Improved Hepatic Profile in Hepatitis C Virus (HCV) Genotype (GT) 4-Infected Egyptian Patients With Compensated Cirrhosis Receiving Ombitasvir/Paritaprevir/Ritonavir With Ribavirin (AGATE-II)

Imam Waked[1], Gamal E Esmat[2], Rabab Fouad[2], Naglaa Allam[1], Mohamed Hassany[3], Mohammad Mohey[2], Ayman Yosry[2], Gamal E Shiha[4], Reham Soliman[4], Marisol Martinez[5], Roula B Qaqish[5], Colleen Hall[5], Niloufar Mobashery[5]

[1]National Liver Institute, Menoufiya, Egypt; [2]Cairo University, Cairo, Egypt; [3]National Hepatology and Tropical Medicine Research Institute, Cairo, Egypt; [4]Mansoura University and Egyptian Liver Research Institute and Hospital (ELRIAH), Mansoura, Egypt; [5]AbbVie Inc., North Chicago, Illinois, United States

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

BACKGROUND

- In Egypt, chronic hepatitis C virus (HCV) infection is a leading cause of liver cirrhosis and liver cancer[1]
- Although HCV genotype (GT) 4 accounts for approximately 8% of HCV infections globally, it constitutes 97% of HCV infections in Egypt[2]
- Achievement of sustained virologic response 12 weeks post-treatment (SVR12) is associated with improvement in liver fibrosis and function and reduced risk of liver decompensation and need for transplant[3–5]
- The 2 direct-acting antiviral (2-DAA) regimen of ombitasvir (OBV) co-formulated with paritaprevir and the pharmacokinetic enhancer ritonavir (PTV/r) plus ribavirin (RBV) achieved high SVR12 rates in GT4-infected patients without cirrhosis or with compensated cirrhosis[5–8]
 - In the phase 3 AGATE-I study, OBV/PTV/r + RBV for 12 or 16 weeks achieved high SVR12 rates (97% and 98% for 12- and 16-week regimens, respectively) and improved markers of liver fibrosis and synthetic function in GT4-infected patients with compensated cirrhosis from Europe and North America[5,7]
 - In the phase 3 AGATE-II study, OBV/PTV/r + RBV for 12 or 24 weeks achieved high SVR12 rates in Egyptian GT4-infected patients without cirrhosis (94% for 12-week regimen) or with compensated cirrhosis (97% and 93% for 12- and 24-week regimens, respectively)[8]

2-DAA HCV Regimen

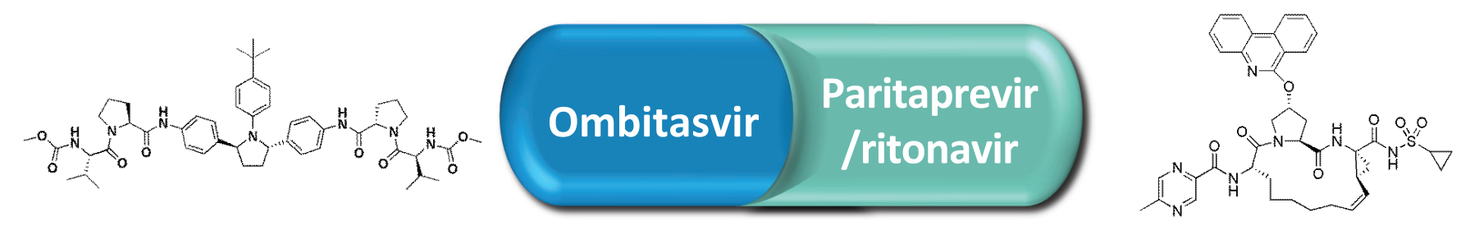
Ombitasvir
Paritaprevir /ritonavir

Ombitasvir (OBV)
NS5A inhibitor

Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)

PTV was identified by AbbVie and Enanta.
Ritonavir does not have antiviral activity against HCV.

OBJECTIVES

- The primary objectives of the AGATE-II study were to assess the efficacy and safety of OBV/PTV/r + RBV in Egyptian GT4-infected patients without cirrhosis or with compensated cirrhosis
- Improvements in biomarkers of liver synthetic function, injury, and fibrosis after treatment with OBV/PTV/r + RBV in Egyptian GT4-infected patients with compensated cirrhosis in AGATE-II are presented here

DISCLOSURES

I Waked: Speaker, Advisory Board Member, and/or Investigator: AbbVie, BMS, Gilead, Janssen, Merck, Onxio, Pharco Pharmaceuticals, Roche.
GE Esmat: Participation in AbbVie-sponsored clinical studies.
R Fouad: Investigator: AbbVie, Gilead, Pharco Pharmaceuticals.
N Allam: Investigator: AbbVie, BMS, Pharco Pharmaceuticals.
M Hassany: Investigator: AbbVie, Janssen, Gilead; Speaker: AbbVie.
M Mohey: Participation in AbbVie-sponsored clinical studies.
A Yosry: Participation in AbbVie-sponsored clinical studies.
GE Shiha: Participation in AbbVie-sponsored clinical studies.
R Soliman: Investigator: AbbVie.
M Martinez, C Hall, and N Mobashery: Employees of AbbVie and may hold stock or options.

METHODS

STUDY DESIGN

- AGATE-II (NCT02247401) was a phase 3, open-label trial conducted at 5 sites in Egypt
- Patients without cirrhosis received once-daily OBV/PTV/r (25 mg/150 mg/100 mg) with weight-based RBV for 12 weeks (Arm A)
- Patients with compensated cirrhosis were randomized 1:1 to the same regimen for either 12 (Arm B) or 24 weeks (Arm C)
- RBV dosage could be reduced if hemoglobin declined <10 g/dL, and interrupted if it declined <8.5 g/dL

KEY ELIGIBILITY CRITERIA

- Eligible patients were adults chronically infected with HCV GT4 for at least 6 months (plasma HCV RNA >1000 IU/mL at screening):
 - HCV treatment-naive or treatment-experienced (prior pegylated-interferon/RBV null responders, partial responders or relapsers)
 - Without cirrhosis or with compensated cirrhosis (Child-Pugh score ≤6 at screening)
- Patients with hepatitis B or HIV virus co-infection, or infection with any HCV GT other than GT4 were excluded
- Patients with a history of hepatic decompensation were also excluded

STUDY ANALYSES

- Efficacy was assessed by the proportion of patients with SVR12 (HCV RNA < lower limit of quantification 12 weeks post-treatment), on-treatment virologic failure and relapse
- In the primary efficacy analyses, all subjects who did not achieve SVR12 for any reason, including missing data, were included in the analysis and counted as failures. A sensitivity analysis for primary efficacy endpoint was completed in which subjects who did not achieve SVR12 for reasons other than virologic failure were excluded
- Mean changes from baseline to post-treatment week (PTW) 24 in biomarkers of liver synthetic function, liver injury, and liver fibrosis were assessed for patients with compensated cirrhosis
 - Hepatic synthetic function or liver's ability to eliminate toxins was assessed by international normalized ratio (INR), albumin, and total bilirubin
 - Liver injury was assessed by alanine aminotransferase (ALT) and aspartate aminotransferase (AST)
 - Liver fibrosis was assessed by platelet count, AST to platelet ratio index (APRI), and fibrosis-4 (FIB-4)
- Treatment-emergent adverse events (TEAEs) and laboratory abnormalities were assessed in all patients receiving at least 1 dose of study drug

RESULTS

PATIENTS

- Patient demographics and baseline disease characteristics for patients with compensated cirrhosis are presented in **Table 1**

Table 1. Patient Demographics and Baseline Disease Characteristics for Patients With Compensated Cirrhosis

Characteristic	Arm B 12 weeks N = 31	Arm C 24 weeks N = 29
Male, n (%)	29 (94)	22 (76)
Age, years, mean (SD)	57 (6)	56 (8)
BMI, kg/m², mean (SD)	29 (4)	31 (5)
HCV RNA, $\log_{10}$ IU/mL, mean (SD)	6.02 (0.62)	5.97 (0.69)
Prior PegIFN/RBV treatment experience, n (%)	16 (52)	14 (48)
Null responder	9 (56)	7 (50)
Partial responder	2 (13)	2 (14)
Relapser	5 (31)	5 (36)
Metavir fibrosis stage, n (%)		
F0–1	0	0
F2	0	0
F3	1 (3)*	0
F4	30 (97)	29 (100)
Platelet count, × 10^9/L, median (range)	141 (46–365)	125 (59–238)
Albumin, g/L, median (range)	41(34–53)	40 (31–54)

BMI, body-mass index; SD, standard deviation.
*One patient without cirrhosis was miscategorized as having compensated cirrhosis at the time of enrollment and was assigned to Arm B.

EFFICACY

- SVR12 rates were comparable between treatment arms in patients with compensated cirrhosis **(Figure 1)**
- Two patients who failed to achieve SVR12 had on treatment virologic failure (1 each from Arm B and C); 1 patient had missing SVR12 data (Arm C)
- No post-treatment relapses were observed

Figure 1. SVR12 Rates Among Patients With Cirrhosis by Treatment Arm

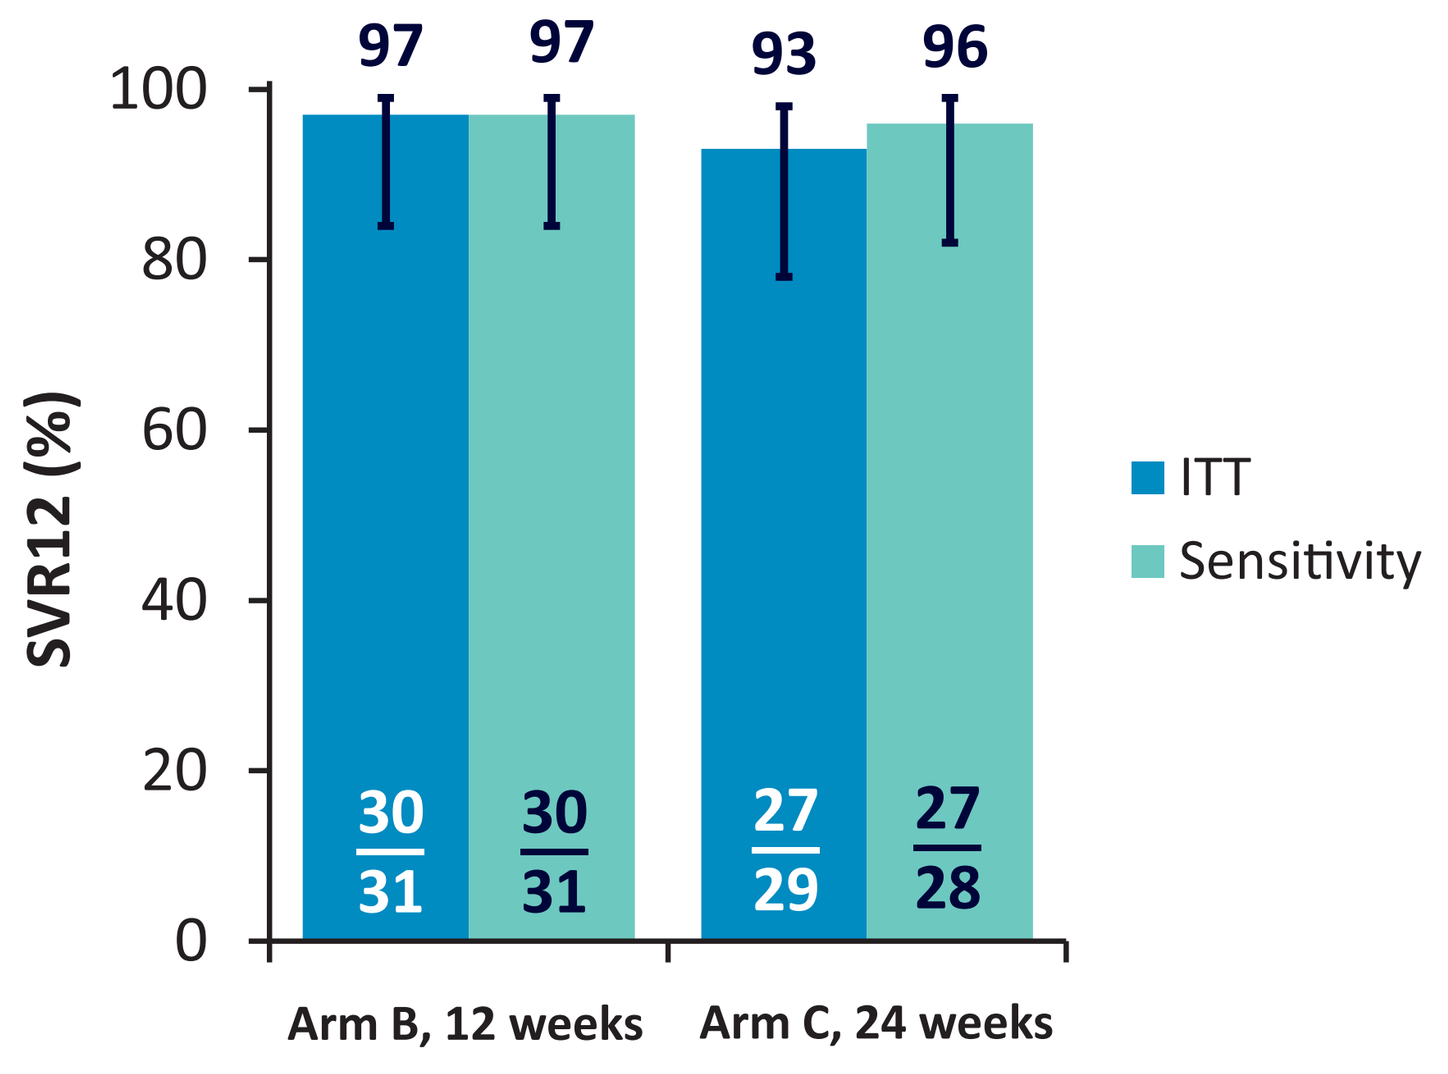
SVR12 (%)
100
80
60
40
20
0
97
97
93
96
30/31
30/31
27/29
27/28
Arm B, 12 weeks
Arm C, 24 weeks
ITT
Sensitivity

ITT, intention to treat; SVR12, sustained virologic response 12 weeks post-treatment. ITT population included all patients who received at least 1 dose of study drug. Sensitivity analysis excluded patients with non-virologic failure (ie, lost to follow-up or missing SVR12 data). Error bars represent 95% confidence intervals.

IMPROVEMENT IN BIOMARKERS OF LIVER SYNTHETIC FUNCTION IN PATIENTS WITH COMPENSATED CIRRHOSIS

- Biomarkers of liver synthetic function at baseline and PTW24 are presented in **Figure 2**

Figure 2. Mean Changes From Baseline to Post-treatment Week 24 in Biomarkers of Liver Synthetic Function in Patients With Compensated Cirrhosis

