen in GT1a-infected patients from the PEARL-IV study who were treated with OBV/PTV/r + DSV without RBV[6]
- **Figure 4** shows the prevalence and impact of baseline polymorphisms on GT1a-infected patients

Figure 4. Prevalence and Impact of Baseline Polymorphisms on GT1a-infected Patients

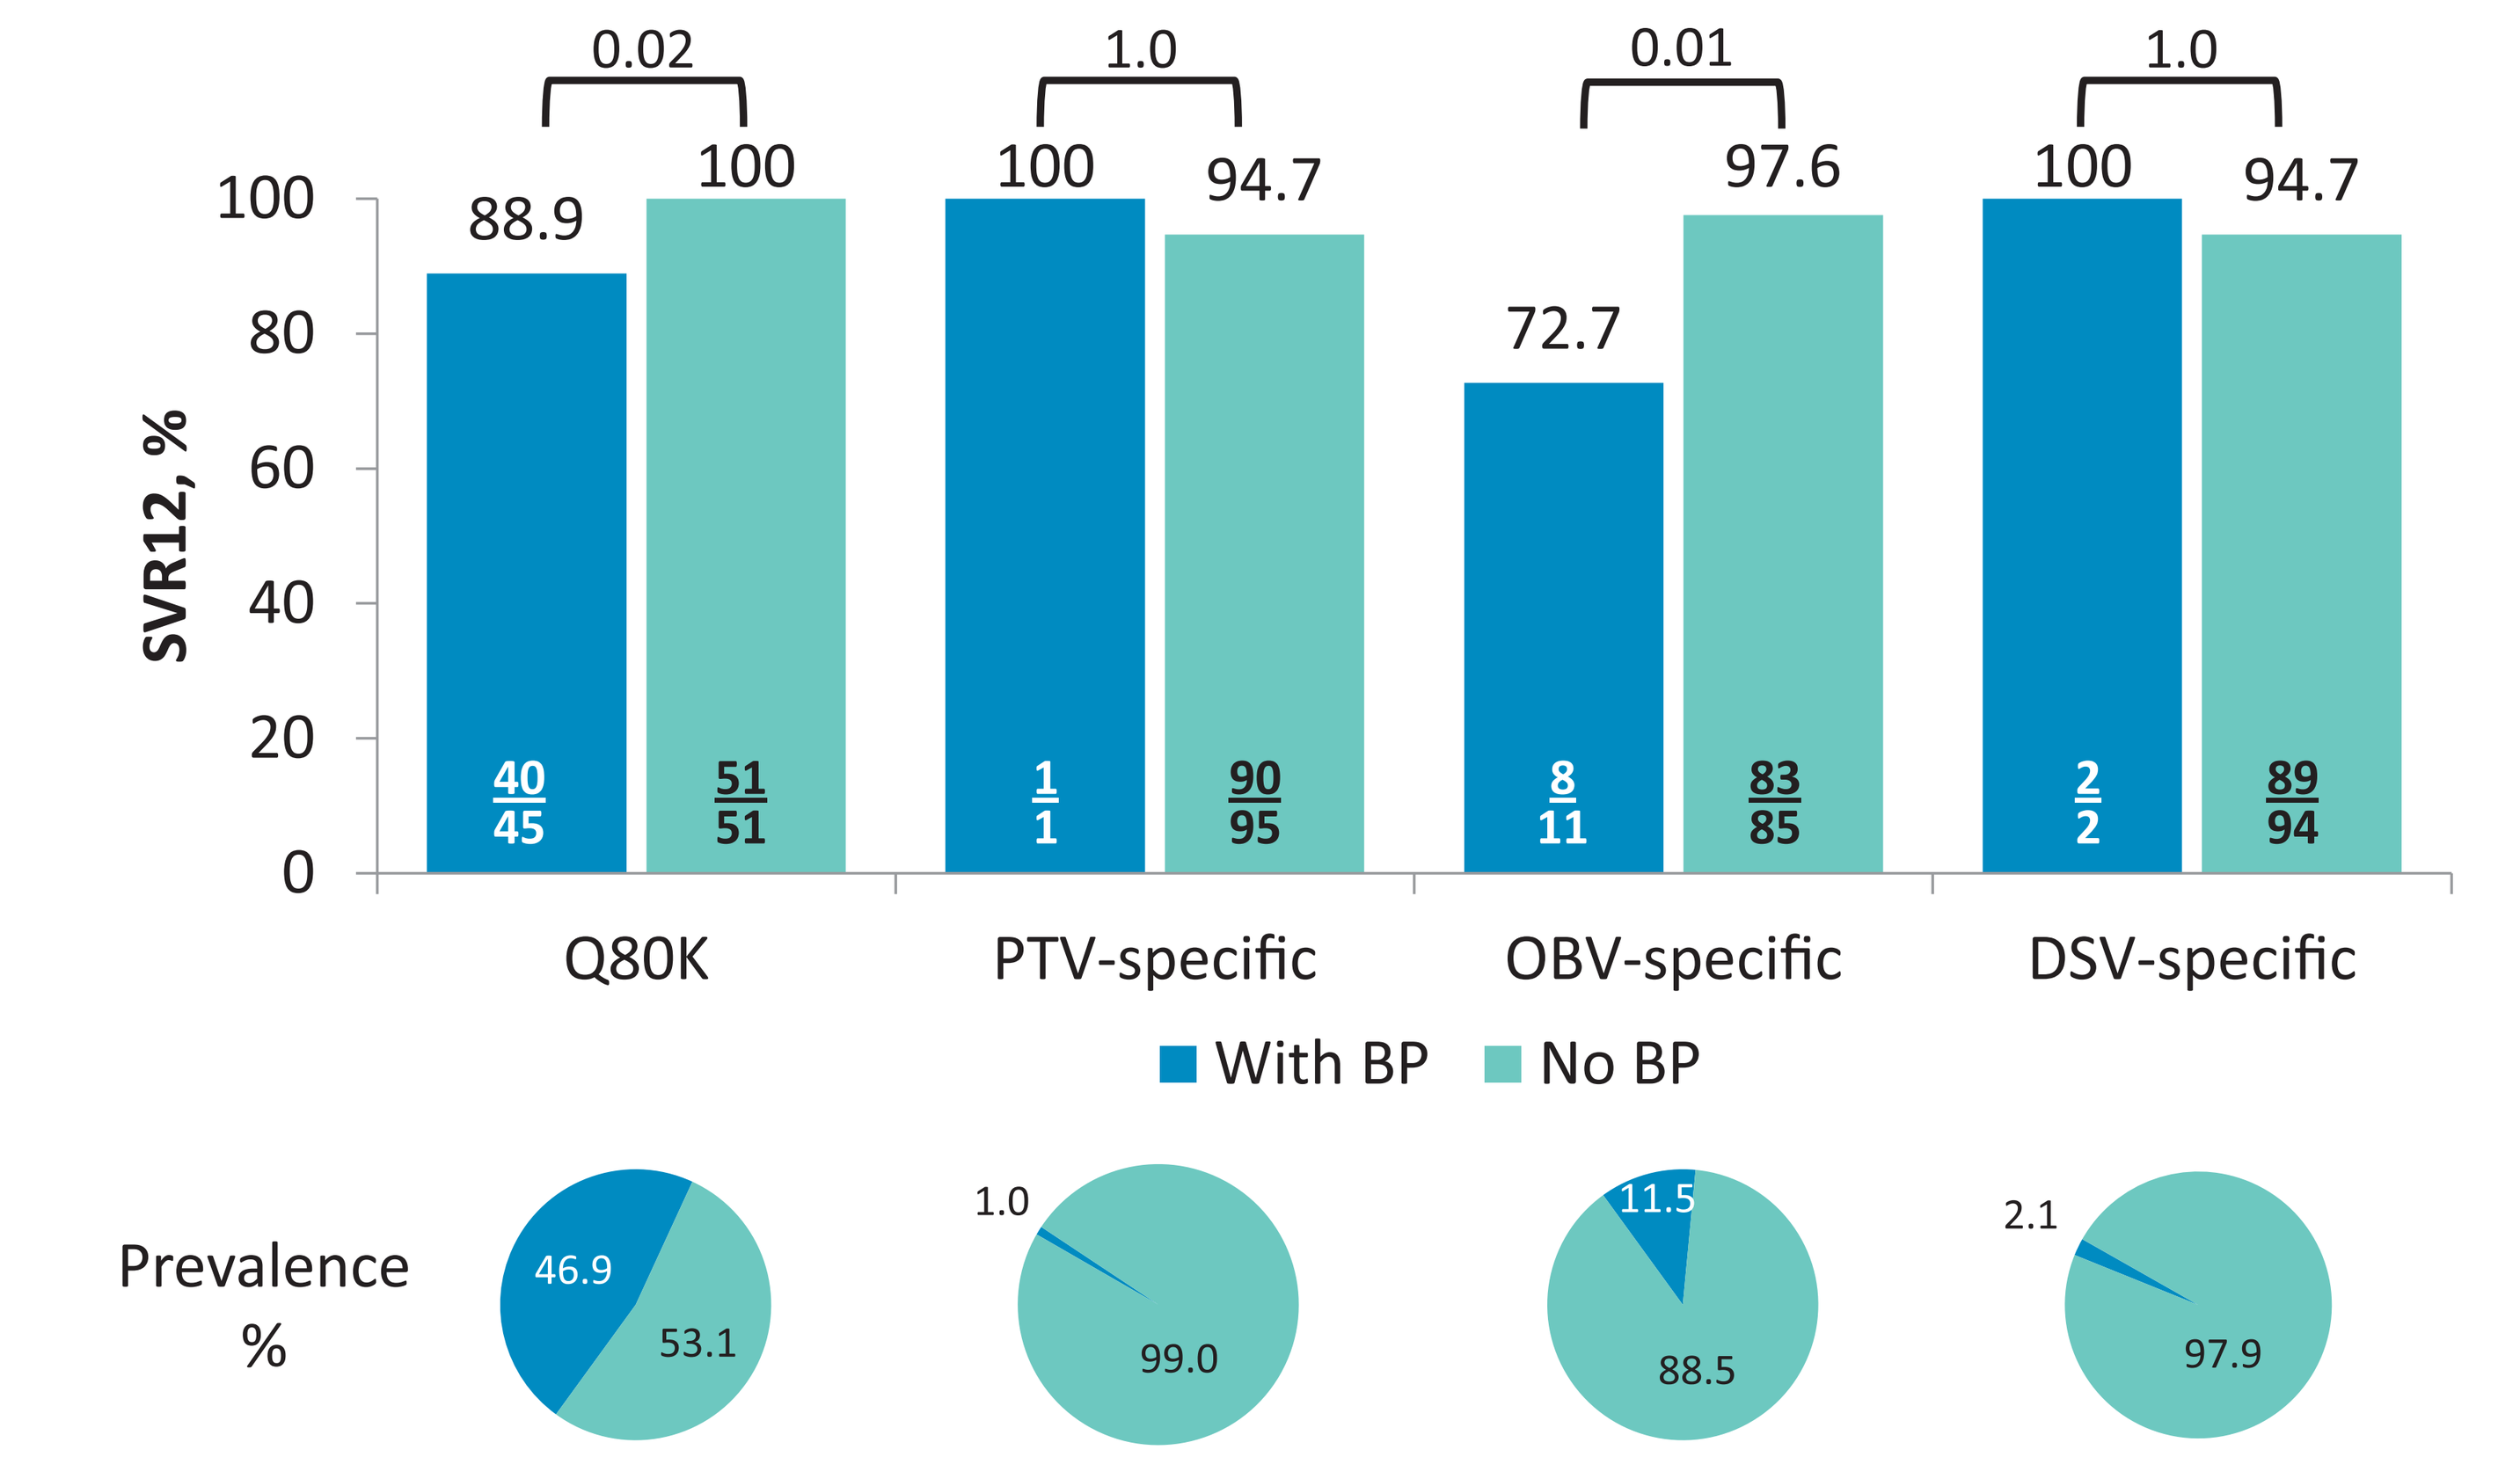
0.02
1.0
0.01
1.0
SVR12, %
100
80
60
40
20
0
88.9
100
100
94.7
72.7
97.6
100
94.7
40
45
51
51
1
1
90
95
8
11
83
85
2
2
89
94
Q80K
PTV-specific
OBV-specific
DSV-specific
With BP
No BP
Prevalence
%
46.9
53.1
1.0
99.0
11.5
88.5
2.1
97.9

Baseline Polymorphisms (BP)	Prevalence % (n/N)	SVR12 rate with BP % (n/N)	SVR12 rate without either BP, % (n/N)
Q80K + OBV-specific	7.3 (7/96)	57.1 (4/7)	
OBV-specific alone	4.2 (4/96)	100 (4/4)	100 (47/47)
Q80K alone	39.6 (38/96)	94.7 (36/38)	

PHARMACOKINETICS

- Trough plasma concentrations of OBV, PTV, r, DSV, and DSV M1 metabolite were generally comparable to the C_{trough} values observed in GT1-infected population receiving 12 weeks of OBV/PTV/r and DSV plus weight-based RBV (BID) in the phase 3 SAPPHIRE-I & II trials. **Table 7** shows RBV trough plasma concentrations
- With the low dose RBV (600 mg QD) in GEODE-II, RBV C_{trough} values (geometric mean: 1032 ng/mL) were approximately 52% to 55% lower than those values (geometric mean: 2170 ng/mL to 2280 ng/mL) from weight-based RBV (1000 mg or 1200 mg BID) regimens in SAPPHIRE-I & II trials
- For patients who had a virologic failure, the concentrations at each visit were generally within the range of concentrations from all patients (but at the lower end of the range for breakthrough patients)

Table 7. RBV Trough Plasma Concentrations From GEODE-II vs Trough Concentrations From SAPPHIRE-I & II

	GEODE-II		SAPPHIRE-I‖		SAPPHIRE-II‖	
Study Drug	N	Geometric mean (arithmetic mean, %CV)	N	Geometric mean (arithmetic mean, %CV)	N	Geometric mean (arithmetic mean, %CV)
Ribavirin*	49	1032 (1094, 36)	274	2280 (2370, 28)	193	2170 (2290, 29)

*Binned time interval of >10–14 hours for GEODE-II, and binned time interval of >22–26 hours for SAPPHIRE-I and II as RBV was dosed as BID.
CV, coefficient of variation.
‖Results from Arm A.

CONCLUSIONS

- SVR12 was achieved by 90% of patients in the ITT population and 95% in the mITT population. Non-inferiority was not established for the primary study end point
- The regimen was well tolerated with mostly mild or moderate AEs reported. Laboratory abnormalities were lower compared to historical controls:
 - Grade ≥2 hemoglobin AEs occurred in 1% of patients (compares favorably to 6.2% in historical controls who received full dose RBV)
 - No Grade ≥3 total bilirubin AEs occurred (compares favorably to 5.7% in historical controls who received full dose RBV)
- Patients with baseline polymorphisms showed numerically lower SVR
- Several patients failed to achieve SVR12 due to loss to follow-up, incarceration, and other underlying behavioral disorders. The patient population in this study shows a high rate of history of IV drug abuse, alcohol abuse, and behavioral disorders (66%, 81%, and 54%, respectively, see **Table 2**) suggesting that a patient support program may provide value to improve treatment adherence and overall clinical outcomes

REFERENCES

1. Webster DP, Klenerman P, Dusheiko GM. Hepatitis C. *Lancet*. 2015;385(9973):1124–35.
2. Messina JP, Humphreys I, Flaxman A, et al. Global distribution and prevalence of hepatitis C virus genotypes. *Hepatology*. 2015;61(1):77–87.
3. Feld JJ, Moreno C, Trinh R, et al. Sustained virologic response of 100% in HCV genotype 1b patients with cirrhosis receiving ombitasvir/paritaprevir/r and dasabuvir for 12 weeks. *J Hepatol*. 2016;64(2):301–7.
4. Ferenci P, Bernstein D, Lalezari J, et al. ABT-450/r-ombitasvir and dasabuvir with or without ribavirin for HCV. *N Engl J Med*. 2014;370(21):1983–92.
5. Andreone P, Colombo MG, Enejosa JV, et al. ABT-450, ritonavir, ombitasvir, and dasabuvir achieves 97% and 100% sustained virologic response with or without ribavirin in treatment-experienced patients with HCV genotype 1b infection. *Gastroenterology*. 2014;147(2):359–65 e351.
6. Sarrazin C, et a. Analysis of Baseline Variants in GT1a-infected Patients Treated with 3D with and without RBV, and Cirrhotic GT1a Patients Treated with 3D plus RBV for 12 or 24 Weeks. Presented at the European Association for the Study of the Liver Special Conference 23–24 September 2016, Paris, France.

ACKNOWLEDGEMENTS

Medical writing support was provided by Maher Quraan, PhD, of AbbVie.

DISCLOSURES

AbbVie sponsored the study (NCT0220708), contributed to its design, the collection, analysis, and interpretation of the data, and participated in the writing, review, and approval of the poster. All authors had access to relevant data.

F Poordad: Grant/Research Support: AbbVie, Achillion Pharmaceuticals, Anadys Pharmaceuticals, Biolex Therapeutics, Boehringer Ingelheim, Bristol-Myers Squibb, Genentech, Gilead Sciences, GlaxoSmithKline, GlobeImmune, Idenix Pharmaceuticals, Idera Pharmaceuticals, Intercept Pharmaceuticals, Janssen, Medarex, Medtronic, Merck, Novartis, Santaris Pharmaceuticals, Scynexis Pharmaceuticals, Vertex Pharmaceuticals, ZymoGenetics. Speaker: Gilead, Kadmon, Merck, Onyx/Bayer, Genentech, GlaxoSmithKline, Salix, Vertex. Consultant/Advisor: AbbVie, Achillion Pharmaceuticals, Anadys Pharmaceuticals, Biolex Therapeutics, Boehringer Ingelheim, Bristol-Myers Squibb, Gilead Sciences, GlaxoSmithKline, GlobeImmune, Idenix, Merck, Novartis, Tibotec/Janssen, Theravance, Vertex. **S Sedghi:** Nothing to disclose. **P Pockros:** Speaker/Consultant/Advisor: Gilead, AbbVie, Janssen, Bristol-Myers Squibb; Research Support: Gilead, AbbVie, Janssen, Bristol-Myers Squibb, Merck, Conatus, Roche Molecular. **N Ravendhran:** AbbVie, Gilead, Merck, Salix, BMS; Research: BMS, Gilead, Merck, Salix. **R Reindollar:** Research Support: AbbVie, BMS, Cepheid, Gilead, Intercept, Janssen; Consultant/Speaker: AbbVie, BMS, Gilead, Janssen. **M Lucey:** Grant/Research Support: AbbVie, Gilead, Salix. **M Epstein:** Consultant Eli Lilly, IMHS, Aspire Bariatric. **L Bank:** Nothing to disclose. **D Bernstein:** Consulting: Merck; Grant/Research Support: GIlead, Pharmasset, Vertex, BMS; Speaking and Teaching: Gilead. **R Trinh, P Krishnan, T Pilot-Matias, A Polepally, R Pothacamury, K Unnebrink, M Martinez:** Employees of AbbVie and may hold stock or stock options. **D Nelson:** Grant/Research Support: Abbot, BMS, Beohringer Ingelheim, Gilead, Genentech, Merck, Bayer, Idenix, Vertex, Jansen.

abbvie

44588-161469-Poordad — AASLD 2016 — AbbVie — proof 1 (rjh) — october 31, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051

CREATED @ 100%

Ombitasvir, Paritaprevir, Ritonavir, Dasabuvir and Ribavirin Pharmacokinetics in HCV-Infected Subjects with Chronic Kidney Disease Stage 4 (Severe Renal Impairment) or Stage 5 (End-Stage Renal Disease)

Diana L. Shuster[1], Rajeev M. Menon[1], Bifeng Ding[2], Hong Li[2], Eric Cohen[3], Daniel E. Cohen[3], Sandeep Dutta[4], Jiuhong Zha[1]

[1] Clinical Pharmacokinetics and Pharmacodynamics, AbbVie, North Chicago, IL, USA, [2] Biometrics, AbbVie, North Chicago, IL, USA, [3] Infectious Disease Development, AbbVie, North Chicago, IL, USA, [4] Clinical Pharmacology and Modeling & Simulation, Amgen, Thousand Oaks, CA, USA

2545396

Presented at The Liver Meeting® 2016, AASLD Annual Meeting, November 11 - November 15, 2016 in Boston, MA

BACKGROUND

- AbbVie's all-oral IFN-free, direct-acting antiviral (DAA) regimens include ombitasvir (OBV; NS5A inhibitor), paritaprevir (PTV, NS3/4A protease inhibitor identified by AbbVie and Enanta), ritonavir (RTV) and dasabuvir (DSV, NS5B non-nucleoside polymerase inhibitor) ± ribavirin (RBV) for treatment of hepatitis C virus (HCV) genotype (GT) 1 infection, and OBV/PTV/RTV + RBV for treatment of HCV GT4 infection.
- HCV is a particular problem among hemodialysis patients, in whom seroprevalence rates as high as 23% have been reported in the US.[1]
- OBV, PTV, RTV and DSV exposures were not meaningfully altered in HCV-uninfected subjects with mild, moderate or severe renal impairment (evaluated chronic kidney disease [CKD] Stages 1-4).[2]
- Two ongoing Phase 3b studies, RUBY-I and RUBY-II, evaluated the safety, pharmacokinetics, and efficacy of the 3-DAA (OBV/PTV/RTV + DSV) and 2-DAA (OBV/PTV/RTV) regimens with and without RBV in HCV GT1- or GT4-infected subjects with Stage 4 CKD or Stage 5 end-stage renal disease (ESRD) without cirrhosis or with compensated cirrhosis.[3,4] To date, subjects from RUBY-I and RUBY-II studies have SVR_{12} rates > 90%.[5,6,7]

OBJECTIVE

To assess the pharmacokinetics (PK) of OBV, PTV, RTV, DSV and RBV in HCV GT1- or GT4-infected subjects with Stage 4 CKD (estimated glomerular filtration rate [eGFR] < 30 - 15 mL/min) or ESRD (eGFR < 15 mL/min or requiring dialysis).

