Poster #464

Lack of Pharmacokinetic Interaction Between HCV Protease Inhibitor MK-5172 and HCV NS5A Inhibitor Daclatasvir in Healthy Volunteers

Wendy W. Yeh,[1] Iain P. Fraser,[1] Marc Bifano,[2] Luzelena Caro,[1] Jennifer Talaty,[1] Zifang Guo,[1] Jennifer McCarthy,[1] Stephen P. Youngberg,[3] Joan R. Butterton[1]

[1]Merck & Co., Inc, Whitehouse Station, NJ; [2]Bristol-Myers Squibb Research and Development, Hopewell, NJ, USA; [3]Celerion, Lincoln, NE, USA

Abstract

Background: MK-5172, a potent, once-daily competitive inhibitor of the hepatitis C virus (HCV) NS3/4A protease with improved potency compared with the approved first generation protease inhibitors, and daclatasvir (DCV), an HCV NS5A replication complex inhibitor with pan-genotypic activity in vitro, could be components of an all-oral combination regimen for the treatment of chronic HCV infection. The aim of the present study was to evaluate potential pharmacokinetic interactions as well as safety and tolerability of MK-5172 and daclatasvir co-administration in healthy subjects.

Methods: This was a single-center, open-label, fixed-sequence, multiple-dose study in 14 healthy adult male and female volunteers, ages 21-49 years. Since MK-5172 in HCV-infected patients demonstrates ~2-fold higher exposure compared with healthy subjects, a 200 mg dose of MK-5172 in healthy subjects was used in this study to match the exposure of a 100 mg dose (the intended clinical dose) in HCV-infected patients. In Period 1, subjects received oral doses of 60 mg daclatasvir once daily on Days 1 to 7. Following a 4-day washout, subjects received oral doses of 200 mg MK-5172 once daily on Days 1 to 7 in Period 2. In Period 3, which commenced immediately after Period 2, subjects were co-administered once daily oral doses of 200 mg MK-5172 and 60 mg daclatasvir on Days 1 to 8. Plasma pharmacokinetic samples were obtained for daclatasvir on Day 7 in Period 1 and Day 8 in Period 3, as well as for MK-5172 on Day 7 in Period 2 and Day 8 in Period 3. Safety assessments included electrocardiograms, vital signs, clinical laboratory tests, physical examination, and adverse event monitoring.

Results: Co-administration of MK-5172 with daclatasvir was safe and well tolerated. Multiple oral doses of MK-5172 did not meaningfully change the steady-state $AUC_{0-\tau}$, C_{max}, or C_{24h} of daclatasvir, with MK-5172+DCV/DCV geometric mean ratios (GMRs) [90% confidence intervals (CIs)] of 1.02 [0.93, 1.11], 0.80 [0.74, 0.86], and 1.23 [1.09, 1.38], respectively. Multiple oral doses of daclatasvir did not meaningfully change the steady-state $AUC_{0-\tau}$, C_{max}, or C_{24h} of MK-5172, with MK-5172+DCV/MK-5172 GMRs [90% CIs] of 1.12 [0.87, 1.44], 1.11 [0.77, 1.60], and 1.04 [0.97, 1.12], respectively.

Conclusions: Co-administration of MK-5172 and daclatasvir in healthy volunteers did not result in clinically significant drug-drug interactions. MK-5172 and DCV were safe and well tolerated when coadministered. These results suggest that no dose adjustments of MK-5172 or daclatasvir are needed for co-administration of these drugs without other direct acting antiviral agents in interferon-free, combination regimens in HCV-infected patients.

Background

- MK-5172 is a potent, once-daily inhibitor of the hepatitis C virus (HCV) NS3/4A protease inhibitor with improved potency compared with the approved first generation protease inhibitors.
- Daclatasvir (DCV), or BMS-790052, is a once-daily HCV NS5A replication complex inhibitor with pan-genotypic activity in vitro.
- MK-5172 and DCV could be components of an interferon-free, all-oral combination regimen for the treatment of chronic HCV infection.