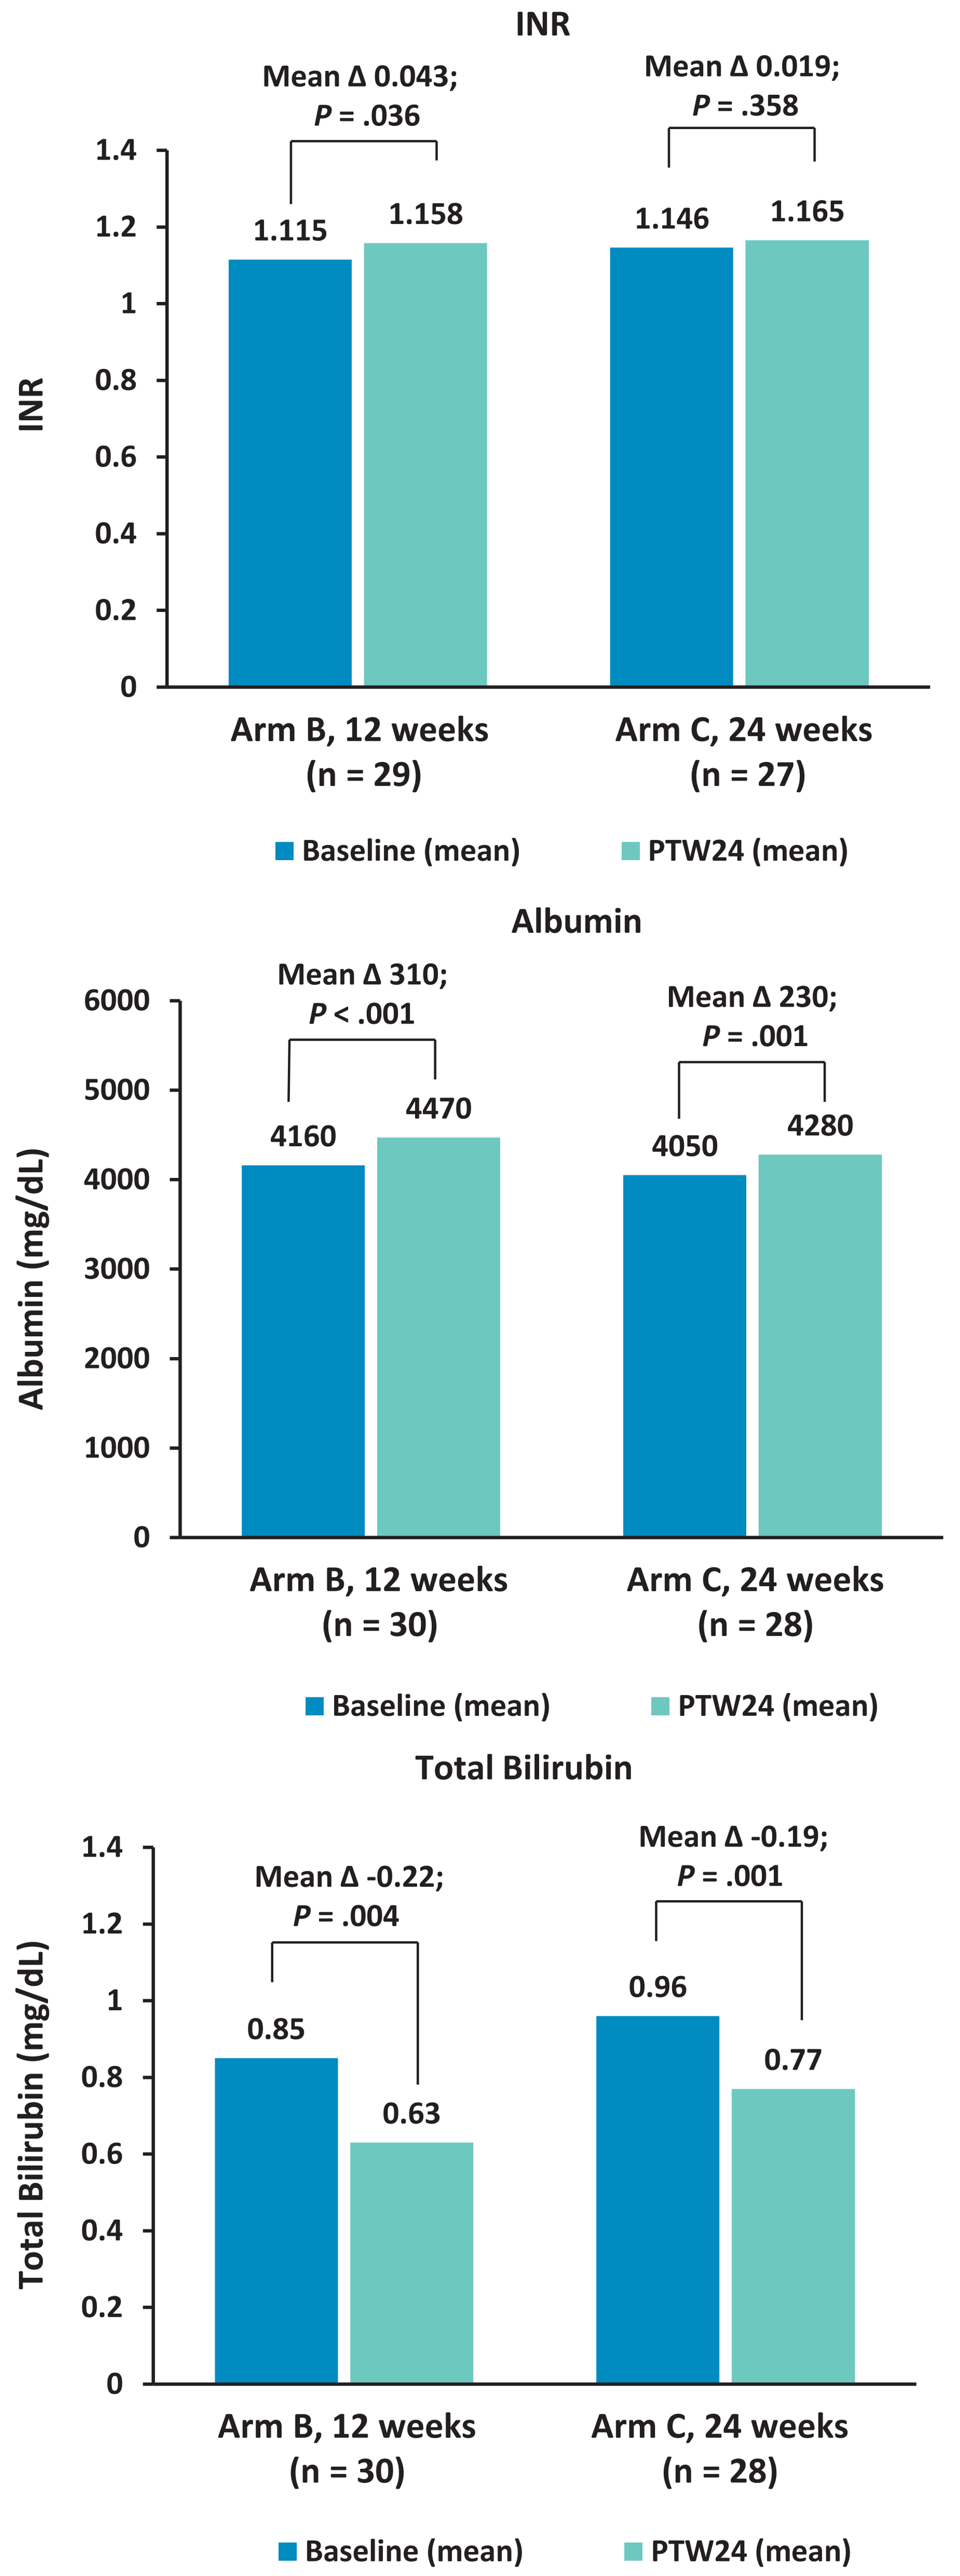
INR
Mean Δ 0.043; P = .036
Mean Δ 0.019; P = .358
1.115
1.158
1.146
1.165
Arm B, 12 weeks (n = 29)
Arm C, 24 weeks (n = 27)
Albumin
Albumin (mg/dL)
Mean Δ 310; P < .001
Mean Δ 230; P = .001
4160
4470
4050
4280
Arm B, 12 weeks (n = 30)
Arm C, 24 weeks (n = 28)
Total Bilirubin
Total Bilirubin (mg/dL)
Mean Δ -0.22; P = .004
Mean Δ -0.19; P = .001
0.85
0.63
0.96
0.77
Arm B, 12 weeks (n = 30)
Arm C, 24 weeks (n = 28)
Baseline (mean)
PTW24 (mean)

INR, international normalized ratio; PTW, post-treatment week.

IMPROVEMENT IN BIOMARKERS OF LIVER INJURY IN PATIENTS WITH COMPENSATED CIRRHOSIS

- Biomarkers of liver injury at baseline and PTW24 are presented in **Figure 3**

Figure 3. Mean Changes From Baseline to Post-treatment Week 24 in Biomarkers of Liver Injury in Patients With Compensated Cirrhosis

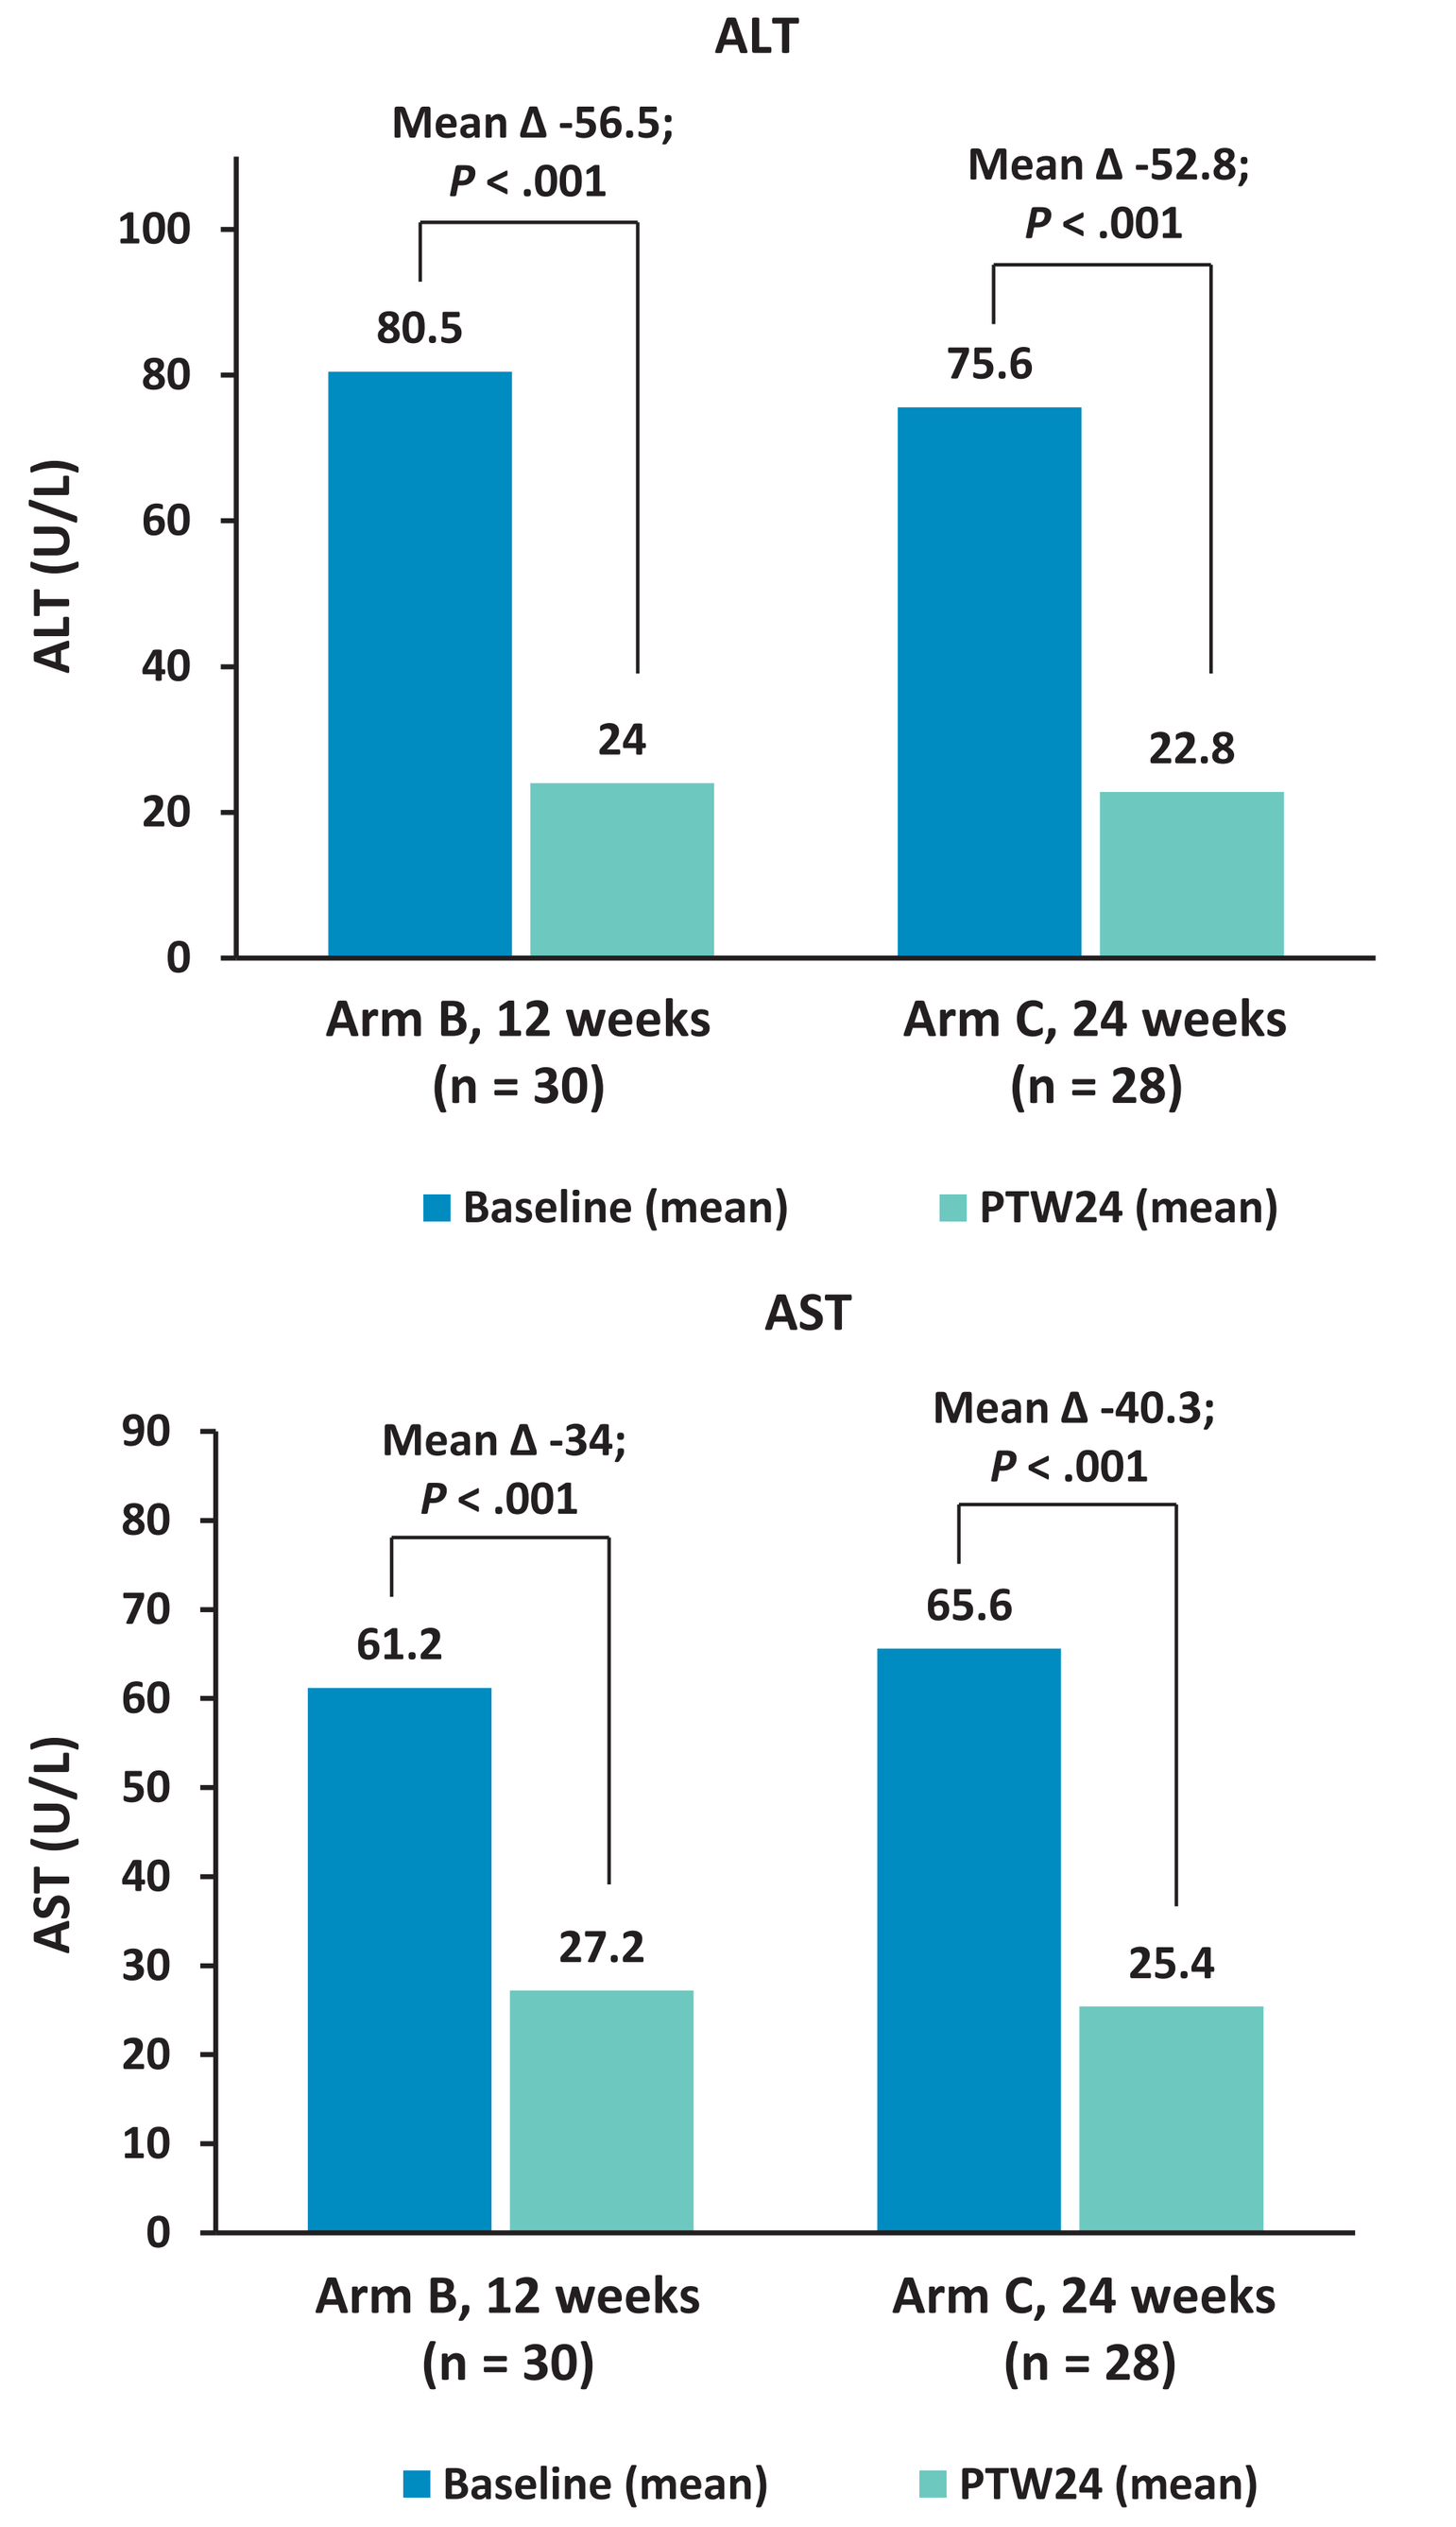
ALT
ALT (U/L)
Mean Δ -56.5; P < .001
Mean Δ -52.8; P < .001
80.5
24
75.6
22.8
Arm B, 12 weeks (n = 30)
Arm C, 24 weeks (n = 28)
AST
AST (U/L)
Mean Δ -34; P < .001
Mean Δ -40.3; P < .001
61.2
27.2
65.6
25.4
Arm B, 12 weeks (n = 30)
Arm C, 24 weeks (n = 28)
Baseline (mean)
PTW24 (mean)

ALT, alanine transaminase; AST, aspartate transaminase; PTW, post-treatment week.