METHODS

Data from two clinical studies in HCV GT1- or GT4-infected subjects were combined for this analysis.[3,4]

Study NCT02207088 (RUBY-I) is an ongoing, open label study evaluating the safety and efficacy of the 3-DAA regimen ± RBV for 12 or 24 weeks in HCV GT1-infected, treatment naïve or previous pegIFN/RBV treatment-experienced subjects without cirrhosis or with compensated cirrhosis with Stage 4 CKD or ESRD.[3] The treatment regimens for RUBY-I are shown below:

Cohort	Arm	N	Genotype	Cirrhosis Status	Treatment[a]	Treatment Duration (Weeks)
1	A	13	1a	without (-) cirrhosis	3-DAA + RBV	12
	B	7	1b	without (-) cirrhosis	3-DAA	12
2	C	28	1a	without (-) cirrhosis	3-DAA + RBV	12
	D	8	1a	with (+) cirrhosis	3-DAA + RBV	24
	E	11	1b	with or without (±) cirrhosis	3-DAA	12

a. 3-DAA (OBV/PTV/RTV 25/150/100 mg QD + DSV 250 mg BID) ± RBV 200 mg QD

METHODS

Study NCT02487199 (RUBY-II) is an ongoing, open label study evaluating the safety and efficacy of the DAA regimens without RBV for 12 weeks in HCV GT1a- or GT4-infected, treatment naïve or previous pegIFN/RBV treatment-experienced non-cirrhotic subjects with Stage 4 CKD or ESRD.[4] The treatment regimens for RUBY-II are shown below:

Arm	N	Genotype	Cirrhosis Status	Treatment[a]	Treatment Duration (Weeks)
1	13	1a	without (-) cirrhosis	3-DAA	12
2	5	4	without (-) cirrhosis	2-DAA	12

a. 3-DAA (OBV/PTV/RTV 25/150/100 mg QD + DSV 250 mg BID); 2-DAA (OBV/PTV/RTV 25/150/100 mg QD)

Pharmacokinetic Sample Collection in RUBY-I and RUBY-II

For all subjects in both studies, single plasma PK samples were collected prior to dosing (0 hour) on Study Day 1 and without regard to time of dosing at all other study visits during treatment period. Subjects could participate in the optional intensive PK sampling on Study Week 4 (open to Stage 4 CKD subjects, ESRD subjects on peritoneal dialysis or ESRD subjects on a non-dialysis day for those receiving hemodialysis) and on the following day, Study Week 4 + 1 day (open to ESRD subjects on hemodialysis only, on the day of a hemodialysis session [dialysis day]). On the Week 4 + 1 day visit, arterial (pre-dialyzer) and venous (post-dialyzer) samples were also collected to evaluate the extraction of DAAs, RTV and RBV during hemodialysis.

Pharmacokinetic Evaluations

The following analyses were completed to evaluate the PK of OBV, PTV, DSV, RTV and RBV:

- Comparison of Stage 4 CKD and ESRD plasma concentrations across all study visits
- Comparison of trough plasma concentrations (Ctrough) (>22 to 26 hours post-dose for QD administered drugs, OBV, PTV, RTV and RBV; >10 to 14 hours post-dose for BID administered DSV) in HCV-infected subjects with normal renal function/mild renal impairment (eGFR ≥ 60 mL/min), Stage 4 CKD or ESRD
- Estimation of individual PK exposures (area under the curve [AUC] and peak plasma concentrations [C_{max}]) on Week 4 and Week 4 + 1 day using non-compartmental analysis (NCA) of intensive PK data
- Estimation of venous/arterial ratios during hemodialysis session on Week 4 + 1 day visit

RESULTS

Subjects

Subject demographics are presented by Study in **Table 1**. Generally there were more treatment-naïve, non-cirrhotic males with ESRD on hemodialysis in both studies.

Table 1. Subject Demographics

Demographic Characteristic	Study NCT02207088 (RUBY-I) N = 67	Study NCT02487199 (RUBY-II) N=18
Sex, N (%)		
Male	56 (84)	12 (67)
Female	11 (16)	6 (33)
Age (years)		
Median (range)	59 (32 - 77)	57 (34 – 76)
Body Weight (kg)		
Median (range)	83 (40 - 137)	80 (50 – 111)
Race, N (%)		
White	27 (40)	15 (83)
Black or African American	40 (60)	3 (17)
Ethnicity, N (%)		
Hispanic or Latino	16 (24)	1 (6)
Other	51 (76)	17 (94)
Cirrhosis, N (%)		
With Compensated Cirrhosis	6 (9)	0 (0)
Without Cirrhosis	61 (91)	18 (100)
Treatment History, N (%)		
Treatment Naïve	57 (85)	15 (83)
Treatment Experienced	10 (15)	3 (17)
Renal Function, N (%)		
Stage 4 CKD	13 (20)	1 (5)
ESRD not on dialysis	1 (1)	0 (0)
ESRD on peritoneal dialysis	7 (10)	5 (28)
ESRD on hemodialysis	46 (69)	12 (67)

N = Number of subjects; CKD = Chronic Kidney Disease; ESRD = End-Stage Renal Disease

Plasma Concentrations of DAAs, RTV and RBV Are Similar Between Stage 4 CKD and ESRD Subjects

Plasma concentrations across all study visits for OBV, PTV, RTV, DSV and RBV are generally comparable between subjects with Stage 4 CKD or ESRD (**Figure 1**).

Figure 1. DAAs, RTV and RBV Plasma Concentration Data Across Study Visits in HCV-Infected Subjects with Stage 4 CKD (Red) or ESRD (Light Blue)

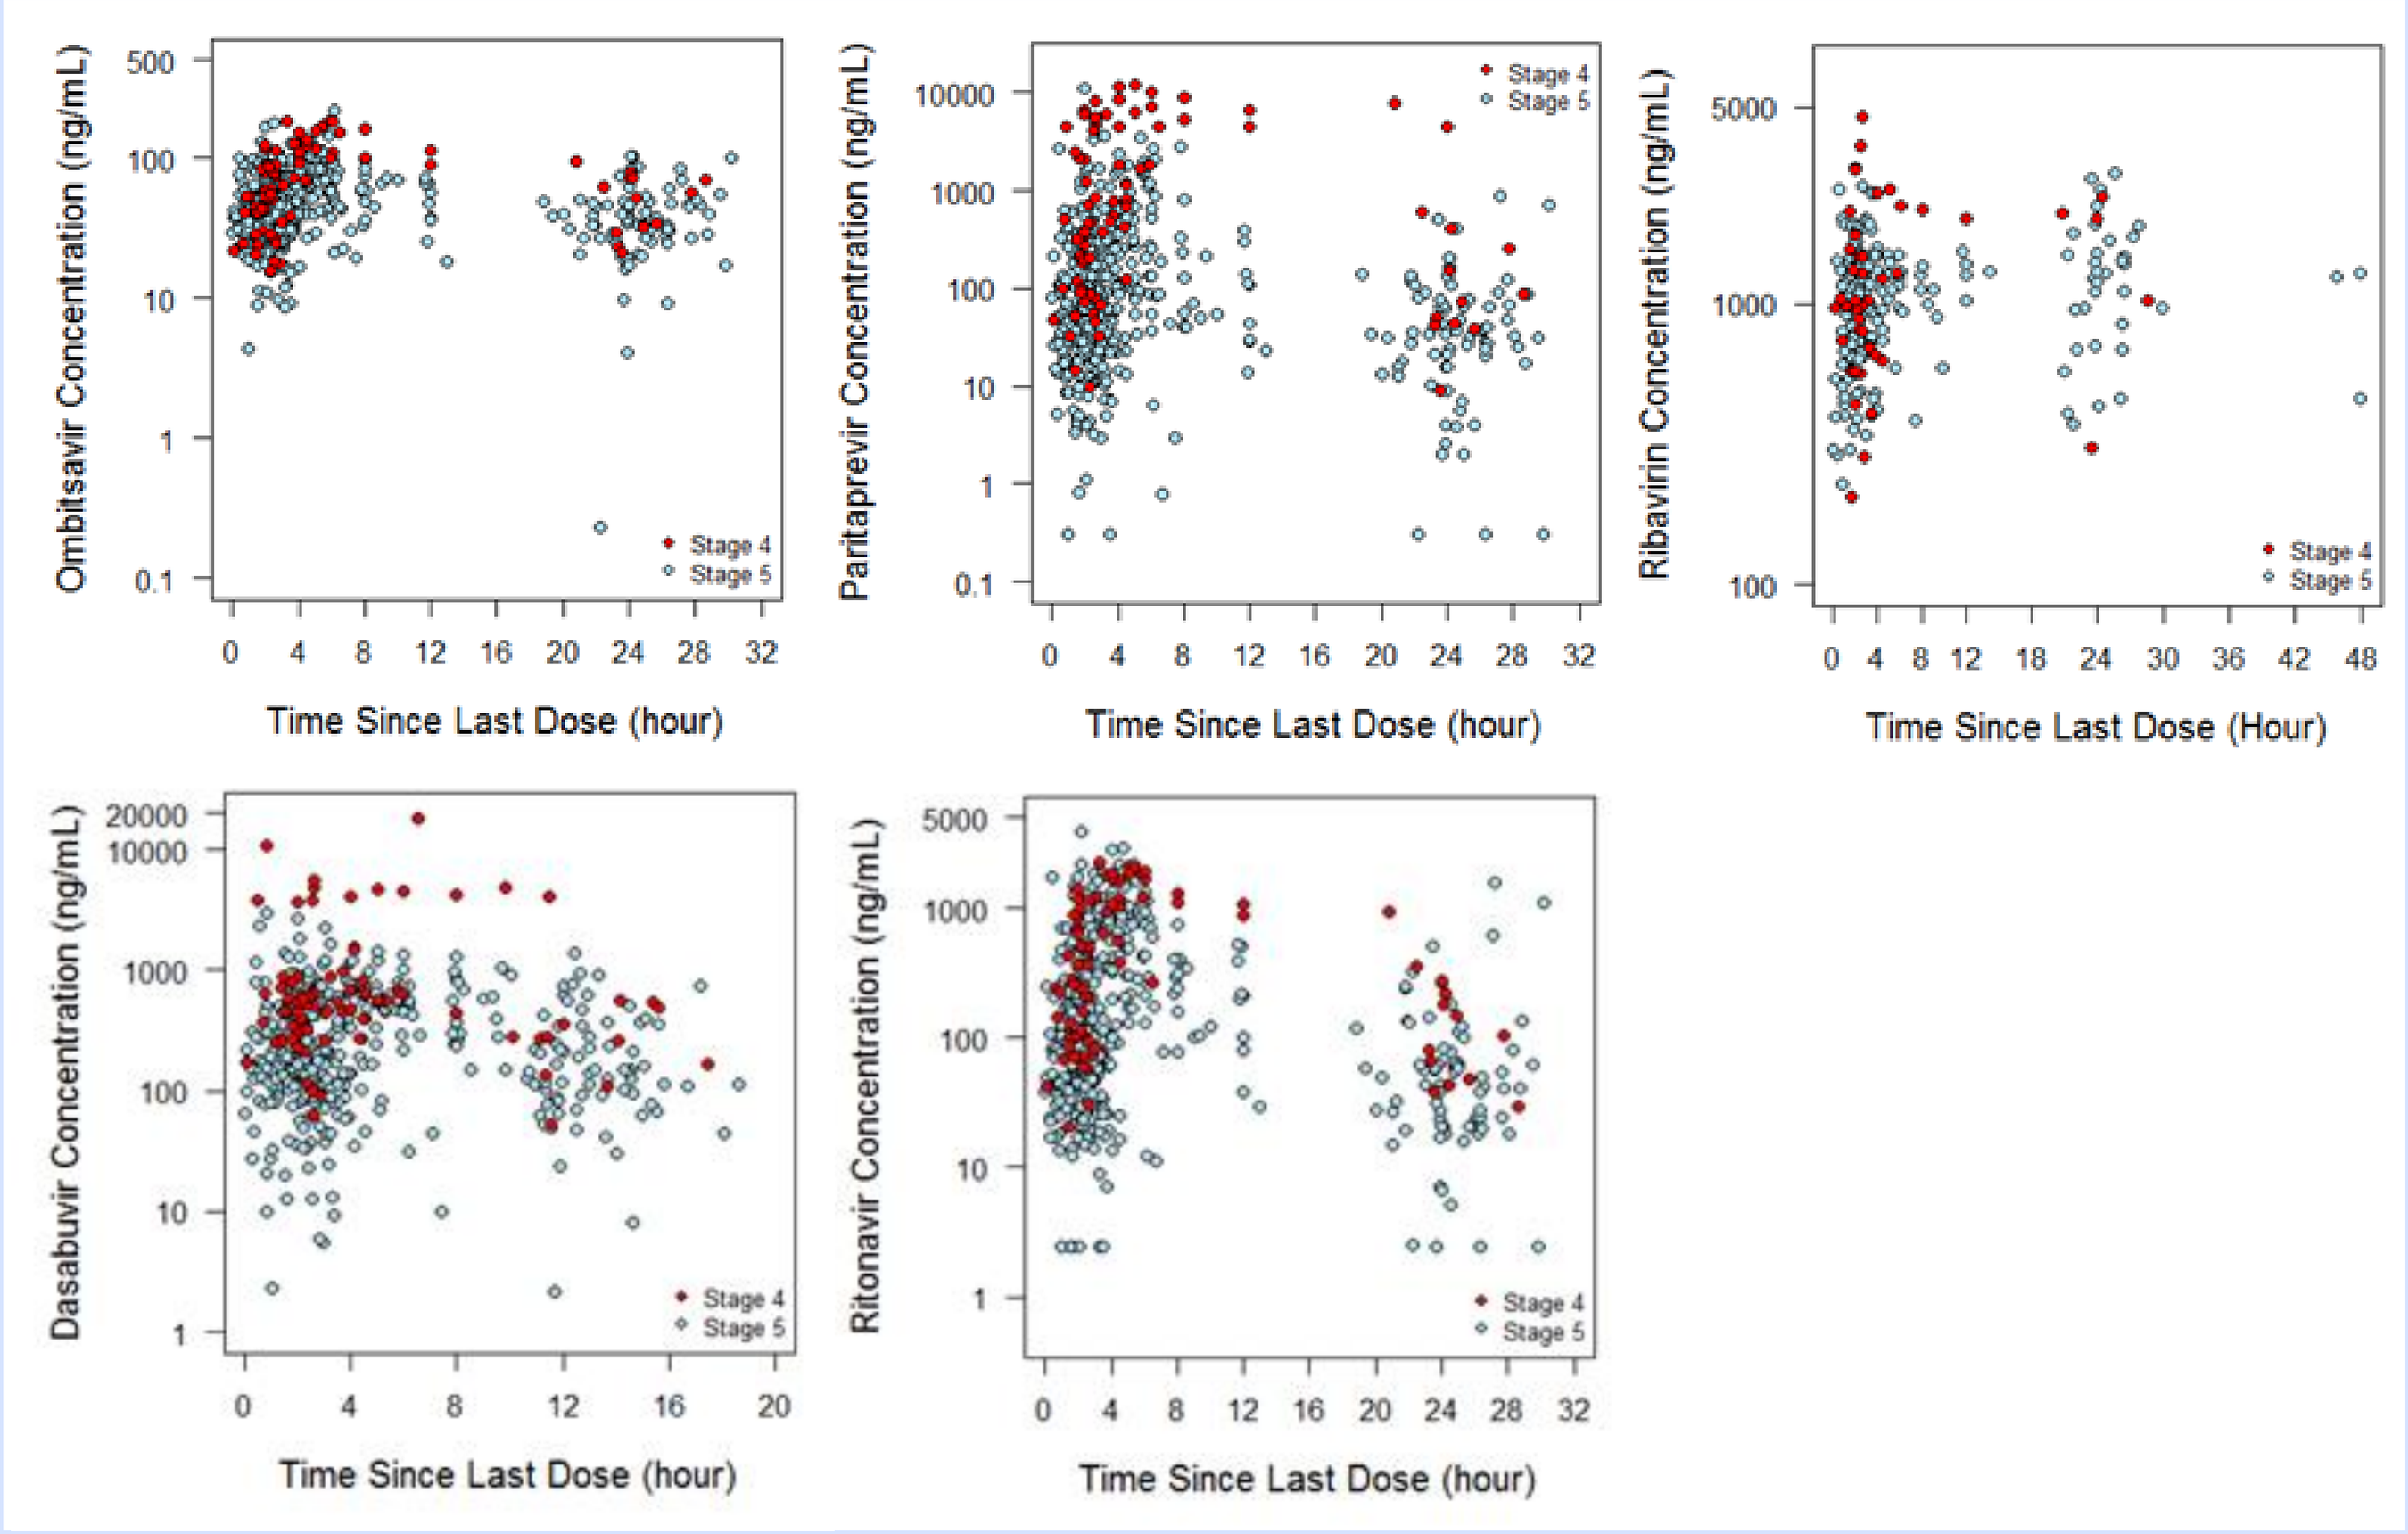
Ombitasvir Concentration (ng/mL)
Paritaprevir Concentration (ng/mL)
Ribavirin Concentration (ng/mL)
Dasabuvir Concentration (ng/mL)
Ritonavir Concentration (ng/mL)
Time Since Last Dose (hour)
Time Since Last Dose (Hour)
Stage 4
Stage 5

C_{trough} Values in HCV-Infected Subjects with Stage 4 CKD or ESRD Are Similar to Those with Normal/Mild Renal Impairment

C_{trough} values of OBV, PTV, RTV, DSV and RBV for RUBY-I and RUBY-II subjects were within the range of subjects with normal renal function or mild renal impairment across eight Phase 3/3b studies in non-cirrhotic and cirrhotic HCV-infected subjects (**Figure 2**).

Figure 2. Individual C_{trough} Values for DAAs, RTV and RBV from RUBY-I, RUBY-II and Phase 3/3b Studies

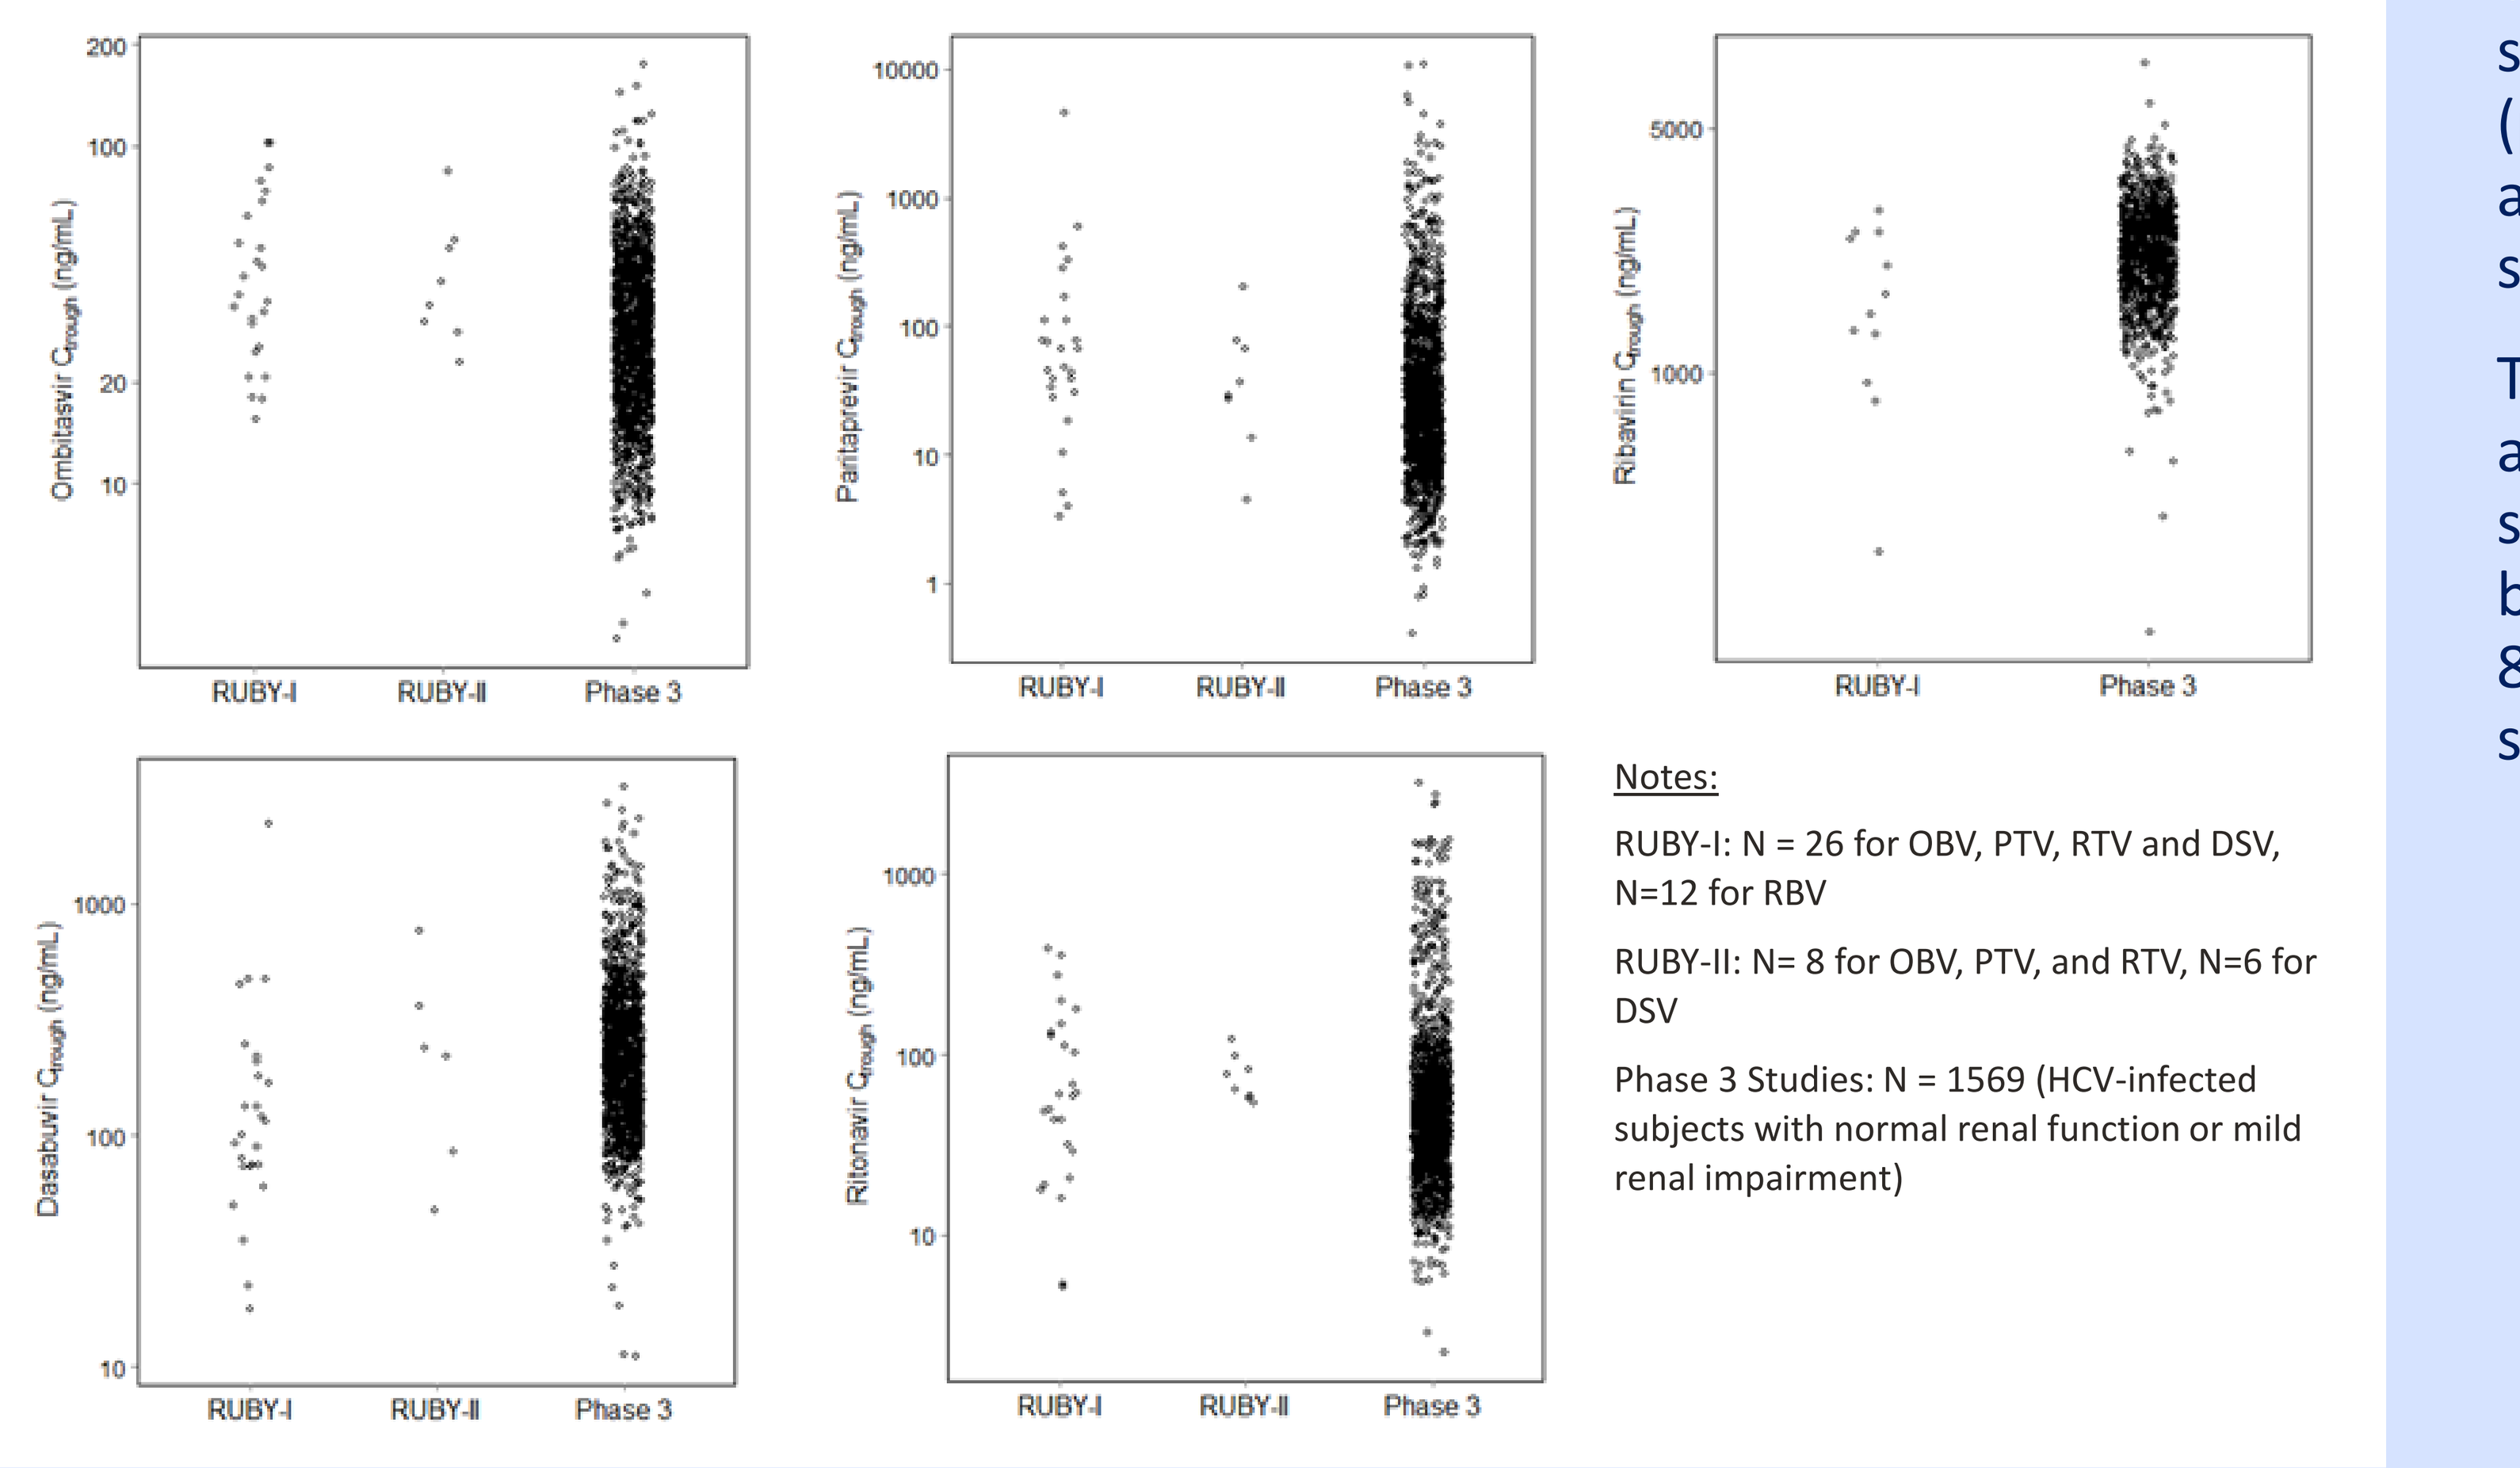
Ombitasvir C_{trough} (ng/mL)
Paritaprevir C_{trough} (ng/mL)
Ribavirin C_{trough} (ng/mL)
Dasabuvir C_{trough} (ng/mL)
Ritonavir C_{trough} (ng/mL)
RUBY-I
RUBY-II
Phase 3