Aims

- To assess the effect of multiple oral doses of MK-5172 on the steady-state pharmacokinetics (eg, $AUC_{0-\tau}$, C_{max}, C_{24h}, and T_{max}) of DCV and the effect of multiple oral doses of DCV on the steady state pharmacokinetics (eg, $AUC_{0-\tau}$, C_{max}, C_{24h}, and T_{max}) of MK 5172.
- To evaluate the safety and tolerability of multiple doses of MK-5172 alone, multiple doses of DCV alone, and multiple doses of MK-5172 co-administered with multiple doses of DCV in healthy volunteers.

Methods

Study Design: This was a single-center, open-label, fixed-sequence, multiple-dose study.

Subjects: Fourteen (14) healthy non-tobacco using adult male and female subjects between 21 and 49 years (inclusive) were enrolled.

Treatments:

- Since MK-5172 in HCV-infected patients demonstrates ~2-fold higher exposure compared to healthy subjects, a 200 mg dose of MK-5172 in healthy subjects was used in this study to match the exposure of a 100 mg dose (the intended clinical dose) in HCV-infected patients.
- DCV 60 mg QD, the clinical dose, was used in this study.
- Period 1: oral doses of 60 mg DCV once daily (QD) on Days 1 to 7 followed by a 4-day washout.
- Period 2: oral doses of 200 mg MK-5172 QD on Days 1 to 7 in Period 2. No washout followed this period.
- Period 3: oral doses of 200 mg MK-5172 and 60 mg DCV on Days 1 to 8 co-administered QD.

Assessments:

- Plasma pharmacokinetic samples were obtained for DCV on Day 7 in Period 1 and Day 8 in Period 3, as well as for MK-5172 on Day 7 in Period 2 and Day 8 in Period 3.
- Safety assessments included electrocardiograms, vital signs, clinical laboratory tests, physical examination, and adverse event monitoring.

Results

Demographics

- 14 subjects were enrolled (9 males, 5 females), and 11 completed the study.
- The majority were white and Hispanic/Latino (n = 11), with a mean age (range) of 35 (21-49) and mean BMI (range) of 25.56 (21.12-28.88).
- Three subjects were discontinued by the Investigator due to reasons unrelated to study drugs; one subject due to clinical AEs not related to study drug and two subjects due to positive urine drug screens.
- All 14 subjects enrolled were included in the evaluation of safety. All available data from all 14 subjects were included in the evaluation of pharmacokinetics.

Pharmacokinetics

MK-5172

- Multiple oral doses of DCV did not meaningfully change the steady-state $AUC_{0-\tau}$, C_{max}, or C_{24h} of MK-5172, with MK-5172+DCV/MK-5172 GMRs [90% CIs] of 1.12 [0.87, 1.44], 1.11 [0.77, 1.60], and 1.04 [0.97, 1.12], respectively.
- The observed MK-5172 median T_{max} of 3.00 hours following administration of 200 mg MK-5172 QD alone was unchanged following the co-administration of 60 mg DCV QD and 200 mg MK-5172 QD. The apparent terminal $t_{½}$ of MK-5172 following the co-administration of 60 mg DCV QD and 200 mg MK-5172 QD was 23.75 hours.

Table 1. Pharmacokinetics of MK-5172 alone and in combination with DCV

MK-5172 PK Parameter	MK-5172 Alone			DCV + MK-5172			DCV + MK-5172/ MK-5172 Alone		Pseudo Within Subject %CV†
	N¶	GM	95% CI	N¶	GM	95% CI	GMR	90% CI	
$AUC_{0-\tau}$‡ (μM•hr)	12	1.89	(1.37, 2.62)	11	2.11	(1.37, 3.24)	1.12	(0.87, 1.44)	32.571
C_{max}‡ (μM)	12	0.447	(0.279, 0.717)	11	0.498	(0.293, 0.846)	1.11	(0.77, 1.60)	47.086
C_{24h}‡ (nM)	12	11.4	(8.51, 15.2)	11	11.9	(8.44, 16.6)	1.04	(0.97, 1.12)	9.743
T_{max}‖ (hr)	12	3.00	(2.00, 4.00)	11	3.00	(2.02, 4.00)	.	.	.
Apparent terminal $t_{½}$§ (hr)	.	.	.	11	23.75	39.72	.	.	.