IMPROVEMENT IN BIOMARKERS OF LIVER FIBROSIS IN PATIENTS WITH COMPENSATED CIRRHOSIS

- Biomarkers of liver fibrosis at baseline and PTW24 are presented in **Figure 4**

Figure 4. Mean Changes From Baseline to Post-treatment in Biomarkers of Liver Fibrosis in Patients With Compensated Cirrhosis

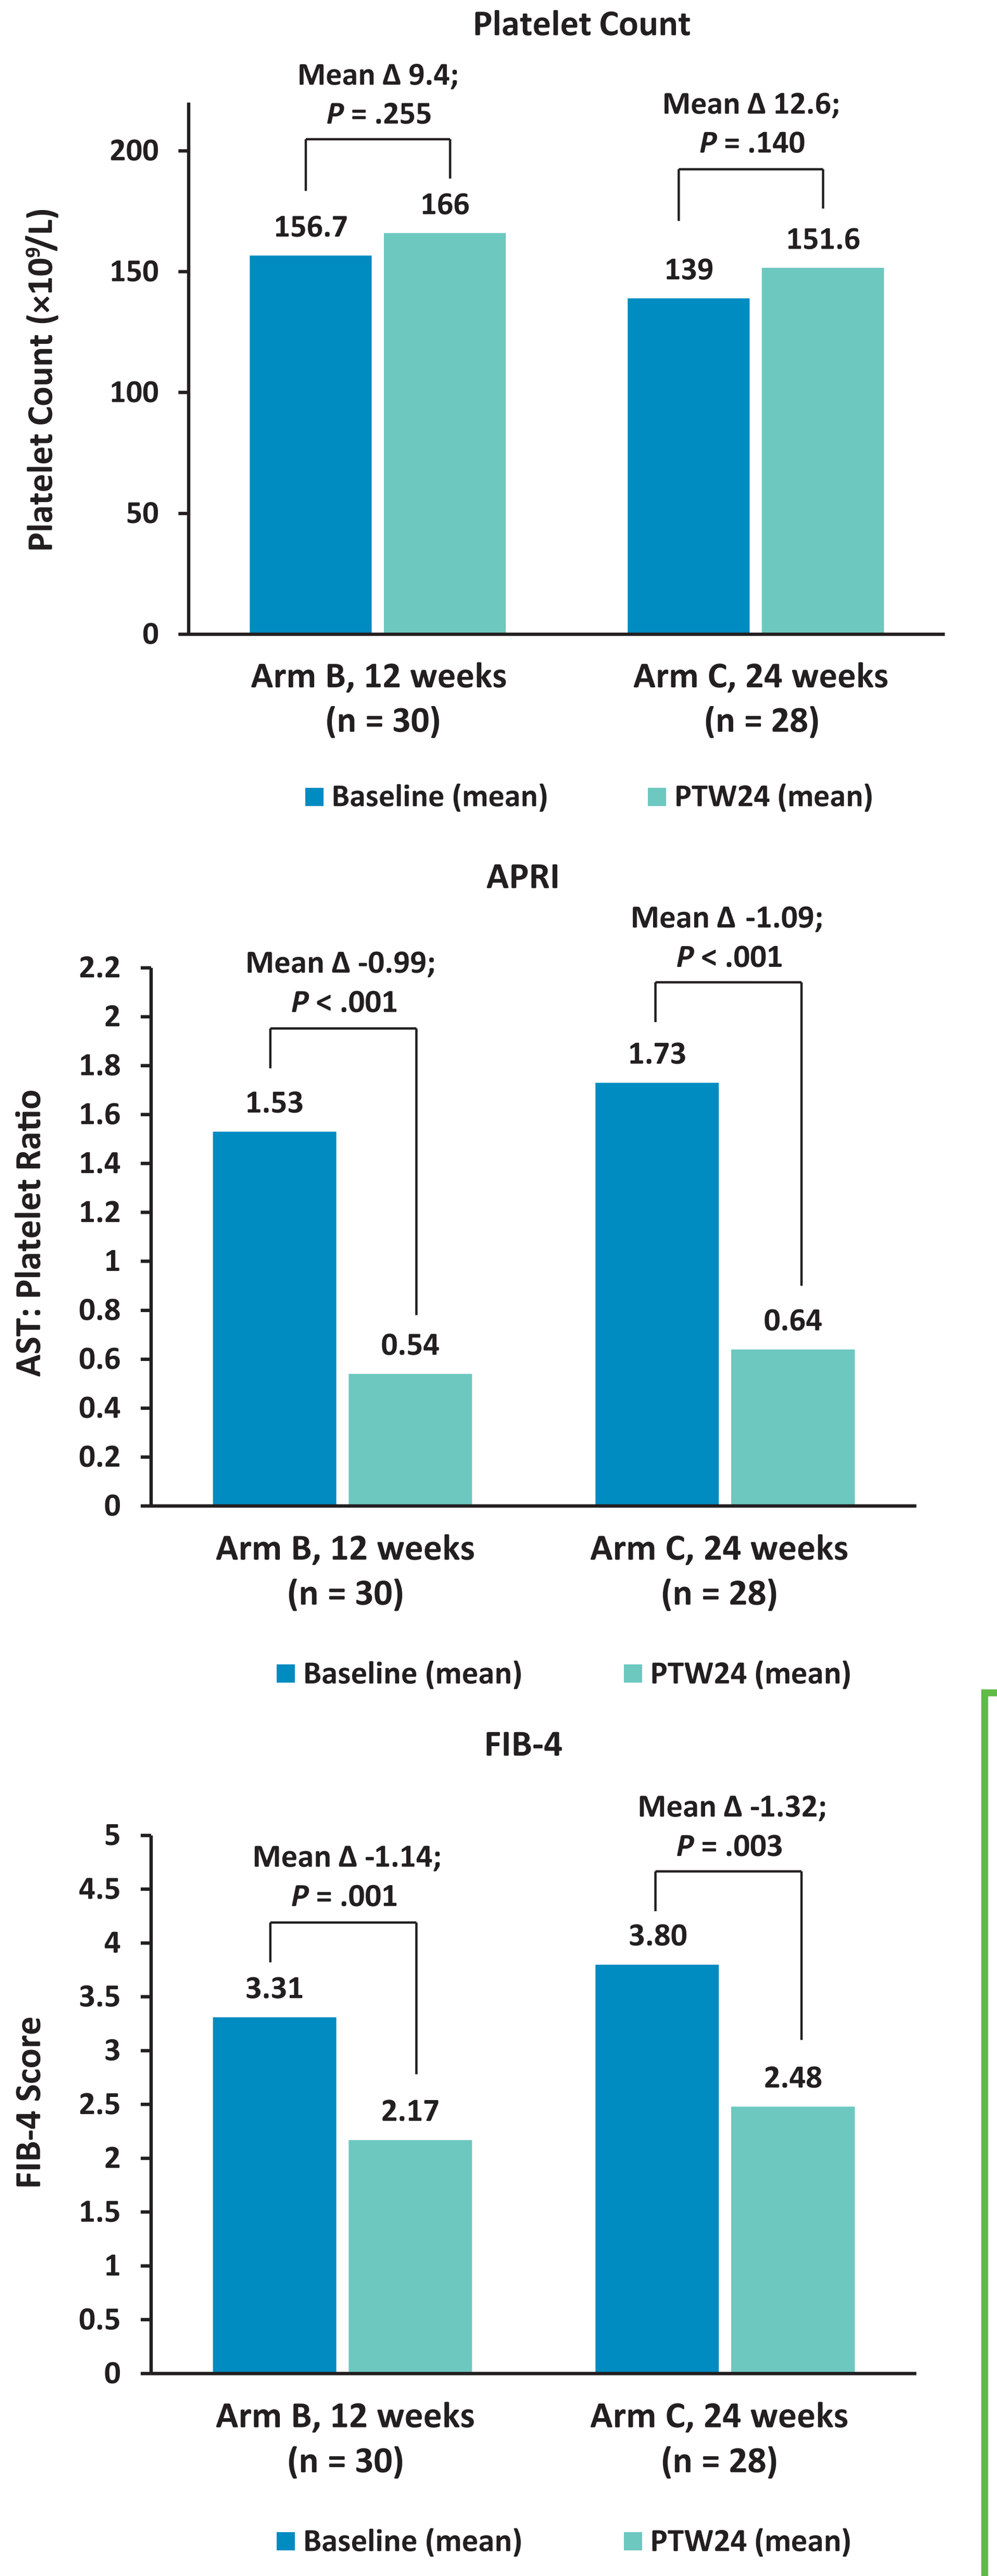
Platelet Count
Platelet Count (×10⁹/L)
Mean Δ 9.4; P = .255
Mean Δ 12.6; P = .140
156.7
166
139
151.6
Arm B, 12 weeks (n = 30)
Arm C, 24 weeks (n = 28)
APRI
AST: Platelet Ratio
Mean Δ -0.99; P < .001
Mean Δ -1.09; P < .001
1.53
0.54
1.73
0.64
Arm B, 12 weeks (n = 30)
Arm C, 24 weeks (n = 28)
FIB-4
FIB-4 Score
Mean Δ -1.14; P = .001
Mean Δ -1.32; P = .003
3.31
2.17
3.80
2.48
Arm B, 12 weeks (n = 30)
Arm C, 24 weeks (n = 28)
Baseline (mean)
PTW24 (mean)

APRI, AST to platelet ratio index; FIB-4, fibrosis-4; PTW, post-treatment week.

SAFETY

- Safety summary for patients with compensated cirrhosis is presented in **Table 2**
- The majority of TEAEs were mild to moderate in severity
- No TEAE led to discontinuation of treatment
- Two patients in Arm C experienced serious TEAEs; neither were considered to be related to study drug
- All patients with an RBV dose modification achieved SVR12
- One patient reported HCC in post-treatment Week 36 and is awaiting liver transplantation:
 - The patient was screened for HCC prior to study by alpha-fetoprotein (AFP) and ultrasound (approximately 3 weeks prior to starting therapy)
 - No baseline or near-baseline CT scan was available to rule out HCC
 - Patient AFP levels were 8.8 ng/ml, 31 ng/ml, and 38ng/ml at baseline, PTW12, and PTW24, respectively
 - Barcelona clinic liver cancer (BCLC) staging at time of HCC diagnosis confirmed early stage-A HCC in this patient
 - The patient achieved SVR

Table 2. Treatment-emergent Adverse Events in Patients With Compensated Cirrhosis

Events, n (%)	Arm B 12 weeks N = 31	Arm C 24 weeks N = 29
Any AE, n (%)	26 (84)	25 (86)
SAE	0	2 (7)*
Death	0	0
AE leading to study drug discontinuation	0	0
RBV dose reduction due to Hb decline	4 (13)	6 (21)

AE, adverse event; Hb, hemoglobin; RBV, ribavirin; SAE, serious adverse event.
*One case of variceal bleeding (post-treatment Day 19) and 1 case of acute cholecystitis requiring cholecystectomy in a patient with previously diagnosed gall stones (treatment Day 137).

CONCLUSIONS

- High SVR rates were achieved with both the 12- and 24-week OBT/PTV/r + RBV regimens in Egyptian HCV GT4-infected patients with compensated cirrhosis
- OBT/PTV/r + RBV treatment resulted in improvements in most biomarkers of liver synthetic function, liver injury, and liver fibrosis
- Liver biomarker improvements were independent of treatment duration
- Treatment was well tolerated with no discontinuations due to AEs

REFERENCES

1. Perz JF, et al. *J Hepatol*. 2006;45(4):529–38.
2. Messina JP, et al. *Hepatology*. 2015;61(1):77–87 (supplementary appendix).
3. van der Meer AJ, et al. *J Hepatol*. 2016;65(Suppl 1):S95–S108.
4. van der Meer AJ, et al. *JAMA*. 2012;308(24):2584–93.
5. Hassanein TI, et al. *J Hepatol*. 2016;64(Suppl 2):S826 (poster presentation).
6. Hezode C, et al. *Lancet*. 2015;385(9986):2502–9.
7. Asselah T, et al. *Lancet Gastroenterol Hepatol*. 2016;1:25–35.
8. Waked I, et al. *Lancet Gastroenterol Hepatol*. 2016;1:36–44.

ACKNOWLEDGEMENTS

AbbVie sponsored the study (NCT02247401), contributed to the design, participated in the collection, analysis, and interpretation of the data, and in the writing, review, and approval of the poster. Medical writing support was provided by Andy Kerr of Medical Expressions, funded by AbbVie.

44587-161467-AGATE-II — AASLD 2016 — AbbVie — proof 4 (rjh) — november 7, 2015
the henderson company 6020 keating avenue, chicago illinois 60646 (847) 979-8051

CREATED @ 100%

885

QUARTZ II-III: Final Efficacy and Safety Results in Patients With HCV Genotype 2 or 3 Infection

Stephen D Shafran[1], David Shaw[2], Mariem Charafeddine[3], Kosh Agarwal[4], Graham R Foster[5], Manal Abunimeh[3], Bo Fu[3], Tami Pilot-Matias[3], Rajvineeth Kumar Pothacamury[3], Jennifer R King[3], Eric Cohen[3], Daniel E Cohen[3], Edward Gane[6]

[1]University of Alberta Hospital, Edmonton, Alberta, Canada; [2]Royal Adelaide Hospital, Adelaide, Australia; [3]AbbVie Inc., North Chicago, Illinois, United States; [4]Institute of Liver Studies, Kings College Hospital, London, United Kingdom; [5]Queen Mary University of London, Barts Health, London, United Kingdom, [6]Liver Unit, Auckland City Hospital, Auckland, New Zealand

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

INTRODUCTION

- Hepatitis C virus (HCV) genotypes (GTs) 2 and 3 account for approximately 40% of HCV infections worldwide and are highly prevalent in Europe, South Asia, and Australasia[1]
- In the direct-acting antiviral (DAA) era, GT3 has emerged as the most difficult to cure, especially in patients with cirrhosis and prior treatment experience[2,3]

2D Regimen + Sofosbuvir

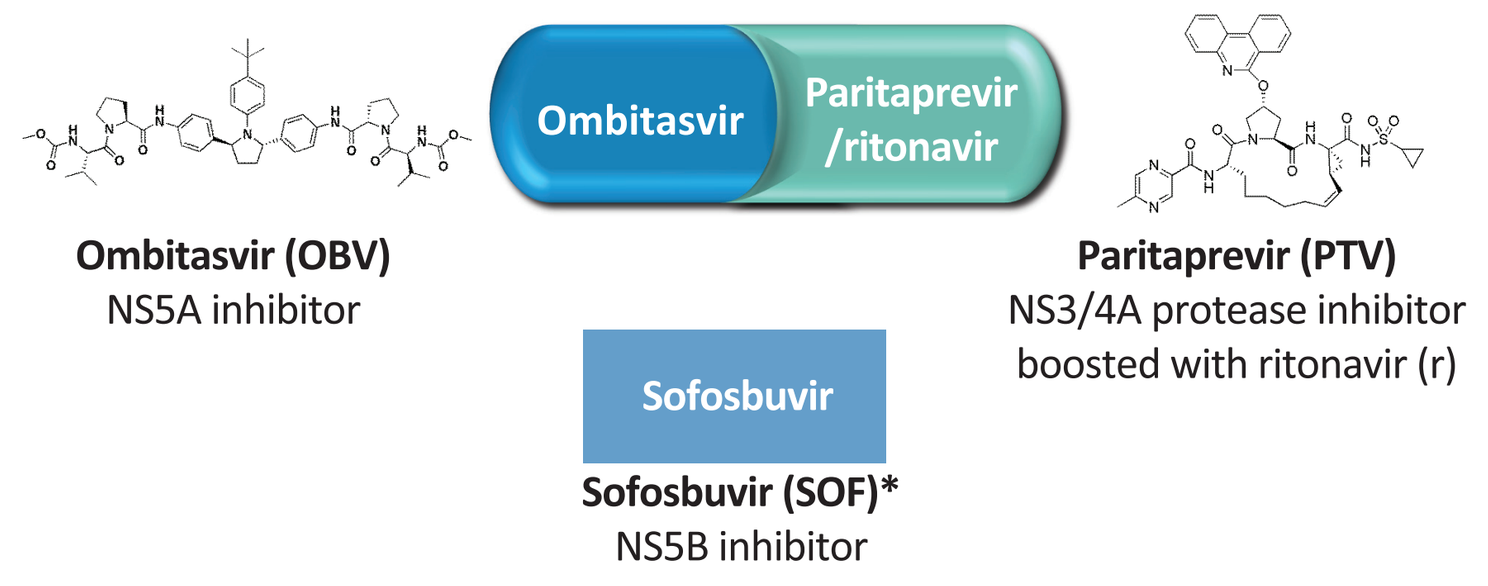
Ombitasvir
Paritaprevir /ritonavir
Ombitasvir (OBV)
NS5A inhibitor
Paritaprevir (PTV)
NS3/4A protease inhibitor boosted with ritonavir (r)
Sofosbuvir
Sofosbuvir (SOF)*
NS5B inhibitor

PTV was identified by AbbVie and Enanta. Ritonavir does not have antiviral activity against HCV.
*SOF was identified by Gilead Sciences.