Notes:

RUBY-I: N = 26 for OBV, PTV, RTV and DSV, N=12 for RBV

RUBY-II: N= 8 for OBV, PTV, and RTV, N=6 for DSV

Phase 3 Studies: N = 1569 (HCV-infected subjects with normal renal function or mild renal impairment)

Exposures (C_{max} and AUC) in HCV-Infected Subjects with Stage 4 CKD or ESRD Are Similar to Those with Normal/Mild Renal Impairment

Ten subjects participated in the intensive PK assessment, 3 with Stage 4 CKD, 2 with ESRD from a non-dialysis day (Week 4 Visit), and 4 with ESRD from both non-dialysis and dialysis days. Exposures (C_{max} and AUC) of OBV, PTV, RTV, and DSV in the Stage 4 CKD and ESRD subjects were compared to the range of individual intensive PK exposures obtained in HCV-infected subjects with normal renal function or mild renal impairment (across three Phase 2/3b studies that had intensive PK assessment), and are presented in **Figure 3**. Individual OBV, PTV, RTV, DSV and RBV exposures were generally within the range of those observed in HCV-infected subjects with normal renal function or mild renal impairment.

Figure 3. Cross-Study Comparison of Individual C_{max} and AUC Values for OBV, PTV, RTV and DSV in Subjects with Stage 4 CKD, ESRD, or Normal/Mild Renal Impairment

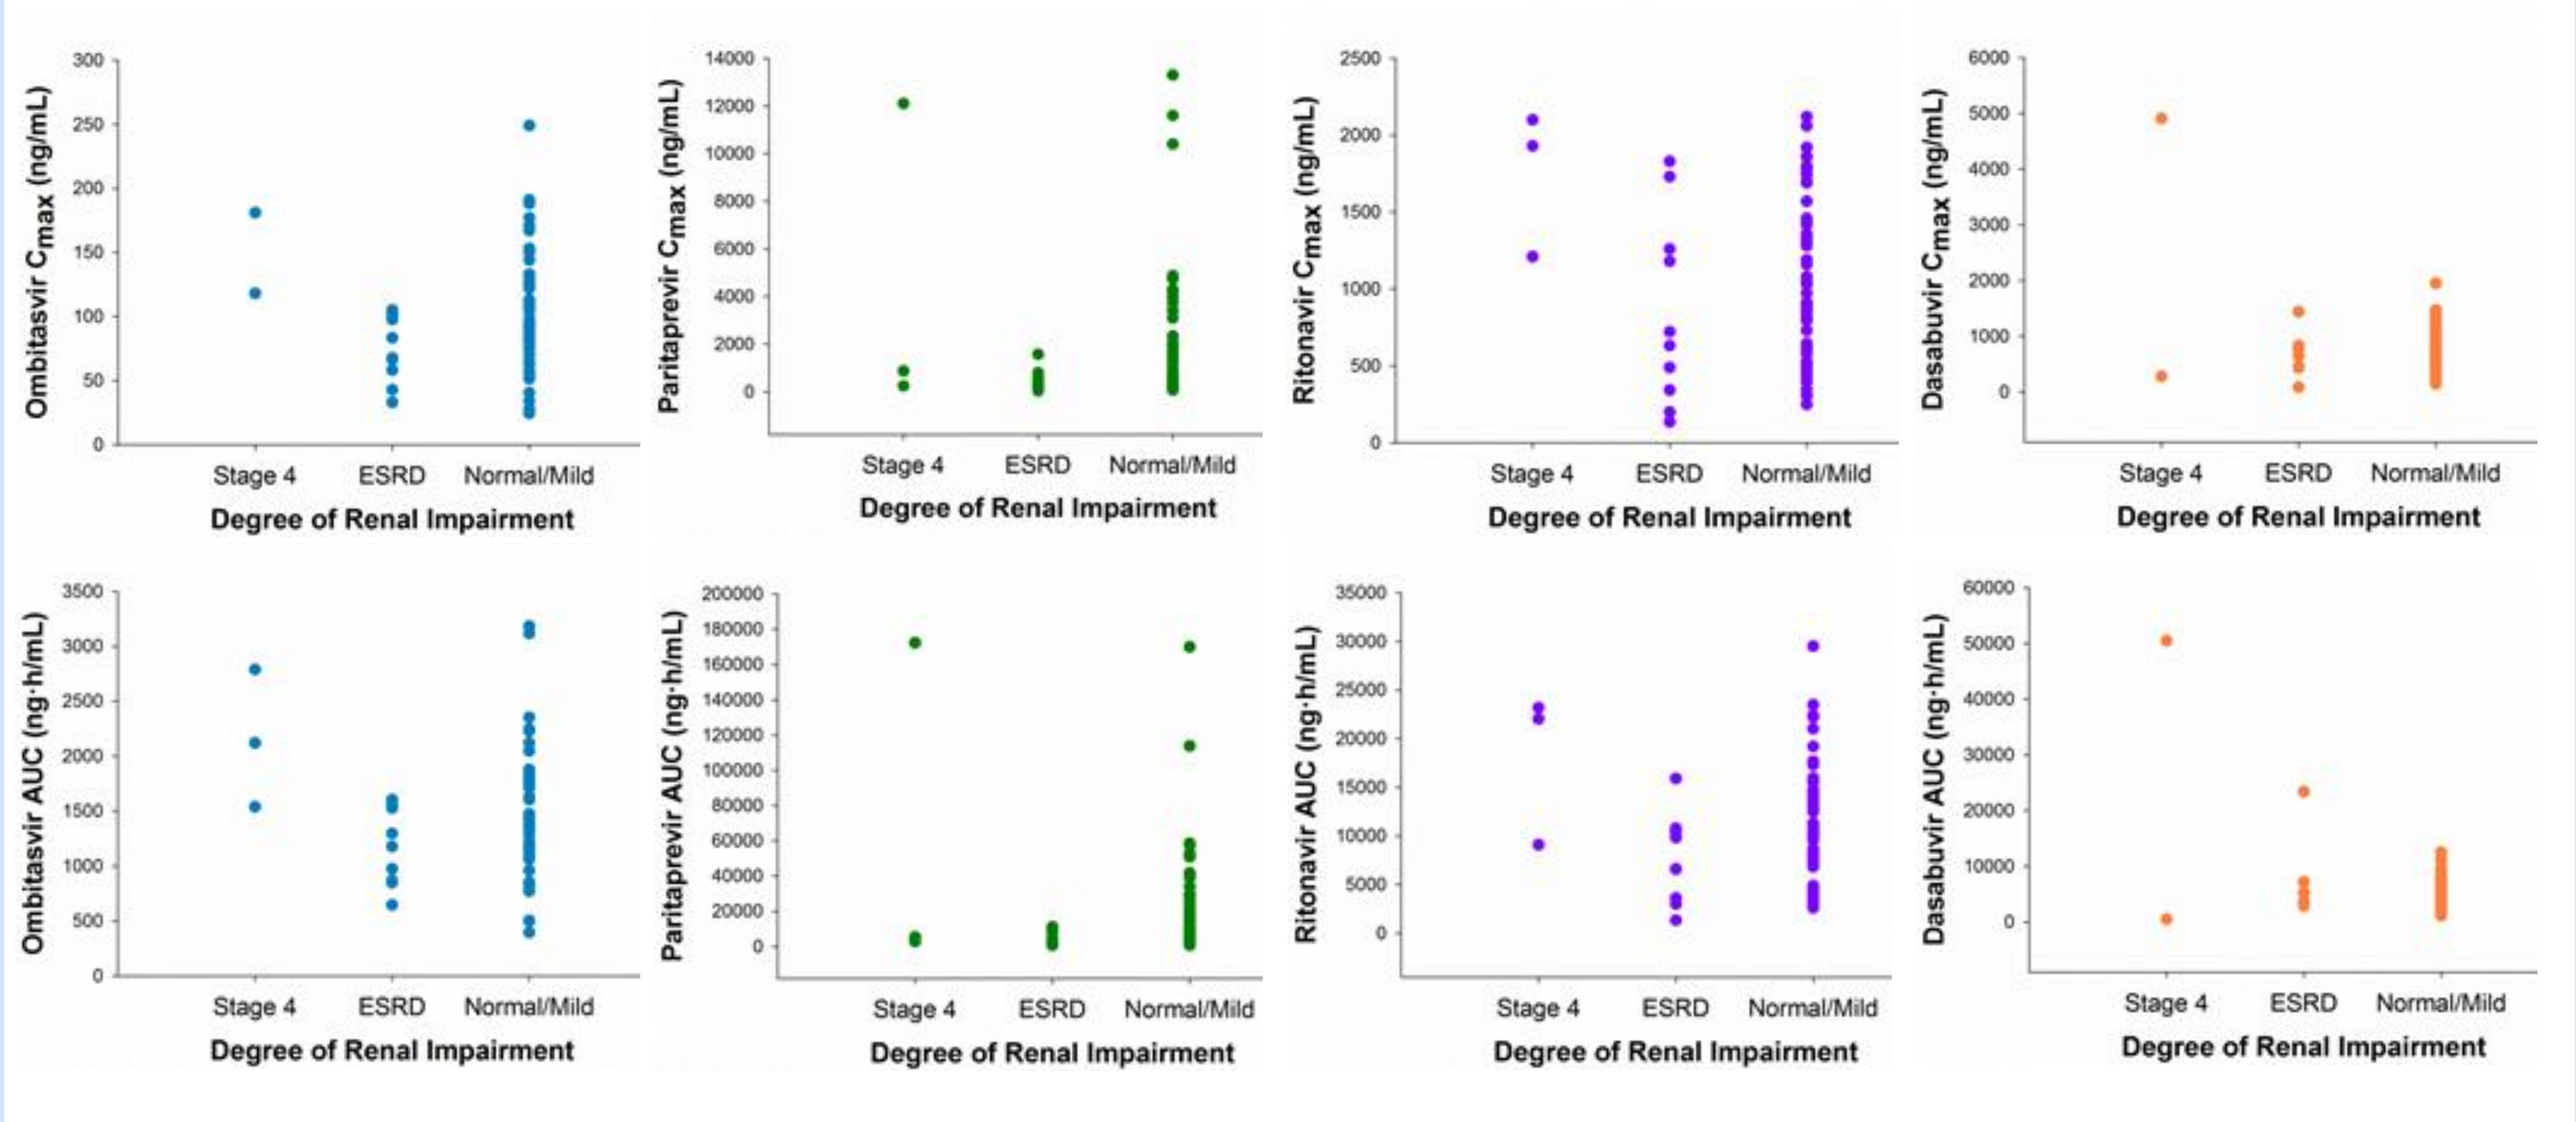
Ombitasvir C_{max} (ng/mL)
Paritaprevir C_{max} (ng/mL)
Ritonavir C_{max} (ng/mL)
Dasabuvir C_{max} (ng/mL)
Ombitasvir AUC (ng·h/mL)
Paritaprevir AUC (ng·h/mL)
Ritonavir AUC (ng·h/mL)
Dasabuvir AUC (ng·h/mL)
Stage 4
ESRD
Normal/Mild
Degree of Renal Impairment

Notes: C_{max} and AUC values for subjects with Stage 4 CKD or ESRD are combined from RUBY-I and RUBY-II. For the ESRD group, C_{max} and AUC values are combined from non-dialysis and dialysis days. C_{max} and AUC values for subjects with normal renal function or mild renal impairment are combined from Phase 2/3b studies.

Hemodialysis Does Not Remove DAAs and RTV from the Bloodstream

Two hemodialysis subjects from RUBY-I completed intensive PK sampling on a dialysis day (Week 4 + 1 Day visit). Venous (post-dialyzer) and arterial (pre-dialyzer) concentrations of DAAs, RTV and RBV were simultaneously collected during hemodialysis, which started approximately 5 hours after the morning dose.

There were no clinically meaningful decreases in venous versus arterial concentrations (≤ 17% decrease) for OBV, PTV, RTV, and DSV, suggesting that hemodialysis does not extract DAAs or RTV from the bloodstream. RBV venous concentrations in one subject were 70% to 81% lower than arterial concentrations, consistent with published data stating that ribavirin is extracted by hemodialysis.[8]

CONCLUSIONS

- The exposures of OBV, PTV, RTV and DSV in HCV-infected subjects with Stage 4 CKD or ESRD were comparable to the exposures observed in HCV-infected subjects with normal renal function or mild renal impairment.
- OBV, PTV, RTV and DSV plasma concentrations are not notably affected during hemodialysis.
- These data support the conclusion that no dose adjustment is necessary for the 2-DAA and 3-DAA regimens when administered to HCV infected subjects with Stage 4 CKD or ESRD.
- RBV exposures based on 200 mg QD dosing in Stage 4 CKD and ESRD subjects were comparable with exposures observed in HCV-infected subjects without Stage 4 CKD or ESRD receiving the regular full dose of ribavirin.[8]

REFERENCES

1. Sivapalasingam S, Malak SF, Sullivan JF, et al. High prevalence of hepatitis C infection among patients receiving hemodialysis at an urban dialysis center. Infect Control Hosp Epidemiol. 2002;23(6):319-24.
2. Khatri A, Dutta S, Marbury T, et al. The Pharmacokinetics and Safety of the Direct Acting Antiviral Regimen of ABT-450/r, Ombitasvir with/without Dasabuvir in Subjects with Mild, Moderate and Severe Renal Impairment Compared to Subjects with Normal Renal Function. The Liver Meeting 2014, Boston.
3. An Open-Label Study to Evaluate the Safety and Efficacy of Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir With or Without Ribavirin (RBV) in Adults With Genotype 1 Chronic Hepatitis C Virus (HCV) Infection, With Severe Renal Impairment or End-Stage Renal Disease (RUBY-I). Clinicaltrials.gov identifier: NCT02207088.
4. An Open-Label Study to Evaluate the Safety and Efficacy of Ombitasvir/Paritaprevir/Ritonavir With or Without Dasabuvir in Adults With Genotype 1a or Genotype 4 Chronic Hepatitis C Virus (HCV) Infection, With Severe Renal Impairment or End-Stage Renal Disease (RUBY-II). Clinicaltrials.gov identifier: NCT02487199.
5. Pockros P, Reddy KR, Mantry P. RUBY-1: Ombitasvir/Paritaprevir/Ritonavir + Dasabuvir +/- Ribavirin in Non-Cirrhotic HCV Genotype 1-Infected Patients with Severe Renal Impairment or End-Stage Renal Disease. AASLD, San Francisco, November, 2015.
6. Vierling J, Lawitz E. RUBY-I: Safety and Efficacy of Ombitasvir/Paritaprevir/ Ritonavir and Dasabuvir with or without Ribavirin in Adults with Genotype 1 Chronic Hepatitis C Virus (HCV) Infection, with Severe Renal Impairment or End-Stage Renal Disease. AASLD, Boston, November 2016.
7. Gane E, et al. RUBY-II: Efficacy and Safety of a Ribavirin-free Ombitasvir/Paritaprevir/Ritonavir ± Dasabuvir Regimen in Patients with Severe Renal Impairment or End-Stage Renal Disease and HCV Genotypes 1a or 4 Infection. AASLD, Boston, November 2016.
8. COPEGUS® (ribavirin) [package insert]. South San Francisco, CA; Hoffmann-La Roche, Inc. c/o Genentech, Inc., 2015.

ACKNOWLEDGEMENTS

The authors kindly acknowledge Subba Rao for creating the datasets used for pharmacokinetic analysis, and the investigators and clinical study teams who contributed to the ongoing efforts of Studies NCT02207088 and NCT02487199.

DISCLOSURES

These studies were funded by AbbVie. AbbVie contributed to the study design, research, and interpretation of data, writing, reviewing, and approving the publication. All authors except SD are AbbVie employees and may hold AbbVie stocks or options. SD is a former AbbVie employee and may own Abbvie stock or stock options.