MK-5172 alone: 200 mg MK-5172 (2 x 100 mg tablets) QD, on Days 1 to 7 in Period 2.

DCV + MK-5172: Co-administration of 60 mg DCV (1 x 60 mg tablet) and 200 mg MK 5172 (2 x 100 mg tablets), QD, on Days 1 to 8 in Period 3.

†Pseudo Within-Subject %CV = 100 x Sqrt[$(\sigma_B^2 + \sigma_C^2 - 2\sigma_{BC})/2$], where σ_B^2 and σ_C^2 are the estimated variances on the log scale for the 2 treatment groups, and σBC is the corresponding estimated covariance, each obtained from the linear mixed-effects model.

‡Back-transformed least-squares mean and confidence interval from a linear mixed-effects model performed on natural log-transformed values, with a fixed effect for treatment, and an unstructured covariance structure for the measurements within each subject.

‖Median (min, max) reported for T_{max}.

§Geometric mean, geometric coefficient of variation reported for apparent terminal $t_{½}$.

GM = Geometric Least-Squares Mean; CI = Confi dence Interval; GMR = Geometric Least-Squares Mean Ratio between treatments.

¶One subject was discontinued from the study on Day 7 of Period 1. A second subject was discontinued on Day -1 of Period 2. A third subject was discontinued on Day -1 of Period 3 (Day 7 of Period 2).

Figure 1. Arithmetic mean plasma concentration-time profiles of MK-5172 following administration of multiple doses of 200 mg MK-5172 QD with or without multiple doses of 60 mg DCV QD in healthy adult subjects (N=12 for MK-5172 alone and N=11 for DCV + MK-5172) (inset = semi-log scale)