- The DAAs OBV, PTV, and SOF have shown antiviral activity against GT2 and GT3 but have not been assessed as a combined regimen
 - In a previous study conducted in healthy volunteers, there were no significant drug-drug interactions between DAAs

Stable HCV Replicon Half Maximal Effective Concentration (EC_{50})	GT2a	GT3a
Ombitasvir (OBV): NS5A inhibitor[4]	12 pM	19 pM
Paritaprevir (PTV): NS3/4A protease inhibitor[4]	5.3 nM	19 nM
Sofosbuvir (SOF): NS5B inhibitor[5]	29 nM	81 nM

- Combining multiple DAAs with different mechanisms of action may enhance antiviral activity, which could yield higher response rates and may allow shorter durations of treatment

OBJECTIVE

- QUARTZ II-III (NCT02292719) is an open-label, multicenter, phase 2 study designed to evaluate the safety and efficacy of 6- and 8-week treatment durations of 2D + SOF + RBV in patients with GT2 infection, and 12-week treatment with 2D + SOF ± RBV in patients with GT3 infection, including those with cirrhosis

MATERIALS AND METHODS

STUDY DESIGN

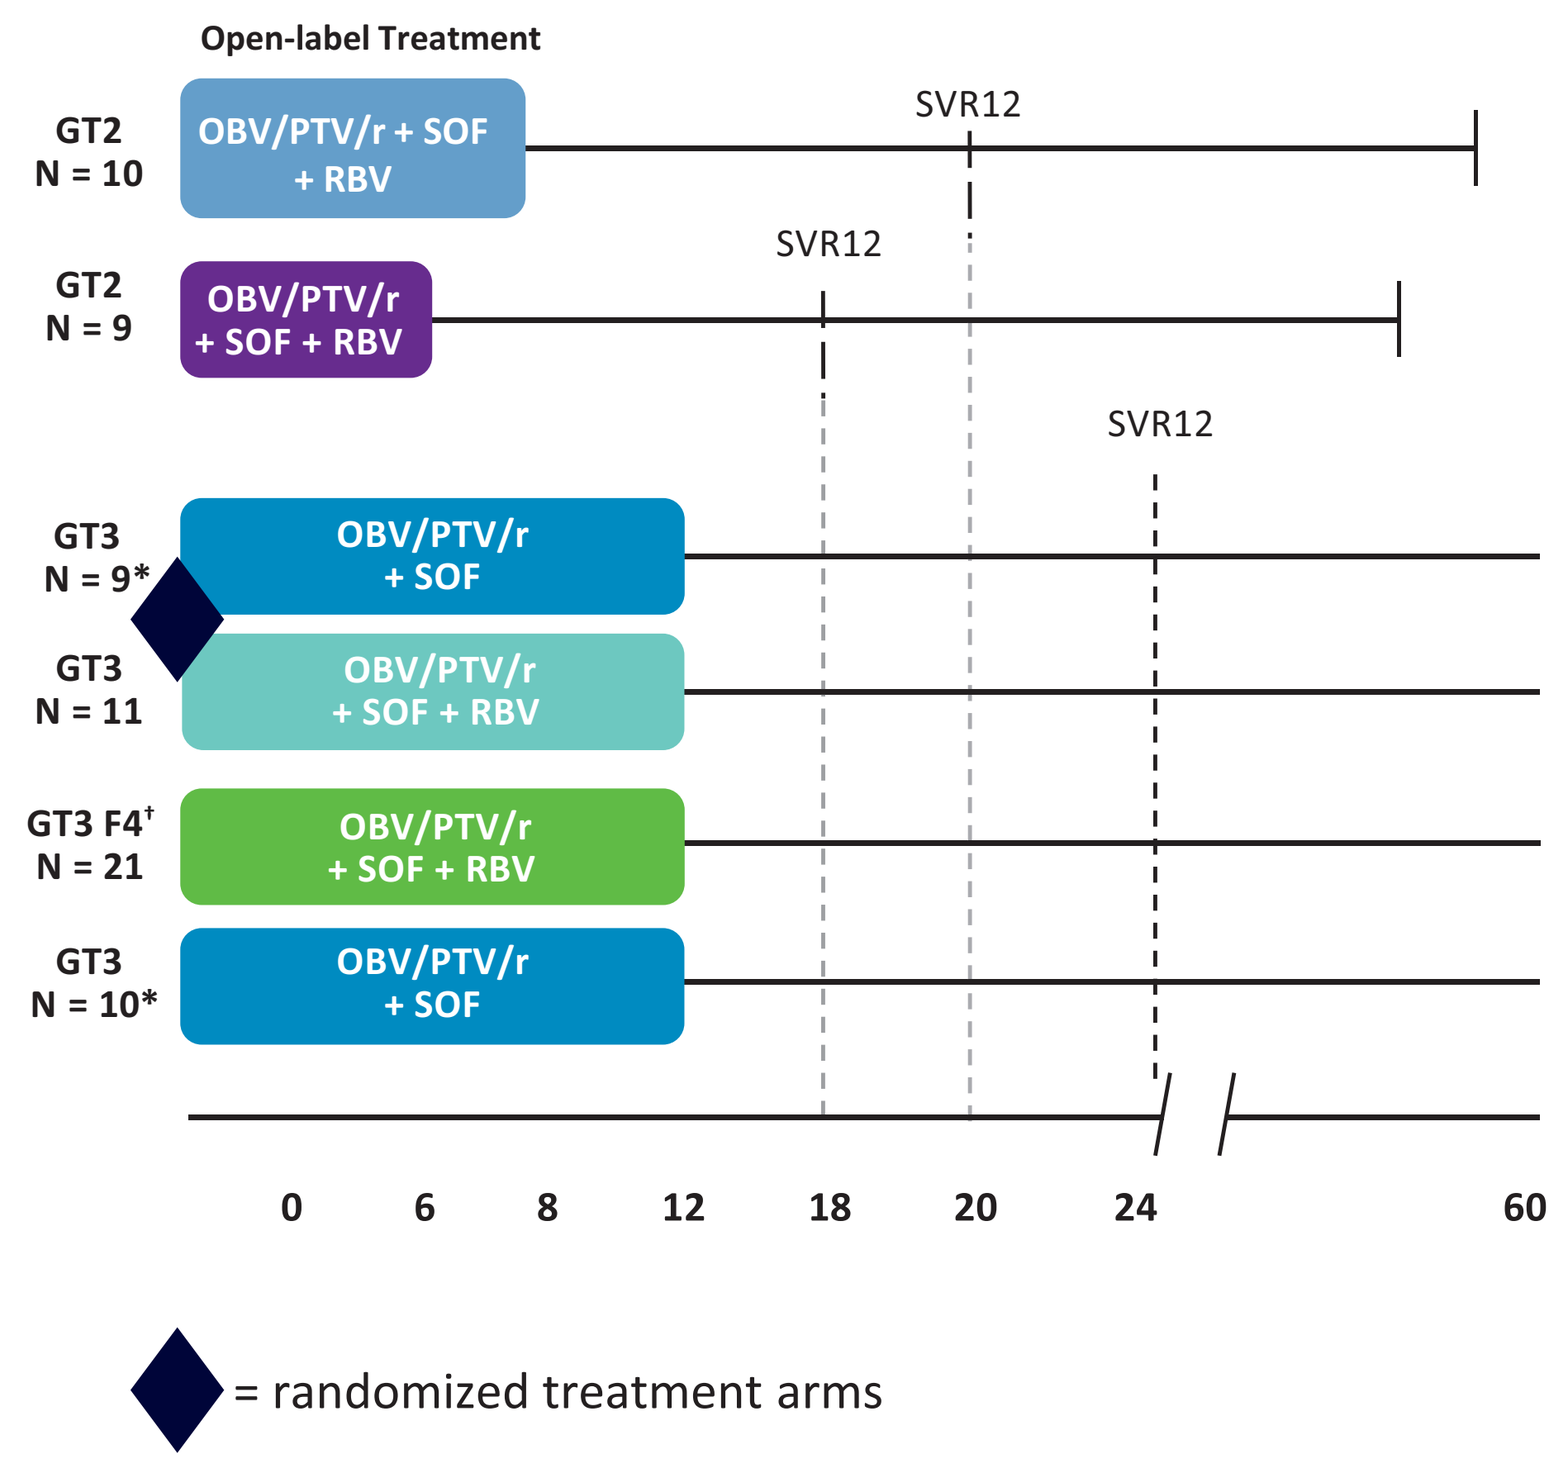
Open-label Treatment
GT2 N = 10
OBV/PTV/r + SOF + RBV
SVR12
GT2 N = 9
OBV/PTV/r + SOF + RBV
SVR12
SVR12
GT3 N = 9*
OBV/PTV/r + SOF
GT3 N = 11
OBV/PTV/r + SOF + RBV
GT3 F4† N = 21
OBV/PTV/r + SOF + RBV
GT3 N = 10*
OBV/PTV/r + SOF
0 6 8 12 18 20 24 60
= randomized treatment arms

SVR12, sustained virologic response at post-treatment Week 12.
*Treatment arms were pooled for analysis.
†All other arms were non-cirrhotic (F0–F3).

MATERIALS AND METHODS (CONTINUED)

KEY ELIGIBILITY CRITERIA

- ≥18 years
- Chronic HCV GT2 or GT3 infection
- HCV RNA >10 000 IU/mL
- For patients with cirrhosis, cirrhosis was documented by liver biopsy, FibroScan score ≥12.5 kPa, or FibroTest score >0.72 and Aminotransferase-to-Platelet Ratio Index >2
- HCV treatment-naïve or treatment-experienced (peginterferon [pegIFN]/RBV, pegIFN/RBV + SOF, or SOF + RBV)
- Patients testing positive for hepatitis B, HIV, or an HCV genotype or subtype other than GT2 or 3 were excluded

ENDPOINTS AND ANALYSES

Primary Efficacy Endpoint	The percentage of patients with SVR12 (HCV RNA <25 IU/mL 12 weeks after the last dose of study drug)
Secondary Efficacy Endpoints	The percentage of patients with on-treatment virologic failure and post-treatment relapse
Safety Assessments	Adverse events (AEs) and laboratory abnormalities were assessed in all patients receiving at least 1 dose of study drug

RESULTS

Table 1. Baseline Demographics and Disease Characteristics

Characteristic	GT2 + RBV 8 Weeks N = 10	GT2 + RBV 6 Weeks N = 9	GT3 No RBV 12 Weeks N = 19	GT3 + RBV 12 Weeks N = 11	GT3 F4 + RBV 12 Weeks N = 21
Male, n (%)	5 (50)	6 (67)	13 (68)	7 (64)	12 (57)
Age, median years (range)	57 (46–66)	62 (50–69)	54 (31–62)	54 (37–62)	56 (38–64)
White race, n (%)	10 (100)	7 (78)	16 (84)	11 (100)	17 (81)
Asian race, n (%)	0	2 (22)	2 (11)	0	1 (5)
BMI, median kg/m^2 (range)	25.4 (21.0–34.3)	24.6 (21.4–32.2)	28.8 (19.9–38.2)	27.5 (24.2–31.2)	27.3 (18.7–38.3)
HCV RNA, median log_{10} IU/mL (range)	6.37 (4.4–7.5)	5.92 (4.2–7.3)	6.79 (3.3–7.4)	6.49 (5.6–7.4)	6.49 (4.1–7.6)
Platelet count, median (range), × 10^9/L	213 (122–281)	229 (131–320)	197 (117–269)	200 (139–365)	128 (56–199)
IL28B non-CC genotype, n (%)	7 (70)	6 (67)	7 (37)	7 (64)	12 (57)
Treatment-experienced, n (%)	2 (20)	2 (22)	9 (47)	5 (45)	12 (57)
Fibrosis stage, n (%)					
F0–1	10 (100)	7 (78)	10 (53)	9 (82)	0
F2	0	0	4 (21)	0	0
F3	0	1 (11)	3 (16)	2 (18)	0
F4	0	0	0	0	21 (100)
Missing	0	1 (11)	2 (11)	0	0
Genotype/subgenotype					
2a	4 (40)	5 (56)			
2b	6 (60)	4 (44)			
3a			19 (100)	11 (100)	21 (100)

RESULTS (CONTINUED)