RUBY-I: Safety and Efficacy of Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir With or Without Ribavirin in Adults With Genotype 1 Chronic Hepatitis C Virus (HCV) Infection With Severe Renal Impairment or End-Stage Renal Disease

John M Vierling[1], Eric Lawitz[2], K Rajender Reddy[3], Eric Cohen[4], Nyingi Kemmer[5], Giuseppe Morelli[6], Philippe J Zamor[7], Michael Bennett[8], David Bernstein[9], Kris Kowdley[10], Parvez S Mantry[11], Paul J Pockros[12], David Wyles[13], Sonal Kumar[14], Kalyan Ram Bhamidimarri[15], Daniel E Cohen[4], Tami Pilot-Matias[4], Zhenzhen Zhang[4], Thomas Podsadecki[4], Tarek Hassanein[16]

[1]Baylor College of Medicine; Advanced Liver Therapies, Houston, Texas, United States; [2]The Texas Liver Institute, University of Texas Health Science Center, San Antonio, Texas, United States; [3]University of Pennsylvania, Philadelphia, Pennsylvania, United States; [4]AbbVie Inc., North Chicago, Illinois, United States; [5]Tampa General Medical Group, Tampa, Florida, United States; [6]University of Florida Health Science Center, Gainesville, Florida, United States; [7]Carolinas Medical Center, Charlotte, North Carolina, United States; [8]Medical Associates Research Group, San Diego, California, United States; [9]North Shore University Hospital, Manhasset, New York, United States; [10]Swedish Medical Center, Seattle, Washington, United States; [11]The Liver Institute at Methodist Dallas, Dallas, Texas, United States; [12]Scripps Clinic, San Diego, California, United States; [13]University of California San Diego, La Jolla, California, United States; [14]Weill Cornell Medical College, New York, New York, United States; [15]University of Miami, Miami, Florida, United States; [16]Southern California GI and Liver Centers and Southern California Research Center, Coronado, California, United States

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

INTRODUCTION

- Ombitasvir/paritaprevir/ritonavir + dasabuvir (OBV/PTV/r + DSV; "3D regimen") is approved for the treatment of GT1a (with RBV) and GT1b HCV infection, including patients with compensated cirrhosis[1]
- However, only limited clinical data have been published on the safety and efficacy of this regimen in patients with severe renal dysfunction, including those on dialysis
- In the first cohort of the RUBY-I study, observed rates of sustained virologic response (intent-to-treat population) at post-treatment Week 12 (SVR12) were 90% (18/20) in treatment-naïve patients with GT1 infection without cirrhosis[2]
- Here we present safety and efficacy data from the second cohort of RUBY-I, which includes patients with compensated cirrhosis and those who failed prior peginterferon/RBV treatment

Multi-targeted 3 Direct-acting Antiviral Regimen (3-DAA)

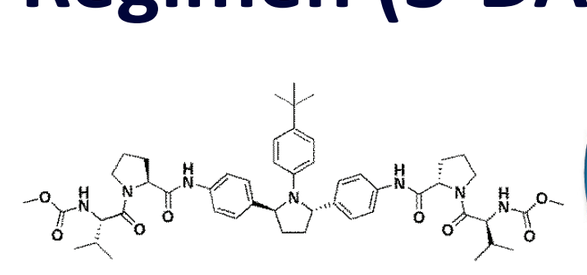

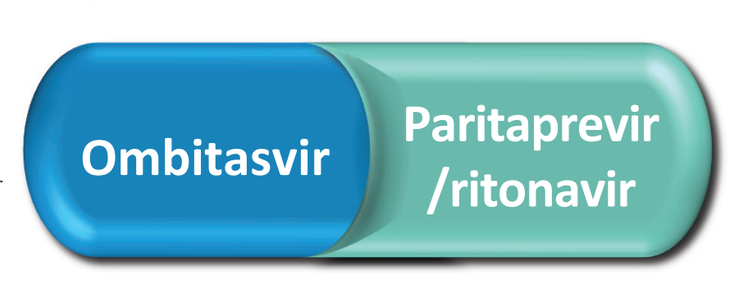
Ombitasvir
Paritaprevir /ritonavir

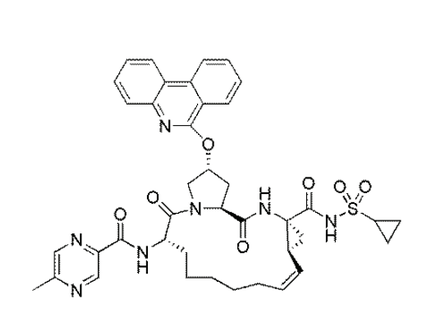

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)

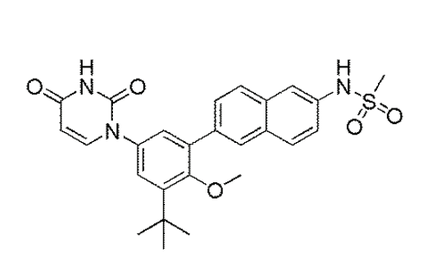

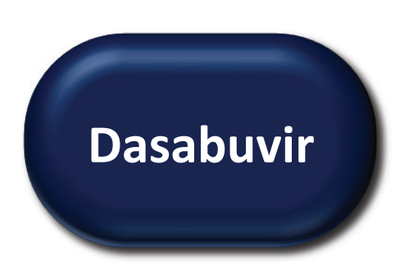
Dasabuvir

Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

PTV was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVE

- Evaluate the safety and efficacy of OBV/PTV/r + DSV ± RBV for 12 or 24 weeks in GT1 HCV-infected patients with severe renal impairment or end-stage renal disease, including those on dialysis

METHODS

STUDY DESIGN

- RUBY-I is a phase 3b, open-label, multi-center study that enrolled patients with CKD stages 4 or 5. Study design and interim findings from Cohort 1 have been presented previously

Figure 1. Study Design for RUBY-I, Cohort 2

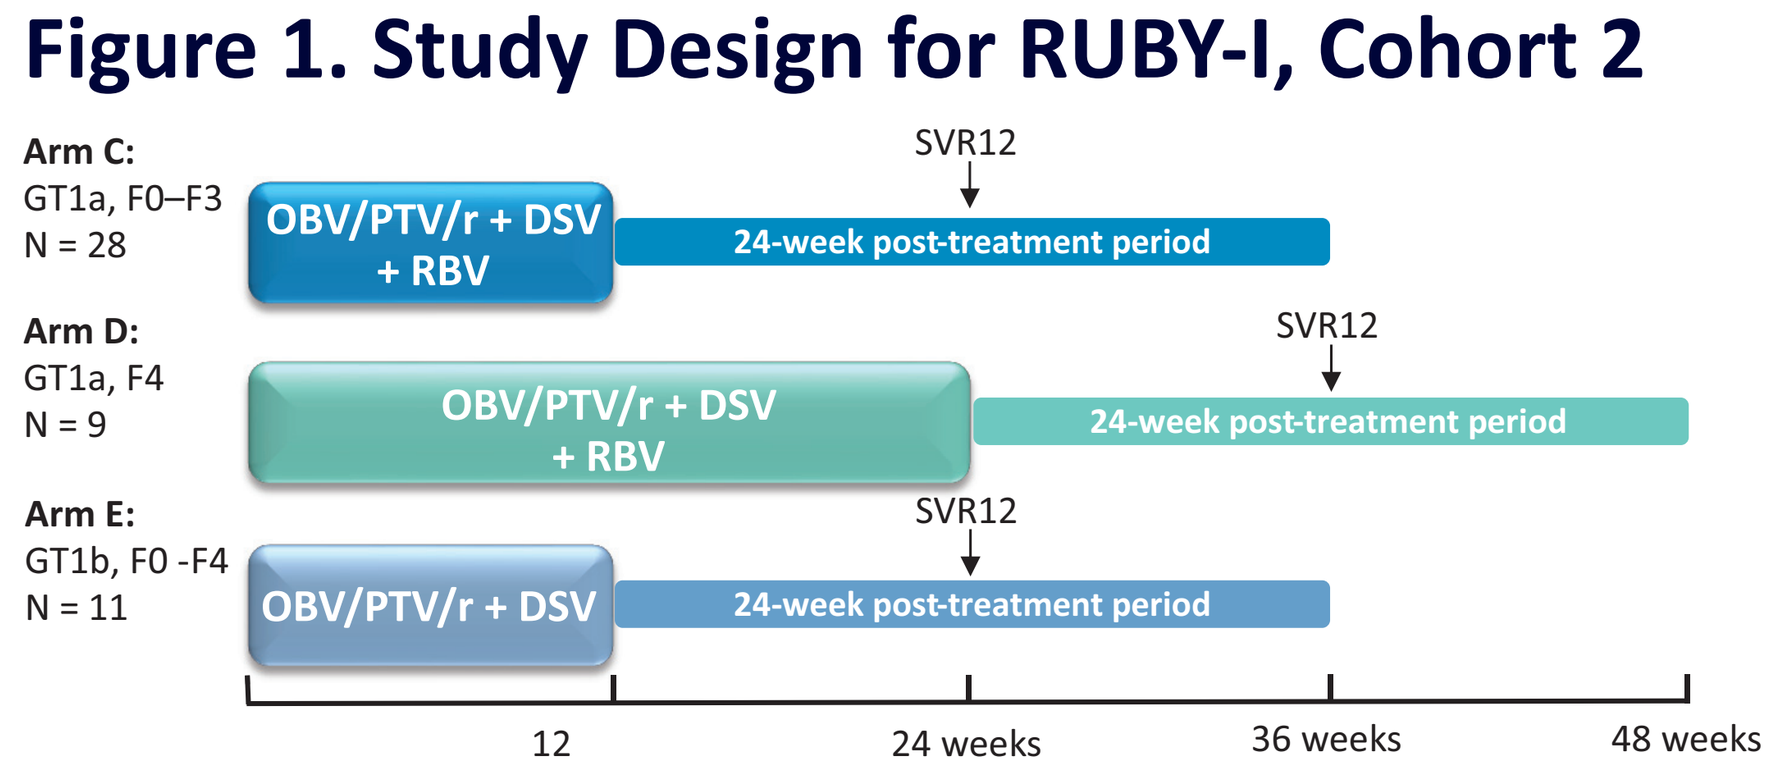
Arm C:
GT1a, F0–F3
N = 28
OBV/PTV/r + DSV + RBV
SVR12
24-week post-treatment period
Arm D:
GT1a, F4
N = 9
OBV/PTV/r + DSV + RBV
SVR12
24-week post-treatment period
Arm E:
GT1b, F0 -F4
N = 11
OBV/PTV/r + DSV
SVR12
24-week post-treatment period
12
24 weeks
36 weeks
48 weeks

METHODS (CONTINUED)

KEY ELIGIBILITY CRITERIA

Key inclusion criteria:

- Male or female at least 18 years of age at time of screening
- Chronic GT1 HCV-infection (HCV RNA >1000 IU/mL)
- Treatment-naïve or treatment-experienced (IFN/pegIFN ± RBV)
- Estimated Glomerular Filtration Rate (eGFR) <30 mL/min/1.73 m^2

Key exclusion criteria:

- Presence of HCV genotype other than GT1
- Current or past evidence of Child Pugh B or C cirrhosis
- Co-infection with the hepatitis B virus or human immunodeficiency virus
- Any primary cause of liver disease other than chronic HCV infection
- Abnormal laboratory result that meets at least 1 the following criteria (see **Table 1**)

Criteria for presence or absence of cirrhosis:

- Absence of cirrhosis was determined using 1 of the following criteria:
 - Liver biopsy demonstrating absence of cirrhosis (eg, Metavir Score of ≤3 or an Ishak score of ≤4)
 - FibroTest score of ≤0.72 and Aspartate Aminotransferase to Platelet Ratio Index (APRI) ≤2
 - FibroScan result of <12.5 kPa
- Presence of cirrhosis was determined using any 1 of the following criteria:
 - Liver biopsy demonstrating cirrhosis (eg, Metavir Score of 4 or an Ishak score of 5–6)
 - FibroScan score ≥14.6 kPa within 6 months of screening or during the screening period
 - FibroTest >0.72 and APRI >2
- In the absence of a qualifying liver biopsy, patients with a screening FibroScan result that is ≥12.5 kPa and <14.6 kPa, a FibroTest result that was ≤0.72 and an APRI >2, or a FibroTest result that was >0.72 and an APRI ≤2, should be evaluated based on the investigator's clinical judgment to determine the presence or absence of cirrhosis

Table 1. Abnormal Laboratory Results Exclusion Criteria

Assessment	Value
Albumin	<2.8 g/dL
International normalized ratio (INR)†	>2.3
Hemoglobin	<10 g/dL
Platelets	<25 000 cells per mm^3
Total Bilirubin	≥3.0 mg/dL

†Subjects with a known inherited blood disorder and INR >2.3 may have been enrolled with permission of the AbbVie study designated physician.

STUDY ASSESSMENTS

Primary Efficacy Endpoint	Percentage of patients with SVR12
Secondary Efficacy Endpoints	Percentage of patients with on-treatment virologic failure and with post-treatment relapse
Safety Assessment	Percentage of patients with treatment-emergent adverse events (AEs) or laboratory abnormalities

RESULTS

PARTICIPANTS

- 48 patients were enrolled at 15 study sites in the US
 - 31% (15/48) of patients had compensated cirrhosis
 - 83% (40/48) of patients had CKD stage 5, and 69% (33/48) were on dialysis

Table 2. Baseline Demographics and Disease Characteristics of Patients Enrolled in this Study

	Arm C GT1a, F0–F3 n = 28	Arm D GT1a, F4 n = 9	Arm E GT1b, F0–F4 n = 11	Total N = 48
Male, n (%)	23 (82)	9 (100)	8 (73)	40 (83)
Race*, n (%)				
White	12 (43)	3 (33)	4 (36)	19 (40)
Black	16 (57)	4 (44)	6 (55)	26 (54)
Other	0	2 (22)	1 (9)	3 (6)
Hispanic or Latino ethnicity*, n (%)	9 (32)	2 (22)	3 (27)	14 (29)
Age, median (range), years	59 (32–76)	56 (44–64)	58 (50–77)	58 (32–77)
BMI, mean ± SD, kg/m^2	27.9 ± 5.9	27.3 ± 4.8	25.0 ± 3.2	27.1 ± 5.2
IL28B non-CC genotype, n (%)	24 (86)	5 (56)	8 (73)	37 (77)
HCV RNA, median (range), $\log_{10}$ IU/mL	6.2 (5.0–7.7)	6.0 (5.3–7.4)	5.8 (3.3–7.3)	6.2 (3.3–7.7)
Fibrosis stage				
F0–F1	14 (50)	0	3 (27)	17 (35)
F2	9 (32)	0	1 (9)	10 (21)
F3	5 (18)	0	1 (9)	6 (13)
F4	0	9 (100)	6 (55)	15 (31)
Chronic kidney disease				
Stage 4	4 (14)	2 (22)	2 (18)	8 (17)
Stage 5	24 (86)	7 (78)	9 (82)	40 (83)
On hemodialysis	18 (64)	7 (78)	8 (73)	33 (69)
Prior IFN/RBV treatment experience, n/N (%)				
Treatment-naïve	24/28 (86)	6/9 (67)	8/11 (73)	38/48 (79)
Null responder	3/4 (75)	1/3 (33)	2/3 (67)	6/10 (60)
Relapser	0	1/3 (33)	1/3 (33)	2/10 (20)
Other	1/4 (25)	1/3 (33)	0	2/10 (20)
Creatinine clearance, mean ± SD, (mg/dL)	15.0 ± 8.9	17.9 ± 8.3	15.0 ± 5.8	15.6 ± 8.1
Albumin, median (range), g/dL	4.0 (3.1–4.8)	4.4 (3.3–4.7)	4.1 (3.2–4.7)	4.1 (3.1 –4.8)
Hemoglobin, median (range), g/dL	11.8 (9.9–16.4)	11.6 (10.1–12.9)	12.3 (10.7–13.7)	11.8 (9.9–16.4)
Total bilirubin, median (range), mg/dL	0.4 (0.2–0.8)	0.6 (0.5–1.4)	0.5 (0.3–0.8)	0.5 (0.2–1.4)
Platelet count, median (range), ×10^9/L	199 (104–346)	127 (73–186)	171 (58–292)	176 (58–346)

BMI = body mass index. SD = standard deviation.
*Race and ethnicity are self-reported.

RESULTS (CONTINUED)

EFFICACY

- SVR12 was achieved by 46/48 (96%) patients (27/28 in Arm C, 8/9 in Arm D, and 11/11 in Arm E) **(Figure 1)**
- Of the 2 patients not achieving SVR12, 1 patient in Arm C had on-treatment breakthrough and 1 patient in Arm D discontinued study drug after 6 days of treatment due to an AE
 - The subject with on-treatment breakthrough reportedly stopped taking all study drugs on Day 73 of treatment
 - The subject who discontinued prematurely had an AE of volvulus assessed by the investigator as not related to study drug

Figure 2. SVR12 Rates for the 3 Groups of Patients Enrolled in this Study as Well as the Total

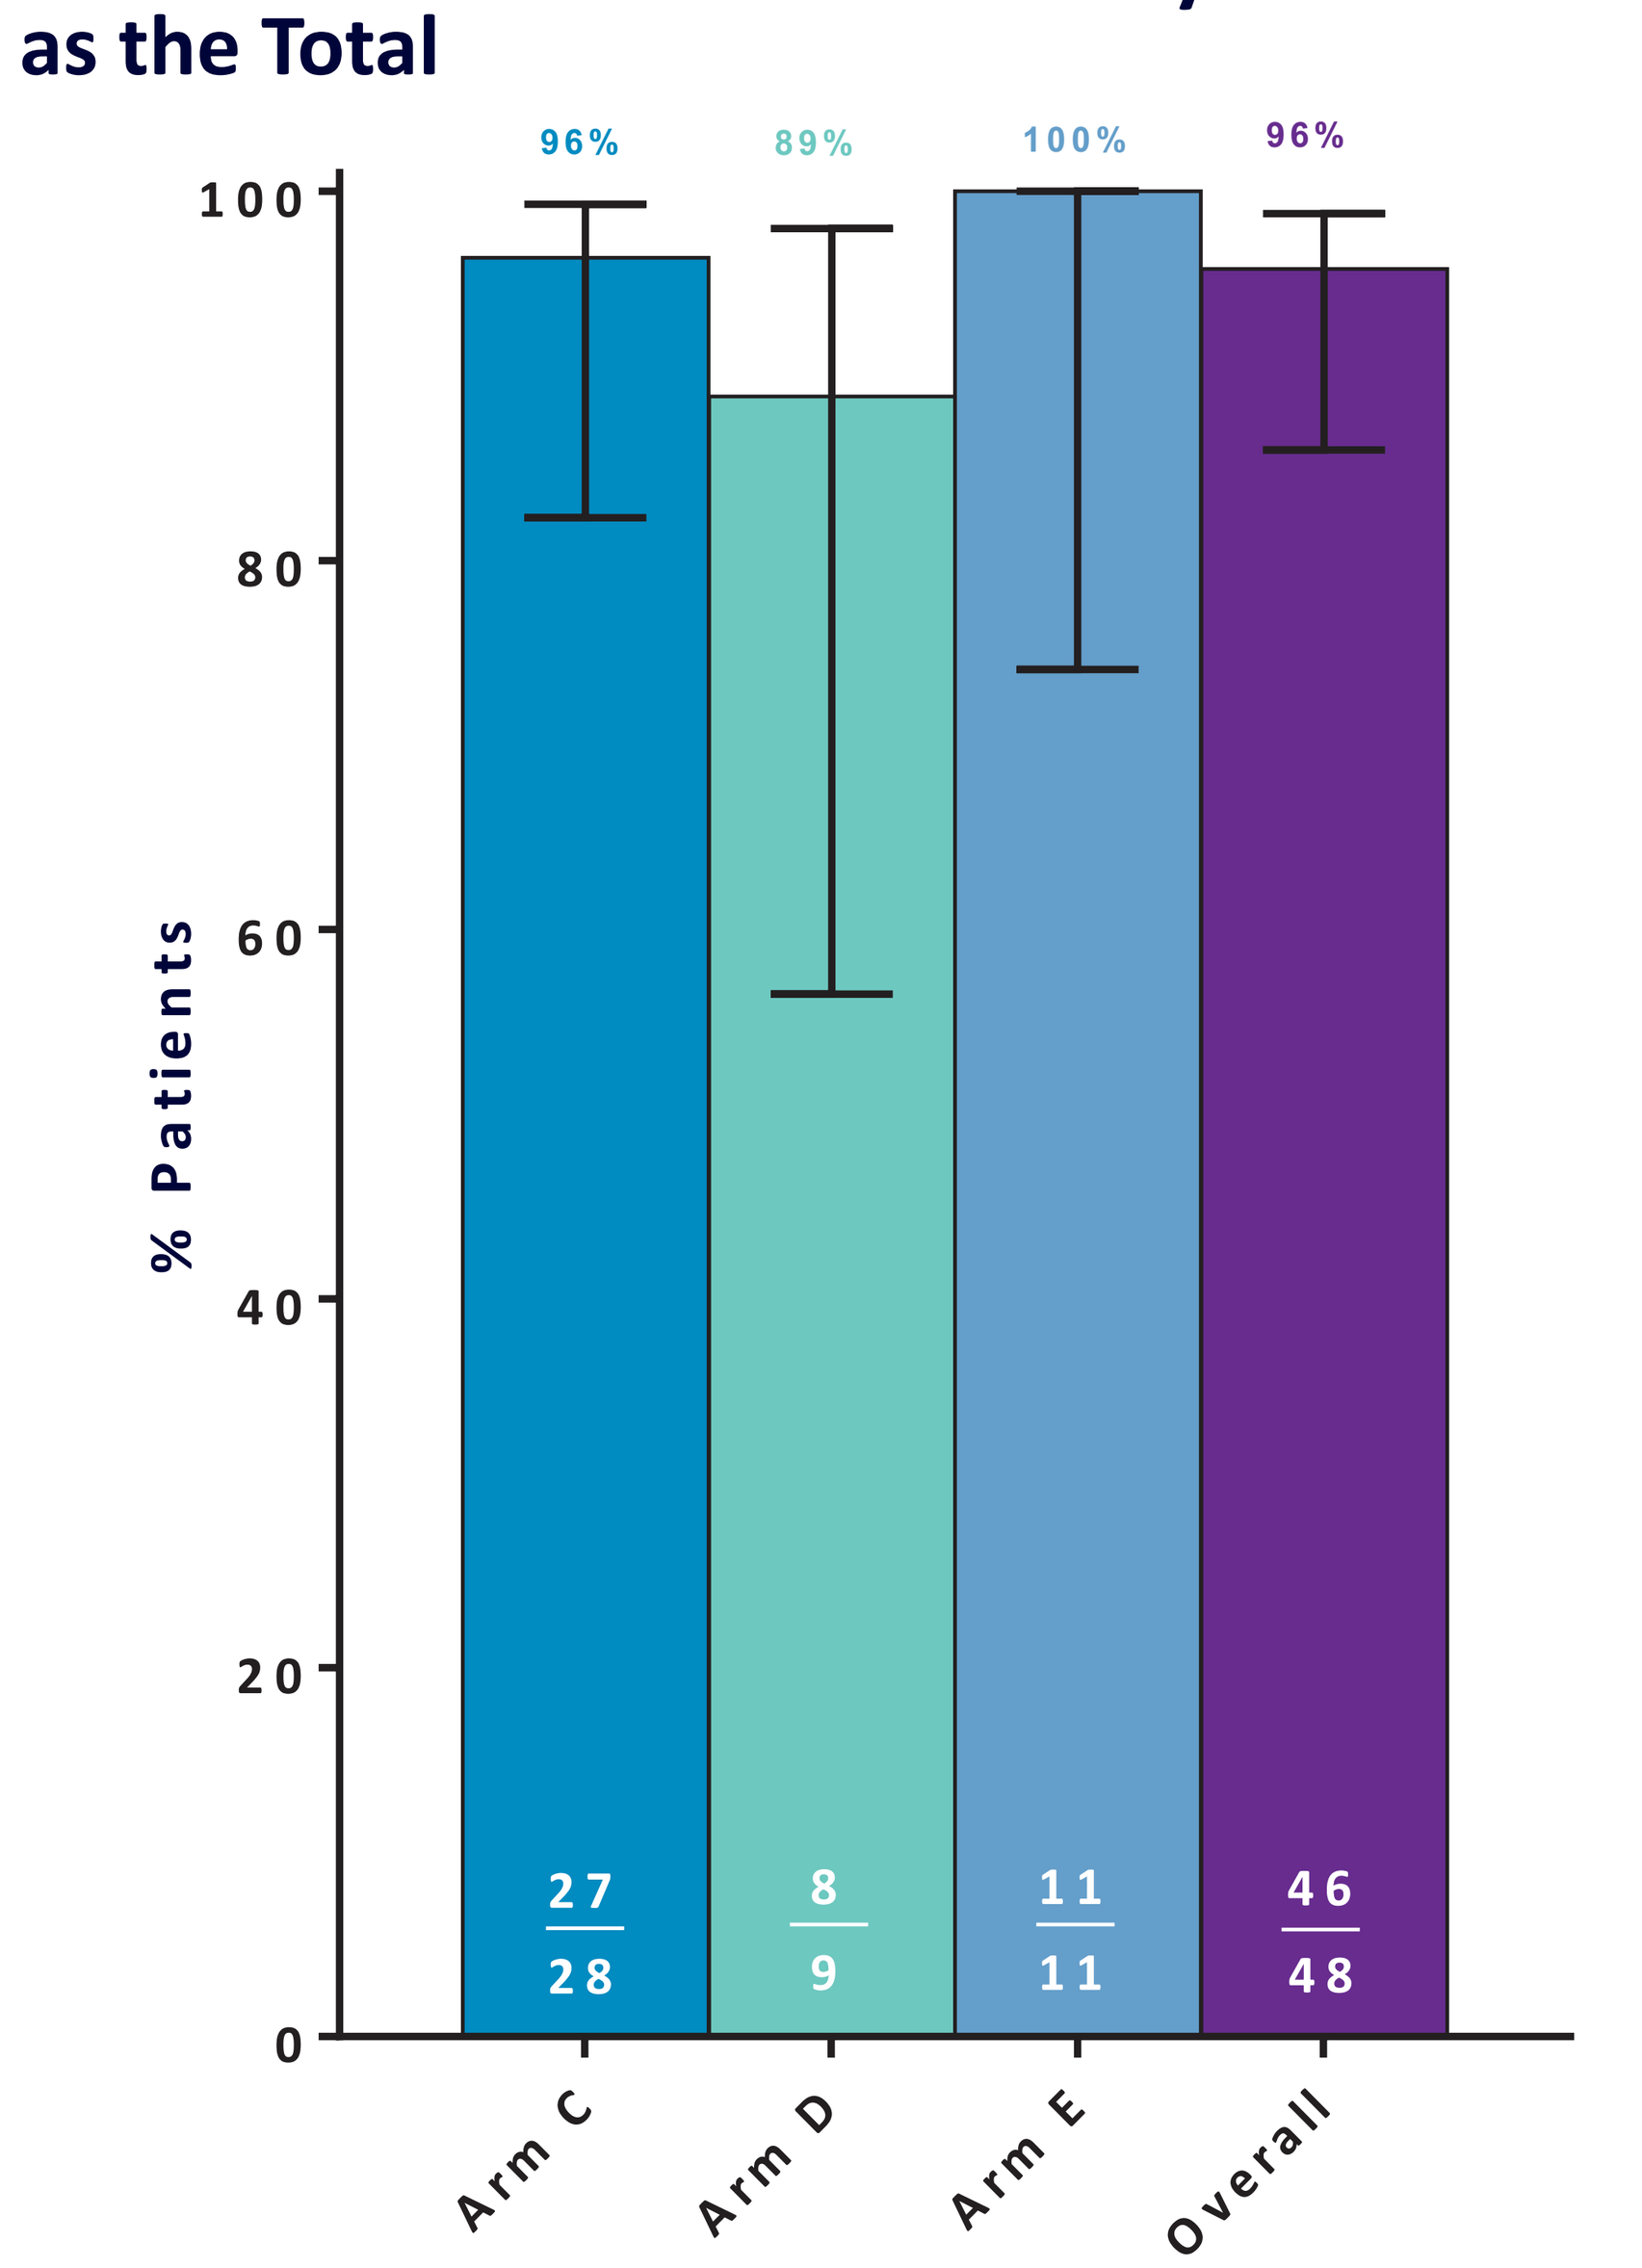
96%
89%
100%
96%
100
80
60
40
20
0
% Patients
27/28
8/9
11/11
46/48
Arm C
Arm D
Arm E
Overall