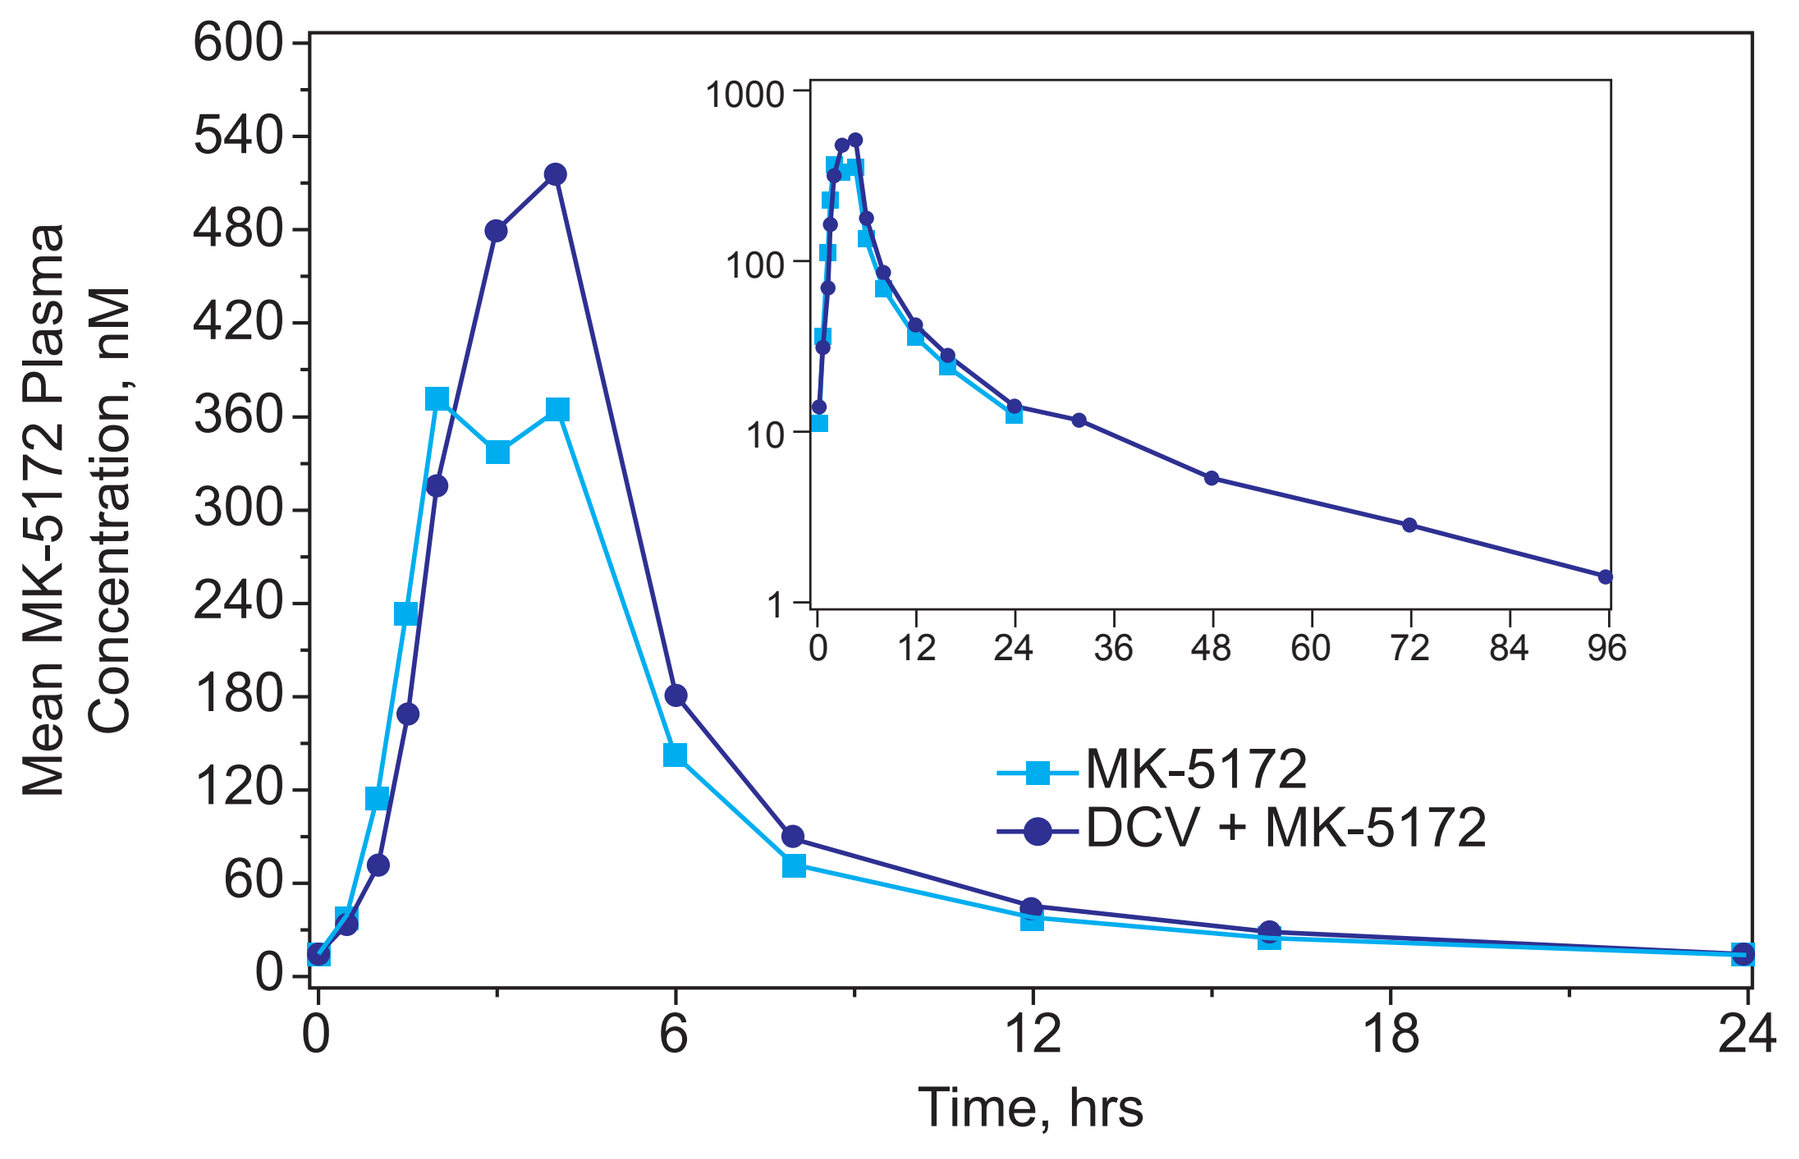

Mean MK-5172 Plasma Concentration, nM
600
540
480
420
360
300
240
180
120
60
0
0
6
12
18
24
Time, hrs
1000
100
10
1
0 12 24 36 48 60 72 84 96
MK-5172
DCV + MK-5172

Daclatasvir

- Multiple oral doses of MK-5172 did not meaningfully change the steady-state $AUC_{0-\tau}$, C_{max}, or C_{24h} of DCV, with MK-5172+DCV/DCV geometric mean ratios (GMRs) [90% confidence intervals (CIs)] of 1.02 [0.93, 1.11], 0.80 [0.74, 0.86], and 1.23 [1.09, 1.38], respectively.
- The observed DCV median T_{max} of 1.50 hours following co administration of 60 mg DCV QD and 200 mg MK-5172 QD was slightly delayed when compared to observed median T_{max} of 1.01 hours following the administration of 60 mg DCV QD alone. The apparent terminal $t_{½}$ of DCV following the co-administration of 60 mg DCV QD and 200 mg MK-5172 QD was 14.20 hours in Period 3.