Figure 1. Efficacy

A. Rates of Virologic Response in Patients With GT3 Infection

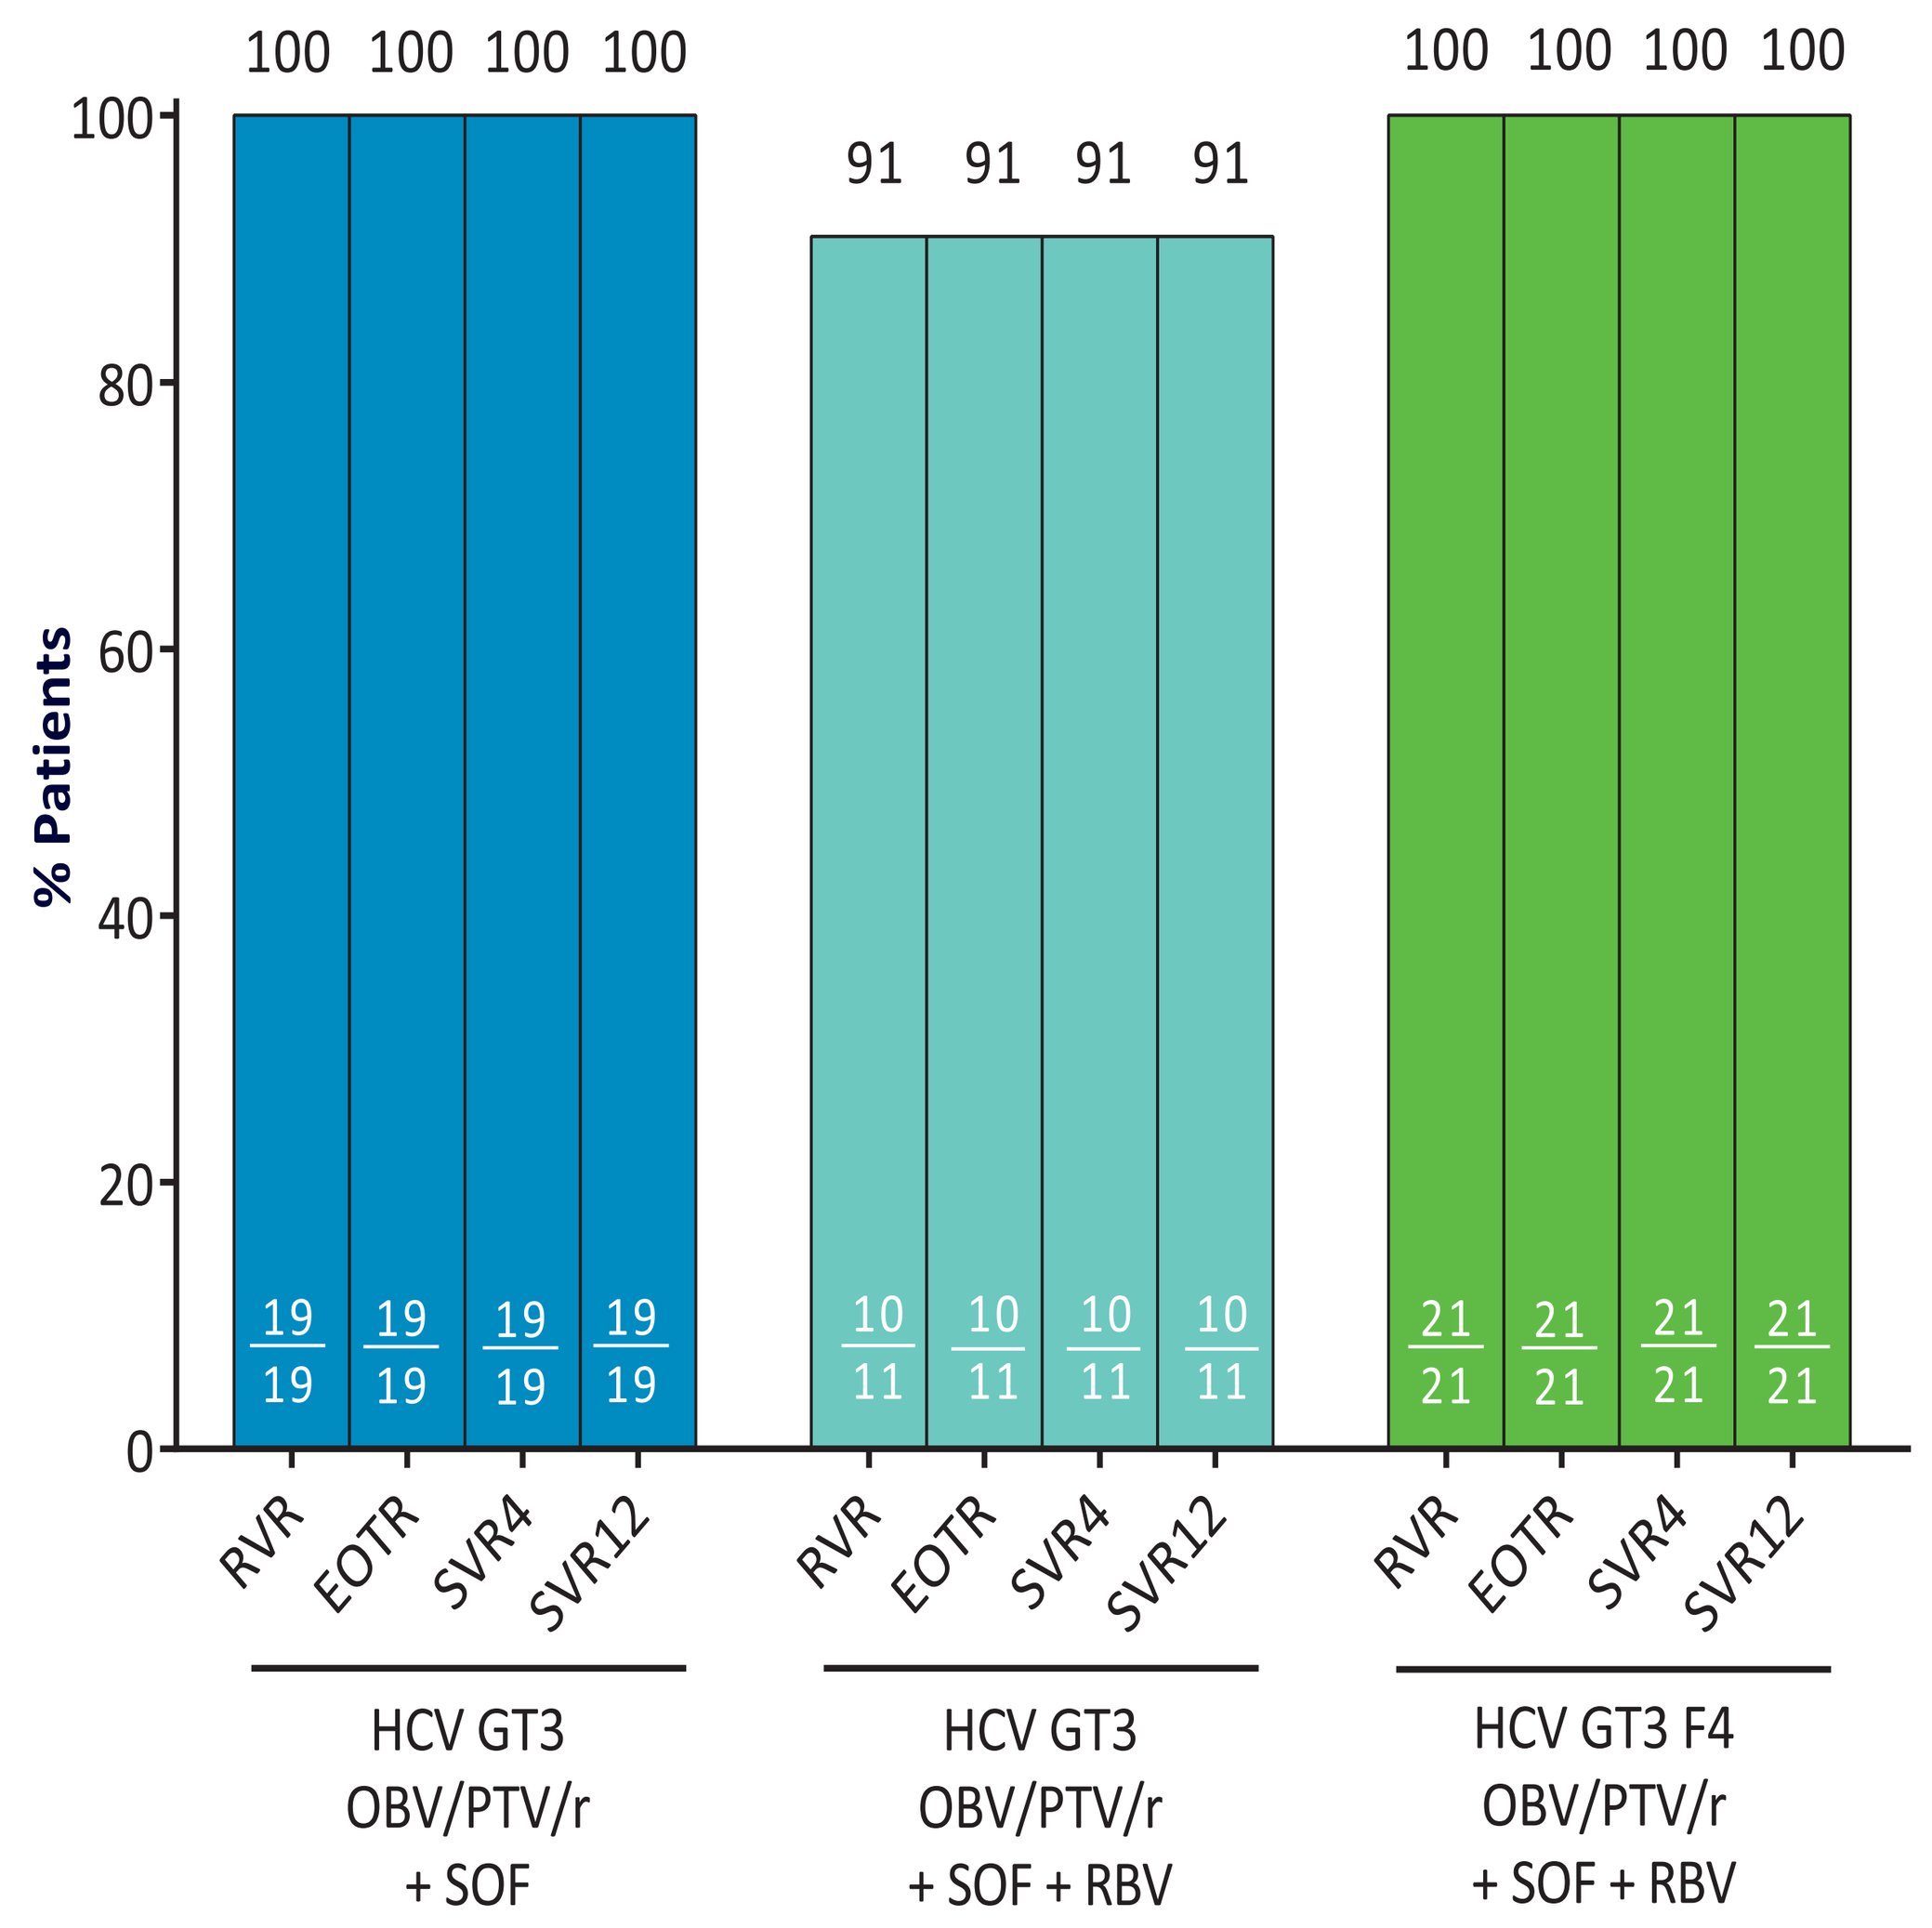
% Patients
0
20
40
60
80
100
100 100 100 100
19/19 19/19 19/19 19/19
91 91 91 91
10/11 10/11 10/11 10/11
100 100 100 100
21/21 21/21 21/21 21/21
RVR EOTR SVR4 SVR12
HCV GT3 OBV/PTV/r + SOF
HCV GT3 OBV/PTV/r + SOF + RBV
HCV GT3 F4 OBV/PTV/r + SOF + RBV

- There were no virologic failures in GT3-infected patients who completed treatment
 - One GT3-infected patient without cirrhosis treated with 12-week 2D + SOF + RBV discontinued treatment on Day 8 due to an adverse event of influenza-like illness deemed unrelated to DAAs; this patient did not achieve SVR12
 - Addition of RBV did not improve treatment response among patients without cirrhosis

B. Rates of Sustained Virologic Response in Patients With GT2 Infection

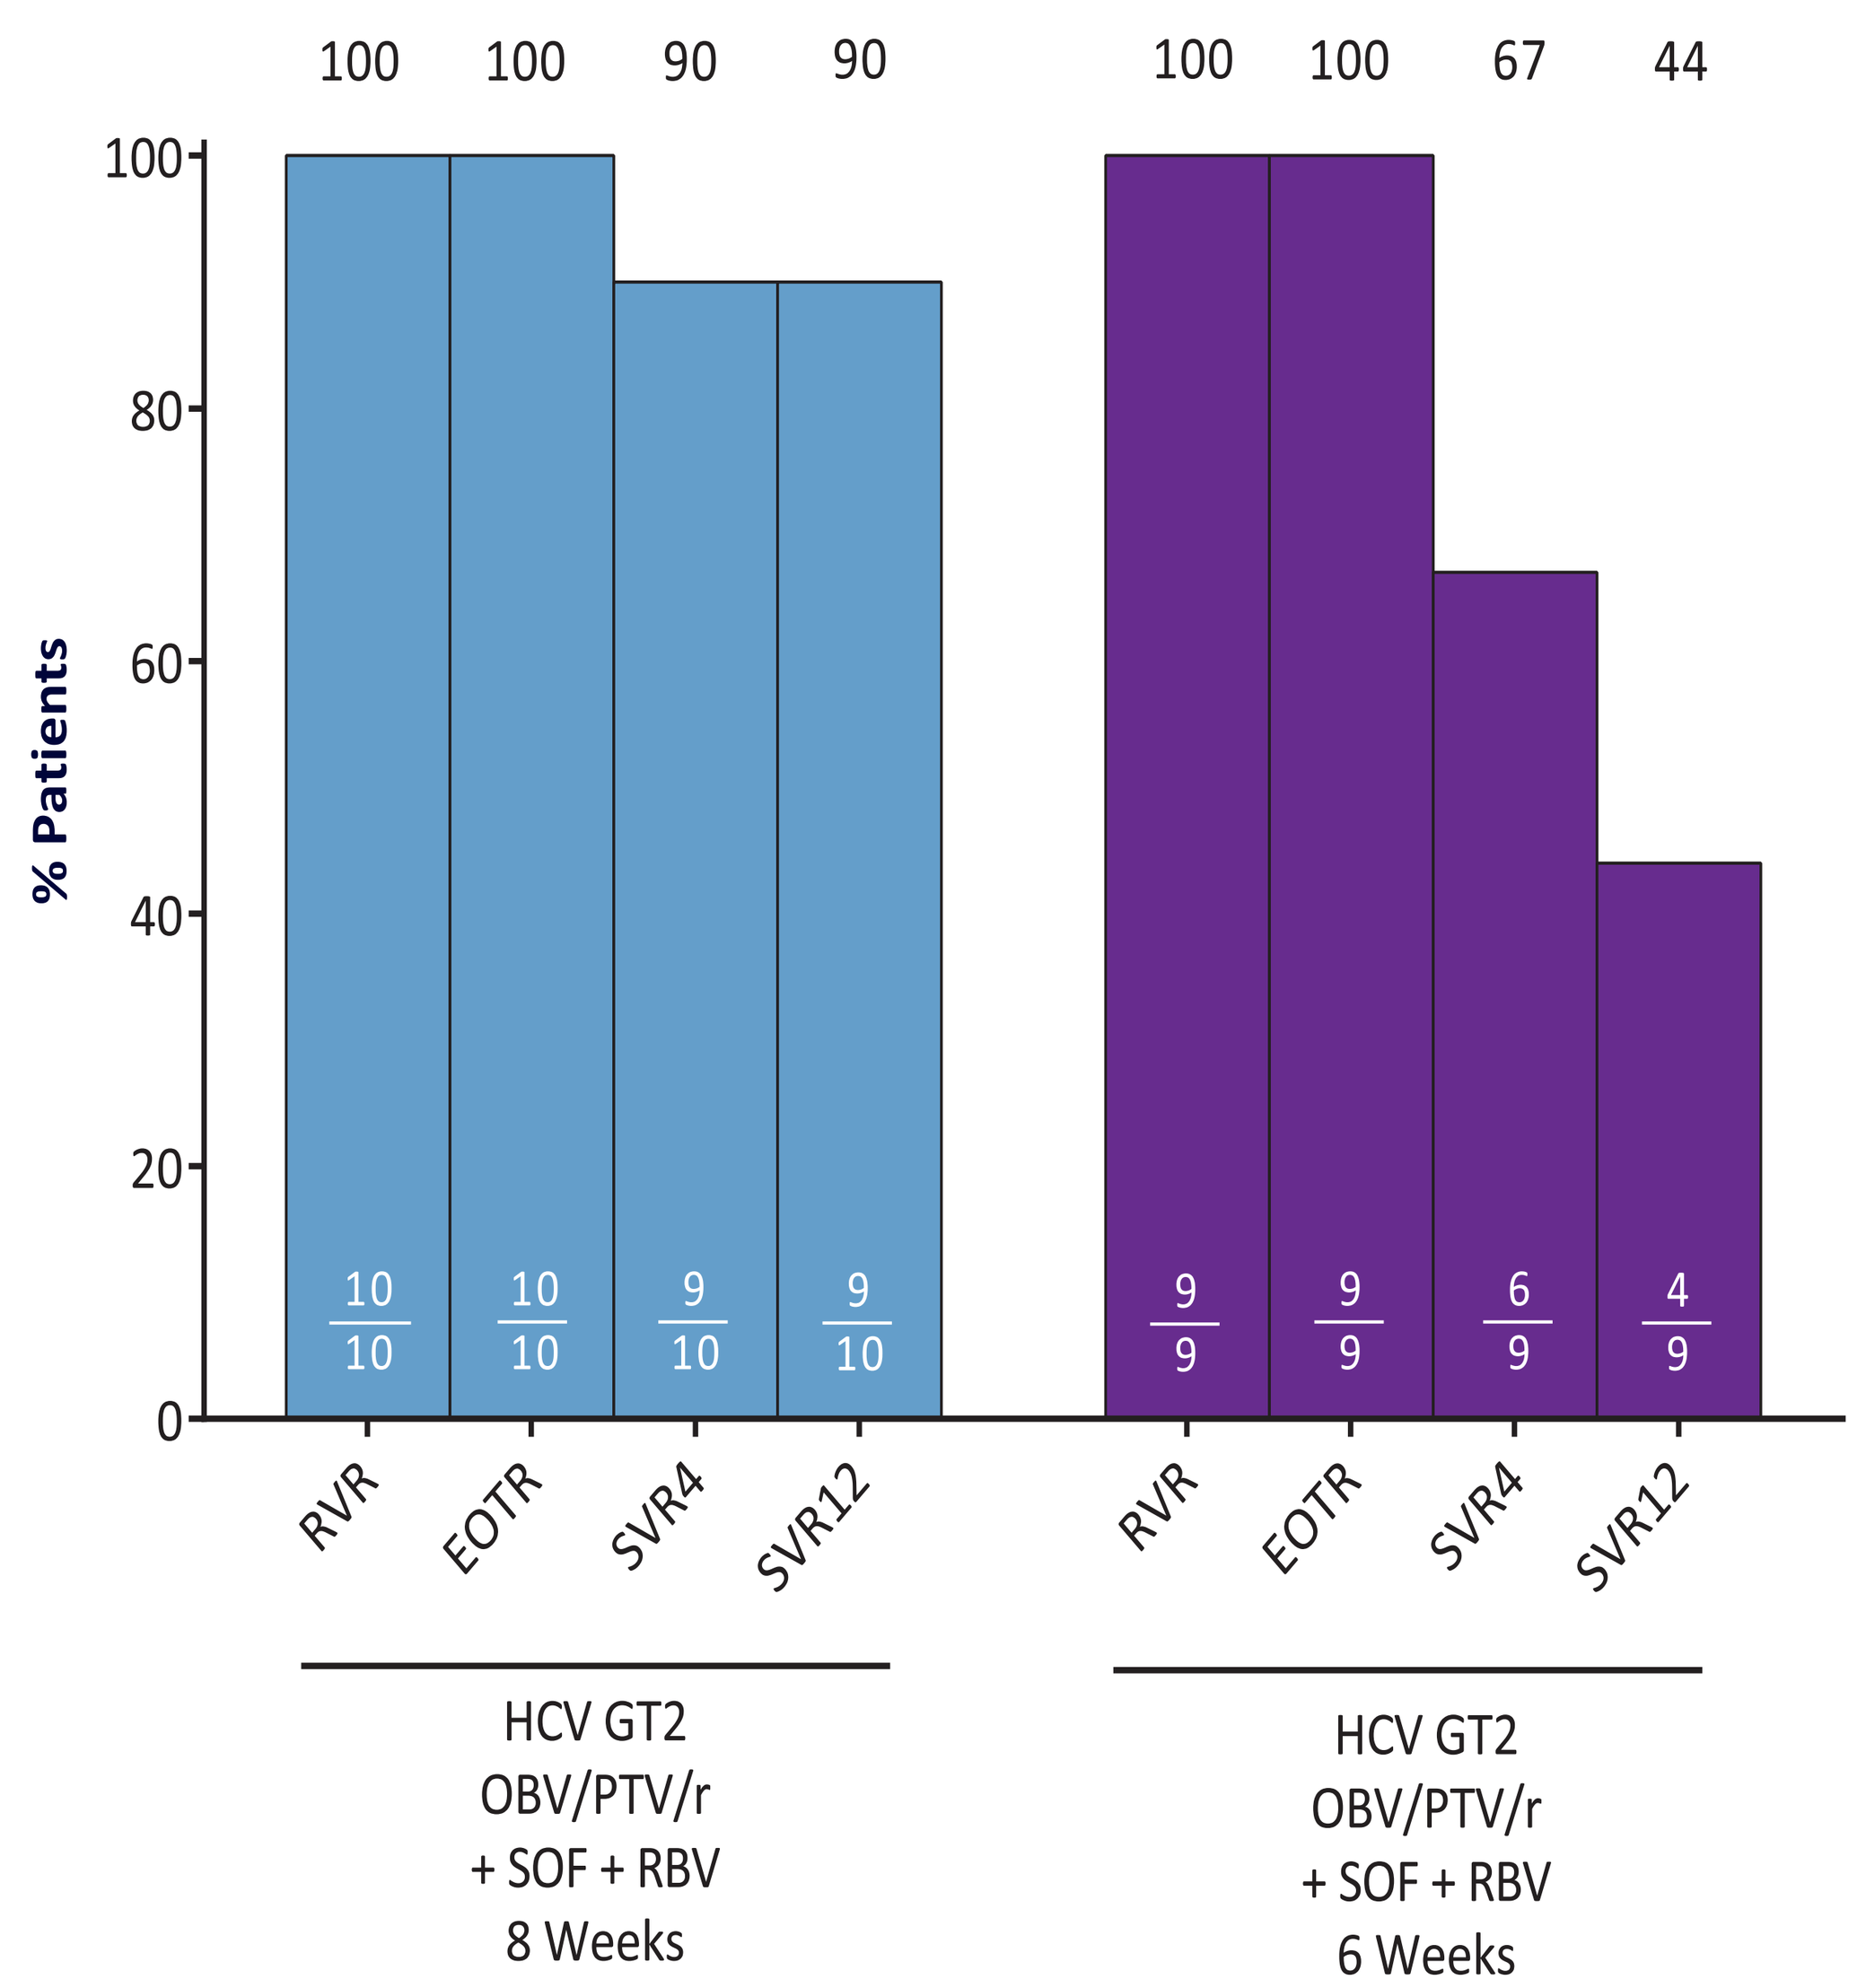
% Patients
0
20
40
60
80
100
100 100 90 90
10/10 10/10 9/10 9/10
100 100 67 44
9/9 9/9 6/9 4/9
RVR EOTR SVR4 SVR12
HCV GT2 OBV/PTV/r + SOF + RBV 8 Weeks
HCV GT2 OBV/PTV/r + SOF + RBV 6 Weeks