VIROLOGY

- Results of resistance testing for the single subject with virologic failure are shown in **Table 4**

Table 4. Resistance at Baseline and After Virologic Failure for the Subject Failing Treatment

Target	Baseline	After virologic failure
NS3	None	D168V
NS5A	None	Q30R
NS5B	S556G	Y448H

SAFETY

- Most AEs were mild or moderate in severity **(Table 3)**
- The most common AEs were anemia (40%), fatigue (27%), and diarrhea (19%)
 - AEs of anemia occurred only in patients receiving RBV
 - Of the 19 cases of anemia, 11 were mild, 6 were moderate, and 2 were severe
 - 2 patients required interruption of study drugs
 - 7 patients received erythropoietin and 2 patients required blood transfusion
 - No patient discontinued DAAs because of anemia
- 13 patients (27%) had serious AEs, 2 (4%) of which were considered possibly related to study drug
 - Diarrhea possibly due to DAAs
 - Mental status change possibly due to RBV
- One patient (Arm D) had an AE that led to premature study drug discontinuation
 - Serious AE of volvulus on Day 6 of treatment, not related to study drug

Table 3. Treatment-emergent Adverse Events

Event, n (%)	Arm C GT1a, F0–F3 n = 28	Arm D GT1a, F4 n = 9	Arm E GT1b, F0–F4 n = 11	Total N = 48
Any adverse event	27 (96)	8 (89)	6 (55)	41 (85)
Adverse events leading to study drug d/c	1 (4)	1 (11)	0	2 (4)
Serious adverse event	8 (29)	4 (44)	1 (9)	13 (27)
Death	0	0	0	0
Adverse events occurring in >10% of the total population				
Anemia	16 (57)	3 (33)	0	19 (40)
Fatigue	9 (32)	3 (33)	1 (9)	13 (27)
Diarrhea	7 (25)	2 (22)	0	9 (19)
Hemoglobin decreased	7 (25)	2 (22)	0	9 (19)
Nausea	5 (18)	3 (33)	0	8 (17)
Vomiting	7 (25)	1 (11)	0	8 (17)
Pruritus	4 (14)	0	2 (18)	6 (13)
Headache	3 (11)	1 (11)	1 (9)	5 (10)

Table 4. Post-baseline Laboratory Abnormalities

	Arm C GT1a, F0–F3 n = 28	Arm D GT1a, F4 n = 9	Arm E GT1b, F0–F4 n = 11	Total N = 48
Hemoglobin				
Grade ≥2	20 (71)	6 (75)	3 (27)	29 (62)
Grade ≥3	1 (4)	2 (25)	0	3 (6)
Alanine aminotransferase				
Grade ≥2	1 (4)	0	0	1 (2)
Grade ≥3	1 (4)	0	0	1 (2)
Aspartate aminotransferase				
Grade ≥2	1 (4)	0	0	1 (2)
Grade ≥3	1 (4)	0	0	1 (2)
Total Bilirubin				
Grade ≥2	1 (4)	3 (38)	0	1 (2)
Grade ≥3	0	1 (13)	0	1 (2)

SUMMARY AND CONCLUSIONS

- OBV/PTV/r + DSV ± RBV resulted in an SVR12 rate of 96% in patients with CKD stages 4 or 5 in cohort 2 of the RUBY-I study
- The regimen was generally well tolerated for this group of patients with severe underlying comorbidities, with 1 possibly DAA-related serious AE of diarrhea and 1 discontinuation due to an AE unrelated to treatment
- A large proportion of patients on RBV required RBV dose modification for anemia
- Most AEs were mild or moderate in severity
- These results of efficacy and safety support the use of this regimen in patients with advanced renal disease, for whom treatment options are limited, and are consistent with recently updated treatment guideline recommendations[3]

REFERENCES

1. Viekira Pak prescribing information. https://www.viekira.com (accessed Oct 6).
2. Pockros PJ, Reddy KR, Mantry PS, et al. Efficacy of Direct-Acting Antiviral Combination for Patients With Hepatitis C Virus Genotype 1 Infection and Severe Renal Impairment or End-Stage Renal Disease. *Gastroenterology*. 2016;150(7):1590–8.
3. European Association for the Study of the Liver. Electronic address eee. EASL Recommendations on Treatment of Hepatitis C 2016. *J Hepatol*. 2016.

ACKNOWLEDGEMENTS

Medical writing support was provided by Maher Quraan, PhD, of AbbVie.

DISCLOSURES

AbbVie sponsored the study (NCT0220708), contributed to its design, the collection, analysis, and interpretation of the data, and participated in the writing, review, and approval of the abstract. All authors had access to relevant data.

J Vierling: Research Support: AbbVie, Biotest, Bristol-Myers Squibb, Gilead, Janssen, Vertex, Merck, Genentech, Genfit, Hyperion, Intercept, Novartis, Ocera, Sundise; Advisory Board: AbbVie, Boehringer Ingelheim, BMS, Gilead, Janssen, Merck, Hyperion, Intercept, Novartis, Sundise.

E Lawitz: Speaker: Gilead, GSK, Kadmon, Merck, Vertex; Research/Grant Support: AbbVie, Achillion, Boehringer Ingelheim, BMS, Gilead Sciences, GSK, Idenix, Intercept Pharmaceuticals, Janssen, Medtronic, Merck, Novartis, Presidio, Roche, Santaris Pharmaceuticals, Vertex; Advisor: AbbVie, Achillion, BioCryst, Biotica, Enanta, Idenix, Janssen, Merck, Novartis, Santaris, Theravance, Vertex.

KR Reddy: Advisor: Gilead, Bristol-Myers Squibb, AbbVie, Merck, Janssen; Research Support (paid to the University of Pennsylvania): Gilead, Bristol-Myers Squibb, AbbVie, Merck, Janssen.

N Kemmer: Nothing to disclose.

G Morelli: Grant/Research Funding: AbbVie.

PJ Zamor: Grant/Research Funding: AbbVie, Gilead, Bristol-Myers Squibb, Merck; Speaker: Gilead, AbbVie, Janssen; Advisory: AbbVie, Bristol-Myers Squibb.

M Bennett: Stock: AbbVie.

D Bernstein: Research Support: AbbVie, Bristol-Myers Squibb, Gilead, Janssen, Merck, Genentech; Consultant/Speaker: AbbVie, Gilead, Janssen, Merck.

K Kowdley: Advisory Committees or Review Panels: Achillion, BMS, Evidera, Gilead, Merck, Novartis, Trio Health, AbbVie; Grant/Research Support: Evidera, Gilead, Immuron, Intercept, Tobira; Speaking and Teaching: AbbVie, Gilead

PS Mantry: Grant/Research Funding: AbbVie, Boehringer Ingelheim, Bristol-Myers Squibb, Gilead, Mass Biologics, Merck, Bayer-Onyx, Santaris, Salix, Vital Therapies, Vertex; Consultant/Advisor: AbbVie, Bristol-Myers Squibb, Gilead, Johnson and Johnson, Merck, Salix, Genentech, Vertex; Fees for Non-CME/CE services: Bayer-Onyx, Gilead, Janssen, Merck, Salix, Vertex.

PJ Pockros: Speaker/Consultant/Advisor: Gilead, AbbVie, Janssen, Bristol-Myers Squibb; Research Support: Gilead, AbbVie, Janssen, Bristol-Myers Squibb, Merck, Conatus, Roche Molecular.

D Wyles: Grant/Research Support: AbbVie, Bristol-Myers Squibb, Gilead, Merck, Tacere Therapeutics; Consultant/Advisor: AbbVie, BMS, Gilead, Merck, Janssen.

S Kumar: Research Support: AbbVie, BMS, Gilead, Intercept Pharmaceuticals, Merck; Speaker: AbbVie, Gilead, Intercept Pharmaceuticals; Advisory Board: Gilead, Janssen, Intercept, AbbVie.

K Bhamidimarri: Scientific Advisory Board: Bristol-Myers Squibb, Gilead Sciences, AbbVie, Salix/Valeant; Research Grant Support: Vital Therapies Inc., Biotest Pharmaceuticals, Ocera Therapeutics Inc., Salix Pharmaceuticals, Synageva/Alexion.

T Hassanein: Grants/Research Support: AbbVie (Advisory Board), Boehringer Ingelheim, Bristol-Myers Squibb (Advisory Board), Eisai, Gilead Sciences, Janssen, Idenix, Ikaria, Mochida, Roche, Ocera, Taigen, Takeda, Salix, Sundise, Vertex; Speaker: Baxter, Bristol-Myers Squibb, Gilead, Salix.

E Cohen, Z Zhang, T Pilot-Matias, DE Cohen, T Podsadecki: Employees of AbbVie; may hold AbbVie stock or options.

abbvie

44588-161469-Vierling — AASLD 2016 — AbbVie — proof 1 (rjh) — october 31, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051

CREATED @ 100%

ONYX-I: Safety and Efficacy of Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir in Asian Adults With Genotype 1b Chronic Hepatitis C Virus Infection – A Randomized, Double-Blind, Placebo-Controlled Study

873

Lai Wei[1], Jinlin Hou[2], Yan Luo[3], Jeong Heo[4], Chi-Jen Chu[5], Zhongping Duan[6], Mong Cho[7], Jun Cheng[8], Jun Li[9], Jidong Jia[10], Wenjing Lu[3], Tami Pilot-Matias[3], Niloufar Mobashery[3], Wan-Long Chuang[11]

[1]Peking University Peoples Hospital, Beijing, China; [2]Nanfang Hospital, Guangzhou, China; [3]AbbVie Inc., North Chicago, Illinois, United States; [4]Department of Internal Medicine, College of Medicine, Pusan National University and Medical Research Institute, Pusan National University Hospital, Busan, Republic of Korea; [5]Taipei Veterans General Hospital, Taipei City, Taiwan; [6]Beijing Youan Hospital Capital Medical University, Beijing, China; [7]Pusan National University Yangsan Hospital, Yangsan, Korea; [8]Beijing Di Tan Hospital, Capital Medical University, Beijing, China; [9]The First Affiliated Hospital with Nanjing Medical University, Nanjing, Jiangsu Province, China; [10]Beijing Friendship Hospital, Capital Medical University, Beijing, China; [11]Kaohsiung Medical University, Kaohsiung City, Taiwan

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

INTRODUCTION

- In Asia, more than 100 million people may be chronically infected with the hepatitis C virus (HCV)[1]
- The prevalence of chronic HCV infection in China was found to be greater than in the Americas and Europe combined[2]
- HCV genotype 1b (GT1b) is the most prevalent sub-genotype in China,[2,3] South Korea[2] and Taiwan[2,4]
- Despite recent advances in the development of all-oral, direct-acting antiviral (DAA) regimens for HCV, interferon (IFN)/pegylated IFN (pegIFN) and ribavirin (RBV) remain the current standard of care in some Asian countries where GT1b has high prevalence[5]
- Paritaprevir (PTV, formerly ABT-450) is a potent HCV NS3/4A protease inhibitor (identified by AbbVie and Enanta); ombitasvir (OBV, formerly ABT-267) is a potent HCV NS5A inhibitor; dasabuvir (DSV, formerly ABT-333) is a non-nucleoside HCV NS5B RNA polymerase inhibitor
- Previous multinational Phase 3 studies have demonstrated that a 12-week treatment with OBV/PTV/ritonavir (r) + DSV (3-DAA) was well tolerated and achieved rates of sustained virologic response at post-treatment Week 12 (SVR12) of 100% in patients infected with GT1b HCV with and without compensated cirrhosis; these studies were conducted in North America, Europe, and Australia with a majority of non-Asian patients[6,7]

Multi-targeted 3 Direct-acting Antiviral Regimen (3-DAA)

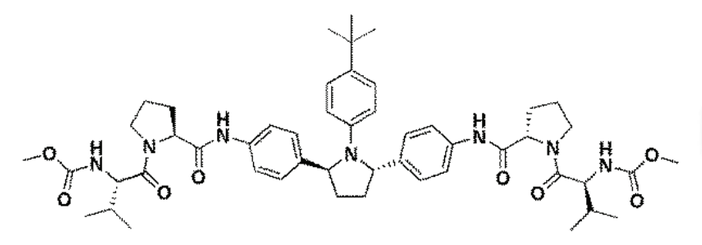

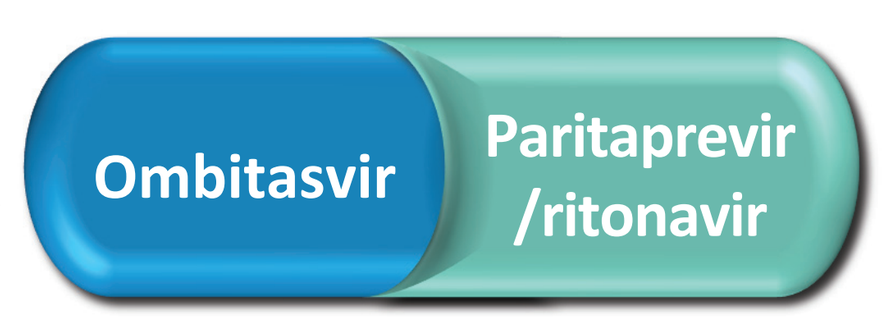
Ombitasvir
Paritaprevir
/ritonavir

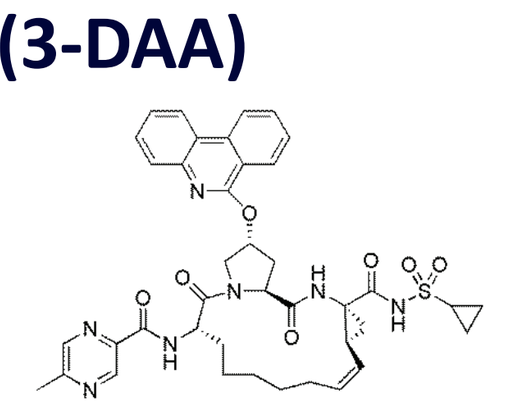

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)

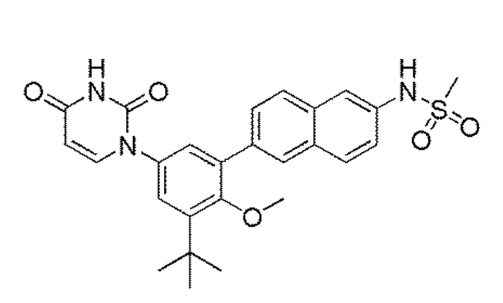

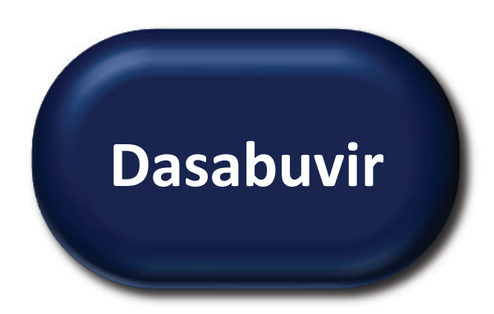
Dasabuvir

Dasabuvir (DSV)
non-nucleoside NS5B RNA polymerase inhibitor

PTV was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVE

Evaluate the safety and efficacy of OBV/PTV/r plus DSV in adults with GT1b HCV infection in China, South Korea, and Taiwan following 12 weeks of therapy.