Table 2. Pharmacokinetics of DCV alone and in combination with MK-5172

Pharmaco-kinetic Parameter	DCV Alone			DCV + MK-5172			DCV + MK-5172/ DCV Alone		Pseudo Within Subject %CV†
	N¶	GM	95% CI	N¶	GM	95% CI	GMR	90% CI	
$AUC_{0-\tau}$‡ (ng•hr/mL)	13	13679.54	(10,901.50, 17,165.50)	11	13,885.31	(11,430.01, 16,868.04)	1.02	(0.93, 1.11)	12.143
C_{max}‡ (ng/mL)	13	1481.62	(1226.68, 1789.55)	11	1183.85	(948.98, 1476.85)	0.80	(0.74, 0.86)	10.010
C_{24h}‡ (ng/mL)	13	182.75	(131.31, 254.35)	11	224.13	(172.27, 291.60)	1.23	(1.09, 1.38)	15.750
T_{max}‖ (hr)	13	1.01	(1.00, 3.01)	11	1.50	(1.01, 4.01)	.	.	.
Apparent terminal $t_{½}$§ (hr)	13	13.36	21.83	11	14.20	23.83	.	.	.

DCV alone: 60 mg DCV (1 x 60 mg tablet) QD on Days 1 to 7 in Period 1.

DCV + MK-5172: Co-administration of 60 mg DCV (1 x 60 mg tablet) and 200 mg MK 5172 (2 x 100 mg tablets) QD on Days 1 to 8 in Period 3.