- With 8 weeks of treatment, 1/10 patients with GT2 infection relapsed, but with the shorter 6-week treatment duration, 5/9 patients relapsed
 - None of the 6 patients had resistance-associated polymorphisms at baseline in NS3, NS5A, or NS5B, nor did any have treatment-emergent substitutions at the time of failure
- Exposures of the DAAs and ritonvair were comparable between non-cirrhotic patients who achieved SVR12 and those who failed treatment with 2D + SOF

Table 3. Safety and Laboratory Abnormalities

	GT2 + RBV 8 Weeks N = 10	GT2 + RBV 6 Weeks N = 9	GT3 No RBV 12 Weeks N = 19	GT3 + RBV 12 Weeks N = 11	GT3 F4 + RBV 12 Weeks N = 21
Any AE	10 (100)	9 (100)	16 (84)	10 (91)	20 (95)
Serious AE	1 (10)*	0	0	0	2 (10)†
AE leading to RBV dose modification	2 (20)	0	N/A	2 (18)	3 (14)
AE leading to D/C	0	0	0	1 (9)‡	1 (5)§
AEs occurring in ≥10% of total patients					
Fatigue	3 (30)	6 (67)	7 (37)	3 (27)	10 (48)
Headache	6 (60)	3 (33)	3 (16)	2 (18)	9 (43)
Pruritus	0	2 (22)	2 (11)	2 (18)	7 (33)
Nausea	3 (30)	0	3 (16)	1 (9)	6 (29)
Diarrhea	3 (30)	2 (22)	3 (16)	1 (9)	2 (10)
Dizziness	1 (10)	2 (22)	3 (16)	1 (9)	3 (14)
URTI	4 (40)	0	1 (5)	0	5 (24)
Insomnia	2 (20)	0	2 (11)	0	5 (24)
Laboratory abnormalities					
Hemoglobin					
Grade 2 (<10–8 g/dL)	1 (10)	0	0	1 (9)	1 (5)
Grade ≥3 (<8 g/dL)	0	0	0	0	0
Bilirubin Grade ≥3 (>3 × ULN)	0	0	0	0	1 (5)
ALT					
Grade ≥3 (>5 × ULN)	1 (10)	0	0	0	2 (10)
AST					
Grade ≥3 (>5 × ULN)	0	0	0	0	1 (5)

*Pneumonia on Day 26 deemed unrelated to DAAs.
†One patient experienced respiratory tract infection on Day 33; a second patient experienced anemia on Day 28; both were deemed unrelated to DAAs.
‡D/C'd on Day 8 due to a non-serious AE of influenza-like symptoms deemed unrelated to DAAs; this patient did not achieve SVR12.
§D/C'd on Day 78 due to a non-serious AE of vertigo deemed possibly related to DAAs.
GT, genotype; AE, adverse event; RBV, ribavirin; N/A, not applicable; D/C, discontinuation; URTI, upper respiratory tract infection; ULN, upper limit of normal; ALT, alanine aminotransferase; AST, aspartate aminotransferase.

- No serious AEs were related to DAAs
- There were no Grade 4 or higher laboratory abnormalities

CONCLUSIONS

- High SVR rates were seen among GT3-infected patients without cirrhosis treated with 2D + SOF with or without RBV for 12 weeks
 - 100% of GT3-infected patients with compensated cirrhosis achieved SVR12 following 12-week 2D + SOF + RBV
- An 8-week regimen of 2D + SOF + RBV achieved an SVR rate of 90% in GT2-infected non-cirrhotic patients
 - Shortening treatment duration to 6 weeks resulted in a high rate of relapse
- 2D + SOF ± RBV had a favorable safety profile consistent with previous observed regimens of 2D or SOF ± RBV
- These results suggest that 2D + SOF ± RBV may be a useful treatment option for patients with GT3 infection with or without cirrhosis

REFERENCES

1. Messina JP, Humphreys I, Flaxman A, et al. Global distribution and prevalence of hepatitis C virus genotypes. *Hepatology*. 2015;61(1):77–87.
2. Goossens N, Negro F. Is genotype 3 of the hepatitis C virus the new villain? *Hepatology.* 2014;59(6):2403–12.
3. Ampuero J, Romero-Gomez M, Reddy KR. Review article: HCV genotype 3–the new treatment challenge. *Alimentary Pharmacology & Therapeutics.* 2014;39(7):686–98.
4. VIEKIRA PAK (ombitasvir, paritaprevir, and ritonavir tablets; dasabuvir tablets) [package insert] October 2015. www.rxabbvie.com/pdf/viekirapak_pi.pdf (accessed).
5. Gilead Sciences. SOVALDI (sofosbuvir) tablets, for oral use [package insert] August 2015. (accessed).

ACKNOWLEDGEMENTS

The authors would like to express their gratitude to the patients and their families who participated in this study. Medical writing support was provided by Zoë Hunter, PhD, of AbbVie.

DISCLOSURES

The design, study conduct, analysis, and financial support of the QUARTZ (NCT02292719) study were provided by AbbVie. AbbVie participated in the interpretation of data, review, and approval of the content. All authors had access to all relevant data and participated in writing, review, and approval of this presentation.

SD Shafran: Research Funding: AbbVie, BMS, Gilead, Janssen, Merck; Advisory Boards/Speaker: BMS, Gilead, Merck, Pfizer.

D Shaw: Nothing to disclose.

K Agarwal: Speaker Bureau/Consultancy: Achillion, Arbutus, BMS, Gilead, GSK, Intercept, Janssen, Merck, Novartis, Roche; Research Support: AbbVie, BMS, Gilead, Roche.

GR Foster: Grant/Research Support: AbbVie, BMS, Merck, Roche/Genentech, Gilead, Novartis, Janssen; Consultant/Advisor: AbbVie, Vertex, BMS, Merck, Roche/Genentech, Gilead, GSK, Janssen, Virco, Novartis.

E Gane: Advisor: AbbVie, Gilead, Achillion, Idenix, Novartis, Roche, Merck, Janssen.

M Charafeddine, M Abunimeh, B Fu, T Pilot-Matias, RK Pothacamury, JR King, DE Cohen, E Cohen: AbbVie employees and may own AbbVie stock or options.

Combined Resistance, Demographic, and Phylogenetic Analyses of HCV Genotype 4-Infected Patients Treated With Ombitasvir/Paritaprevir/r ± Ribavirin in the PEARL-I and AGATE-I Studies

866

Gretja Schnell, Rakesh Tripathi, Jill Beyer, Thomas Reisch, Coleen Hall, Yao Yu, Niloufar Mobashery, Tami Pilot-Matias, Christine Collins

[1]Research & Development, AbbVie Inc., North Chicago, Illinois, United States

Presented at the 67th Annual Meeting of the American Association for the Study of Liver Diseases, November 11–15, 2016, Boston, Massachusetts

BACKGROUND

- The HCV regimen containing ombitasvir (NS5A inhibitor) and paritaprevir (NS3/4A protease inhibitor identified by AbbVie and Enanta) with ritonavir [OBV/PTV/r] plus ribavirin (RBV) is highly efficacious for the treatment of chronic HCV genotype (GT) 4 infection
- HCV GT4 comprises approximately 13% of infections worldwide[1] and includes 17 confirmed subtypes.[2] GT4 is most prevalent in Europe, the Middle East, North Africa, and central and eastern sub-Saharan Africa.[3,4] In Egypt, GT4 accounts for approximately 90% of HCV infections with subtype 4a predominating[3]
- AbbVie has a large dataset of NS5A sequences containing diverse HCV GT4 subtypes isolated from patients treated with OBV/PTV/r ± RBV in the PEARL-I and AGATE-I studies **(Figure 1)**

OBJECTIVE

- Utilize our HCV GT4 NS5A sequence database to assess genetic diversity by geographic region, analyze patient demographics and baseline sequence variability across GT4 subtypes, and report the development of viral resistance in virologic failure (VF) patients treated with OBV/PTV/r ± RBV

METHODS

Figure 1. PEARL-I and AGATE-I Study Design and SVR12 Rates for HCV GT4-infected Patients

A. PEARL-I, HCV GT4-infected Patients Without Cirrhosis

Regimen, Duration	N	SVR12[5]
OBV/PTV/r 12 weeks, TN	44	90.9%
OBV/PTV/r + RBV 12 weeks, TN	42	100%
OBV/PTV/r + RBV 12 weeks, TE	49	100%

B. AGATE-I, HCV GT4-infected Patients With Compensated Cirrhosis

Regimen, Duration	N	SVR12[6]
OBV/PTV/r + RBV 12 weeks, TN or TE	59	97.0%
OBV/PTV/r + RBV 16 weeks, TN or TE	61	98.0%

TN, treatment-naïve; TE, treatment-experienced; OBV/PTV/r (25/150/100 mg QD). The Versant HCV Genotype Inno-LiPA Assay v2.0 was used to determine genotype for enrollment of patients in each study.

- Phylogenetic analyses were conducted on sequences from the baseline samples of HCV GT4-infected patients from PEARL-I and AGATE-I in order to assign subtype **(Figure 2)**

Figure 2. Methodology for HCV GT4 Subtype Assignment in the PEARL-I and AGATE-I Studies

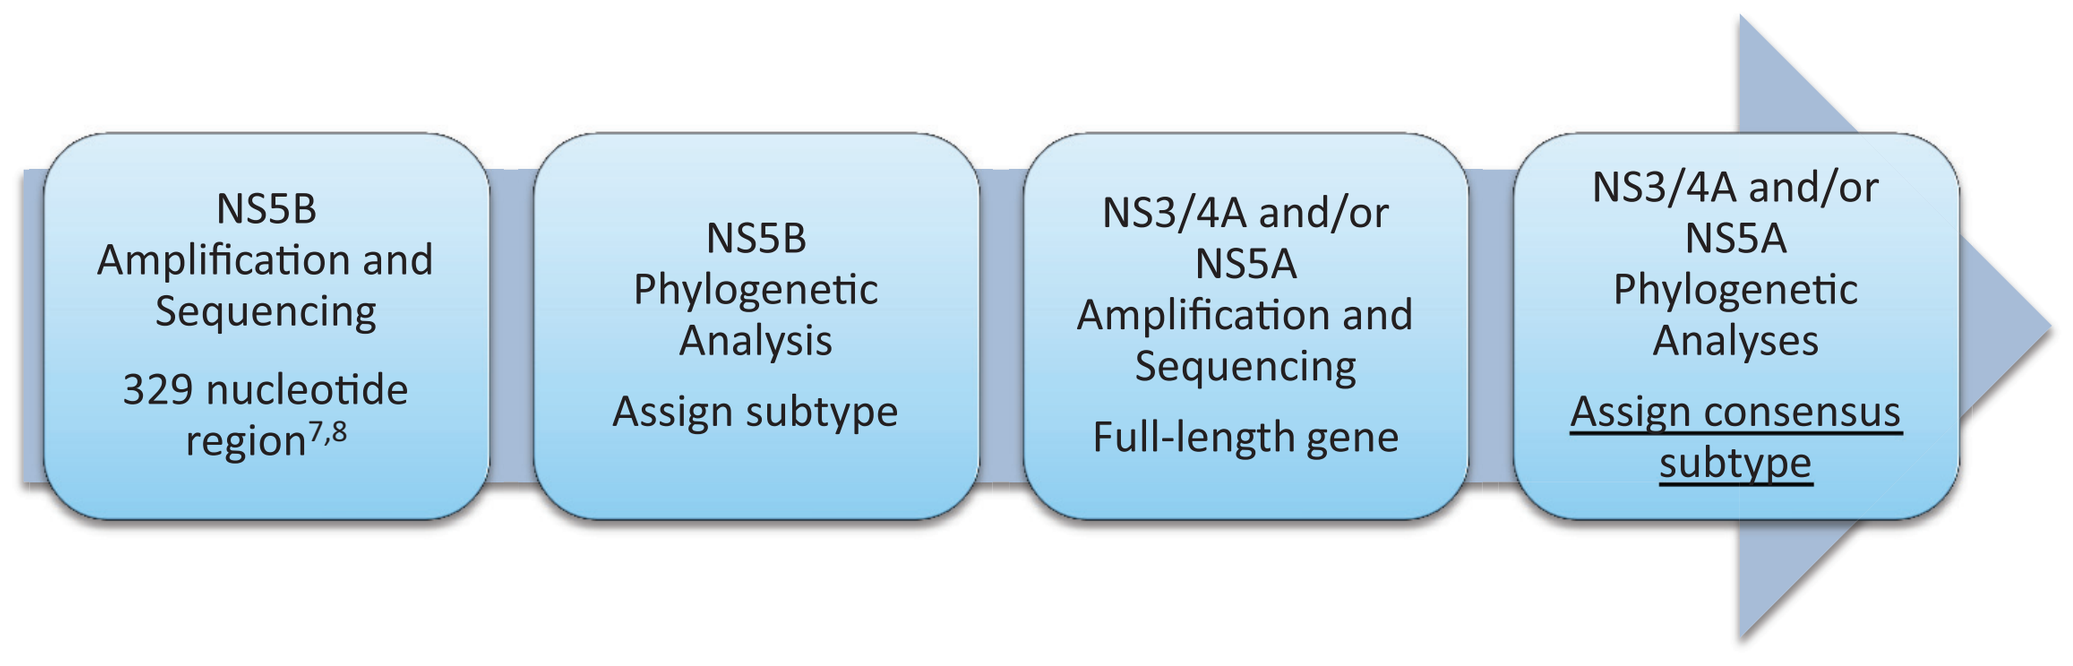
NS5B Amplification and Sequencing
329 nucleotide region[7,8]
NS5B Phylogenetic Analysis
Assign subtype
NS3/4A and/or NS5A Amplification and Sequencing
Full-length gene
NS3/4A and/or NS5A Phylogenetic Analyses
Assign consensus subtype

Nucleotide alignments and phylogenetic trees were generated using the Geneious and MEGA software packages.

- The full-length NS5A gene was sequenced by population or next-generation sequencing (NGS) from the baseline samples of 132/135 non-cirrhotic (PEARL-I) or 118/120 cirrhotic (AGATE-1) GT4-infected patients
- The full-length NS3/4A gene was sequenced by population sequencing from baseline samples of GT4-infected patients in PEARL-I
- NS5A sequences were included in a phylogenetic analysis to assess genetic relationships among and within GT4 subtypes by country
- Prevalence of baseline polymorphisms in NS5A was analyzed using population and NGS data with a detection threshold of 15%. Baseline polymorphisms and treatment-emergent substitutions in NS3 and NS5A were analyzed for patients who experienced VF
- The following NS3 and NS5A amino acid positions were analyzed for GT4
 - NS3 amino acid positions 56, 155, 156, and 168
 - NS5A amino acid positions 24, 28, 29, 30, 31, 32, 58, 92, and 93

RESULTS

HCV GT4 SUBTYPE PREVALENCE BY GEOGRAPHIC REGION

Table 1. HCV GT4 Subtype Prevalence by Country of Enrollment (PEARL-I and AGATE-I)

	Country of Enrollment										
Subtype	Austria	Belgium	France	Germany	Greece	Italy	Poland	Spain	Canada	United States	Total
4a	12	2	39	2	9	1		16	11	22	114
4b			3								3
4c			4							1	5
4d	1	6	21		2	18	16	32	2	1	99
4e			1								1
4f			8	1				1			10
4g/4k			1								1
4h		1									1
4k		2	2								4
4l		1			1						2
4n			1		1						2
4o			1						1		2
4p		1	1								2
4q		1									1
4r		1									1
4t		1									1
4[a]			1								1
Not available		1		1				2		1	5

[a]GT4 subtype could not be determined by phylogenetic analysis.