METHODS

- ONYX-I is an ongoing Phase 3, multicenter, randomized, double-blind, placebo-controlled trial conducted in China, South Korea, and Taiwan in treatment naïve and treatment experienced, noncirrhotic HCV GT1b-infected adults
- Patients were randomized 1:1 to Arm A and Arm B. Patients in Arm A received the active drugs during the 12-week double-blind period, while patients in Arm B received placebo during the same period followed by the active drug treatment during the 12-week open-label period **(Figure 1)**. All patients are to be followed up for 48 weeks post-treatment

Figure 1. Study Design

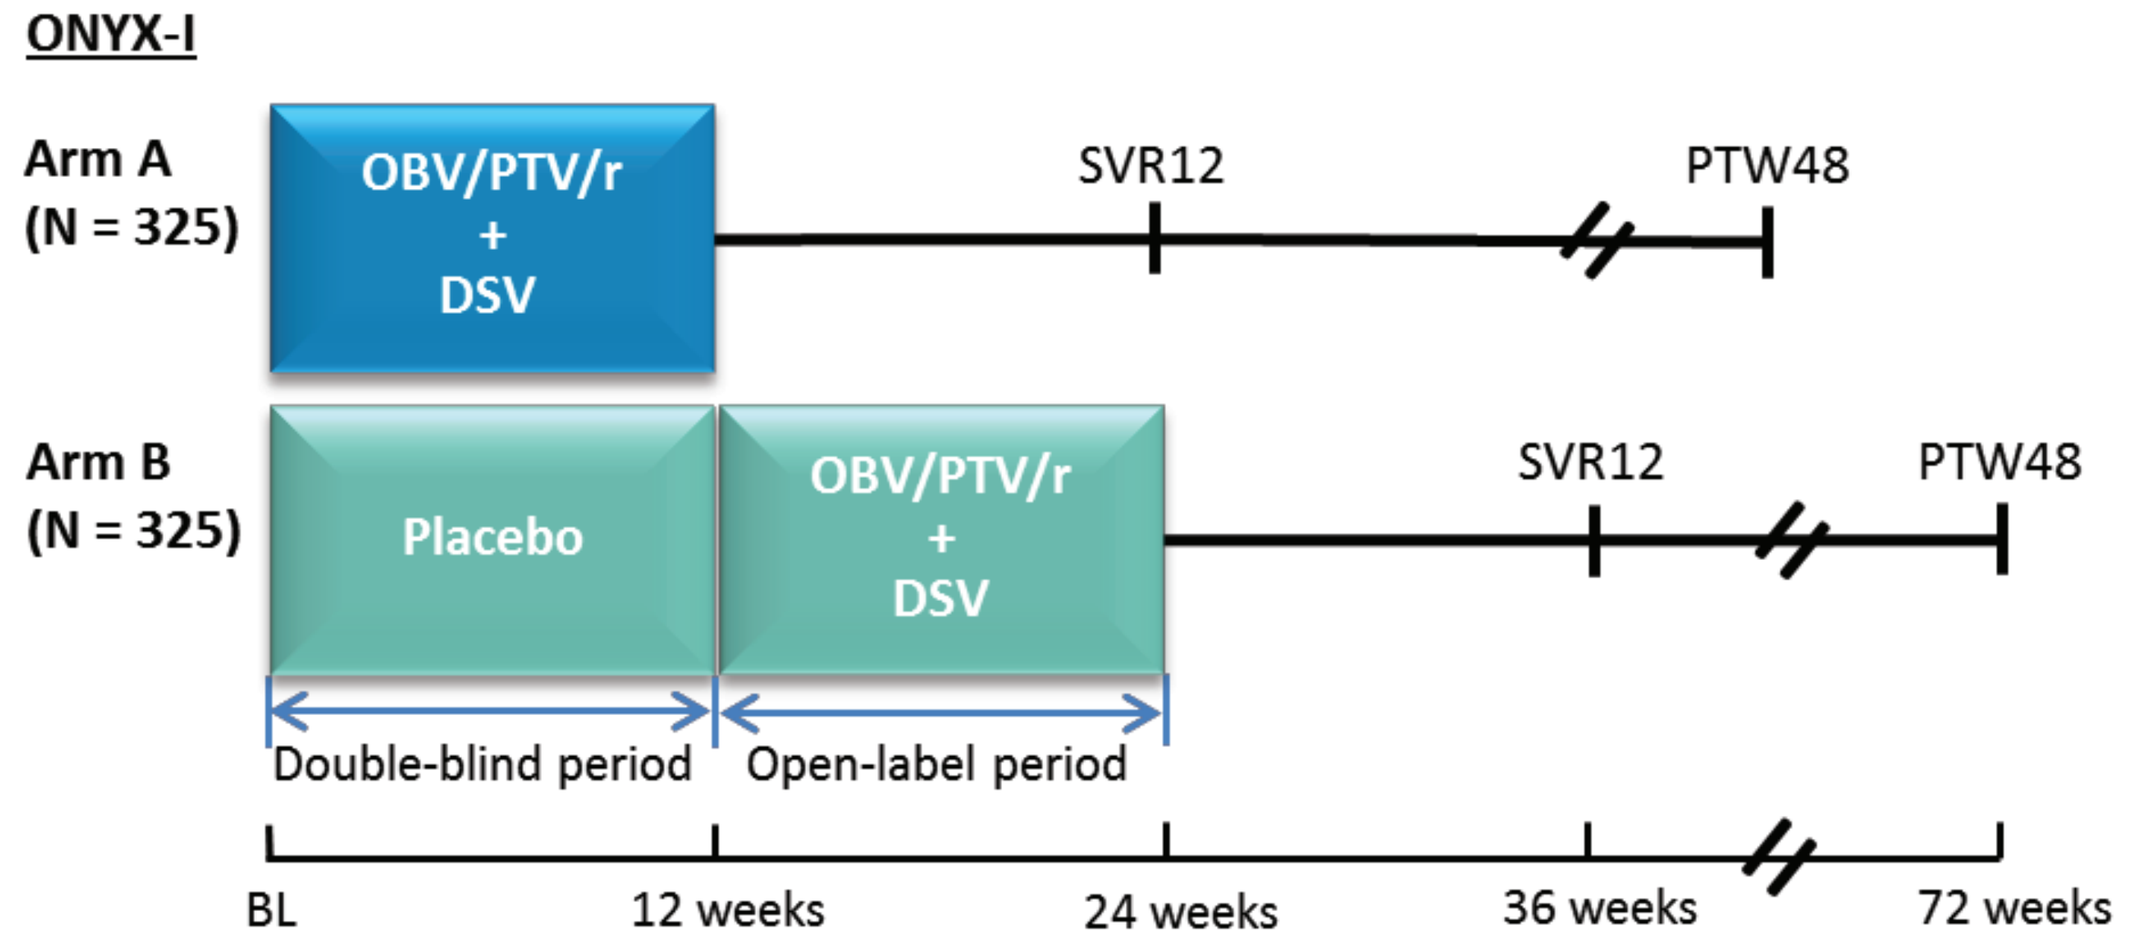
ONYX-I
Arm A
(N = 325)
OBV/PTV/r
+
DSV
SVR12
PTW48
Arm B
(N = 325)
Placebo
OBV/PTV/r
+
DSV
SVR12
PTW48
Double-blind period
Open-label period
BL
12 weeks
24 weeks
36 weeks
72 weeks

METHODS (CONTINUED)

KEY ELIGIBILITY CRITERIA

Main inclusion criteria:

- Chinese, South Korean, and Taiwanese patients with full Chinese, South Korean, and Taiwanese parentage, respectively
- 18 to 70 years of age inclusive, at time of screening
- Chronic GT1b HCV-infection
- Absence of Cirrhosis using 1 of the following criteria:
 - Liver biopsy demonstrating absence of Cirrhosis (eg, Metavir Score of ≤3 or an Ishak score of ≤4)
 - FibroTest score of ≤0.72 and Aspartate Aminotransferase to Platelet Ratio Index (APRI) ≤2
 - FibroScan result of <9.6 kPa
- Treatment-naïve or treatment-experienced (IFN/pegIFN and RBV)

Main exclusion criteria:

- Presence of HCV genotypes/subgenotype other than GT1b
- Co-infection with the hepatitis B virus (HBV) or human immunodeficiency virus (HIV)
- Any current or past clinical evidence of cirrhosis (such as ascites or esophageal varices, or prior liver biopsy showing cirrhosis (eg, a Metavir score of >3 or Ishak score of >4)
- Any primary cause of liver disease other than chronic HCV infection
- Abnormal laboratory result that meets at least 1 the following criteria (see **Table 1**)

Table 1. Abnormal Laboratory Results Exclusion Criteria

Assessment	Value
Alanine Aminotransferase	>5 × ULN
Aspartate Aminotransferase	>5 × ULN
Estimated Glomerular Filtration Rate (eGFR)*	<50 mL/min/1.73m^2
Albumin	<LLN
International normalized ratio (INR)†	>1.5
Hemoglobin	<LLN
Platelets	<100 000 cells per mm^3
Absolute Neutrophil Count (ANC)	<1500 cells/μL
Indirect Bilirubin	>1.5 × ULN
Direct Bilirubin	>ULN

LLN = lower limit of normal, ULN = upper limit of normal.
*Modified for the Chinese population as follows: eGFR = 175 × (Serum Creatinine)$^{-1.234}$ × (Age)$^{-0.179}$ × (0.79 if Female).
†Subjects with a known inherited blood disorder and INR >1.5 may have been enrolled with permission of the AbbVie study designated physician.

STUDY ASSESSMENTS

Primary Efficacy Endpoint	Percentage of TN and TE Arm A patients with SVR12 compared to historical SVR rates for telaprevir (TLV) plus pegIFN/RBV therapy*
Secondary Efficacy Endpoints	Percentage of TN and TE Arm A patients with on-treatment virologic failure and post-treatment relapse
Safety Assessment	Percentage of patients with treatment-emergent adverse events (AEs) and laboratory abnormalities during the double-blind and open-label periods†

*Superiority achieved if lower bound of 95% CI for Arm A rate exceeds 84% for treatment-naïve (TN) patients and 75% for treatment-experienced (TE) patients.
†Safety of the 3DAA regimen is compared statistically to placebo during the double-blind period.

RESULTS

PARTICIPANTS

- A total of 650 patients were enrolled in this study and were randomized 1:1 to Arms A and B
 - China: 410; South Korea: 120; Taiwan: 120
 - All patients were Asian
 - 44% were treatment-experienced

RESULTS (CONTINUED)

Table 2. Baseline Demographics and Disease Characteristics of Patients Enrolled in the Study

	Arm A				Arm B			
	China n = 205	South Korea n = 60	Taiwan n = 60	Total N = 325	China N = 205	South Korea N = 60	Taiwan N = 60	Total N = 325
Female, n (%)	103 (50)	32 (53)	40 (67)	175 (54)	110 (54)	34 (57)	33 (55)	177 (55)
Age, median (range), years	47 (20–68)	51 (18–71)	55 (28–70)	50 (18–71)	44 (19–69)	53.5 (22–68)	56.5 (27–69)	48 (19–69)
BMI, mean ± SD, kg/m^2	23.3 ± 3.2	23.6 ± 3.4	23.9 ± 2.9	23.4 ± 3.2	23.5 ± 3.3	24.7 ± 3.4	24.6 ± 3.3	23.9 ± 3.4
IL28B CC genotype, n (%)	143 (70)	47 (78)	43 (72)	233 (72)	135 (66)	46 (77)	44 (75)*†	225 (69)*†
HCV RNA, median (range), log_{10} IU/mL	6.5 (4.5–7.4)	6.5 (3.6–7.1)	6.4 (4.6–7.2)	6.4 (3.6–7.4)	6.4 (1.6–7.6)	6.3 (2.6–7.2)	6.4 (4.2–7.2)	6.4 (1.6–7.6)
Fibrosis Stage,† n (%)								
F0–F1	172 (84)	53 (88)	41 (70)	266 (82)	175 (85)	49 (82)	48 (80)	272 (84)
F2	24 (12)	4 (7)	15 (25)	43 (13)	20 (10)	8 (13)	8 (13)	36 (11)
≥F3	8 (4)	3 (5)	3 (5)	14 (4)	10 (5)	3 (5)	4 (7)	17 (5)
Missing	1	0	1	2	0	0	0	0
Treatment-naïve, n (%)	104 (51)	40 (67)	40 (67)	184 (57)	102 (50)	40 (67)	40 (67)	182 (56)
Treatment-experienced, n (%)	101 (49)	20 (33)	20 (33)	141 (43)	103 (50)	20 (33)	20 (33)	143 (44)
Non-responder, n/N (%)	63/101 (62)	6/20 (30)	9/20 (45)	78/141 (55)	54/103 (52)	7/20 (35)	7/20 (35)	68/143 (48)
Relapser, n/N (%)	38/101 (38)	14/20 (70)	11/20 (55)	63/141 (45)	49/103 (48)	13/20 (65)	13/20 (65)	75/143 (52)

Data from one subject was missing. †Percentages calculated on non-missing values.

EFFICACY

- In Arm A, SVR12 was achieved in 183 of 184 (99.5%) treatment-naïve patients and all (100%) 141 treatment-experienced patients. The lower bound of the 95% confidence interval (LCB) was above the 84% superiority threshold for treatment-naïve patients and the 75% superiority threshold for treatment-experienced patients. The 3-DAA regimen demonstrated superiority to the historical telaprevir plus pegIFN/RBV SVR rate, irrespective of prior treatment status **(Figure 2)**
- The SVR12 rates were 99.0%, 100%, and 100% in Arm A treatment-naïve patients from China, South Korea, and Taiwan, respectively, and 100% in Arm A treatment-experienced patients from each region **(Figure 2)**
- One patient in the Chinese cohort of Arm A experienced on-treatment virologic failure (failed to suppress)
 - Pharmacokinetic analysis showed that this patient had concentrations of 0 ng/mL for all components of the 3 DAA regimen at all study visits
 - **Table 3** shows the resistance data for this patient at baseline and at the time of virologic failure (VF)

Table 3. Resistance Data for the Patient Not Achieving SVR12

NS3		NS5A		NS5B	
Baseline	VF	Baseline	VF	Baseline	VF
None	None	None	Y93H	C316N	C316N

Figure 2. SVR12 Rates (and 95% CIs) for Treatment-naïve (TN) and Treatment-experienced (TE) Patients in Arm A

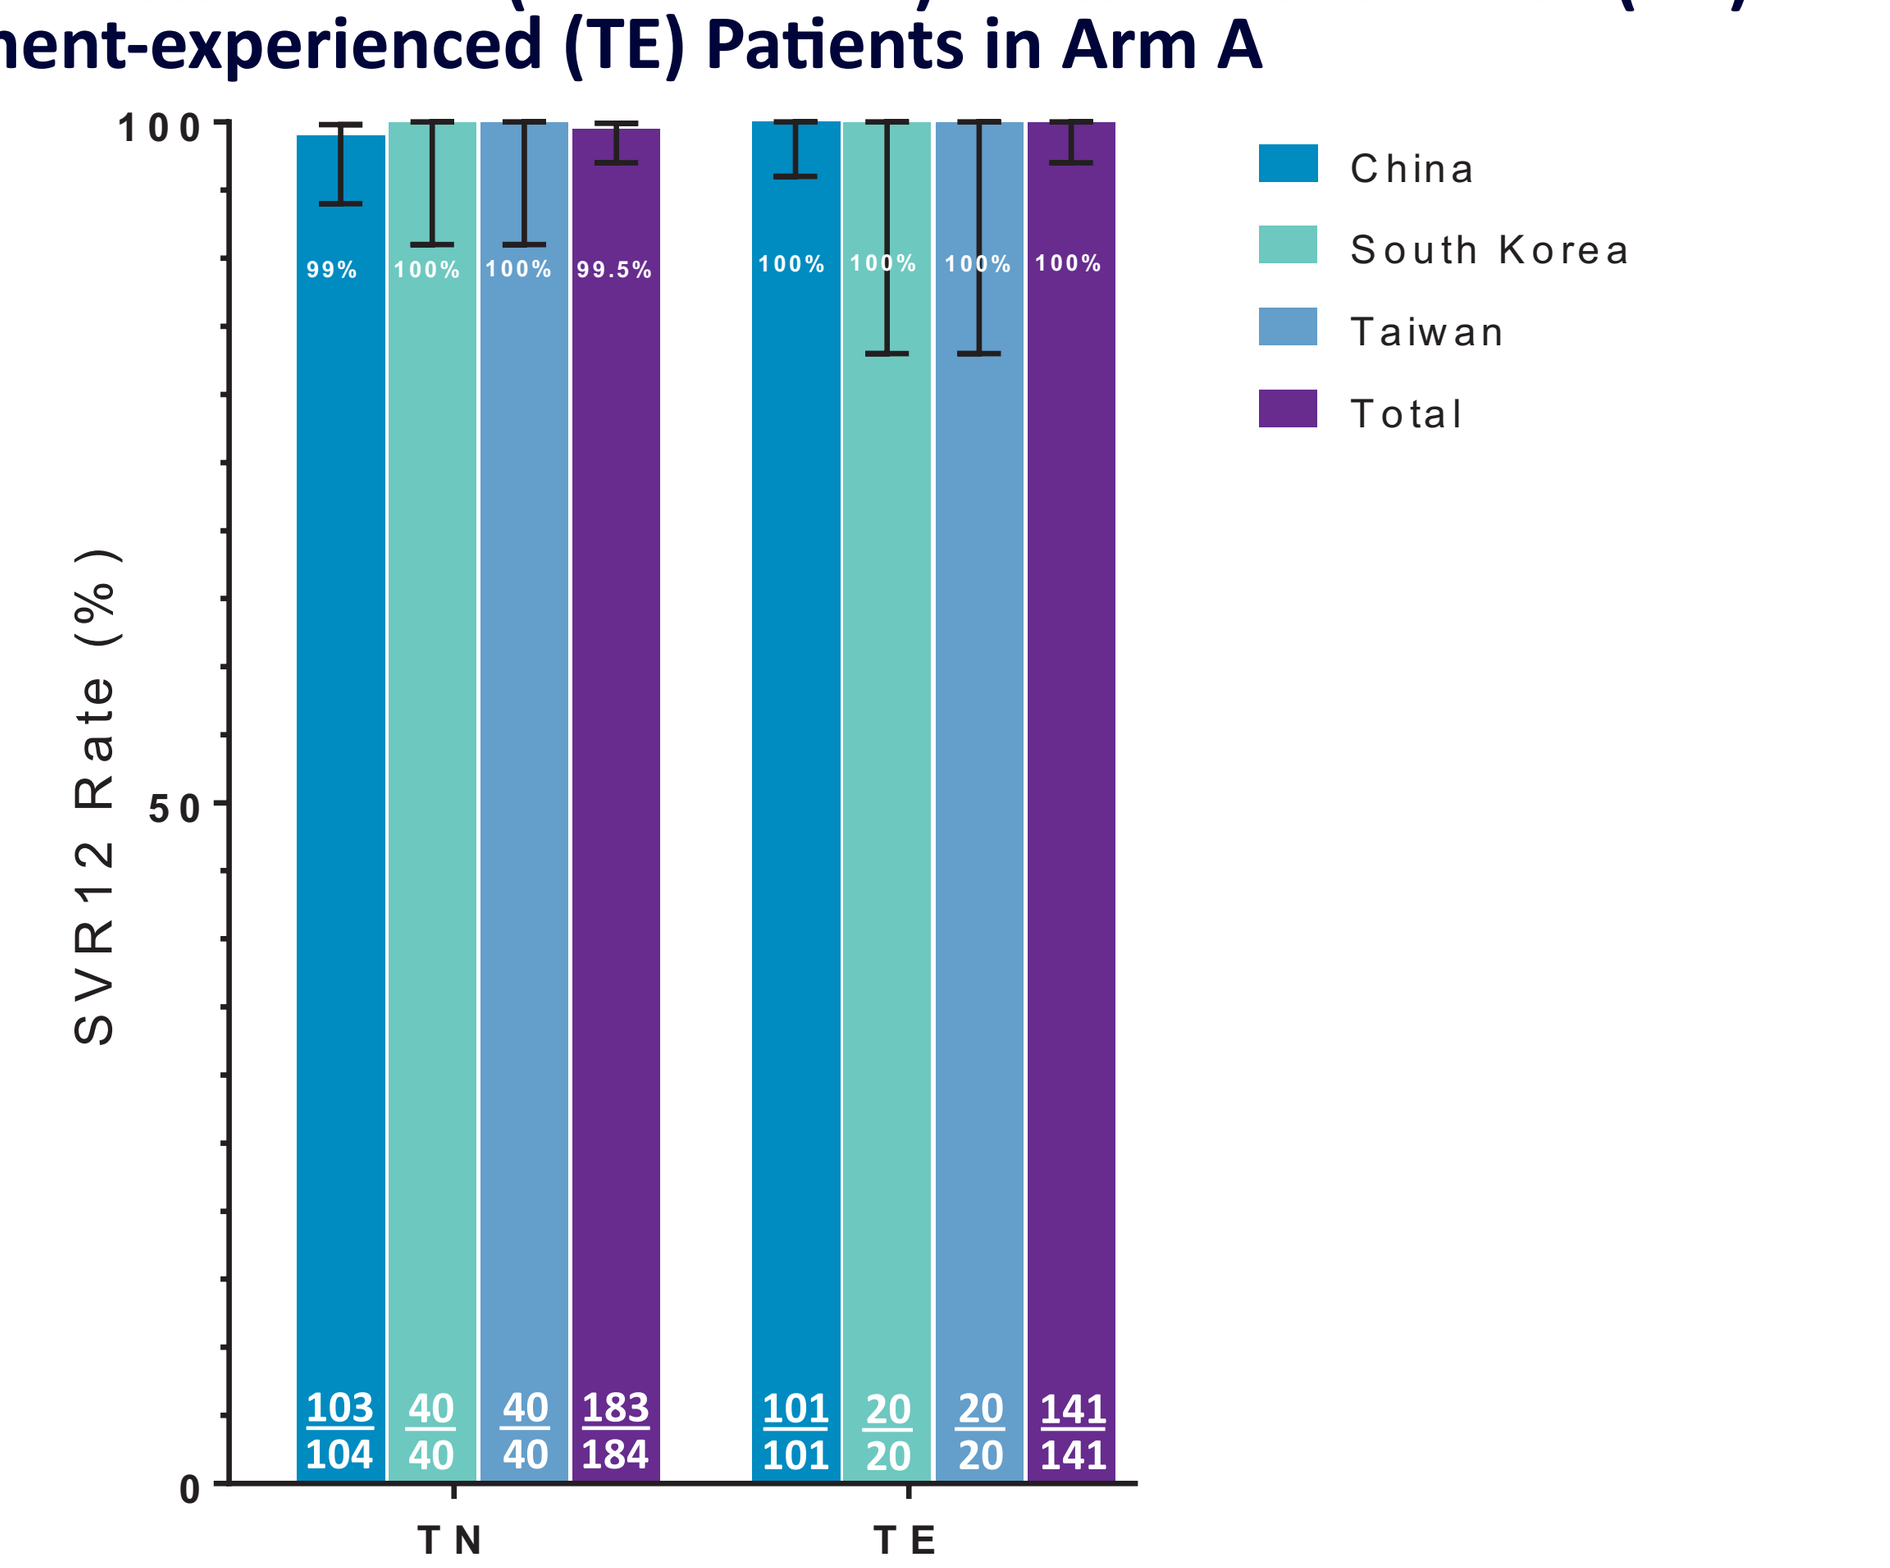
100
50
0
SVR12 Rate (%)
99%
100%
100%
99.5%
100%
100%
100%
100%
103
104
40
40
40
40
183
184
101
101
20
20
20
20
141
141
TN
TE
China
South Korea
Taiwan
Total

SAFETY

- Most AEs were mild in severity during both the double-blind period in Arm A and the open-label period in Arm B
- The most common AEs for patients receiving the 3-DAA regimen was upper respiratory tract infection reported in 10.5% of patients in Arm A during the double-blind period **(Table 4)** and 9.6% in Arm B during the open-label period **(Table 5)**
- One patient in the Chinese cohort of Arm A had serious AEs (increased alanine and aspartate aminotransferase) assessed by the investigator as having a reasonable possibility of being related to study drug; this subject continued with uninterrupted treatment and achieved SVR12
- Post-baseline laboratory abnormalities with grade ≥3 were rare in patients treated with the 3-DAA regimen in both the double-blind period **(Table 6)** and the open-label period **(Table 7)**

Table 4. Treatment-emergent Adverse Events During the Double-blind Period

	Arm A: 3-DAA				Arm B: Placebo			
Event, n (%)	China n = 205	South Korea n = 60	Taiwan n = 60	Total N = 325	China N = 205	South Korea N = 60	Taiwan N = 60	Total N = 325
Any AE*	116 (56.6)	35 (58.3)	40 (66.7)	191 (58.8)	102 (49.8)	27 (45.0)	30 (50.0)	159 (48.9)
AEs leading to drug d/c	0	0	0	0	3 (1.5)	0	0	3 (0.9)
Serious AE†	4 (2.0)	1 (1.7)	2 (3.3)	7 (2.2)	2 (1.0)	0	0	2 (0.6)
Death	0	0	0	0	0	0	0	0
Adverse events with ≥10% frequency in any Arm A group								
Upper respiratory tract infection	23 (11.2)	3 (5.0)	8 (13.3)	34 (10.5)	18 (8.8)	3 (5.0)	8 (13.3)	29 (8.9)
Headache	3 (1.5)	8 (13.3)	9 (15.0)	20 (6.2)	4 (2.0)	5 (8.3)	4 (6.7)	13 (4.0)
Fatigue	2 (1.0)	7 (11.7)	7 (11.7)	16 (4.9)	5 (2.4)	2 (3.3)	1 (1.7)	8 (2.5)
Pruritus*	8 (3.9)	6 (10.0)	2 (3.3)	16 (4.9)	4 (2.0)	1 (1.7)	1 (1.7)	6 (1.8)

*Statistically significant (*P* ≤.05) difference between Arm A and Arm B for Total.
†One patient in the Chinese cohort of Arm A had serious AEs that were assessed by the study investigator as being related to study drug (increased ALT and AST).