†Pseudo Within-Subject %CV = 100 x Sqrt[$(\sigma_A^2 + \sigma_C^2 - 2\sigma_{AC})/2$], where σ_A^2 and σ_C^2 are the estimated variances on the log scale for the 2 treatment groups, and σAC is the corresponding estimated covariance, each obtained from the linear mixed-effects model.

‡Back-transformed least-squares mean and confidence interval from a linear mixed-effects model performed on natural log-transformed values, with a fixed effect for treatment, and an unstructured covariance structure for the measurements within each subject.

‖Median (min, max) reported for T_{max}.

§Geometric mean, geometric coefficient of variation reported for apparent terminal $t_{½}$.

GM = Geometric least-squares mean; CI = Confi dence interval; GMR = Geometric least-squares mean ratio between treatments.

¶One subject was discontinued from the study on Day 7 of Period 1. A second subject was discontinued on Day -1 of Period 2. A third subject was discontinued on Day -1 of Period 3 (Day 7 of Period 2).

Figure 2. Arithmetic mean plasma concentration-time profiles of DCV following administration of multiple doses of 60 mg DCV QD with or without multiple doses of 200 mg MK-5172 QD in healthy adult subjects (N=13 for DCV alone and N=11 for DCV + MK-5172) (inset = semi-log scale)