Table 2. HCV GT4 Subtype Prevalence by Geographic Region of Origin in Study AGATE-I

	Subtype by geographic region of origin				
Subtype	Egypt and Middle East	Africa	Europe	North America	Total
4a	36	2	21	5	64
4c		4			4
4d		1	29	1	31
4e		1			1
4f		3			3
4h		1			1
4k		4			4
4l	1	1			2
4n	2				2
4o	1				1
4p		1	1		2
4q		1			1
4r		1			1
4t		1			1
Not available		1	1		2

- Sixteen GT4 subtypes were identified in the combined NS5A sequence dataset **(Table 1)**
- The majority of patients enrolled in North America were subtype 4a, patients enrolled in Europe were predominantly 4a or 4d, and patients infected with non-4a/4d subtypes were mostly enrolled in Belgium and France **(Table 1)**
- In study AGATE-1, in addition to country of enrollment, the patient-reported geographic country of origin was also collected and revealed that the geographic region of origin correlated with the known distribution of GT4 subtypes worldwide **(Table 2)**

BASELINE DEMOGRAPHICS BY GT4 SUBTYPE

Table 3. Patient Baseline Demographic Characteristics by GT4 Subtype in PEARL-I and AGATE-I

	Number (% of subtype) of patients by GT4 subtype										
	PEARL-I, non-cirrhotics			AGATE-I, cirrhotics			Combined dataset				
Baseline Characteristic	4a	4d	non-4a/4d	4a	4d	non-4a/4d	4a	4d	non-4a/4d	Total	P-value[a]
Patients	50	68	14	64	31	23	114	99	37	250	
Birth Cohort											<.001
Pre 1950	3 (6)	1 (2)	2 (14)	12 (19)	3 (10)	9 (39)	15 (13)	4 (4)	11 (30)	30	
1950–1959	18 (36)	15 (22)	8 (57)	20 (31)	8 (26)	9 (39)	38 (33)	23 (23)	17 (46)	78	
1960–1969	19 (38)	33 (49)	2 (14)	29 (45)	19 (61)	4 (17)	48 (42)	52 (53)	6 (16)	106	
1970 and after	10 (20)	19 (28)	2 (14)	3 (5)	1 (3)	1 (4)	13 (11)	20 (20)	3 (8)	36	
Race											<.001
White	46 (92)	68 (100)	4 (29)	58 (91)	30 (97)	6 (26)	104 (91)	98 (99)	10 (27)	212	
Black	2 (4)	0	10 (71)	2 (3)	0	17 (74)	4 (4)	0	27 (73)	31	
Other	2 (4)	0	0	4 (6)	1 (3)	0	6 (5)	1 (1)	0	7	

[a]P-value calculated using the combined dataset from the comparison of proportions across subtypes (4a, 4d, and non-4a/4d) using Fisher's exact test.

HCV GT4 PHYLOGENETIC ANALYSIS BY GEOGRAPHIC REGION

Figure 3. Phylogenetic Analysis of HCV GT4 NS5A Sequences Revealed Clustering by Country of Origin in Subtype 4a

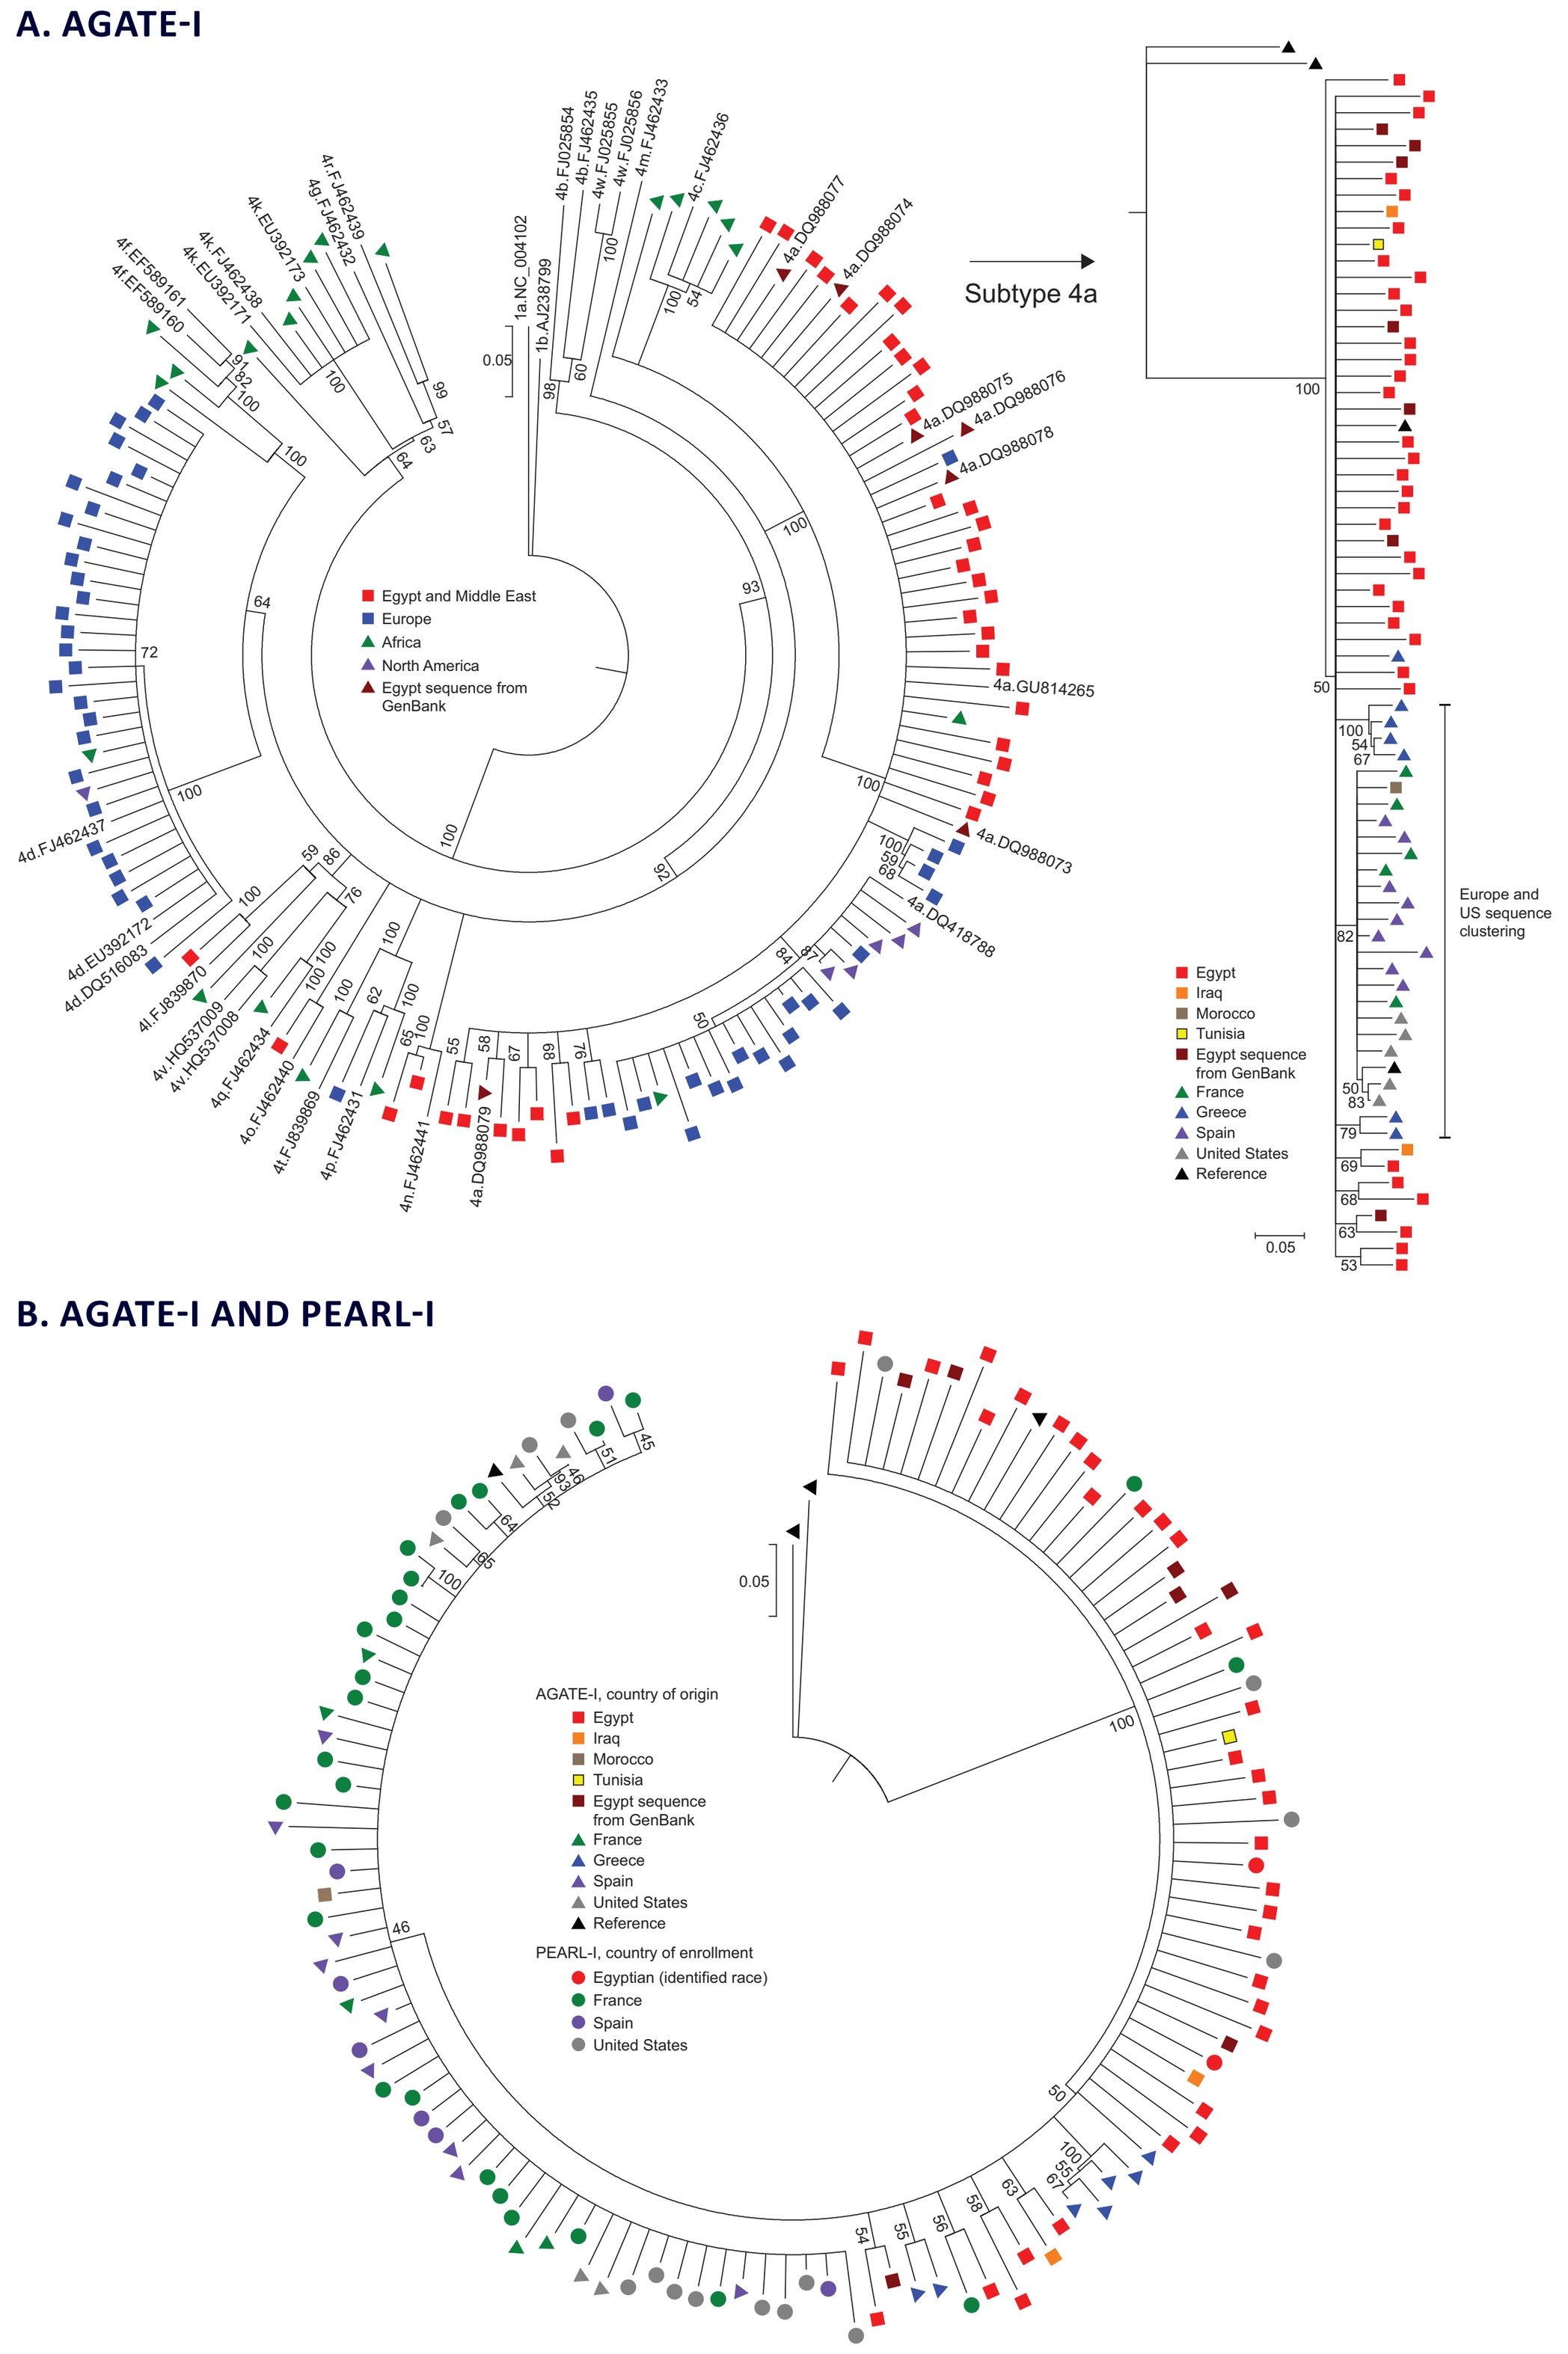
A. AGATE-I
Subtype 4a
Egypt and Middle East
Europe
Africa
North America
Egypt sequence from GenBank
Egypt
Iraq
Morocco
Tunisia
Egypt sequence from GenBank
France
Greece
Spain
United States
Reference
Europe and US sequence clustering
B. AGATE-I AND PEARL-I
AGATE-I, country of origin
Egypt
Iraq
Morocco
Tunisia
Egypt sequence from GenBank
France
Greece
Spain
United States
Reference
PEARL-I, country of enrollment
Egyptian (identified race)
France
Spain
United States

Neighbor-joining phylogenetic trees displayed in circular format. (A) NS5A sequences from AGATE-I, and (B) NS5A sequences from GT4a-infected patients in PEARL-I and AGATE-I. Reliability of the tree topology was examined using 1000 bootstrapping replicates, and bootstrap values ≥45 are listed at appropriate nodes. The genetic distance scale bar indicates the number of nucleotide substitutions per site between sequences. Reference sequences are labeled by the subtype followed by the GenBank accession number. HCV patient isolates are represented by a colored circle, square, or triangle indicating the geographic region identified for each sequence.