Table 5. Treatment-emergent Adverse Events During the Open-label Period

	Arm B: 3-DAA			
Event, n (%)	China n = 204*	South Korea n = 60	Taiwan n = 60	Total N = 324*
Any adverse event	111 (54.4)	33 (55.0)	39 (65.0)	183 (56.5)
Adverse events leading to drug d/c	1 (0.5)	0	0	1 (0.3)
Serious adverse event†	2 (1.0)	3 (5.0)	0	5 (1.5)
Death	0	0	0	0
Adverse events with ≥10% frequency in any Arm B group				
Upper respiratory tract infection	17 (8.3)	9 (15.0)	5 (8.3)	31 (9.6)

*One patient in the Chinese cohort of Arm B did not enter the open-label period due to AEs.
†All serious AEs were assessed by the study investigators as not being related to study drug.

Table 6. Post-baseline Laboratory Abnormalities During the Double-blind Period

	Arm A: 3-DAA				Arm B: Placebo			
	China n = 205	South Korea n = 60	Taiwan n = 60	Total N = 325	China N = 205	South Korea N = 60	Taiwan N = 60	Total N = 325
Hemoglobin								
Grade ≥2	1 (0.5)	0	1 (1.7)	2 (0.6)	2 (1.0)	0	0	2 (0.6)
Grade ≥3	0	0	0	0	0	0	0	0
Alanine aminotransferase (ALT)								
Grade ≥2	8 (3.9)*	1 (1.7)*	0	9 (2.8)*	29 (14.1)	15 (25.0)	5 (8.3)	49 (15.1)
Grade ≥3	2 (1.0)*	0	0	2 (0.6)*	10 (4.9)	2 (3.3)	1 (1.7)	13 (4.0)
Aspartate aminotransferase (AST)								
Grade ≥2	5 (2.4)*	1 (1.7)	0	6 (1.8)*	17 (8.3)	4 (6.7)	1 (1.7)	22 (6.8)
Grade ≥3	3 (1.5)	1 (1.7)	0	4 (1.2)	5 (2.4)	3 (5.0)	0	8 (2.5)
Total Bilirubin								
Grade ≥2	24 (11.7)*	5 (8.3)	4 (6.7)	33 (10.2)*	8 (3.9)	0	0	8 (2.5)
Grade ≥3	0	1 (1.7)	0	1 (0.3)	0	0	0	0

*Difference from Arm B statistically significant (*P* ≤.05).

Table 7. Post-baseline Laboratory Abnormalities During the Open-label Period

	Arm B: 3-DAA			
	China n = 204*	South Korea n = 60	Taiwan n = 60	Total N = 324*
Hemoglobin				
Grade ≥2	5 (2.5)	0	0	5 (1.5)
Grade ≥3	1 (0.5)	0	0	1 (0.3)
Alanine aminotransferase (ALT)				
Grade ≥2	2 (1.0)	1 (1.7)	1 (1.7)	4 (1.2)
Grade ≥3	0	0	1 (1.7)	1 (0.3)
Aspartate aminotransferase (AST)				
Grade ≥2	2 (1.0)	0	1 (1.7)	3 (0.9)
Grade ≥3	0	0	1 (1.7)	1 (0.3)
Total Bilirubin				
Grade ≥2	15 (7.4)	0	6 (10.0)	21 (6.5)
Grade ≥3	1 (0.5)	0	0	1 (0.3)

*One patient in the Chinese cohort of Arm B did not enter the open-label period due to AEs.

CONCLUSIONS

- In Arm A, SVR12 was achieved in 183 of 184 (99.5%) treatment-naïve patients and all 141 (100%) treatment-experienced patients
- The regimen was well tolerated; AEs were mostly mild in severity
- One serious AE in the Chinese cohort of Arm A was assessed as being study-drug related
- Post-baseline laboratory abnormalities with a severity grade ≥3 were rare
- The efficacy and safety profiles observed in this Asian regional Phase 3 study are similar to those in the global Phase 3 studies

REFERENCES

1. Mohd Hanafiah K, Groeger J, Flaxman AD, Wiersma ST. Global epidemiology of hepatitis C virus infection: new estimates of age-specific antibody to HCV seroprevalence. *Hepatology*. 2013;57(4):1333–42.
2. Sievert W, Altraif I, Razavi HA, et al. A systematic review of hepatitis C virus epidemiology in Asia, Australia and Egypt. *Liver Int.* 2011;31 Suppl 2:61–80.
3. Rao H, Wei L, Lopez-Talavera JC, et al. Distribution and clinical correlates of viral and host genotypes in Chinese patients with chronic hepatitis C virus infection. *J Gastroenterol Hepatol*. 2014;29(3):545–53.
4. Lee MH, Yang HI, Lu SN, et al. Hepatitis C virus genotype 1b increases cumulative lifetime risk of hepatocellular carcinoma. *Int J Cancer*. 2014;135(5):1119–26.
5. Duan Z, Jia JD, Hou J, et al. Current challenges and the management of chronic hepatitis C in mainland China. *J Clin Gastroenterol*. 2014;48(8):679–86.
6. Dore GJ, Conway B, Luo Y, et al. Efficacy and safety of ombitasvir/paritaprevir/r and dasabuvir compared to IFN-containing regimens in genotype 1 HCV patients: The MALACHITE-I/II trials. *J Hepatol*. 2016;64(1):19–28.
7. Feld JJ, Moreno C, Trinh R, et al. Sustained virologic response of 100% in HCV genotype 1b patients with cirrhosis receiving ombitasvir/paritaprevir/r and dasabuvir for 12weeks. *J Hepatol*. 2016;64(2):301–7.

ACKNOWLEDGEMENTS

AbbVie sponsored the studies, contributed to their designs, participated in the collection, analysis, and interpretation of data, and in the writing, reviewing, and approval of the publication.

We thank all the patients and investigators who participated in this study. We thank AbbVie employees Angela Nilius, Nancy Shulman, Tahira Payne, Jim A Watson, Brian Oliver, Olga Furman, Rajvineeth Kumar Pothacamury, Kristine M Cwik, Jun Sun, Felecia Hughey, Danijela Dobirc, Chunyan Yuan, Jennifer King, Jiuhong Zha, Diana Shuster, Rakesh Tripathi, Gretja Schnell, and Katia Alvis.

Medical writing support was provided by Maher Quraan, PhD, of AbbVie.

DISCLOSURES

AbbVie sponsored the study (M13-767) contributed to its design, collection, analysis, and interpretation of the data, and participated in the writing, review, and approval of the abstract. All authors had access to relevant data.

L Wei: Received research support from Bristol-Myers Squib, and Roche; Advisory board for Abbott, AbbVie, BMS, Galmed, and Gilead. J Hou: Received research support from Bristol-Myers Squib, Novartis, GSK and Roche; Advisory board for AbbVie, Novartis, and Gilead. Y Luo, W Lu, T Pilot-Matias, N Mobashery, and WL Chuang: employees of AbbVie and may hold stock or stock options. J Heo: Received a grant from GSK; Research support from BMS and Hoffmann-La Roche; Advisor for AbbVie, BMS, Gilead Sciences, Pharma Essentia, SillaJen, and Johnson & Johnson. CJ Chu: Speaker for Gilead, Bristol-Meyers Squibb, MSD, and Roche. Z Duan: Participated in an AbbVie sponsored clinical study. M Cho: Participated in an AbbVie sponsored clinical study. J Cheng: Participated in an AbbVie sponsored clinical study. J Li: Participated in an AbbVie sponsored clinical study. J Jia: Participated in an AbbVie sponsored clinical study. W Chuang: Advisory board for Gilead, AbbVie, Roche; Speaker for Gilead, BMS, MSD, Roche, Novartis.

44588-161469-873 — AASLD 2016 — AbbVie — proof 3 (rjh) — november 2, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051
CREATED @ 100%

875

ONYX-II: Safety and Efficacy of Ombitasvir/Paritaprevir/Ritonavir and Dasabuvir Coadministered With Ribavirin in Asian Adults With Genotype 1b Chronic Hepatitis C Virus Infection and Compensated Cirrhosis

Lai Wei[1], Guiqiang Wang[2], Yan Luo[3], Chi-Jen Chu[4], Seung Woon Paik[5], Jinlin Hou[6], Jun Cheng[7], Qing Xie[8], Zhongping Duan[9], Jia-Horng Kao[10], Bo Fu[3], Niloufar Mobashery[3], Jeong Heo[11]

[1]Peking University Peoples Hospital, Beijing, China; [2]Peking University First Hospital, Beijing, China; [3]AbbVie Inc., North Chicago, Illinois, United States; [4]Taipei Veterans General Hospital, Taipei City, Taiwan; [5]Samsung Medical Center, Sungkyunkwan University, Seoul, Republic of Korea; [6]Nanfang Hospital of Southern Medical University, Guangzhou, China; [7]Beijing Di Tan Hospital, Capital Medical University, Beijing, China; [8]Ruijin Hospital Shanghai Jiaotong, University School of Medicine, Shanghai, China; [9]Beijing Youan Hospital Capital Medical University, Beijing, China; [10]National Taiwan University Hospital, Zhongzheng District, Taiwan; [11]Department of Internal Medicine, College of Medicine, Pusan National University and Medical Research Institute, Pusan National University Hospital, Busan, Republic of Korea

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

INTRODUCTION

- In Asia, more than 100 million people may be chronically infected with the hepatitis C virus (HCV)[1]
- HCV genotype 1b (GT1b) is the most prevalent sub-genotype in China,[2,3] South Korea,[2] and Taiwan[2,4]
- Patients with chronic HCV infection and cirrhosis have a higher risk for liver-related complications such as hepatocellular carcinoma (HCC) and are more difficult to cure than patients without cirrhosis[5]
- Five-year survival of liver-cirrhosis patients is 74% (for those who do not undergo a liver transplant) mostly attributed to HCC which has the highest incidence in GT1b HCV infection[6]
- Interferon (IFN)/pegylated IFN (pegIFN) and ribavirin (RBV) remain the current standard of care in some Asian countries where GT1b has high prevalence[7]
- Paritaprevir (PTV, formerly ABT-450) is a potent HCV NS3/4A protease inhibitor (identified by AbbVie and Enanta); ombitasvir (OBV, formerly ABT-267) is a potent HCV NS5A inhibitor; dasabuvir (DSV, formerly ABT-333) is a non-nucleoside HCV NS5B RNA polymerase inhibitor
- Previous multinational Phase 3 studies in North America, Australia, and Europe have demonstrated that 12-week treatment with the 3 direct-acting antiviral agents (3-DAA) OBV/PTV/ritonavir (r) + DSV ± ribavirin (RBV) was well tolerated and achieved sustained virologic response at post-treatment Week 12 (SVR12) in 100% of patients chronically infected with GT1b HCV with compensated cirrhosis[5]

Multi-targeted 3 Direct-acting Antiviral Regimen (3-DAA)

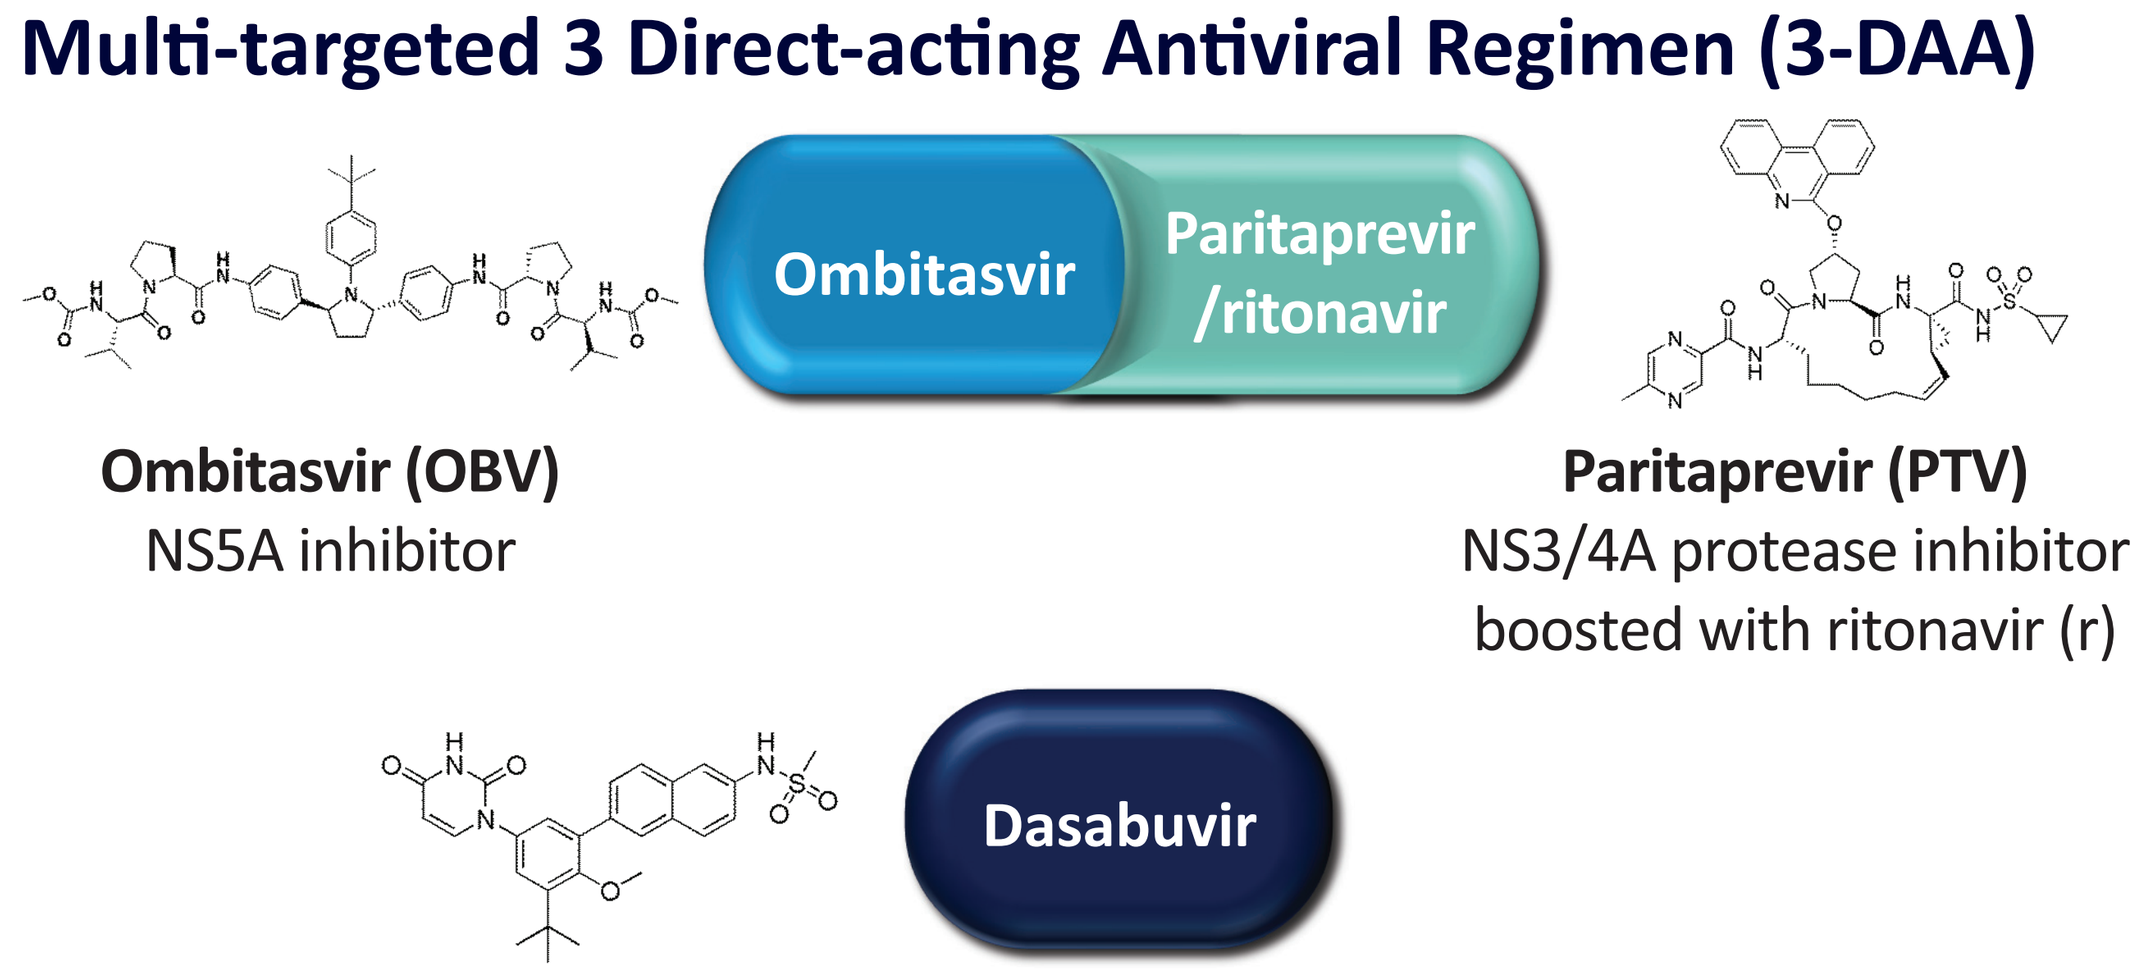
Ombitasvir
Paritaprevir /ritonavir
Dasabuvir

Ombitasvir (OBV) NS5A inhibitor

Paritaprevir (PTV) NS3/4A protease inhibitor boosted with ritonavir (r)

Dasabuvir (DSV) non-nucleoside NS5B RNA polymerase inhibitor

PTV was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVE

Evaluate the safety and efficacy of OBV/PTV/r plus DSV with RBV in adults with GT1b HCV infection and compensated cirrhosis in China, South Korea, and Taiwan following 12 weeks of therapy.

METHODS

- ONYX-II is an ongoing Phase 3, multicenter, single-arm trial conducted in China, South Korea, and Taiwan
- Patients received the 3-DAA + RBV regimen for 12 weeks and are to be followed for 48 weeks post-treatment

Figure 1. ONYX-II Study Design

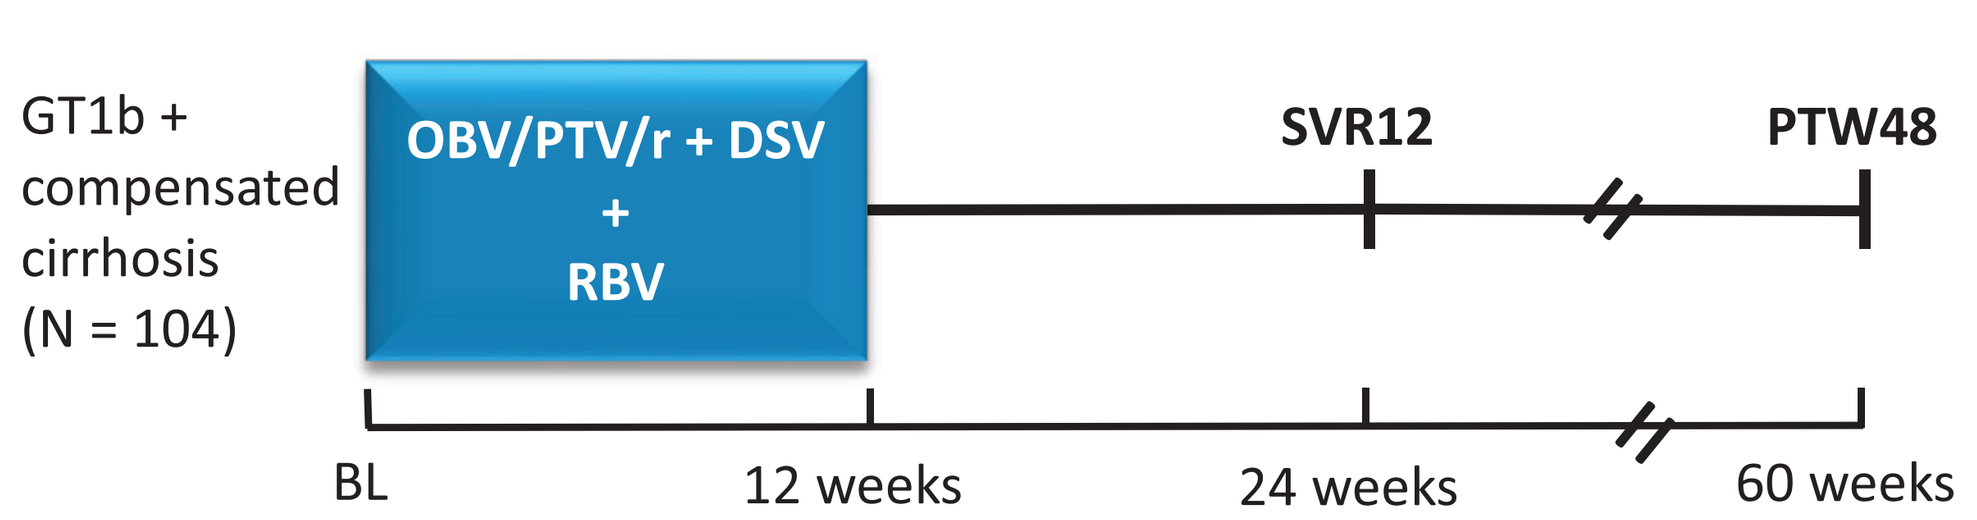
ONYX-II
GT1b + compensated cirrhosis (N = 104)
OBV/PTV/r + DSV + RBV
SVR12
PTW48
BL
12 weeks
24 weeks
60 weeks

METHODS (CONTINUED)

KEY ELIGIBILITY CRITERIA

Main inclusion criteria:

- Chinese, South Korean, and Taiwanese patients with full Chinese, South Korean, and Taiwanese parentage, respectively
- 18 to 70 years of age, inclusive, at time of screening
- Chronic GT1b HCV-infection
- Child-Pugh Score of ≤6 at Screening and documentation of cirrhosis by
 - Liver biopsy demonstrating Cirrhosis (eg, Metavir Score of >3 or an Ishak score of >4)
 - FibroScan score ≥14.6 kPa within 6 months of Screening or during the Screening Period
- Treatment-naïve or treatment-experienced (IFN/pegIFN and RBV)

Main exclusion criteria:

- Presence of HCV genotypes/subgenotype other than GT1b
- Co-infection with the hepatitis B virus (HBV) or human immunodeficiency virus (HIV)
- Any current or past clinical evidence of Child-Pugh B or C classification or clinical history of liver decompensation including ascites, variceal bleeding, or hepatic encephalopathy
- Confirmed presence of hepatocellular carcinoma (HCC)
- Any primary cause of liver disease other than chronic HCV infection
- Abnormal laboratory result that meets at least 1 of the following criteria (see **Table 1**)

Table 1. Abnormal Laboratory Results Exclusion Criteria

Assessment	Value
Alanine Aminotransferase	>7 × ULN
Aspartate Aminotransferase	>7 × ULN
Estimated Glomerular Filtration Rate (eGFR)*	<50 mL/min/1.73m²
Albumin	<2.8 g/dL
International normalized ratio (INR)†	>2.3
Hemoglobin	<LLN
Platelets	<60 000 cells per mm³
Absolute Neutrophil Count (ANC)	<1200 cells/µL
Total Bilirubin	≥3.0 mg/dL

LLN = lower limit of normal, ULN = upper limit of normal.
*Modified for the Chinese population as follows: eGFR = 175 × (Serum Creatinine)$^{-1.234}$ × (Age)$^{-0.179}$ × (0.79 if Female).
†Subjects with a known inherited blood disorder and INR >2.3 may have been enrolled with permission of the AbbVie study designated physician.