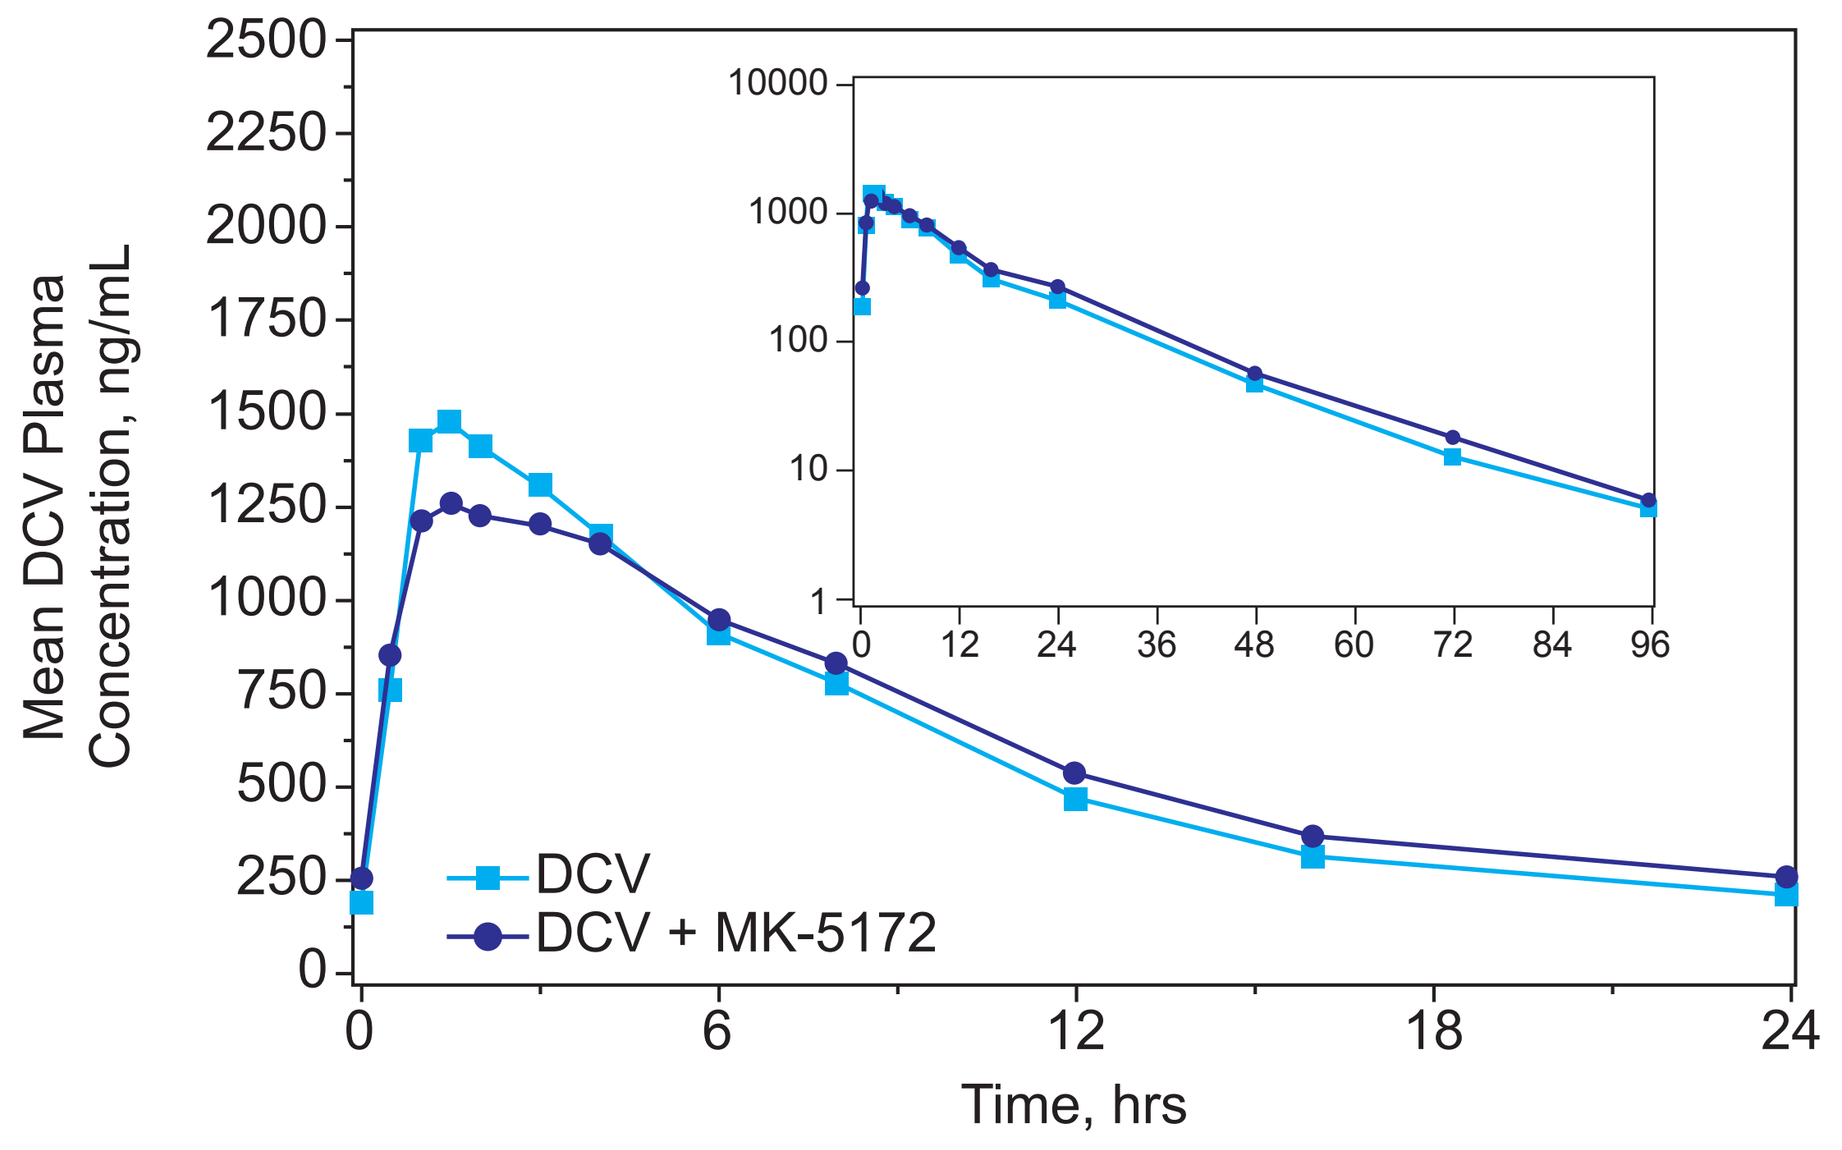

Mean DCV Plasma Concentration, ng/mL
2500
2250
2000
1750
1500
1250
1000
750
500
250
0
0
6
12
18
24
Time, hrs
10000
1000
100
10
1
0 12 24 36 48 60 72 84 96
DCV
DCV + MK-5172

Safety and Tolerability

- Co-administration of MK-5172 with DCV was safe and well tolerated in the healthy adult males and females in this study. No deaths, serious adverse experiences, or laboratory adverse experiences were reported during the study.
- The investigator discontinued one subject on Day 7 of Period 1 due to multiple moderate adverse experiences that were considered unrelated to study drug.
- Ten (10) subjects reported a total of 46 clinical adverse experiences, 5 of which were considered drug-related (all related to DCV alone). All adverse experiences were of mild to moderate intensity. The most common adverse experiences in this study were dyspepsia, nausea, and oropharyngeal pain. The only drug related adverse experience reported in the study was diarrhea (reported by 1 subject on 5 occasions following DCV alone).
- There were no consistent treatment related changes in laboratory, vital signs, or ECG safety parameters.

Discussion and Conclusions

Discussion

- The mean steady-state $AUC_{0-\tau}$, C_{max}, and C_{24h} of MK-5172 are unchanged when daily doses of 200 mg MK 5172 are co-administered with daily doses of 60 mg DCV compared with daily doses of 200 mg MK-5172 administered alone.
- The mean steady-state $AUC_{0-\tau}$ of DCV is unchanged when daily doses of 60 mg DCV are co-administered with daily doses of 200 mg MK-5172 compared with daily doses of 60 mg DCV administered alone. The mean steady state C_{max} of DCV decreased by ~20% and mean C_{24h} increased by ~20%.