- In study AGATE-I, phylogenetic analysis of NS5A sequences revealed 2 sequence clusters within subtype 4a which were found to segregate by patient-reported country of origin **(Figure 3a)** but not by country of enrollment
 - One cluster contained sequences from Egypt and the Middle East, while the second cluster contained sequences from Europe and the United States
- Although country of origin was not collected for study PEARL-I, a combined phylogenetic analysis of NS5A sequences from GT4a-infected patients in PEARL-I and AGATE-I also identified 2 sequence clusters within subtype 4a **(Figure 3b)**
 - The majority of PEARL-I sequences from patients enrolled in France, Spain, and the United States were found to cluster with AGATE-1 sequences isolated from patients who originated from Europe or the United States
 - Two GT4a-infected patients who identified as Egyptian in PEARL-I had NS5A sequences that clustered with AGATE-I sequences isolated from patients who originated from Egypt
 - Based on NS5A phylogenetic analysis, 7 PEARL-I patients enrolled in France or the United States were infected with a strain of HCV subtype 4a that was genetically similar to virus detected in patients who originated from Egypt

- The distributions of patient birth cohort and race were significantly different across GT4 subtypes 4a, 4d, and non-4a/4d (*P*-values <.001)
 - A larger proportion of the GT 4d-infected patients were born after 1959 (73%), while most patients infected with a non-4a/4d subtype were born prior to 1960 (76%)
 - Patients of white race comprised 91%, 99%, and 27%, and patients of black race comprised 4%, 0%, and 73% of the 4a, 4d, and non-4a/4d-infected populations, respectively

PREVALENCE OF BASELINE POLYMORPHISMS IN NS5A

Figure 4. Prevalence of NS5A Baseline Polymorphisms by HCV GT4 subtype

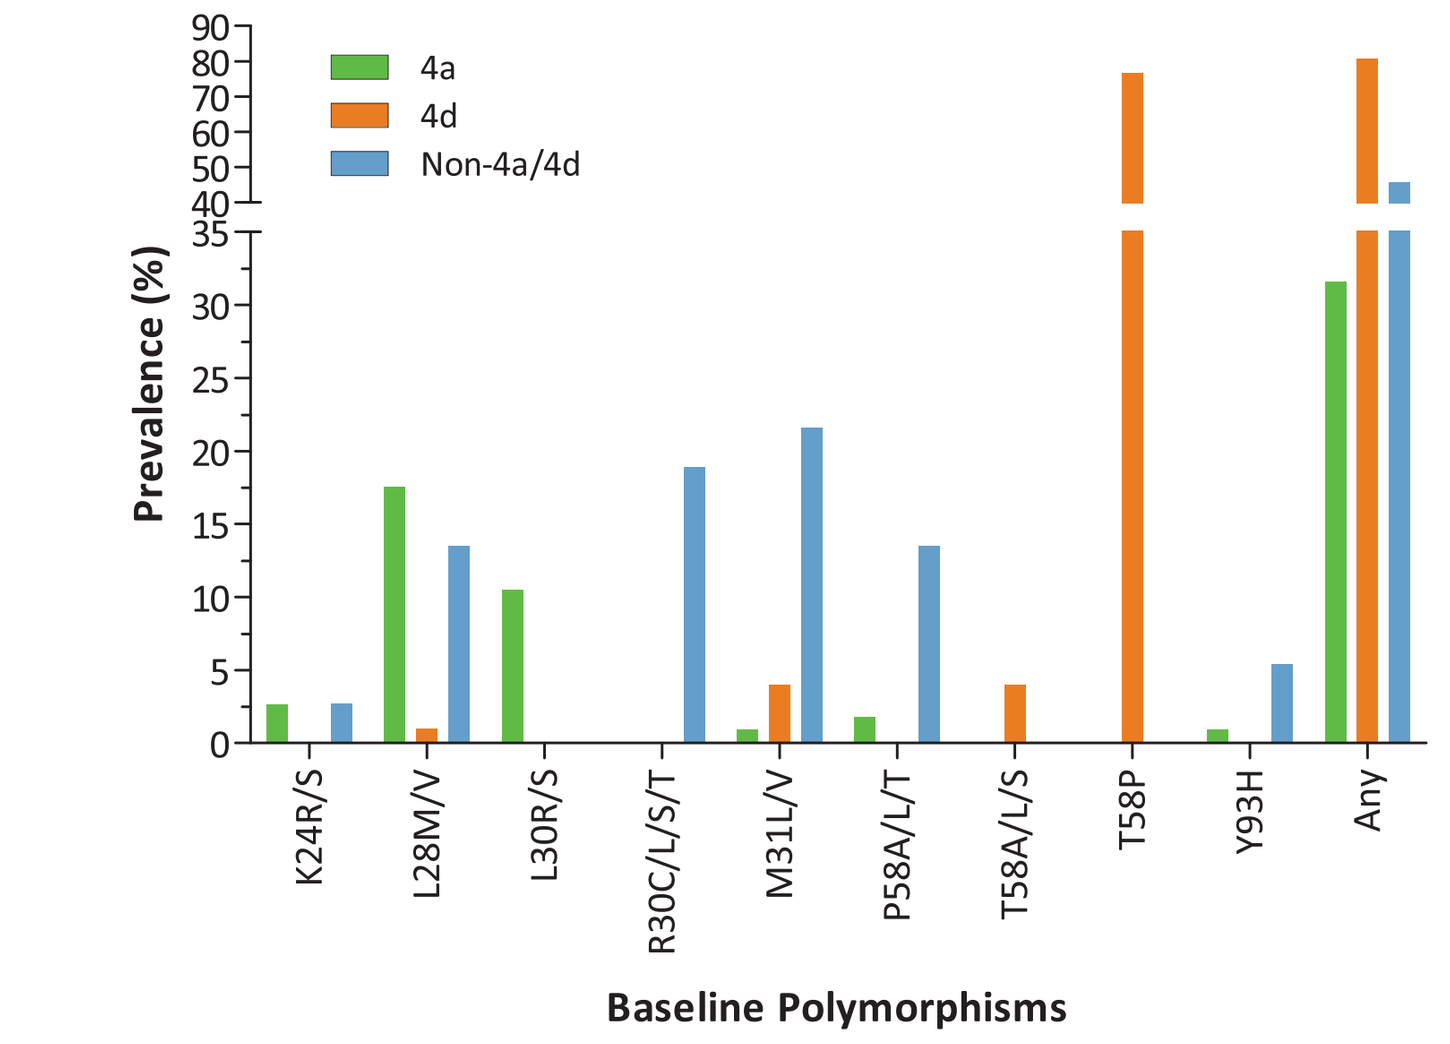
4a
4d
Non-4a/4d
Prevalence (%)
K24R/S
L28M/V
L30R/S
R30C/L/S/T
M31L/V
P58A/L/T
T58A/L/S
T58P
Y93H
Any
Baseline Polymorphisms

The following reference sequences were used in the analysis: GT 4a sequences were compared to 4a-ED43 (GenBank accession GU814265), GT 4d sequences were compared to 4d-QC382 (GenBank accession FJ462437), and non-4a/4d sequences were compared to 4c-QC381 (GenBank accession FJ462436).

- Baseline polymorphisms at resistance-associated amino acid positions in NS5A were present in 53.2% (133/250) of the GT4 samples. The most prevalent polymorphisms were L28M in GT 4a (20/114), T58P in GT 4d (76/99), and M31L in non-4a/4d (8/37) sequences **(Figure 4)**

Figure 5. Impact of NS5A Baseline Polymorphisms on SVR12 Rates

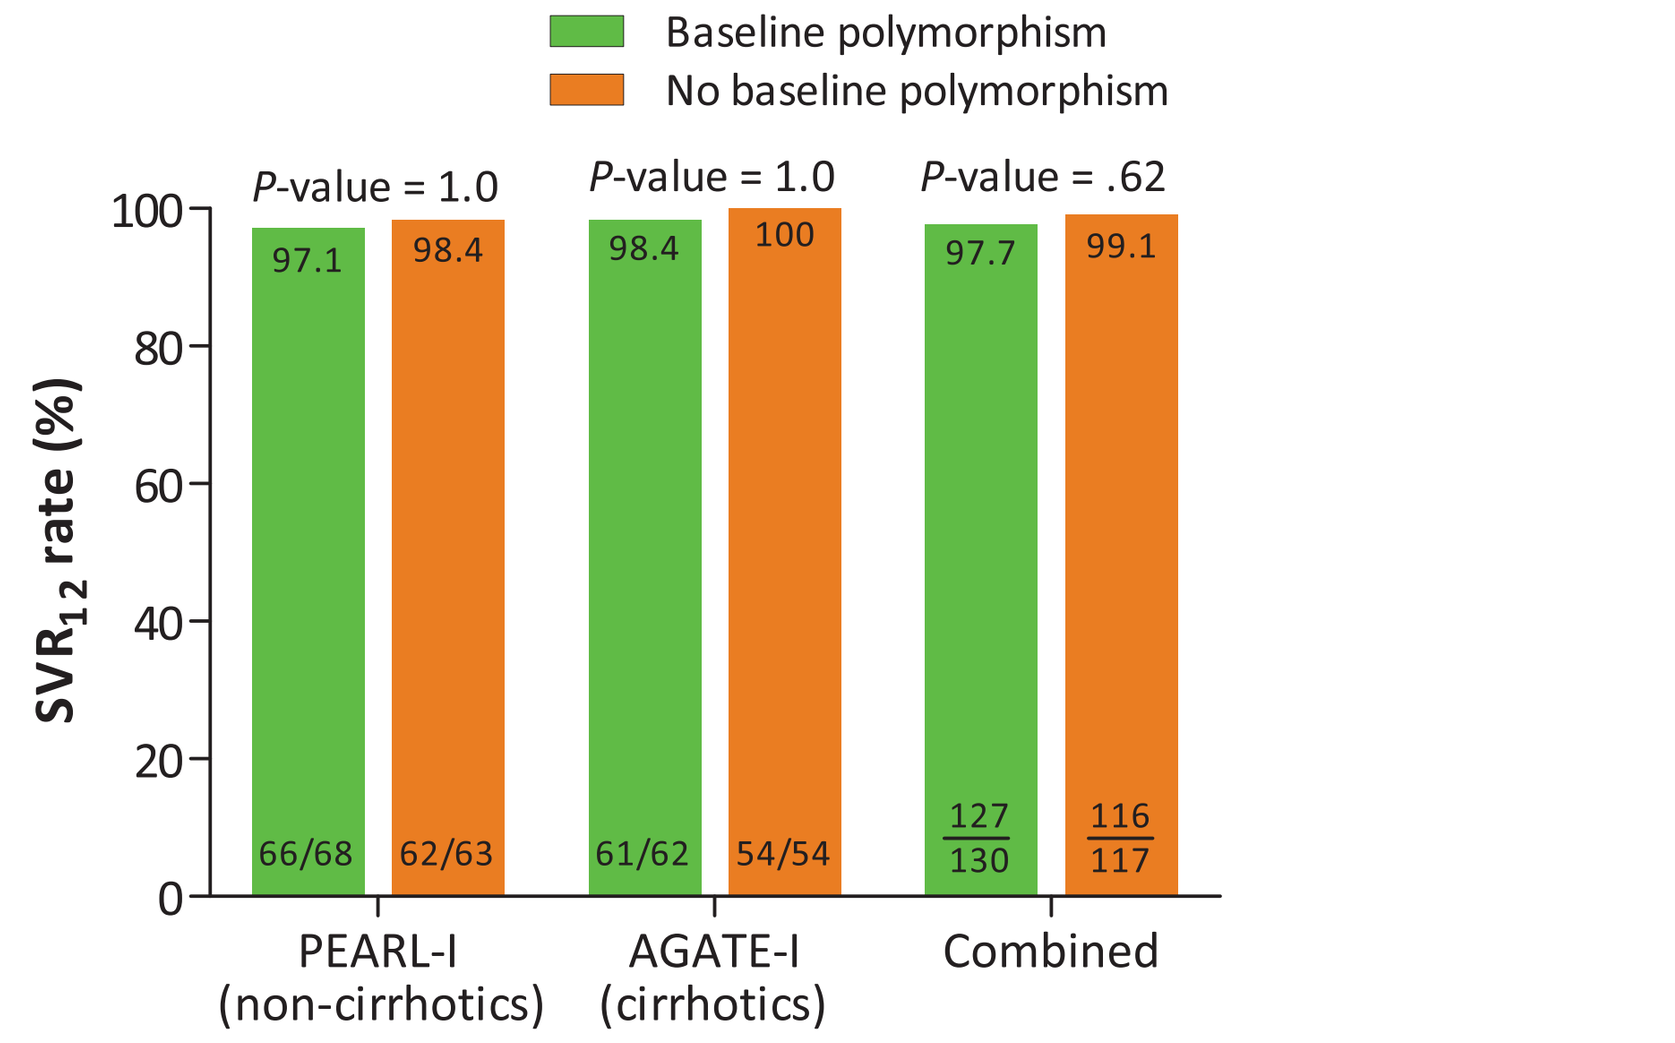
Baseline polymorphism
No baseline polymorphism
P-value = 1.0
P-value = 1.0
P-value = .62
SVR_{12} rate (%)
97.1
98.4
98.4
100
97.7
99.1
66/68
62/63
61/62
54/54
127/130
116/117
PEARL-I (non-cirrhotics)
AGATE-I (cirrhotics)
Combined

P-values were calculated using a 2-sided Fisher's exact test. Patients who did not achieve SVR12 for reasons other than virologic failure were excluded from the analysis. One patient in study AGATE-I achieved SVR12 and later relapsed at PTW24; this patient had the common T58P polymorphism in NS5A at baseline.

- There was no apparent impact of NS5A baseline polymorphisms on treatment outcome **(Figure 5)**
- Baseline polymorphisms at resistance-associated amino acid positions were not detected in NS3/4A sequences from PEARL-I; NS3/4A was not sequenced for patients who achieved SVR in study AGATE-I

TREATMENT-EMERGENT SUBSTITUTIONS IN PATIENTS WHO EXPERIENCED VIROLOGIC FAILURE IN PEARL-I AND AGATE-I

Table 4. Baseline Polymorphisms and Treatment-emergent Substitutions in NS3 and NS5A in Patients With Virologic Failure

				NS3[a]		NS5A[a]	
Patient	Study	GT	Cirrhosis	Baseline	Virologic Failure	Baseline	Virologic Failure
1[b]	PEARL-I	4d	N	None	Y56H + D168V	None	L28V
2[c]	PEARL-I	4d	N	None	D168V	T58P	L28S + T58P, M31I/M
3[c]	PEARL-I	4d	N	None	D168V	T58S/T	L28V + T58S
4[b]	AGATE-I	4a	Y	None	A156K	P58L	L28I + P58L
5[c]	AGATE-I	4d	Y	None	D168V	T58P	K24Q + T58P + Y93H

[a]Includes amino acid substitutions detected at NS3 positions 56, 155, 156, or 168, and NS5A positions 24, 28, 29. 30, 31, 32, 58, 92, or 93 by population sequencing or NGS at a 15% detection threshold. [b]Breakthrough. [c]Relapse.

- Predominant treatment-emergent substitutions at the time of VF in GT 4d-infected patients were D168V in NS3, and L28S/V or K24Q + Y93H in NS5A. Substitutions A156K in NS3 and L28I in NS5A were detected in the GT 4a-infected patient at the time of VF

Table 5. Resistance Conferred by Amino Acid Substitutions in NS3 or NS5A to Paritaprevir or Ombitasvir in GT4 Subgenomic Transient Replicons

Target	GT4 Subtype	Amino Acid Substitutions	Mean EC_{50} ± SD (nM)	Fold Change in EC_{50}
NS3	4a	Wild type	0.048 ± 0.007	—
	4d	Wild type	0.015 ± 0.001	—
		Y56H	0.12 ± 0.03	8.0
		D168V	4.7 ± 0.9	313
		Y56H + D168V	188 ± 50	12533
NS5A	4a	Wild type	0.00035 ± 0.00007	—
		L28I	0.0028 ± 0.00072	8.0
		L28M	0.00018 ± 0.00003	0.5
		P58L	0.0010 ± 0.00016	2.9
		L28I + P58L	1.328 ± 0.0386	3795
	4d	Wild Type	0.00038 ± 0.00006	—
		L28S	NA	—
		L28V	0.118 ± 0.0163	310
		M31I	0.00096 ± 0.00012	2.5
		M31L	0.00039 ± 0.00008	1.0
		T58A	0.00053 ± 0.00002	1.4
		T58P	0.00042 ± 0.00004	1.1
		T58S	0.00052 ± 0.00009	1.4
		Y93H	NA	—
		K24Q + T58P	0.00045 ± 0.00005	1.2
		L28S + M31I	NA	—
		L28V + T58S	0.289 ± 0.0295	760
		T58P + Y93H	NA	—
		K24Q + T58P + Y93H	0.417 ± 0.145	1098

NA = not available, the EC_{50} could not be determined due to low replication efficiency of the replicon containing the amino acid substitution.

CONCLUSIONS

- Overall, high SVR12 rates were observed among patients infected with 16 HCV GT4 subtypes in studies PEARL-I and AGATE-I with no impact of NS5A baseline polymorphisms on treatment outcome. Baseline polymorphisms at resistance-associated amino acid positions were frequently detected in NS5A (53.2% of the samples)
- The 5 patients who experienced virologic failure were infected with HCV GT 4a (n = 1) or GT 4d (n = 4)
- The distributions of patient birth cohort and race were significantly different across GT4 subtypes (*P*-values <.001)
- Phylogenetic analysis of NS5A sequences revealed 2 sequence clusters within subtype 4a which were found to segregate by country of origin and suggest a genetically distinct strain of 4a circulating in Egypt vs Europe and North America

REFERENCES

1. Gower E, Estes C, Blach S, et al. Global epidemiology and genotype distribution of the hepatitis C virus infection. *J Hepatol.* 2014;61:S45–57.
2. Smith DB, Bukh J, Kuiken C, et al. Expanded classification of hepatitis C virus into 7 genotypes and 67 subtypes: updated criteria and genotype assignment web resource. *Hepatology.* 2014;59:318–26.
3. Kamal SM. Improving outcome in patients with hepatitis C virus genotype 4. *Am J Gastroenterol.* 2007;102:2582–8.
4. Kamal SM, Nasser IA. Hepatitis C genotype 4: what we know and what we don't yet know. *Hepatology.* 2008;47:1371–83.
5. Hézode C, Asselah T, Reddy KR, et al. Ombitasvir plus paritaprevir plus ritonavir with or without ribavirin in treatment-naive and treatment-experienced patients with genotype 4 chronic hepatitis C virus infection (PEARL-I): a randomized, open-label trial. *Lancet.* 2015;385:2502–9.
6. Asselah T, Hezode C, Qaqish RB, et al. Ombitasvir, paritaprevir, and ritonavir plus ribavirin in adults with hepatitis C virus genotype 4 infection and cirrhosis (AGATE-I): a multicentre, phase 3, randomised open-label trial. *Lancet Gastroenterol Hepatol.* 2016;1:25–35.
7. Koletzki D, Dumont S, Vermeiren H, et al. Development and evaluation of an automated hepatitis C virus NS5B sequence-based subtyping assay. *Clin Chem Lab Med.* 2010;48(8):1095–102.
8. Murphy DG, Willems B, Deschenes M, et al. Use of sequence analysis of the NS5B region for routine genotyping of hepatitis C virus with reference to C/E1 and 5' untranslated region sequences. *JCM.* 2007;45:1102–12.

DISCLOSURES

All authors are employees of AbbVie, and may own AbbVie stock.

The design, study conduct, and financial support for this study were provided by AbbVie. AbbVie participated in the interpretation of data, review, and approval of the publication.

abbvie