STUDY ASSESSMENTS

Primary Efficacy Endpoint	Percentage of patients with SVR12 compared to historical SVR rate for telaprevir (TLV) plus pegIFN/RBV therapy*
Secondary Efficacy Objectives	Percentage of patients with on-treatment virologic failure and post-treatment relapse
Safety Assessment	Percentage of patients with treatment-emergent adverse events (AEs) and laboratory abnormalities

*Superiority is achieved if the lower bound of the 95% CI for Arm A SVR12 rate exceeds 67%.

RESULTS

PARTICIPANTS

- A total of 104 patients were enrolled in this study
 - China: 63; South Korea: 21; Taiwan: 20
 - 100% were of Asian
 - 58% were treatment-experienced

Table 2. Patient Demographics and Baseline Characteristics

Characteristic	China n = 63	South Korea n = 21	Taiwan n = 20	Total N = 104
Female, n (%)	43 (68)	10 (48)	11 (55)	64 (62)
Age, median (range), years	55 (24–69)	60 (47–69)	60 (33–68)	56 (24–69)
BMI, mean ± SD, kg/m²	24.6 ± 2.9	26.6 ± 3.3	26.6 ± 2.9	25.4 ± 3.1
HCV RNA, median (range), $\log_{10}$ IU/mL	6.3 (4.4–7.2)	6.3 (5.0–7.0)	6.2 (4.1–6.8)	6.3 (4.1–7.2)
IL28B CC genotype, n (%)	44 (70)	15 (71)	13 (65)	72 (69)
Treatment-naïve, n (%)	31 (49)	6 (29)	7 (35)	44 (42)
Treatment-experienced, n (%)	32 (51)	15 (71)	13 (65)	60 (58)
Non-responder, n/N (%)	13/32 (41)	8/15 (53)	4/13 (31)	25/60 (42)
Relapser, n/N (%)	16/32 (50)	5/15 (33)	8/13 (62)	29/60 (48)
IFN intolerant, n/N (%)	3/32 (9)	2/15 (13)	1/13 (8)	6/60 (10)

EFFICACY

- SVR12 was achieved by 100% of patients. The lower bound of the 95% confidence interval (LCB) was above the 67% superiority threshold; therefore, the 3-DAA regimen demonstrated superiority to the historical telaprevir plus pegIFN/RBV SVR rate **(Figure 2)**

Figure 2. SVR12 Rates (and 95% CIs)

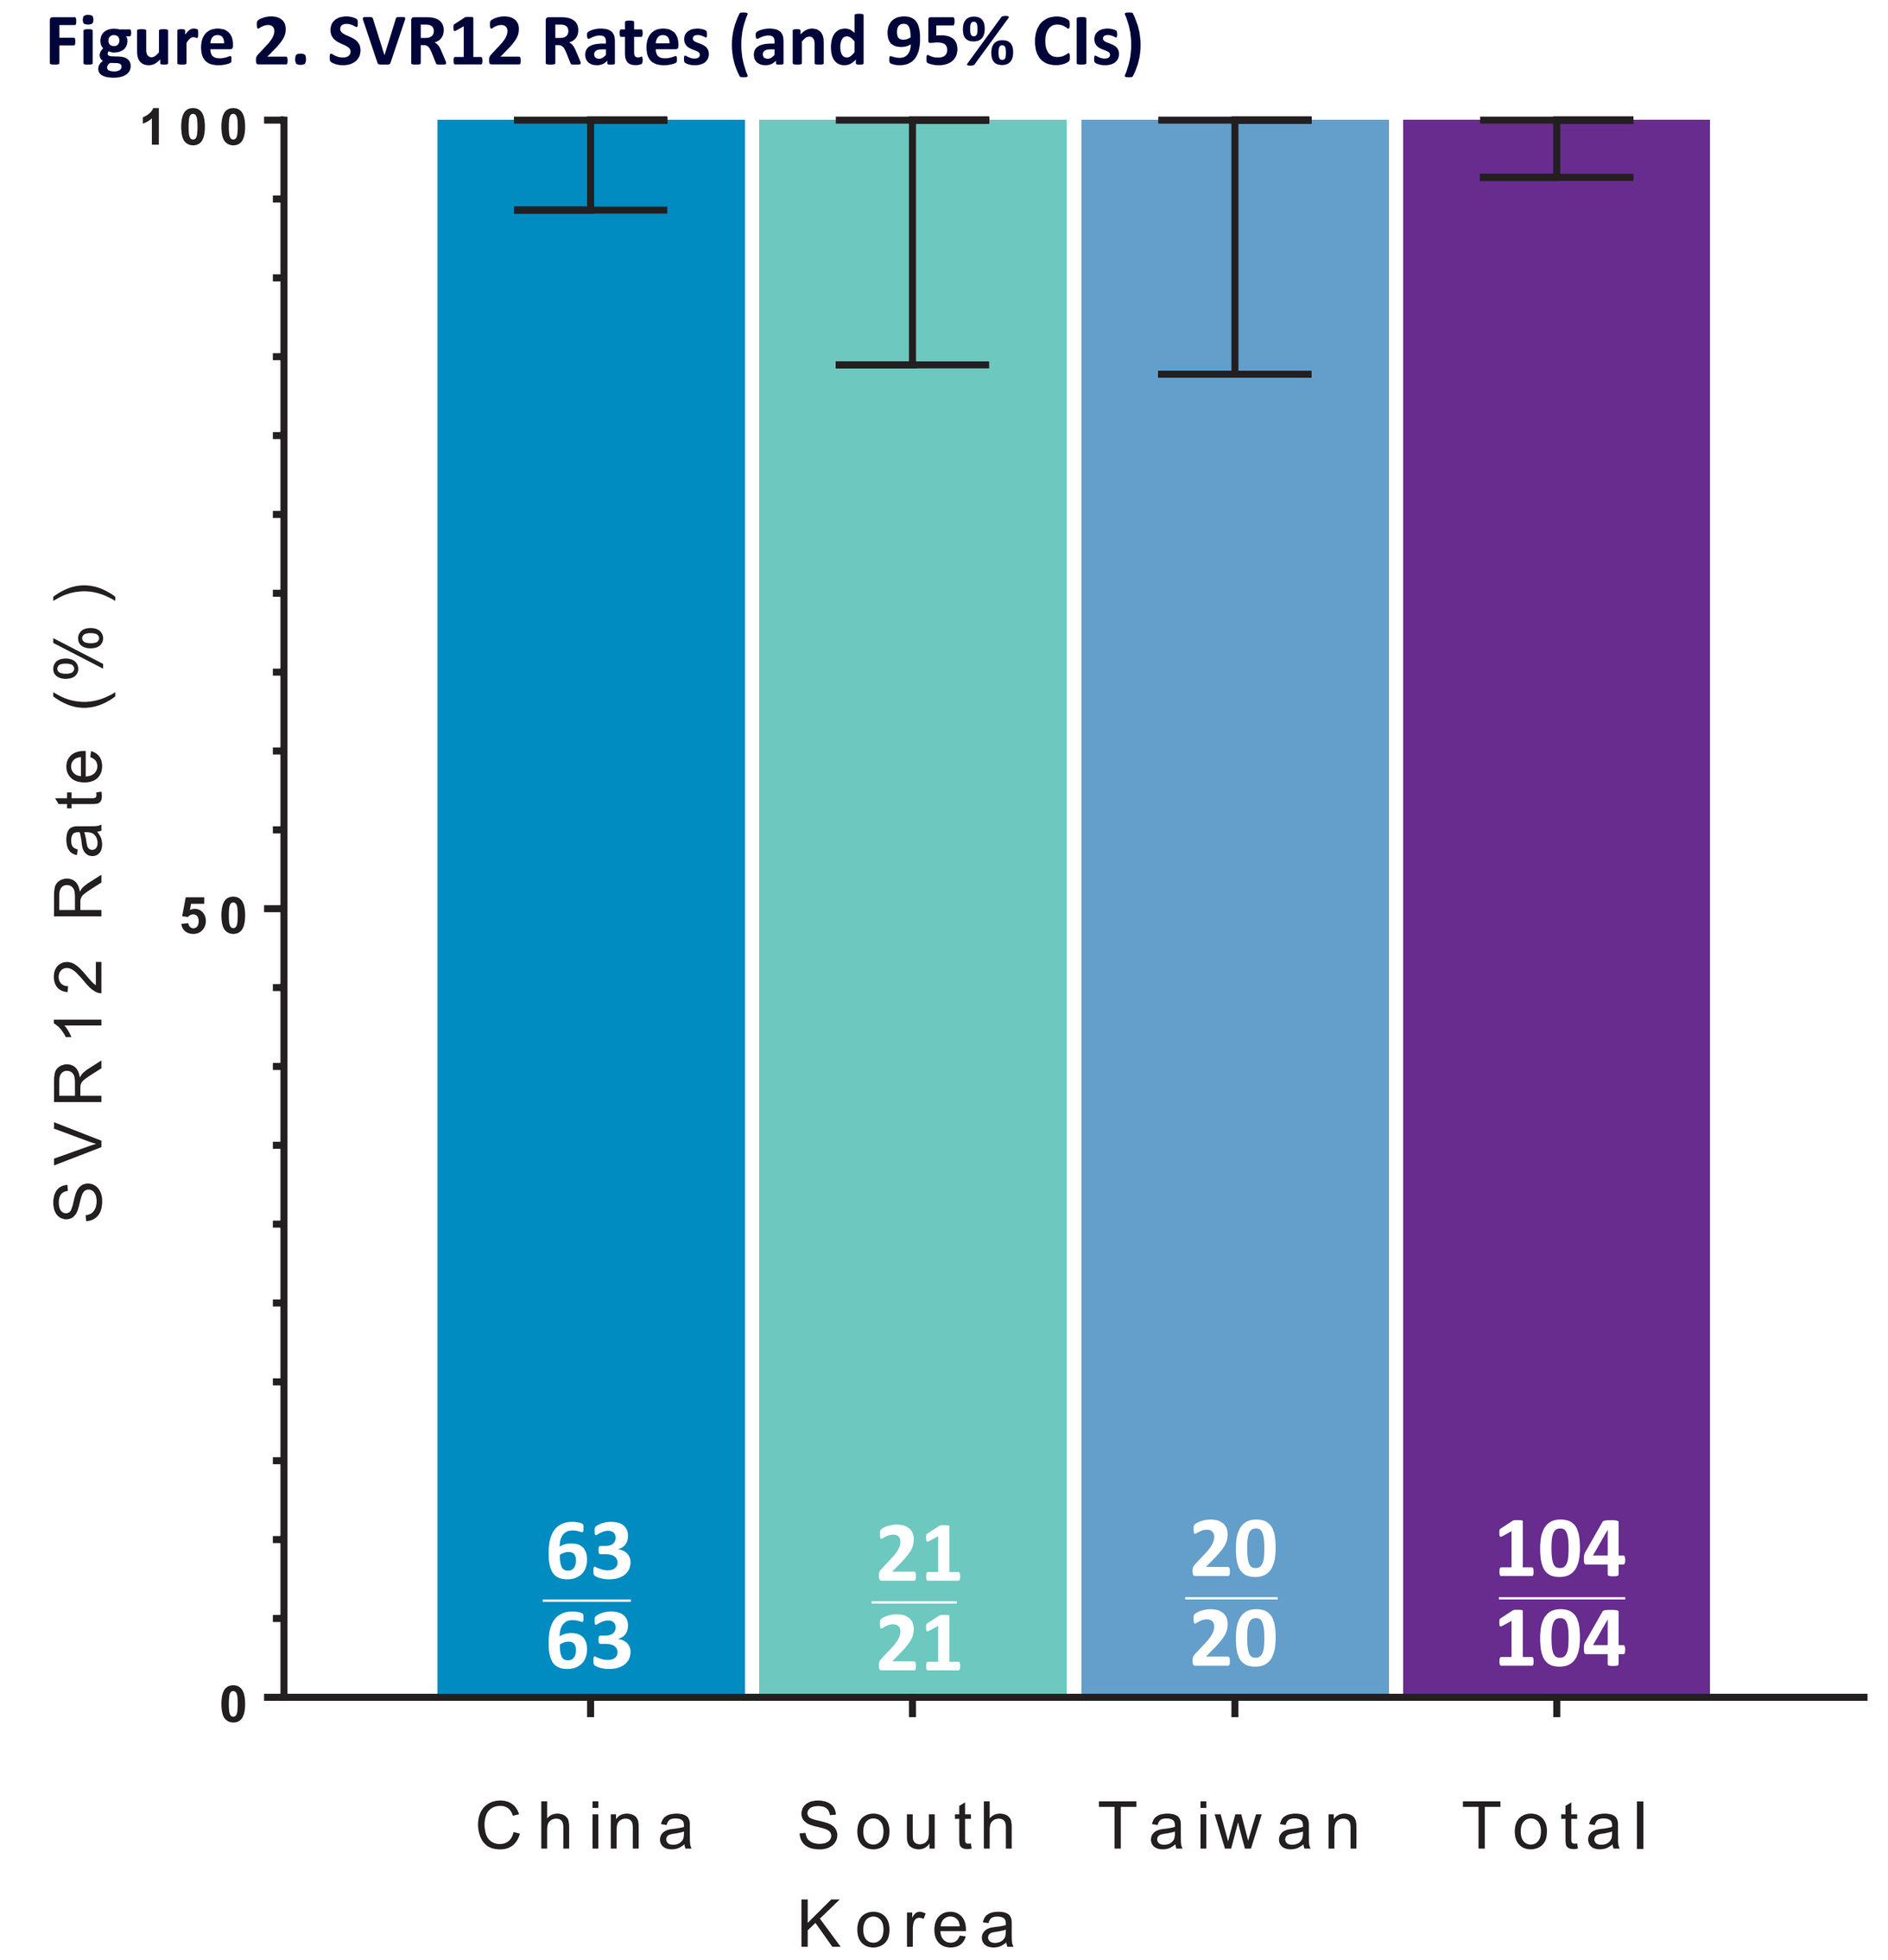
SVR12 Rate (%)
100
50
0
63/63
21/21
20/20
104/104
China
South Korea
Taiwan
Total

SAFETY

- Most AEs were mild in severity
- The 3 most common AEs (see **Table 3** for a list) were increase in blood bilirubin (25%), pruritus (15%), and anemia (14%), with most cases of anemia being considered RBV-related
- In general, cases of bilirubin increase were asymptomatic, transient and did not result in study drug interruption or discontinuation. They were mainly driven by indirect bilirubin increase and were not associated with other abnormal liver function test results
- Serious AE were reported in 4/104 (4%) of patients, none of which was assessed as being DAA-related
- One patient in the Chinese cohort had a serious AE (ventricular extrasystoles) that was assessed as having a reasonable possibility of being RBV related but not 3-DAA-related
- One patient in the South Korean cohort discontinued study drug due to increases in alanine and aspartate aminotransferase and total bilirubin after 3 weeks of treatment, and went on to achieve SVR12
- RBV dose was reduced in 11 (11%) patients mostly due to anemia and hemoglobin decreases
- Study drug was interrupted in 1 Chinese patient due to intermittent AEs (vomiting and dizziness)
- No cases of liver decompensation (such as ascites, variceal bleeding or hepatic encephalopathy) were reported

Table 3. Treatment-emergent Adverse Events

Event, n (%)	China n = 63	South Korea n = 21	Taiwan n = 20	Total N = 104
Any AE	55 (87)	12 (57)	18 (90)	85 (82)
AEs leading to drug d/c	0	1 (5)*	0	1 (1)
Serious AE†	3 (5)	1 (5)	0	4 (4)
Death	0	0	0	0
AEs with >10% frequency in the total population				
Blood bilirubin increased¶	23 (37)	2 (10)	1 (5)	26 (25)
Pruritus	6 (10)	4 (19)	6 (30)	16 (15)
Anemia	8 (13)	1 (5)	5 (25)	14 (14)
Asthenia	12 (19)	0	0	12 (12)
Conjugated bilirubin increased§,¶	11 (18)	0	1 (5)	12 (12)
Unconjugated bilirubin increased‖,¶	12 (19)	0	0	12 (12)
Dizziness	4 (6)	0	7 (35)	11 (11)
Fatigue	2 (3)	1 (5)	8 (40)	11 (11)

*Due to increases in alanine and aspartate aminotransferase and total bilirubin. Patient achieved SVR12.
†One serious AE (ventricular extrasystoles) in the Chinese cohort was assessed as being RBV-related.
§10 patients (9 in the Chinese cohort and 1 in the Taiwanese cohort) also had an AE of blood bilirubin increase reported.
‖9 patients also had an AE of blood bilirubin increase reported, 9 also had an AE of conjugated bilirubin increase reported and 7 had all 3 AEs reported, all being from the Chinese cohort.
¶Higher AE reporting in China vs South Korea and Taiwan was noted; however, the incidence of laboratory abnormalities by grade was comparable among the 3 countries (see **Table 4**).

Table 4. Post-baseline Laboratory Abnormalities

Post-baseline Laboratory Abnormalities	China n = 63	South Korea n = 21	Taiwan n = 20	Total N = 104
Hemoglobin				
Grade ≥2	4 (6)	2 (10)	4 (20)	10 (10)
Grade ≥3	0	0	0	0
Alanine aminotransferase				
Grade ≥2	2 (3)	1 (5)	1 (5)	4 (4)
Grade ≥3	1 (2)	1 (5)	1 (5)	3 (3)
Aspartate aminotransferase				
Grade ≥2	0	1 (5)	1 (5)	2 (2)
Grade ≥3	0	1 (5)	1 (5)	2 (2)
Total bilirubin				
Grade ≥2	31 (49)	10 (48)	11 (55)	52 (50)
Grade ≥3	5 (8)	0	2 (10)	7 (7)

CONCLUSIONS

- SVR12 was achieved by 100% of patients enrolled in this study
- The regimen was well tolerated; AEs were mostly mild
- One patient had AEs leading to study drug discontinuation
- One serious AE in the Chinese cohort was assessed as being RBV-related but not DAA-related
- Post-baseline laboratory abnormalities with a severity grade ≥3 were rare
- The efficacy and safety profiles observed in this Asian regional Phase 3 study are similar to those in the global Phase 3 studies

ACKNOWLEDGEMENTS

AbbVie sponsored the studies, contributed to their designs, participated in the collection, analysis, and interpretation of data, and in the writing, reviewing, and approval of the publication.

We thank all the patients and investigators who participated in this study. We thank AbbVie employees Angela Nillus, Nancy Shulman, Tahira Payne, Jim A Watson, Olga Furman, Vilma V Kaus, Kristine M Cwik, Jun Sun, Felecia Hughey, Danijela Dobirc, Chunyan Yuan, Jennifer King, Jiuhong Zha, Diana Shuster, Tami Pilot-Matias, Rakesh Tripathi, Gretja Schnell, and Katia Alvis.

Medical writing support was provided by Maher Quraan, PhD, of AbbVie.

REFERENCES

1. Mohd Hanafiah K, Groeger J, Flaxman AD, Wiersma ST. Global epidemiology of hepatitis C virus infection: new estimates of age-specific antibody to HCV seroprevalence. *Hepatology*. 2013;57(4):1333–42.
2. Sievert W, Altraif I, Razavi HA, et al. A systematic review of hepatitis C virus epidemiology in Asia, Australia and Egypt. *Liver Int.* 2011;31 Suppl 2:61–80.
3. Rao H, Wei L, Lopez-Talavera JC, et al. Distribution and clinical correlates of viral and host genotypes in Chinese patients with chronic hepatitis C virus infection. *J Gastroenterol Hepatol*. 2014;29(3):545–53.
4. Lee MH, Yang HI, Lu SN, et al. Hepatitis C virus genotype 1b increases cumulative lifetime risk of hepatocellular carcinoma. *Int J Cancer*. 2014;135(5):1119–26.
5. Feld JJ, Moreno C, Trinh R, et al. Sustained virologic response of 100% in HCV genotype 1b patients with cirrhosis receiving ombitasvir/paritaprevir/r and dasabuvir for 12 weeks. *J Hepatol*. 2016;64(2):301–7.
6. Bennett H, Waser N, Johnston K, et al. A review of the burden of hepatitis C virus infection in China, Japan, South Korea, and Taiwan. *Hepatol Int*. 2015;9(3):378–90.
7. Duan Z, Jia JD, Hou J, et al. Current challenges and the management of chronic hepatitis C in mainland China. *J Clin Gastroenterol*. 2014;48(8):679–86.
8. Kumada H, Chayama K, Rodrigues L, Jr., et al. Randomized phase 3 trial of ombitasvir/paritaprevir/ritonavir for hepatitis C virus genotype 1b-infected Japanese patients with or without cirrhosis. *Hepatology*. 2015;62(4):1037–46.

DISCLOSURES

AbbVie sponsored the study (M14-491) contributed to its design, collection, analysis, and interpretation of the data, and participated in the writing, review, and approval of the abstract. All authors had access to relevant data.

L Wei: Received research support from Bristol-Myers Squib, and Roche; Advisory board for Abbott, AbbVie, BMS, Galmed, and Gilead. G Wang: Participated in an AbbVie sponsored clinical study. CJ Chu: Speaker for Gilead, Bristol-Meyers Squibb, MSD, and Roche. SW Paik: Received research support from AbbVie, Bristol-Meyers Squibb, Gilead, GlaxoSmithKline, MSD, Novartis, and Roche. J Hou: Received research support from Bristol-Meyers Squib, Novartis, GSK, and Roche; Advisory board for AbbVie, Novartis, and Gilead. J Cheng: Participated in an AbbVie sponsored clinical study. Q Xie: Participated in an AbbVie sponsored clinical study. Z Duan: Participated in an AbbVie sponsored clinical study. JH Kao: Consultant for Abbvie, Bristol-Meyers Squibb, Gilead, Novartis, Roche; Speaker for Roche, Bristol-Meyers Squibb, Novartis, Gilead Sciences. Y Luo, B Fu, N Mobashery: employees of AbbVie and may hold stock or stock options. J Heo: Research support from BMS and Hoffmann-La Roche; Advisor for AbbVie, BMS, Gilead Sciences, Pharma Essentia, SillaJen, and Johnson & Johnson.

44588-161469-875 — AASLD 2016 — AbbVie — proof 2 (rjh) — november 2, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051

CREATED @ 100%