- Multiple oral doses of DCV alone, MK-5172 alone, and MK-5172 co-administered with DCV were generally safe and well tolerated in healthy adult male and female subjects.

Conclusions

- Co-administration of MK-5172 and DCV in healthy volunteers did not result in clinically significant drug-drug interactions.
- Co-administration of MK-5172 and DCV was safe and well tolerated.
- These results suggest that no dose adjustments of MK-5172 or DCV are needed if co-administered in interferon-free, combination regimens without other direct-acting antiviral agents in HCV-infected patients.

Acknowledgements:

Bioanalytical analysis coordination work was provided by Monika Martinho at Merck Sharp & Dohme Corp. Pharmacokinetic analysis was provided by Nadia Cardillo, Daria Stypinski, Xiaopeng Li, and Bruce DeGroot of Celerion (Lincoln, NE). This assistance was funded by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Whitehouse Station, NJ.

Disclosures:

This study was funded by Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Whitehouse Station, NJ.

WWY, IPF, LC, JT, ZG, JM, and JRB are current employees of Merck Sharp & Dohme Corp., a subsidiary of Merck & Co., Inc., Whitehouse Station, NJ. MB is a current employee of Bristol-Myers Squibb Research and Development, Hopewell, NJ.

SPY is currently employed by Celerion, Inc., Lincoln NE.

Contact Information:

Wendy W. Yeh, M.D.
Merck Research Laboratories
Merck & Co., Inc.
BMB3-423
33 Avenue Louis Pasteur
Boston, MA 02115
Phone: 617-992-3237
wendy.yeh@merck.com

Presented at the 64th Annual Meeting of the American Association for the Study of Liver Diseases, November 1 – 5, 2013, Washington, DC

Copyright © 2013 Merck Sharp & Dohme Corp., a subsidiary of **Merck & Co., Inc.** All Rights Reserved.